THR-520

Mifepristone for the Successful Treatment of Cushing's Syndrome After Multiple Failed Interventions

M. Tufail Ijaz, MD[1]; Neven Pesa, RN, CPNP[2]; Dat Nguyen, PharmD[2]

[1]Genesis HealthCare System, Zanesville, OH; [2]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Surgical treatment for Cushing's disease (CD) is not always successful and repeat surgery may not be an option for some patients due to the increased risk and lowered success rate with each reoperation. Even in cases with postoperative biochemical remission, clinical and metabolic manifestations (eg, cushingoid appearance, obesity, diabetes) may still persist. There is a need for therapies that can offer sustained clinical and metabolic benefits. The following case reviews a patient with refractory CD despite multiple treatment modalities and who subsequently responded to treatment with a competitive glucocorticoid receptor antagonist, mifepristone.

Clinical Case: A 44-yr-old female patient presented in 2012 with uncontrolled CD after several unsuccessful interventions.

Past treatments included 2 failed transsphenoidal surgeries (TSS), radiation therapy, bromocriptine, and ketoconazole.

Her first TSS was performed in 2008 but her preoperative cushingoid appearance was unchanged a year later. Her BMI (39.1), A1c (6.4%) and urine cortisol levels (85 mcg/24 h [nl<45]) were high while taking bromocriptine 2.5 mg BID. Her MRI showed a 1.1 X 1.4 cm pituitary mass. Ketoconazole 200 mg BID was initiated but stopped after 1 month due to elevated ALT (97 units/L) and AST (52 units/L). A second TSS was performed in 2010 followed by Intensity Modulated Radiation Therapy 2 months later. A year after the 2nd TSS, the ACTH (54 pg/mL) and urine cortisol (74 mcg/24 h) were elevated. Her BMI (38.4) remained high and she continued to exhibit cushingoid features. In 2012, the patient's AM cortisol (28 mcg/dL) and ACTH (108 pg/mL) remained high. UFC was normal despite signs and symptoms of Cushing's syndrome (central obesity, BMI 37, moon facies, striae, easy bruising, thin extremities, and depression). Her BP was stable, and her A1c was 6%. Concomitant medications included levothyroxine 100 mcg/d, sitagliptin/metformin 50 mg/500 mg BID, rosuvastatin 10 mg/d, fenofibrate 145 mg/d.

Mifepristone (Korlym®, Corcept Therapeutics) was initiated at 300 mg/d and currently stabilized at 900 mg/d. After ~2 yrs of treatment, improvements in her signs and symptoms were observed: BMI 31.6, A1c 5.2% (without the use of diabetic medications), waist circumference decreased by 11 cm to 108 cm. Physical appearance was normal with minimal striae. The patient describes feeling 'completely normal'; her depression resolved with mifepristone.

Conclusion: Control of metabolic and clinical manifestation of CD is often difficult to achieve and maintain. Prior to FDA approval, multiple therapies (eg, 2xTSS, radiation, bromocriptine, ketoconazole) were tried without success in this patient with CD. The addition of mifepristone therapy resulted in several beneficial and sustained outcomes - including reductions in A1c, weight, and elimination of anti-diabetes medications.

Figure 1. Patient photographs showing progressive weight gain over time from early 1990s to 2008 before any treatment for CD

A. Patient in her 20s

B. Patient in her early 30s

C. Patient in her late 30s

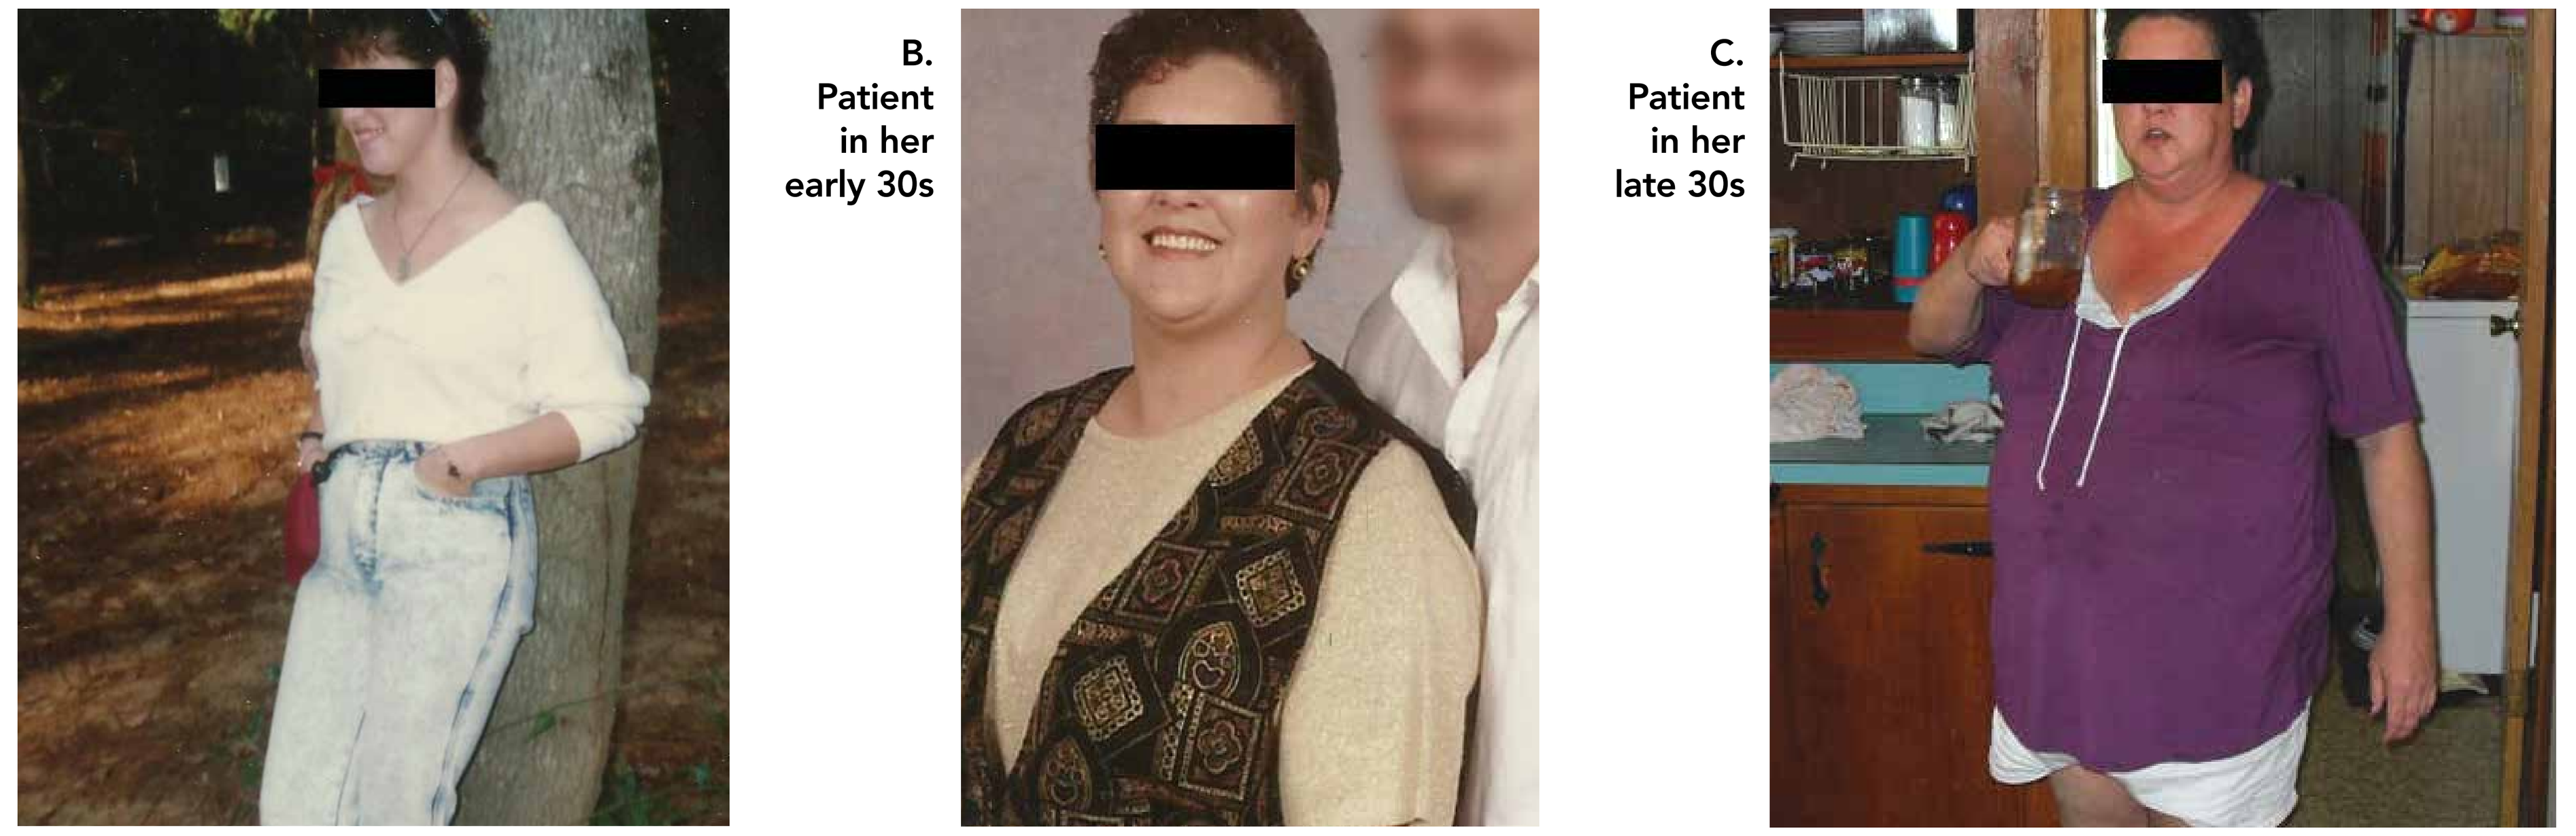

Patient provided signed release to share photographs.

INTRODUCTION

- Endogenous Cushing's syndrome (CS) is a complex multisystemic condition resulting from prolonged exposure to cortisol, most commonly caused by an adrenocorticotropin hormone (ACTH)-secreting secreting pituitary adenoma, and is then referred to as Cushing's disease (CD)[1,2]
- CD is associated with substantial metabolic dysfunction, including weight gain, hypertension, insulin resistance, diabetes mellitus, and dyslipidemia[2-4]
- Surgical resection of the underlying tumor remains the treatment of choice for most patients
 - However, 10% to 35% of patients will have persistent hypercortisolism postoperatively depending on the circumstances of the disease,[5-8] and the 5-year recurrence rate is 26%[9]
- Repeat surgical procedures may be performed, but are associated with lower success rates and increased postoperative risks[5]
- Alternatively, adjunctive radiotherapy for ACTH-secreting pituitary adenomas may take years to normalize excess cortisol[5,10]
- Thus, there is a need for therapy that can help maintain remission from hypercortisolism and potentially ameliorate metabolic risk factors
- Ketoconazole is widely used off-label for CD and produces biochemical remission in approximately 50% of patients[11]
 - However, up to 21% of patients develop intolerance to the treatment[11]
 - It is also associated with liver toxicity, which has been fatal in some cases,[12,13] prompting the Food and Drug Administration to issue a black box warning in 2013[14]
- The glucocorticoid receptor antagonist mifepristone is approved for patients with all forms of CS who have diabetes or glucose intolerance and have failed or are not candidates for surgery
 - In addition to improvement in glucose parameters, significant clinical improvement was noted in 87% of patients treated with mifepristone in the 24-week SEISMIC trial[15]
- We describe the case of a patient with persistent signs and symptoms of CD after multiple therapies who was successfully treated with mifepristone

CLINICAL PRESENTATION

- A 41-year-old woman with a 10-year history of CD presented for disease follow-up and management in September 2009
- Past medical history was significant for pituitary adenoma and type 2 diabetes
- Patient photographs show progressive weight gain up to 2008 before any treatment for CD (**Figure 1**)

Assessments

- MRI in July 2009 showed a 1.1 x 1.4 cm pituitary mass
- Laboratory test results obtained in October 2009 revealed elevated 24-hr urinary free cortisol (UFC) (85 mcg/24 hr, normal <45 mcg/24 hr), ACTH (106 pg/mL, normal 6-58 pg/mL), serum AM cortisol (28.2 mcg/dL, normal 4.5-22.7 mcg/dL), and failure to suppress cortisol with a 1 mg dexamethasone suppression test (DST) (17 mcg/dL), which suggested ongoing CD
- At that time, the patient's weight was 221 lbs, her BMI was 39.1 kg/m^2, and her blood pressure was stable at 130/84 mmHg
- Physical examination showed central obesity, thin extremities, cushingoid facies, dorsocervical fat pad, erythema on the cheeks, and purple striae on the abdomen; patient reported bruising and weakness in her extremities

TREATMENT COURSE

- An overview of the patient's treatment course is shown in **Figure 2**

Figure 2. Patient treatment course

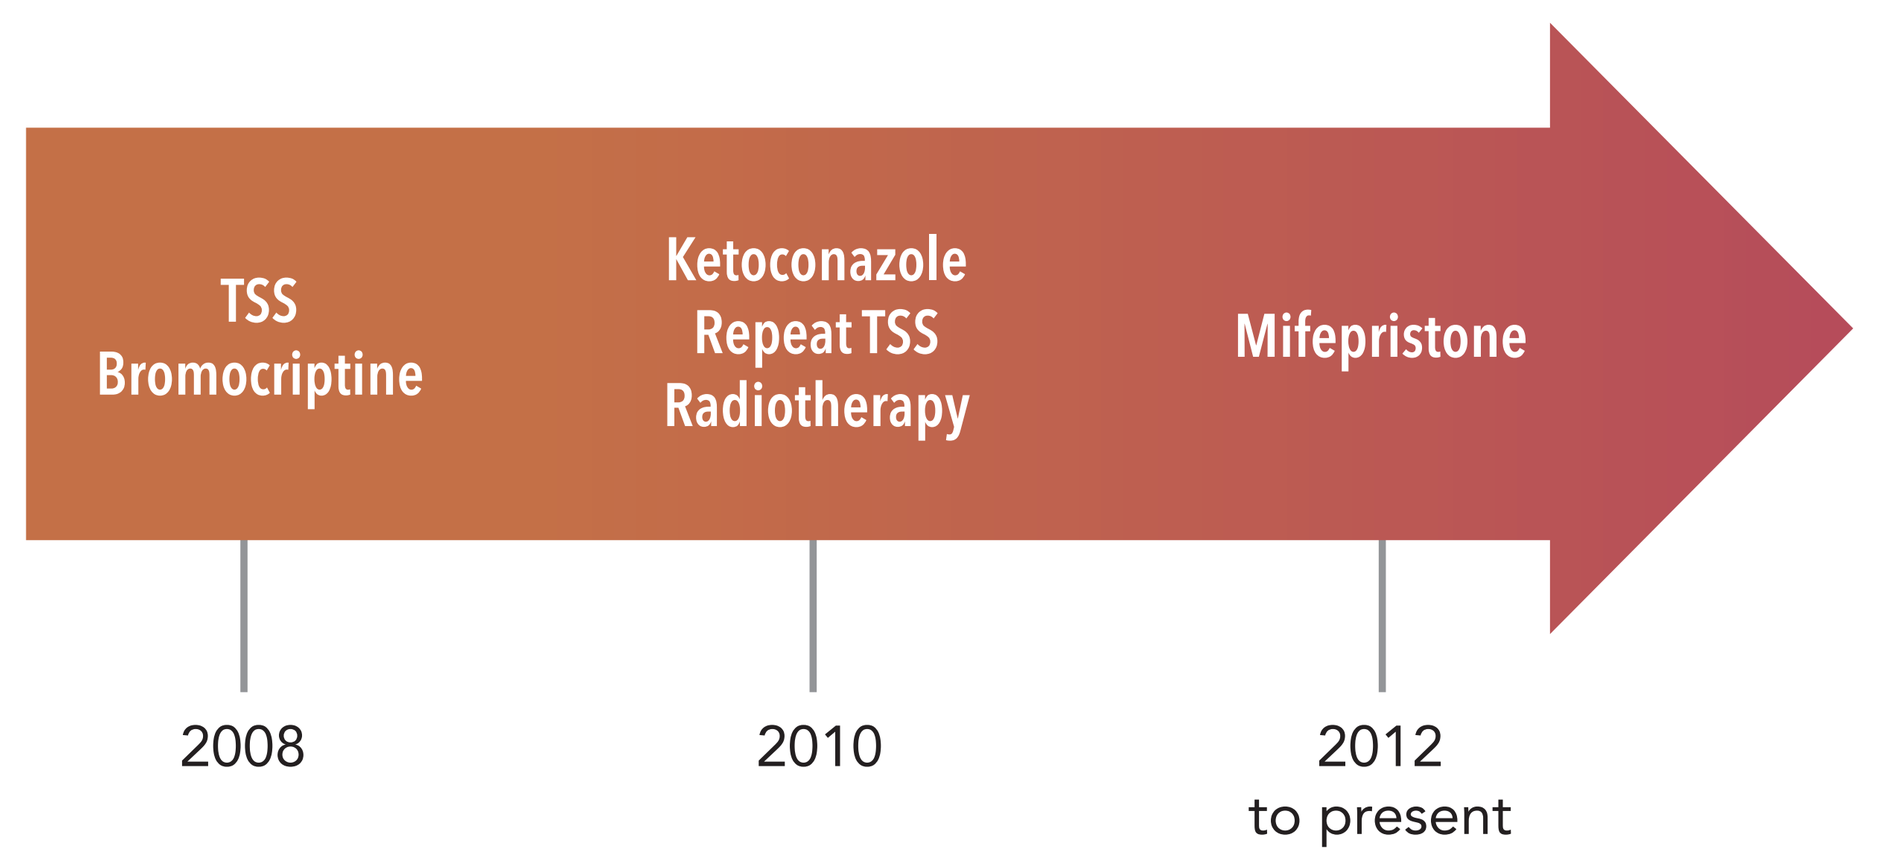
TSS
Bromocriptine
Ketoconazole
Repeat TSS
Radiotherapy
Mifepristone
2008
2010
2012
to present

- The patient underwent her first transsphenoidal surgery (TSS) in 2008 and was taking bromocriptine 2.5 mg twice daily
- Ketoconazole 200 mg twice daily was started, but was discontinued after 1 month because of elevated ALT (97 units/L) and AST (52 units/L)
- A repeat TSS was performed in May 2010
 - Immediate postoperative cortisol assessment was as follows: AM cortisol 17 mcg/dL and midnight salivary cortisol 0.349 mcg/dL (normal <0.112 mcg/dL)
- Intensity Modulated Radiation therapy was initiated 2 months after surgery and was completed in September 2010
- Following repeat TSS and radiotherapy, persistent elevations in ACTH, 24-hr UFC, or AM cortisol were observed over the next 2 years (**Table 1**)

Table 1. Patient ACTH, UFC, and AM cortisol levels following repeat TSS and radiotherapy (2010-2012)

	Sept 2010	Dec 2010	Apr 2011	Aug 2011	Feb 2012	Jun 2012	Dec 2012
ACTH (pg/mL, normal 6-58)	**88**	**96**	54	**87**	**108**	**67**	**74**
UFC (mcg/24 hr, normal <45)	**146**	**126**	**74**	44.6	19.7	22.4	29
AM cortisol (mcg/dL, normal 4.5-22.7)	**25**	19.2	19.6	18	**28**	20.5	19

Values above the normal range are in bold font.
ACTH=adrenocorticotropin hormone; TSS=transsphenoidal surgery; UFC=urinary free cortisol.

- Cushingoid features remained present, along with little change in weight or BMI
- In 2012, repeat DSTs showed failure to suppress with 1 mg DST (cortisol 7.2 mcg/dL) and normal suppression with 8 mg DST (2 mcg/dL, baseline 23 mcg/dL)
- In December 2012, the patient reported body aches, fatigue, and depressed mood
- Concomitant medications are listed in **Table 2**

Table 2. Concomitant medications (2012)

Medications	Dose
Levothyroxine	100 mcg daily
Sitagliptin/metformin	50 mg/500 mg twice daily
Lovastatin*	40 mg daily
Fenofibrate	145 mg daily

*Lovastatin was changed to rosuvastatin 10 mg upon initiation of mifepristone to prevent potential drug-drug interaction.

- Treatment with mifepristone was initiated at a dose of 300 mg/d, which was titrated up to 900 mg/d

Treatment Outcomes After Mifepristone

- Following 1.5 years of treatment with mifepristone, several clinical improvements in signs and symptoms of CD were noted (**Table 3**)
 - The patient lost 31 lbs and her waist circumference decreased by 11 cm to 108 cm
 - Blood glucose and hemoglobin A1c (HbA1c) decreased and antidiabetes medications were stopped in April 2014
 - Physical improvements included resolution of cushingoid facies and reduction in striae

Table 3. Changes in metabolic parameters before initiation of mifepristone and after 20 months of treatment

	Before mifepristone	After mifepristone
Weight, lbs	209.5	178
BMI, kg/m^2	37.1	31.6
HbA1c	6	5.2
Blood glucose, mg/dL	127	109
Blood pressure, mmHg	133/93	134/90

HbA1c=hemoglobin A1c; BMI=body mass index.

Additional Outcomes

- Blood pressure remained relatively stable before and after initiation of mifepristone therapy (**Table 3**)
- As of April 2014, the patient reported "feeling completely normal" and was extremely happy
- Some fatigue and nausea were reported by the patient
- Patient continues to be monitored

SUMMARY AND CONCLUSIONS

- Long-term control of metabolic and clinical manifestation of CS is often difficult to achieve
- With CD, surgical treatment is not always successful and repeat surgery may not be an option for some patients due to the increased risk and lowered success rate with each reoperation
- In this report, a patient with persistent CD despite multiple therapies (eg, 2xTSS, radiation, bromocriptine, ketoconazole) was initiated on mifepristone therapy
- The addition of mifepristone 2 years after radiation therapy resulted in several beneficial and sustained outcomes, including reductions in body weight, BMI, and HbA1c and elimination of blood glucose-lowering medications. The therapy was well tolerated.
- Mifepristone is a useful treatment option for patients with CS who have failed surgery or as adjunctive treatment to radiation therapy

REFERENCES

1. Newell-Price J, et al. *Lancet.* 2006;367:1605-17.
2. Nieman LK, et al. *J Clin Endocrinol Metab.* 2008;93:1526-40.
3. Faggiano A, et al. *J Clin Endocrinol Metab.* 2003;88:2527-33.
4. Valassi E, et al. *Eur J Endocrinol.* 2011;165:383-92.
5. Biller BMK, et al. *J Clin Endocrinol Metab.* 2008;93:2454-62.
6. Tritos NA, Biller BM. *J Neurooncol.* 2014;117:407-14.
7. Dimopoulou C, et al. *Eur J Endocrinol.* 2014;170:283-92.
8. Hamrahian AH, et al. *Endocr Pract.* 2014;20:746-57.
9. Patil CG, et al. *J Clin Endocrinol Metab.* 2008;93:358-62.
10. Loeffler JS, Shih HA. *J Clin Endocrinol Metab.* 2011;96:1992-2003.
11. Castinetti F, et al. *J Clin Endocrinol Metab.* 2014;99:1623-30.
12. Zollner E, et al. *J Pediatr Endocrinol Metab.* 2001;14:335-8.
13. McCance DR, et al. *Lancet.* 1987;1:573.
14. US Food and Drug Administration. http://www.fda.gov/drugs/drugsafety/ucm362415.htm.
15. Fleseriu M, et al. *J Clin Endocrinol Metab.* 2012;97:2039-49.

ACKNOWLEDGEMENT

Funding for development, editorial, design, and production support was provided by Corcept Therapeutics, to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors contributed to the development of the poster, reviewed the material, and provided approval of the final version.

DISCLOSURE

MTI: Speaker, Astra Zeneca, Boehringer Ingelheim, Bristol-Myers Squibb, Corcept, Eli Lilly, Merck, Pamlab.

NP: Employee, Corcept Therapeutics.

DN: Employee, Corcept Therapeutics.

Presented at ENDO 2015 • San Diego, California • March 5-8, 2015