SAT-391

Pre-surgical Use of Mifepristone for Ectopic Cushing's in a High-Risk Bilateral Adrenalectomy Candidate

Ivy-Joan Madu, MD[1]; Dat Nguyen, PharmD[2]

[1]Diabetes Associates Medical, Orange, CA; [2]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Surgical resection of the underlying tumor remains the definitive therapy for Cushing's syndrome (CS). Hypertension (HTN), obesity, hyperglycemia, and other cardiometabolic manifestations of CS can increase surgical risk. Preoperative intervention to improve metabolic control in a patient scheduled for a BLA is described.

Case: A 42-yr-old female with CS due to an ACTH producing carcinoid with lung metastasis was referred for preoperative optimization prior to a scheduled BLA. Chest CT showed numerous bilateral pulmonary nodules (largest up to 3.6 cm). She presented with moon facies, purple striae, and weakness requiring a cane for ambulation.

Co-morbid conditions included obesity (BMI 44), HTN (152/114 mmHg), diabetes (A1c 8.8%, blood glucose 266 mg/dL). She was treated with: octreotide LAR injection, Levemir insulin 10 units/d, metformin 1000 mg BID, glipizide 10 mg BID, amlodipine 10 mg/d, hydrochlorothiazide 12.5 mg/d, metoprolol 50 mg BID, losartan 100 mg/d.

Due to the progressive nature of the lung masses and the uncontrolled cortisol secretion, a BLA was scheduled. Preoperative evaluation categorized the patient as high risk due to her co-morbid conditions, high probability of frail tissues and poor wound healing due to prolonged hypercortisolism. Preoperative optimization goals were to reduce blood sugars, BP and the effects of hypercortisolism in order to improve wound healing, reduce the perioperative complications such as DVT and electrolyte imbalance.

Mifepristone (Korlym®, Corcept Therapeutics) 300 mg/d, a competitive glucocorticoid receptor antagonist, was utilized for a total of 2 months, terminating right before surgery. After one month of mifepristone therapy, blood glucose ranged between 90-150 mg/dL and Levemir insulin was discontinued. Two months later and just prior to surgery, BMI decreased (42.5); improvements were documented in BP (143/101 mmHg), A1c (8.0%) and glucose (149 mg/dL). No adrenal insufficiency was observed.

Despite her high risk, there were no major postoperative complications requiring extended hospitalization after the BLA (via the posterior approach). Only episodes of nausea and vomiting were observed. BP and blood glucose improved, allowing amlodipine and glipizide to be discontinued. Metformin was reduced to 500 mg/d and eventually discontinued. Patient was placed on steroid replacement therapy with hydrocortisone 20 mg BID and fludrocortisone 0.1 mg daily.

Nine months post-surgery - BMI 43.8, BP 135/80 mmHg, A1c 6.8%, glucose 118 mg/dL, Na 142 mEq/L, K 4.4 mEq/L.

Conclusion: Two months' therapy with mifepristone improved preoperative clinical status in a high-risk BLA patient by reducing weight and improving diabetic control. Clinical trials with longer duration of therapy are needed to evaluate the impact of mifepristone on surgical risks, outcomes, and length of hospitalization.

INTRODUCTION

- Severe Cushing's syndrome (CS) resulting from ectopic adrenocorticotropin (ACTH) secretion in association with metastatic cancer can progress rapidly[1,2]
 - In these patients, hypercortisolism may be treated with either medical therapy or bilateral adrenalectomy (BLA)[2]
- Severe or prolonged hypercortisolism is associated with increased risk of infection, hypokalemia, and cardiometabolic derangements such as obesity, hypertension, and hyperglycemia, all of which can increase surgical risk[3-5]
- Medical therapy may be used to treat hypercortisolism and improve metabolic indices prior to surgery, although data are limited[2]
- Mifepristone is a glucocorticoid receptor antagonist approved for control of hyperglycemia in all forms of CS in patients who have failed or are not candidates for surgery and who have diabetes or glucose intolerance
 - In addition to improvement in diabetes and/or hypertension, significant clinical response (assessed as improvement in glucose, blood pressure, lipids, weight/BMI, cushingoid appearance, depression, cognition, and quality of life) was noted in 87% of patients treated with mifepristone in the 24-week phase 3 SEISMIC trial[6]
- We describe the case of a 42-year-old woman with an ACTH-producing carcinoid tumor and lung metastasis who was treated preoperatively with mifepristone before undergoing a scheduled BLA

CLINICAL PRESENTATION

History of Present Illness

- The patient was a 42-year-old woman with a history of an ectopic ACTH-producing carcinoid tumor and lung metastasis who was scheduled for a BLA
- Past medical history
 - Carcinoid diagnosed in 2003; right lower lung lobectomy (2003)
 - Gastroesophageal reflux disease
 - Hypertension (2010)
 - Diabetes mellitus (2013); started on oral hypoglycemic agents
 - Diagnosed with CS resulting from ACTH-producing carcinoid in June 2013; treating physician recommended BLA to control cortisol secretion
- Preoperative evaluation categorized the patient as high-risk for perioperative complications because of the presence of co-morbid conditions and likelihood of impaired wound healing due to prolonged hypercortisolism
- The patient was referred for preoperative treatment of cardiometabolic derangements secondary to hypercortisolism to reduce surgical risk

Assessments

- The patient's chest CT showed numerous bilateral pulmonary nodules up to 3.6 cm in size; adrenals CT scans in 2012 and 2013 showed progression of bilateral hyperplasia (**Figure 1A** and **1B**)

Figure 1. Patient CT images of lungs A) and adrenals B)

A. Multiple bilateral lung nodules

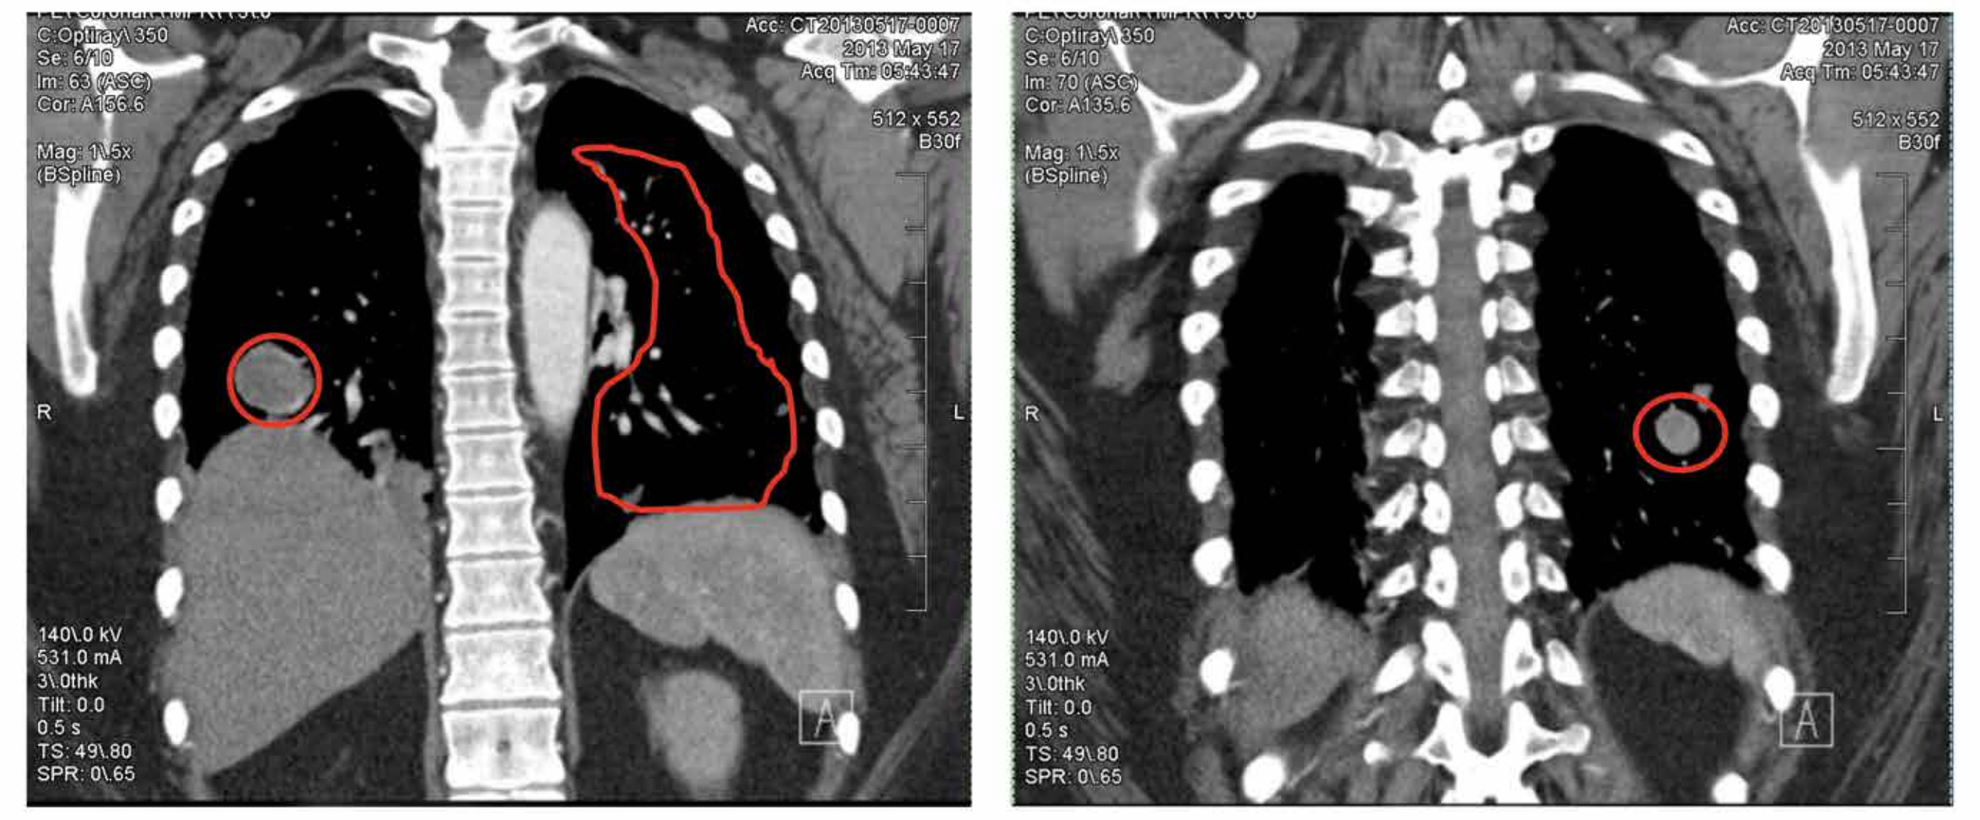

2013

B. Progression of bilateral hyperplastic adrenals

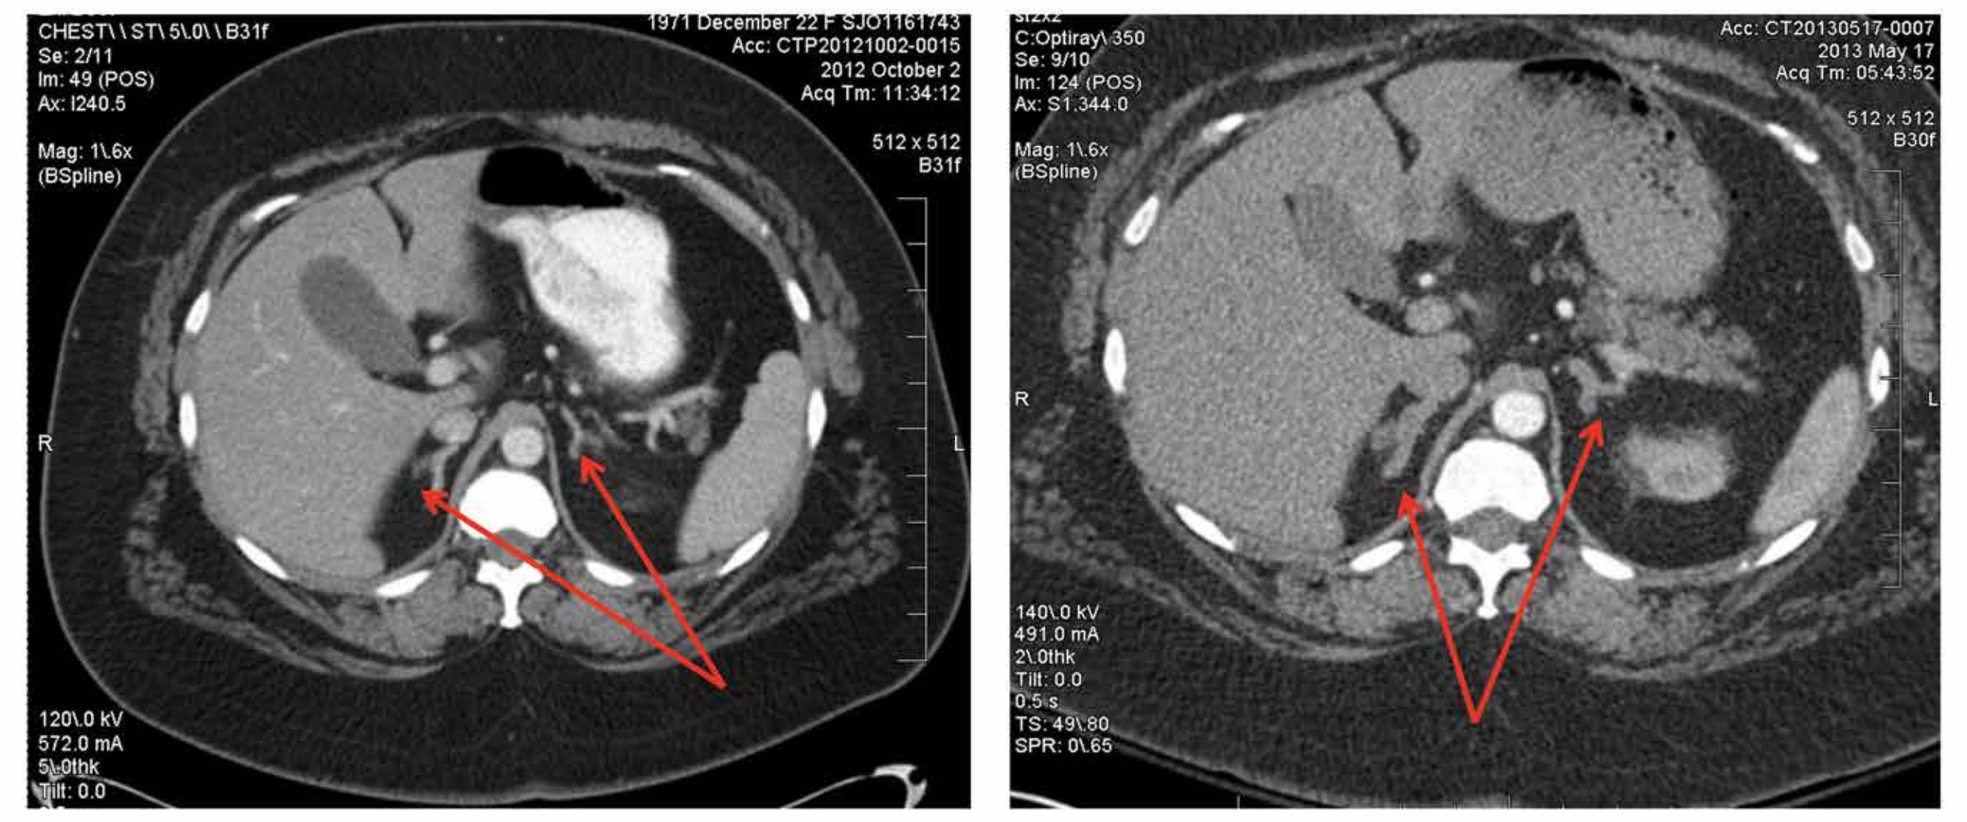

2012 2013

- Current vital signs and laboratory assessments included a body mass index (BMI) of 44 kg/m^2, blood pressure of 152/114 mmHg, blood glucose of 266 mg/dL, and hemoglobin A1c (HbA1c) of 8.8%
- Physical assessment revealed the presence of moon facies, purple striae, and weakness requiring the use of a cane for ambulation (**Figure 2**)
- Concomitant medications prior to mifepristone are listed in **Table 1**

Figure 2. Patient image showing cushingoid features

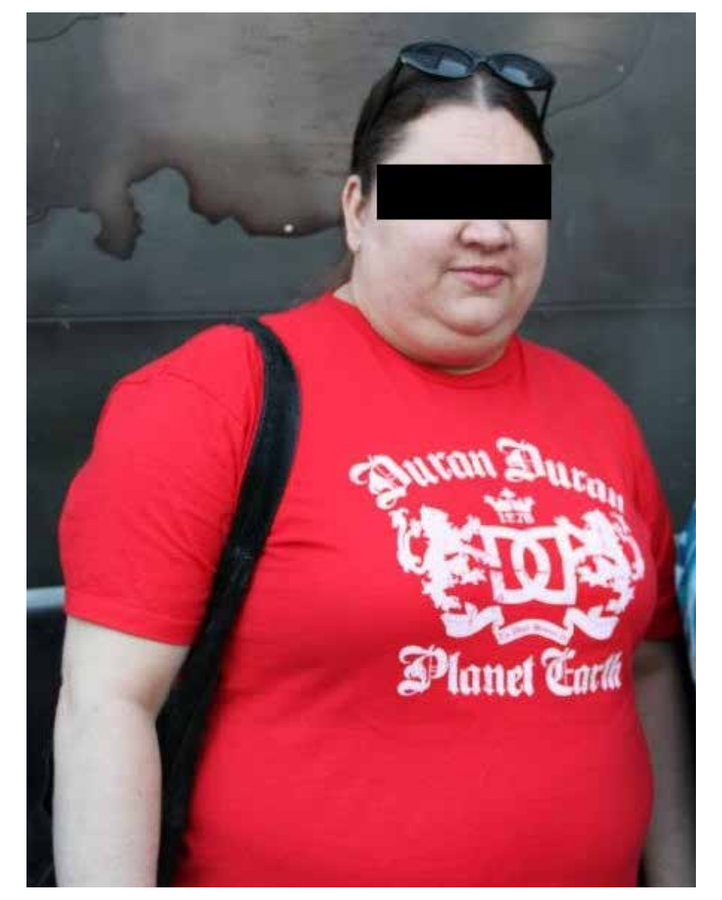

Table 1. Concomitant medications prior to mifepristone initiation

Medication	Dose
Octreotide depot	As directed (for many years)
Insulin (detemir)	10 units daily
Metformin	1000 mg twice daily
Glipizide	10 mg twice daily
Amlodipine	10 mg daily
Hydrochlorothiazide	12.5 mg daily
Metoprolol	50 mg twice daily
Losartan	100 mg daily

TREATMENT COURSE

- Mifepristone was initiated at a dose of 300 mg/d
- After 1 month of treatment with mifepristone, improvements in blood glucose were observed (ranging between 90-150 mg/dL), and insulin (detemir) was discontinued
- After 2 months of mifepristone therapy (just before surgery), additional improvements in weight and cardiometabolic parameters were noted (**Table 2**)

Table 2. Metabolic parameters before and 2 months after initiation of mifepristone therapy

Parameter	Before Mifepristone	After Mifepristone
BMI, kg/m^2	44	42.5
HbA1c	8.8	8
Blood pressure, mmHg	152/114	143/101
Blood glucose, mg/dL	266	149

- The patient stated that she "felt better" after mifepristone was added to her treatment regimen
- A BLA via the posterior approach was performed
- The patient's recovery was unremarkable with the exception of episodes of nausea and vomiting
 - The patient received post-operative medications for pain including morphine sulfate, oxycodone, and hydromorphone at various times (history of intolerance [severe nausea and vomiting] to hydrocodone)
- There were no major postoperative complications requiring extended hospitalization
- Postoperative improvements in blood pressure and blood glucose were noted, leading to discontinuation of amlodipine and glipizide
 - Metformin dosage was reduced to 500 mg/d and eventually discontinued
- Steroid replacement therapy included hydrocortisone 20 mg twice daily and fludrocortisone 0.1 mg daily
- 9 months after surgery, further improvements in several cardiometabolic parameters were noted (**Table 3**)
 - Her sodium and potassium were within normal range (sodium 142 mEq/L, potassium 4.4 mEq/L)

Table 3. Metabolic parameters 9 months post-surgery

Parameter	Post-surgery
BMI, kg/m^2	43.8
HbA1c	6.8
Blood pressure, mmHg	135/80
Blood glucose, mg/dL	118

DISCUSSION

- Patients with ectopic ACTH associated with metastatic cancer may have severe or prolonged hypercortisolism that can increase surgical risk
- Treatment to address the effects of hypercortisolism may benefit patients before surgical treatment with BLA
- Cassier et al (2008) published a report of 2 patients with ectopic ACTH due to metastatic endocrine carcinomas who were treated with mifepristone[7]
 - In both patients, marked improvement in diabetes and physical features were noted within 3 months of treatment and continued for approximately 10 months. Both patients later underwent BLA.
- Additional case reports have demonstrated successful use of mifepristone for severe hypercortisolism[8,9]
 - Chu et al (2001) reported the successful long-term treatment (18 months) of an extremely ill patient with refractory Cushing's disease (macroadenoma; cortisol levels >828 nM) with high-dose mifepristone (25 mg/kg/d) while awaiting the effects of pituitary radiation.[8] Of note, the patient's appearance and strength improved dramatically over time, with marked improvement in psychosis and cognition as well.
 - In a case series published by Castinetti et al (2009), a patient with bilateral adrenal hyperplasia had severe clinical signs of hypercortisolism.[9] Treatment with mifepristone (600 mg/d) led to improvement in signs of hypercortisolism and hypertension during the first 3 months of treatment, followed by additional improvement in HbA1c after 6 months of treatment. This patient later underwent a BLA.

SUMMARY AND CONCLUSIONS

- This report describes preoperative treatment of a surgical high-risk ectopic ACTH patient with the glucocorticoid receptor antagonist mifepristone before BLA
- Several clinical improvements were noted with short-term (2-month) therapy, including improvement in blood pressure and reductions in BMI and blood glucose levels resulting in discontinuation of basal insulin
- Treatment with mifepristone was well tolerated and there was no occurrence of adrenal insufficiency
- Mifepristone may be a beneficial preoperative treatment to rapidly improve cardiometabolic status and decrease surgical risks related to the effects of hypercortisolism in patients with CS
- Prospective trials of mifepristone are needed to further evaluate the effects of mifepristone on surgical outcomes

REFERENCES

1. Newell-Price J, et al. *Lancet*. 2006;367:1605-17.
2. Feelders RA, et al. *Neuroendocrinology*. 2010;92(suppl 1):111-5.
3. Takagi S, et al. *Endocr J*. 2009;56:1009-18.
4. Heyn J, et al. *Pituitary*. 2012;15:117-25.
5. Sarlis NJ, et al. *J Clin Endocrinol Metab*. 2000;85:42-7.
6. Fleseriu M, et al. *J Clin Endocrinol Metab*. 2012;97:2039-49.
7. Cassier PA, et al. *Eur J Endocrinol*. 2008;158:935-8.
8. Chu JW, et al. *J Clin Endocrinol Metab*. 2001;86:3568-73.
9. Castinetti F, et al. *Eur J Endocrinol*. 2009;160:1003-10.

ACKNOWLEDGEMENT

Funding for development, editorial, design, and production support was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors contributed to the development of the poster, reviewed the material, and provided approval of the final version.

DISCLOSURES

IJM: Clinical Researcher, Amylin, Andromeda, Astra Zeneca, Grifols, Jansen, Merck, Mylan, Novo Nordisk, Sanofi, Takeda.

DN: Employee, Corcept Therapeutics.

Presented at ENDO 2015 • San Diego, California • March 5-8, 2015