Using Mifepristone to Differentiate Cushing's Disease from Cushing's Syndrome

Joel R L Ehrenkranz, MD, Dept. of Medicine, Intermountain Healthcare, Murray, UT, Coleman Gross*, MD, Corcept Therapeutics, Menlo Park, CA and Joseph K Belanoff*, MD, Corcept Therapeutics, Menlo Park, CA

*Potential conflict of interest may exist. Refer to the abstract.

INTRODUCTION

- Cushing's syndrome (CS), is a serious endocrine disorder that is pituitary (Cushing's disease [CD]) or non-pituitary (eg. ectopic ACTH-secreting tumor) in origin.
- Identifying the cause of excess cortisol production is based upon pituitary ACTH secretory response to pharmacological agents such as dexamethasone.
- Mifepristone is a glucocorticoid receptor (GR) antagonist approved for use in patients with CS.
- We hypothesized that, by blocking GR receptors, mifepristone might distinguish patients with CD from patients with other forms of CS. Patients with CD would be expected to have an increase in ACTH and cortisol following mifepristone administration, while patients with other forms of CS would show no increase in ACTH and cortisol.

METHODS

- Retrospective review of previously collected data from the SEISMIC Study of ACTH and cortisol responses in patients with CD or ectopic ACTH.
- In this analysis:
 - o CD patients (N=41) all had prior transphenoidal surgery:
 - 18 with prior pituitary radiation therapy
 - 23 with no prior pituitary radiation therapy
 - 4 patients with CS secondary to ectopic ACTH secretion.
 - o Patients received mifepristone 300 mg po once daily.
 - o ACTH and cortisol were measured as close to 8 AM as possible using a centralized laboratory (Quest Diagnostics, Collegeville, Pa.) at baseline and 14 days after commencing mifepristone therapy.

RESULTS

- Patients with CD and:
 - o no radiation therapy had a 2.13 ± 0.96 fold difference in rise of ACTH.
 - o radiation therapy had a 1.4 ± 0.45 fold rise in ACTH.
 - o no radiation therapy had a 1.56 ± 0.6 fold rise in cortisol.
 - o radiation therapy had a 1.24 + 0.23 fold rise in cortisol.
- No statistically significant rise in ACTH or cortisol between ectopic ACTH patients and CD patients as a group or in those who had radiation.

Table 1. Mean ± SD cortisol and ACTH.

		CD	CD xrt (N=23)	CD no xrt (N=18)	Ectopic (N=4)
Serum Cortisol (nmol/L)	Baseline	587.1±163.8	562.2±164.9	618.9±161.5	1175.3±395.2
	Day 14	822.7±405.0	874.3±501.7	756.9±227.8	1350.3±277.8
ACTH (ng/L)	Baseline	63.7±52.1	46.1±25.8	86.1±67.7	152.5±140.3
	Day 14	99.2±71.0	93.7±71.6	106.2±71.7	200.0±120.2

Figure 1. Mean ± SD fold increase at Day 14

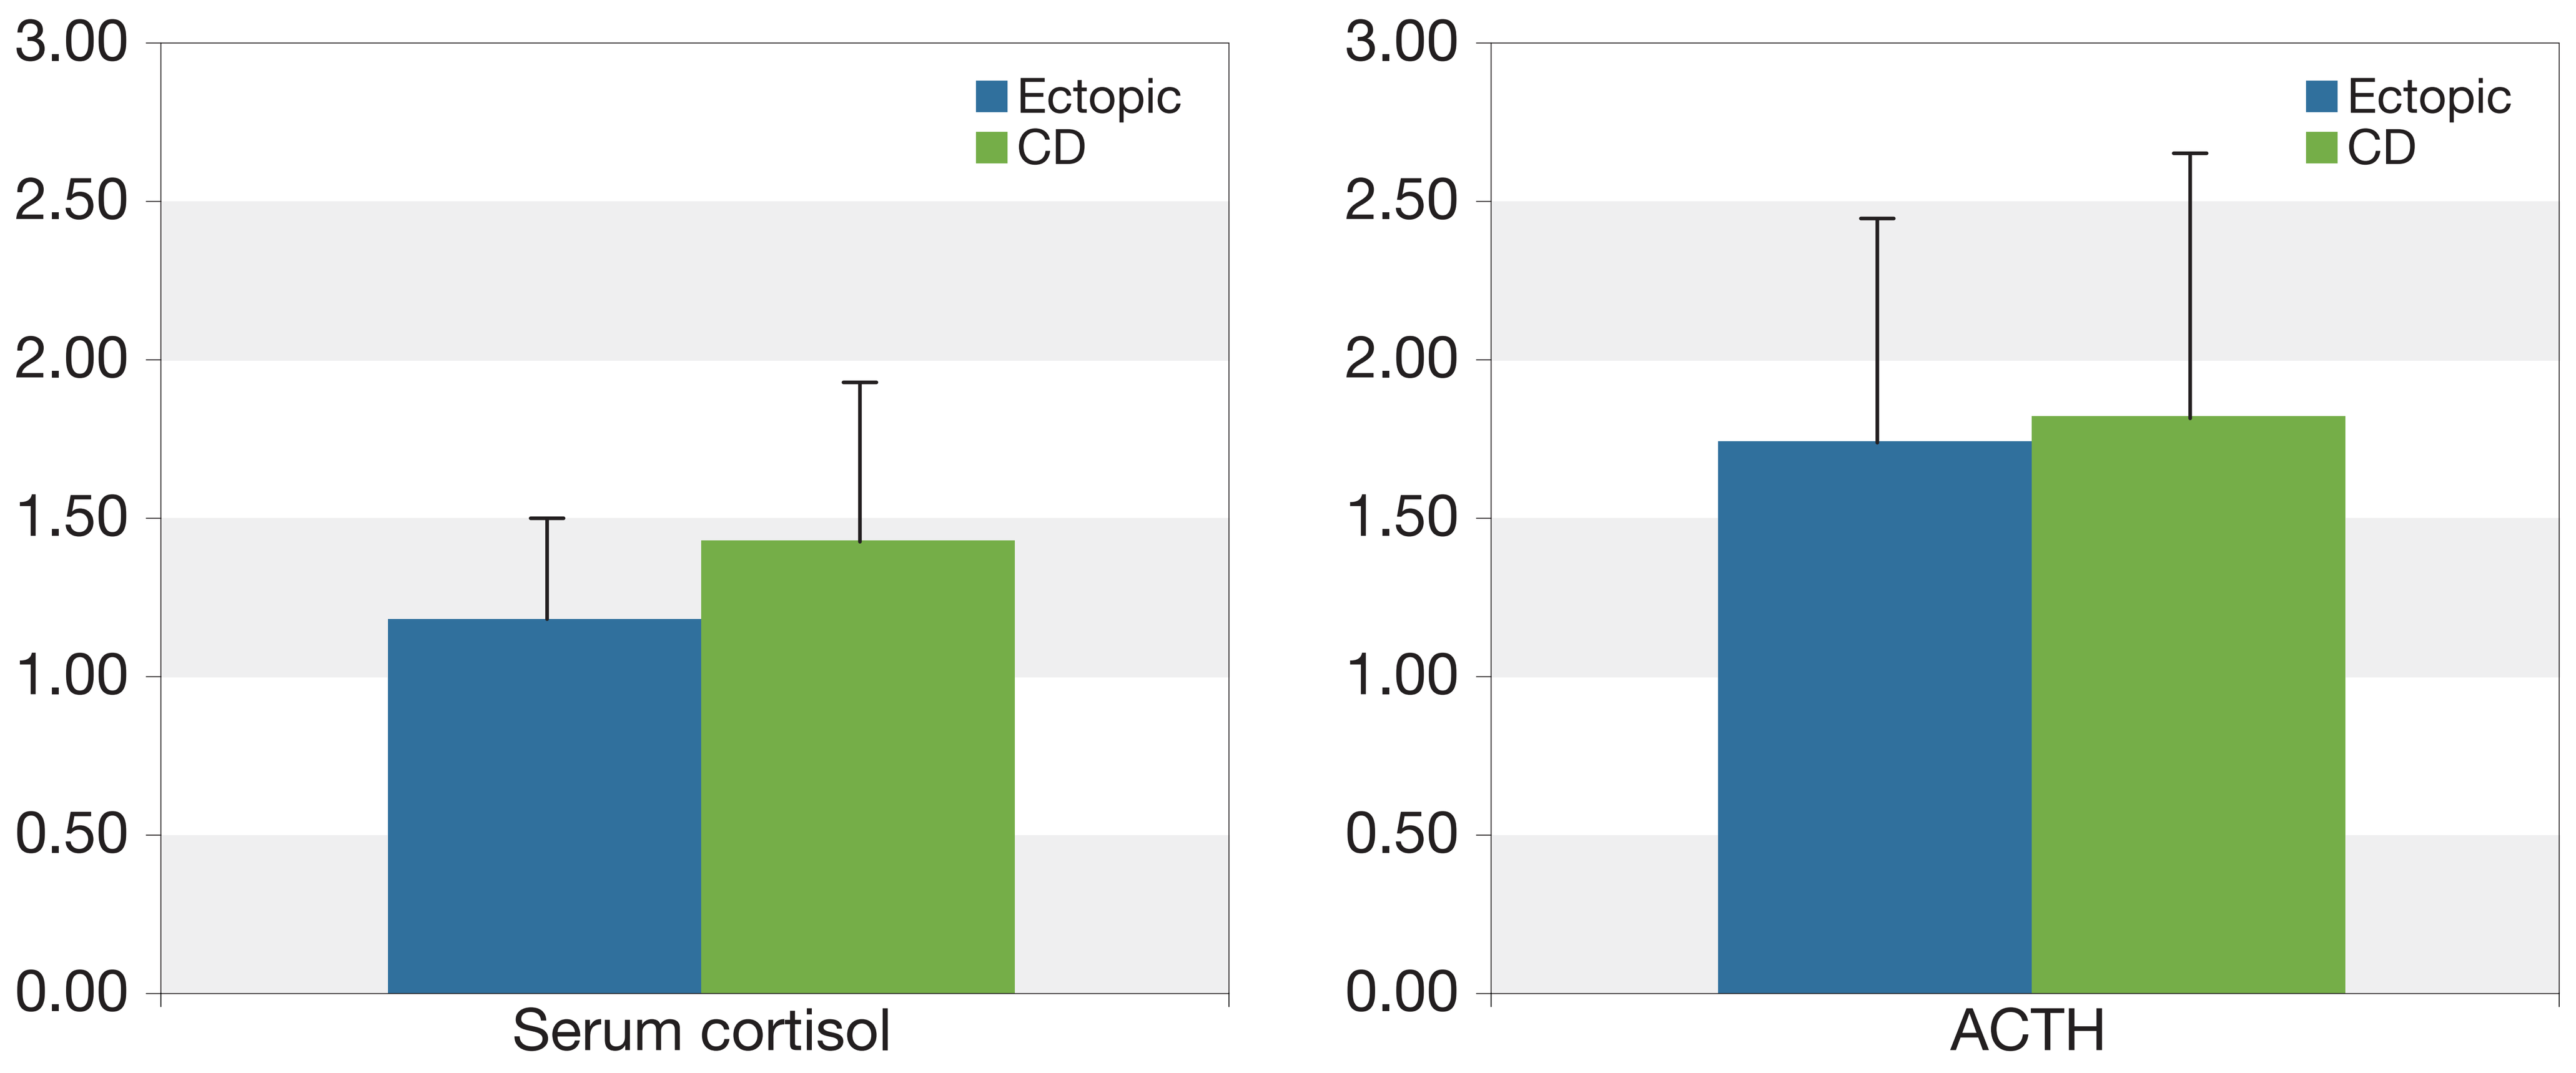
3.00
2.50
2.00
1.50
1.00
0.50
0.00
Ectopic
CD
Serum cortisol
ACTH

Figure 2. Mean ± SD fold increase at Day 14 in CD by XRT

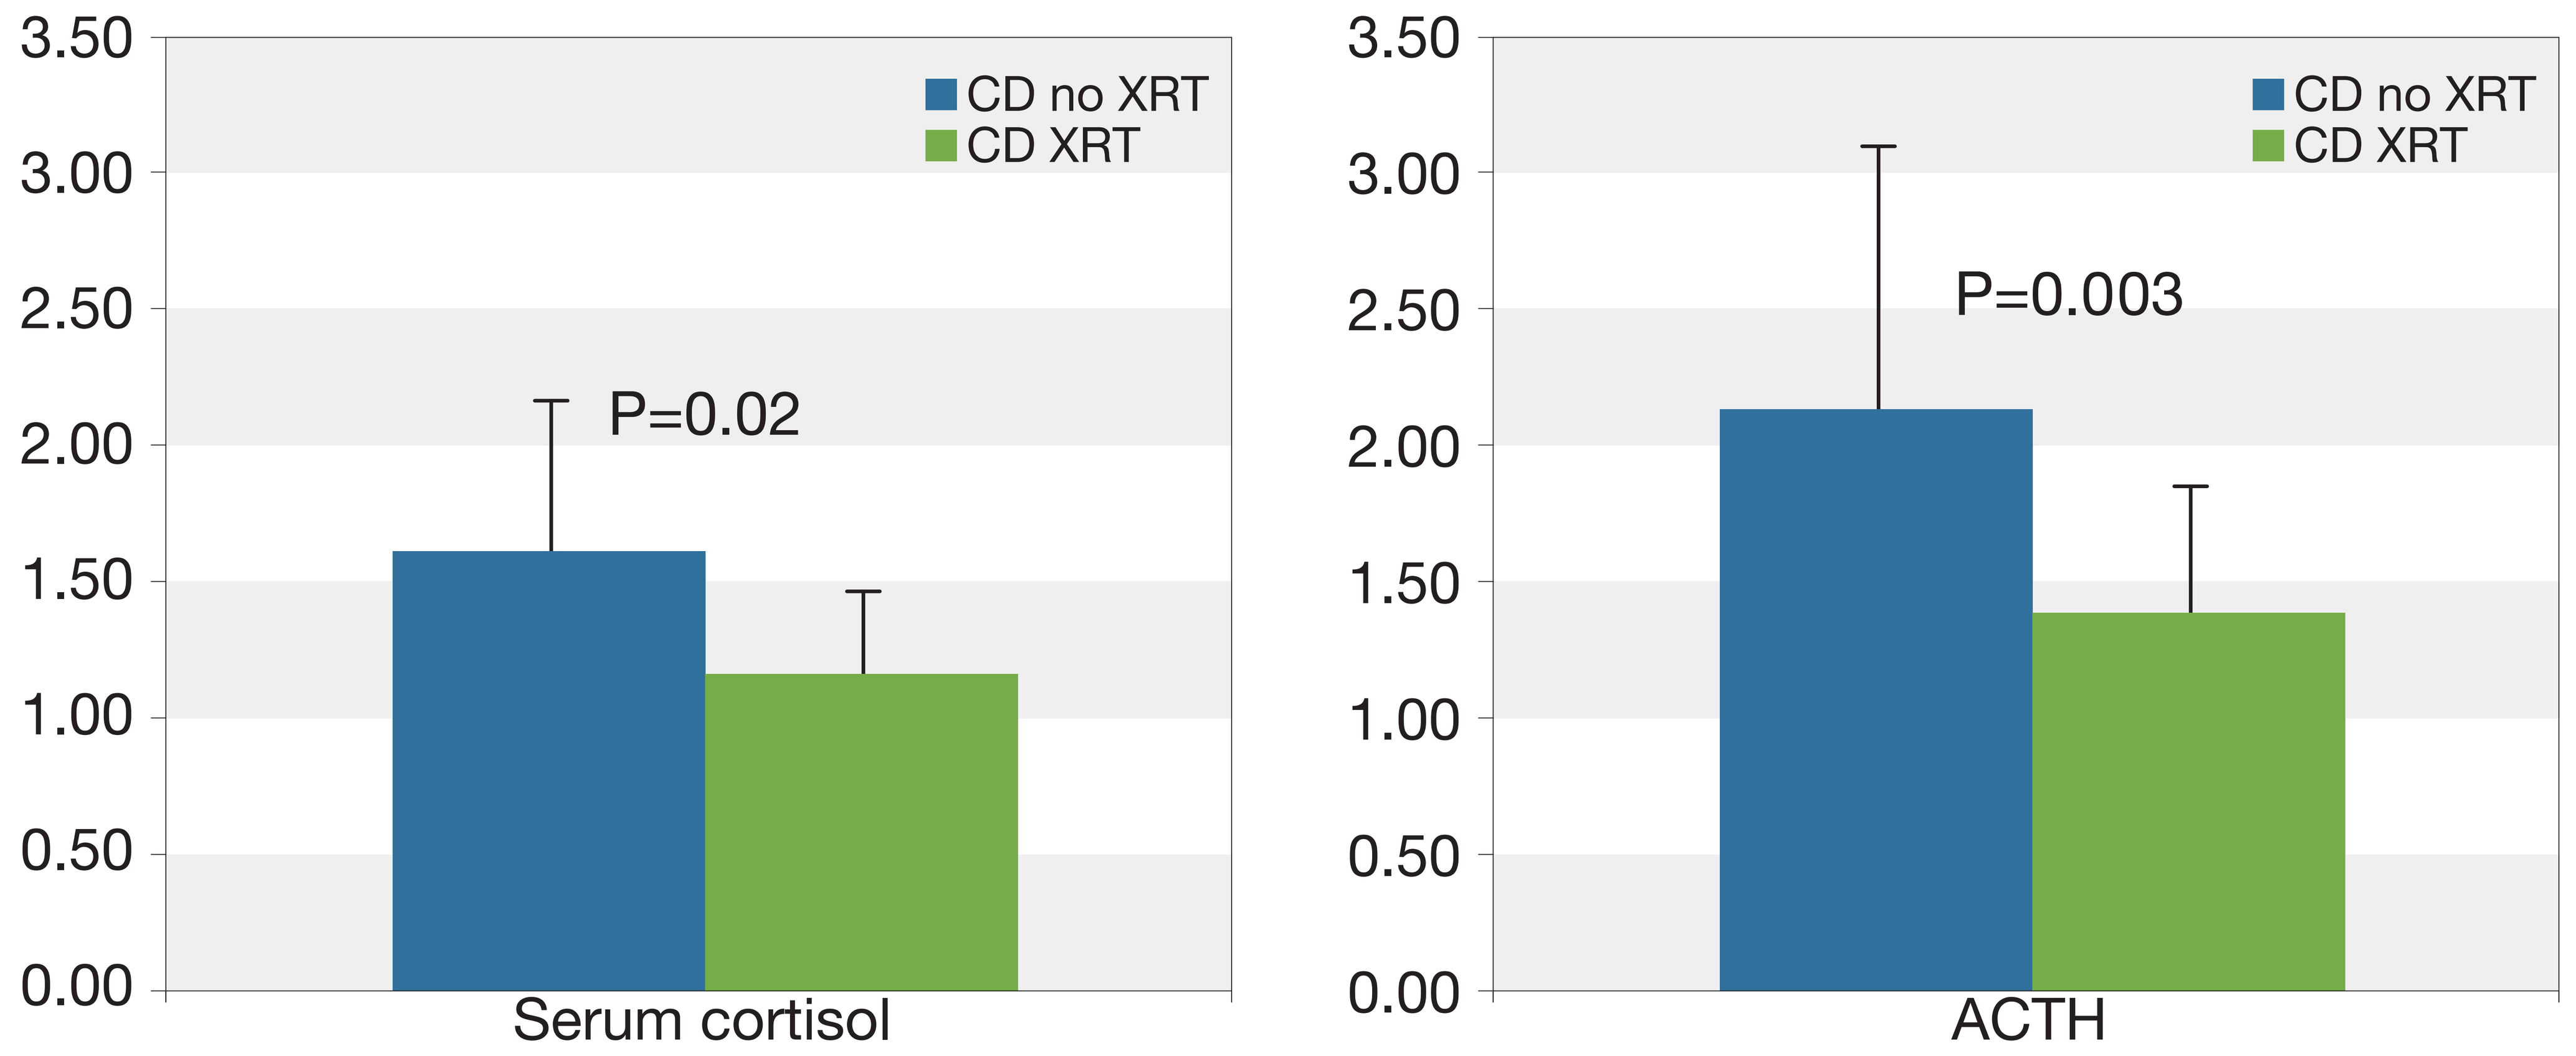
3.50
3.00
2.50
2.00
1.50
1.00
0.50
0.00
CD no XRT
CD XRT
P=0.02
Serum cortisol
P=0.003
ACTH

Figure 3. Mean ± SD absolute change ACTH D14

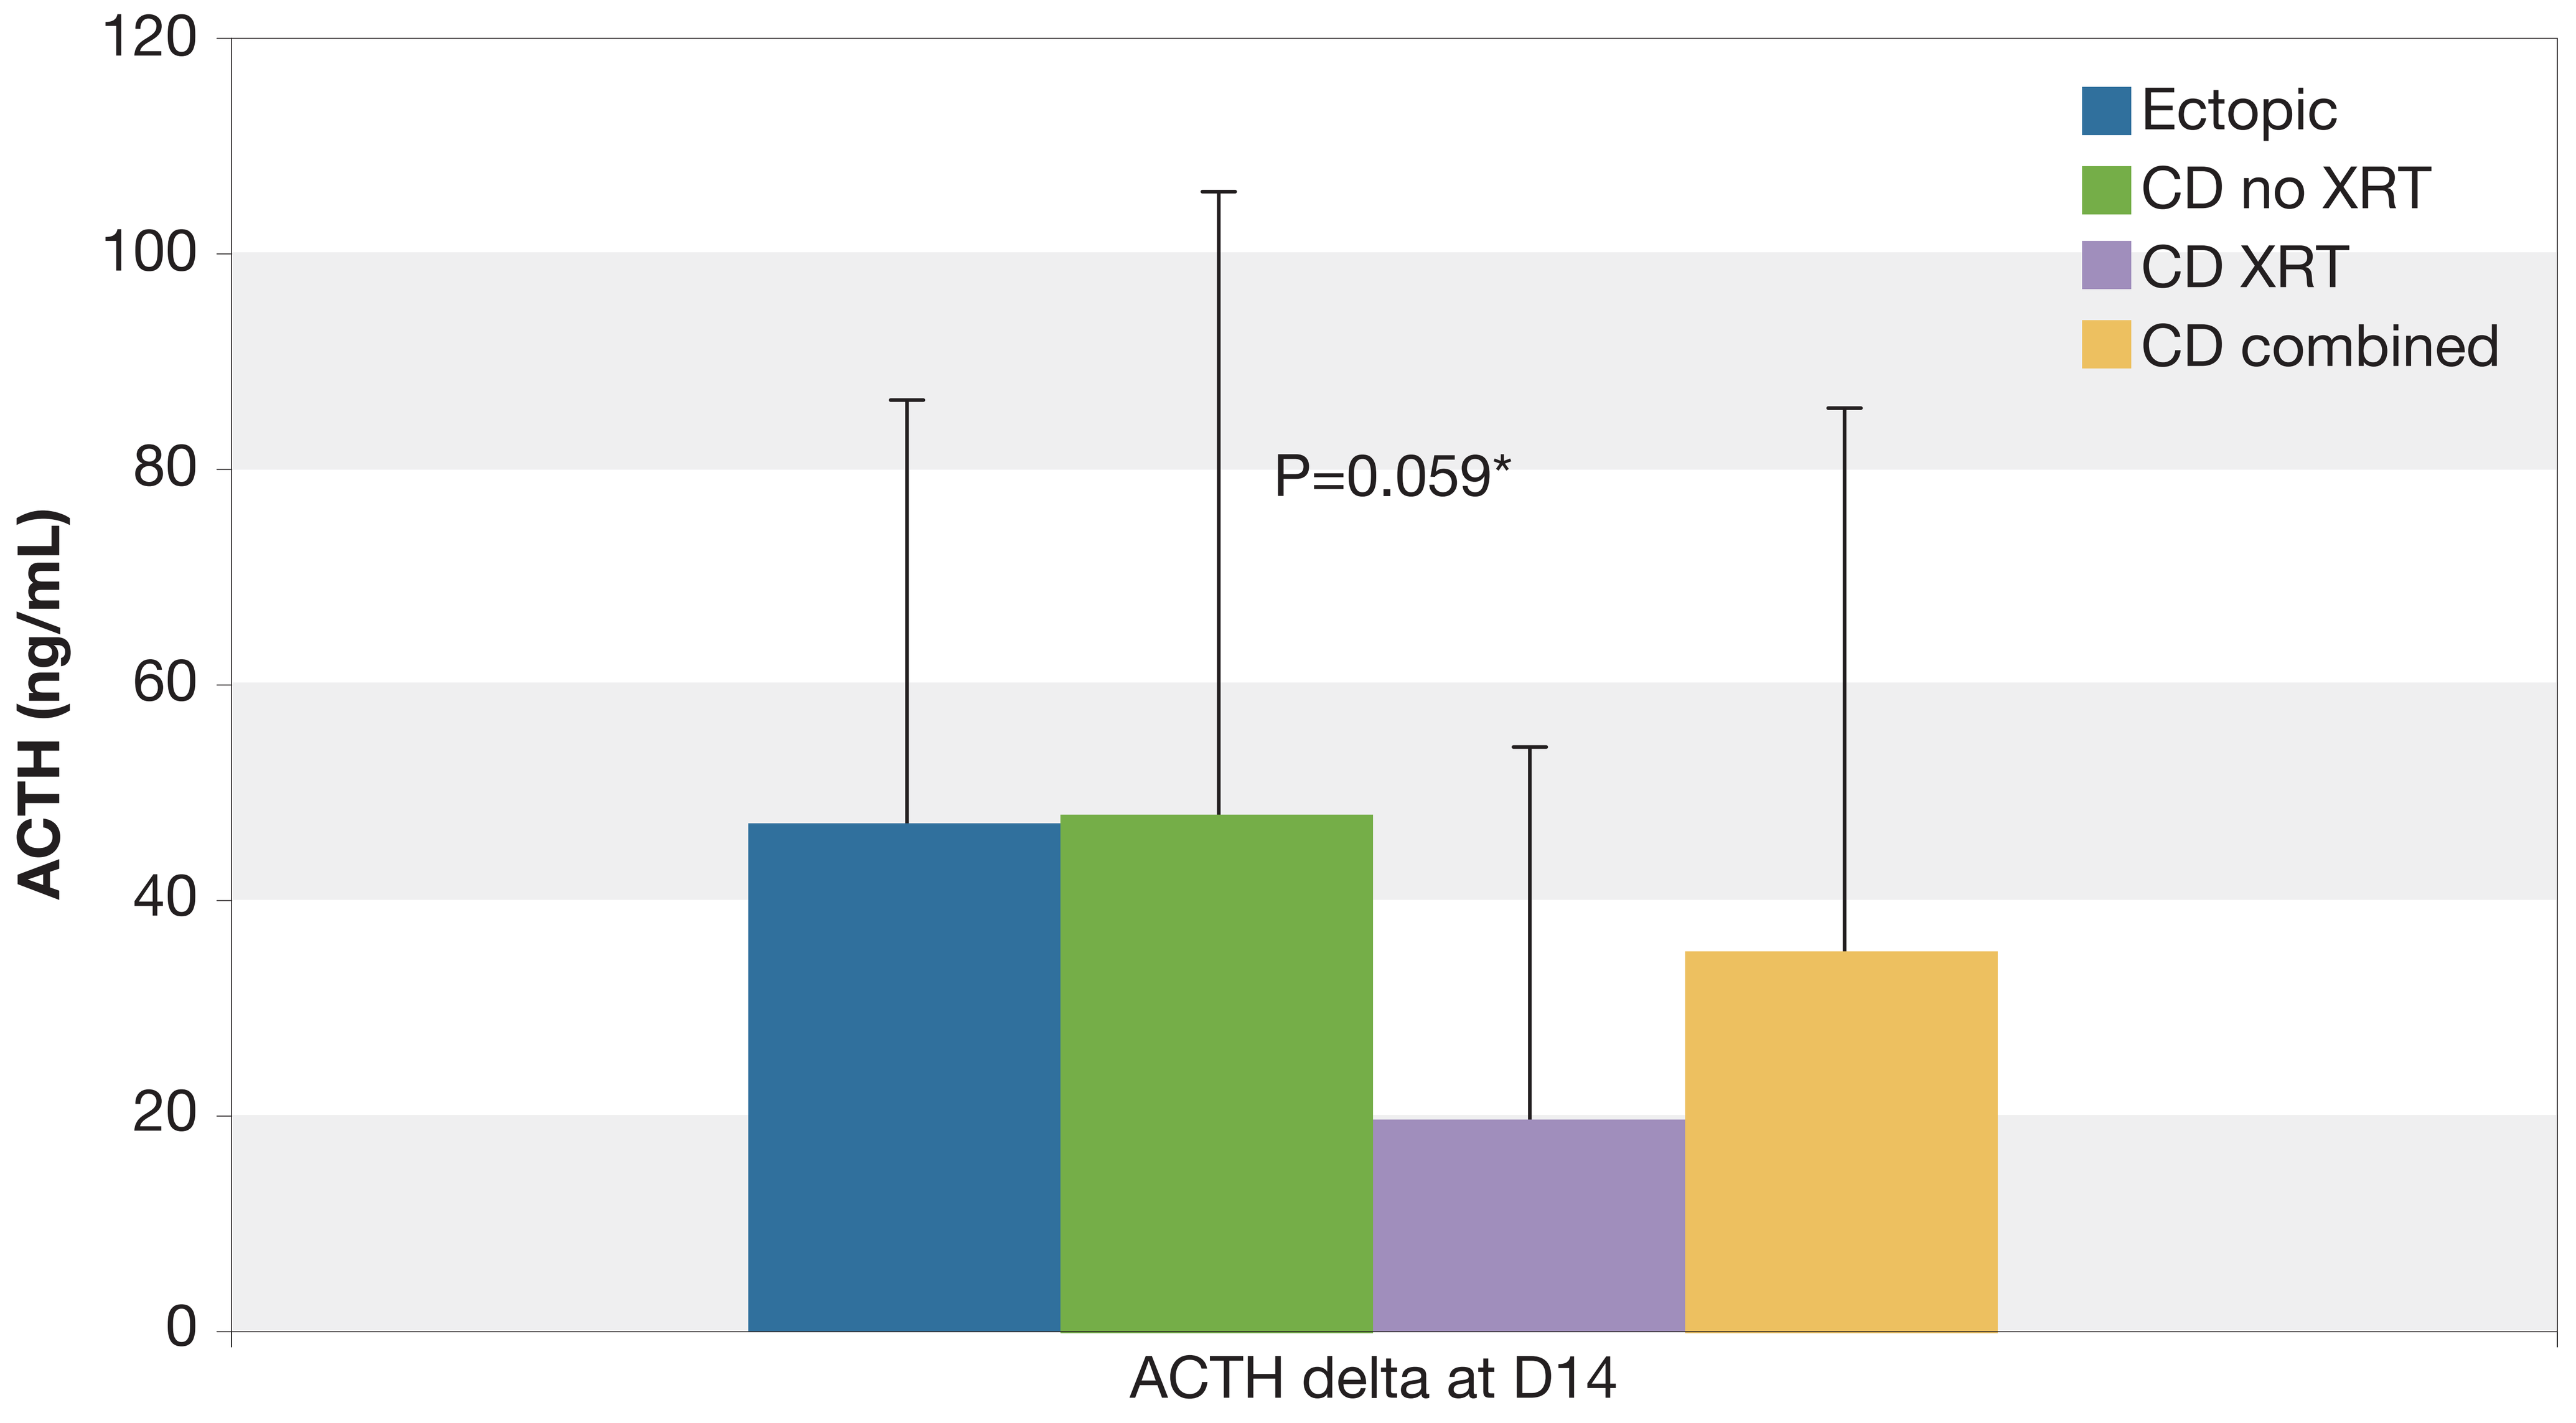
120
100
80
60
40
20
0
ACTH (ng/mL)
Ectopic
CD no XRT
CD XRT
CD combined
P=0.059*
ACTH delta at D14

*CD XRT vs no XRT

CONCLUSION

- After short term mifepristone treatment, ACTH and cortisol rise to a greater extent in radiation-naïve postoperative CD patients than those who have had radiation.
- These findings suggest further study of the use of mifepristone to aid in the differential diagnosis of CS and to test the integrity of the pituitary adrenal axis is warranted.

REFERENCE

[1] Corcept Therapeutics Incorporated. KorlymTM (mifepristone) 300mg Tablets. Available at: http://www.accessdata.fda.gov/drugsatfda_docs/label/2012/202107s000lbl.pdf Accessed May 9, 2013

[2] Fleseriu M, Biller BMK, Findling JW, Molitch ME et al. Mifepristone, a glucocorticoid receptor antagonist, produces clinical and metabolic benefits in patients with Cushing's syndrome. J Clin Endocrinol Metab 2012; 97, 2039-2049.

ACKNOWLEDGEMENTS

Editorial and graphical assistance was funded by Corcept Therapeutics Inc. and was provided by Judy Wiles and Selma Tse of Facet Communications.