LBS-075

Marked Improvement in Liver Enzymes During Medical Therapy With Mifepristone for Cushing's Syndrome

Enzo Ragucci, MD[1]; Michele Lamerson, RN, MS, CPNP[2]

[1]Diabetes and Endocrinology, West Nyack, NY; [2]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Nonalcoholic fatty liver disease is frequent among people with obesity, type 2 diabetes, insulin resistance, and dyslipidemia. These metabolic aberrations are also common in Cushing's syndrome (CS), which has led researchers to investigate the contributions of cortisol. Rockall et al previously confirmed hepatic steatosis on CT scans in 20% of patients with active CS. However, it is not known if successful treatment of CS can reverse the incidence and progression to NASH (nonalcoholic steatohepatitis).

We detail a patient with CS and NASH who saw a significant improvement in liver function tests (LFTs) after successful treatment of CS with mifepristone (Korlym®, Corcept Therapeutics), a competitive glucocorticoid receptor antagonist.

Clinical Case: A 47-yr-old woman with a past medical history of hypertension (142/90 mmHg), pre-diabetes (A1c 5.9%), dyslipidemia (Tchol 274 mg/dL, LDL 203 mg/dL, TG 132 mg/dL) and NASH was found to have a left adrenal adenoma on a routine CT scan she had for evaluation of nephrolithiasis. She previously had a cholecystectomy (due to chronic cholecystitis & cholelithiasis) and liver biopsy showing steatohepatitis and diffuse steatosis. Her medications included amlodipine/olmesartan medoxomil and rosuvastatin calcium.

The CT scan revealed a left adrenal adenoma measuring 2.9 x 1.9 x 2.5 cm with benign radiologic characteristics. She did not have overt features of CS but complained of persistent abnormal weight gain (BMI 30.6 kg/m^2) with central obesity. Biochemical testing was negative for pheochromocytoma and primary aldosteronism. Hormonal testing for autonomous cortisol secretion showed a mildly elevated UFC (62.6 µg/d), failure to suppress on successive 1-mg dexamethasone suppression tests (13.2 µg/dL, 12.5 µg/dL), elevated midnight salivary cortisol (0.19 µg/dl), suppressed ACTH <5 pg/mL (5-45 pg/mL) and low DHEA-S 20 µg/dL (42-290 µg/dL). Baseline LFTs were elevated: AST 110 U/L (nl 2-40 U/L), ALT 232 U/L (nl 2-60 U/L).

She declined adrenalectomy. Mifepristone was initiated at 300 mg/d and increased to 900 mg/d over 20 weeks.

At 4 weeks, antihypertensive medications were discontinued. At week 20, her BP was stable (120/82 mmHg) and her weight decreased from 178 to 160 lbs. Marked improvement in her liver enzymes was noticed during mifepristone therapy with normalization by week 20 (AST 32 U/L [nl, 10-35U/L], ALT 45 U/L [nl 6-29 U/L]).

Conclusion: In a patient with a unilateral cortisol-producing adenoma, medical therapy with mifepristone was effective in reducing associated co-morbidities related to persistent hypercortisolemia, including reductions in weight and normalization of BP with discontinuation of antihypertensive medication. Additionally, successful management of CS led to marked improvement in the liver enzymes of a patient with long-standing NASH.

INTRODUCTION

- Nonalcoholic fatty liver disease is frequent among people with obesity, type 2 diabetes, insulin resistance, and dyslipidemia
- These metabolic aberrations are also common in Cushing's syndrome (CS), a complex, multisystemic condition resulting from prolonged exposure to cortisol
- Because of the metabolic aberrations associated with prolonged elevated cortisol in CS, researchers have investigated the contributions of cortisol to nonalcoholic steatohepatitis (NASH)
- Rockall et al confirmed hepatic steatosis in 20% of patients with active CS, using computed tomography (CT)[1]
 - There was a strong correlation between the presence of increased visceral fat and hepatic steatosis, as measured by CT[1]
- However, it is not known if successful treatment of CS would slow the progression to, and decrease the incidence of, NASH
- Mifepristone (Korlym®, Corcept Therapeutics) is a competitive glucocorticoid receptor antagonist approved for patients with all forms of CS who have diabetes or glucose intolerance and have failed or are not candidates for surgery
 - In patients with CS and type 2 diabetes or hypertension, significant clinical improvement was noted in 87% of patients treated with mifepristone in the 24-week SEISMIC trial, including significant reductions in hemoglobin A1c (HbA1c), fasting plasma glucose (FPG), weight, and diastolic blood pressure (BP)[2]
- We detail a patient with CS and NASH who saw a significant improvement in liver function tests (LFTs) after successful treatment of CS with mifepristone

CLINICAL PRESENTATION

- A 47-year-old woman with medical history of:
 - Hypertension (BP 142/90 mmHg)
 - Pre-diabetes (HbA1c 5.9%)
 - Dyslipidemia (total cholesterol 274 mg/dL, low-density lipoprotein cholesterol 203 mg/dL, high-density lipoprotein cholesterol 47 mg/dL, triglycerides 132 mg/dL)
 - Cholecystectomy (due to chronic cholecystitis and cholelithiasis) and liver biopsy showing steatohepatitis and diffuse steatosis in November 2012
- A routine CT scan for evaluation of nephrolithiasis found a left adrenal adenoma
- Medications
 - Amlodipine/olmesartan medoxomil 10/20 mg, once daily
 - Rosuvastatin calcium 10 mg once daily

Assessments

- A CT scan in May 2014 revealed a left adrenal adenoma measuring 2.9 x 1.9 x 2.5 cm with benign radiologic characteristics (**Figure 1**)

Figure 1. Left adrenal adenoma measuring 2.9 x 1.9 x 2.5 cm

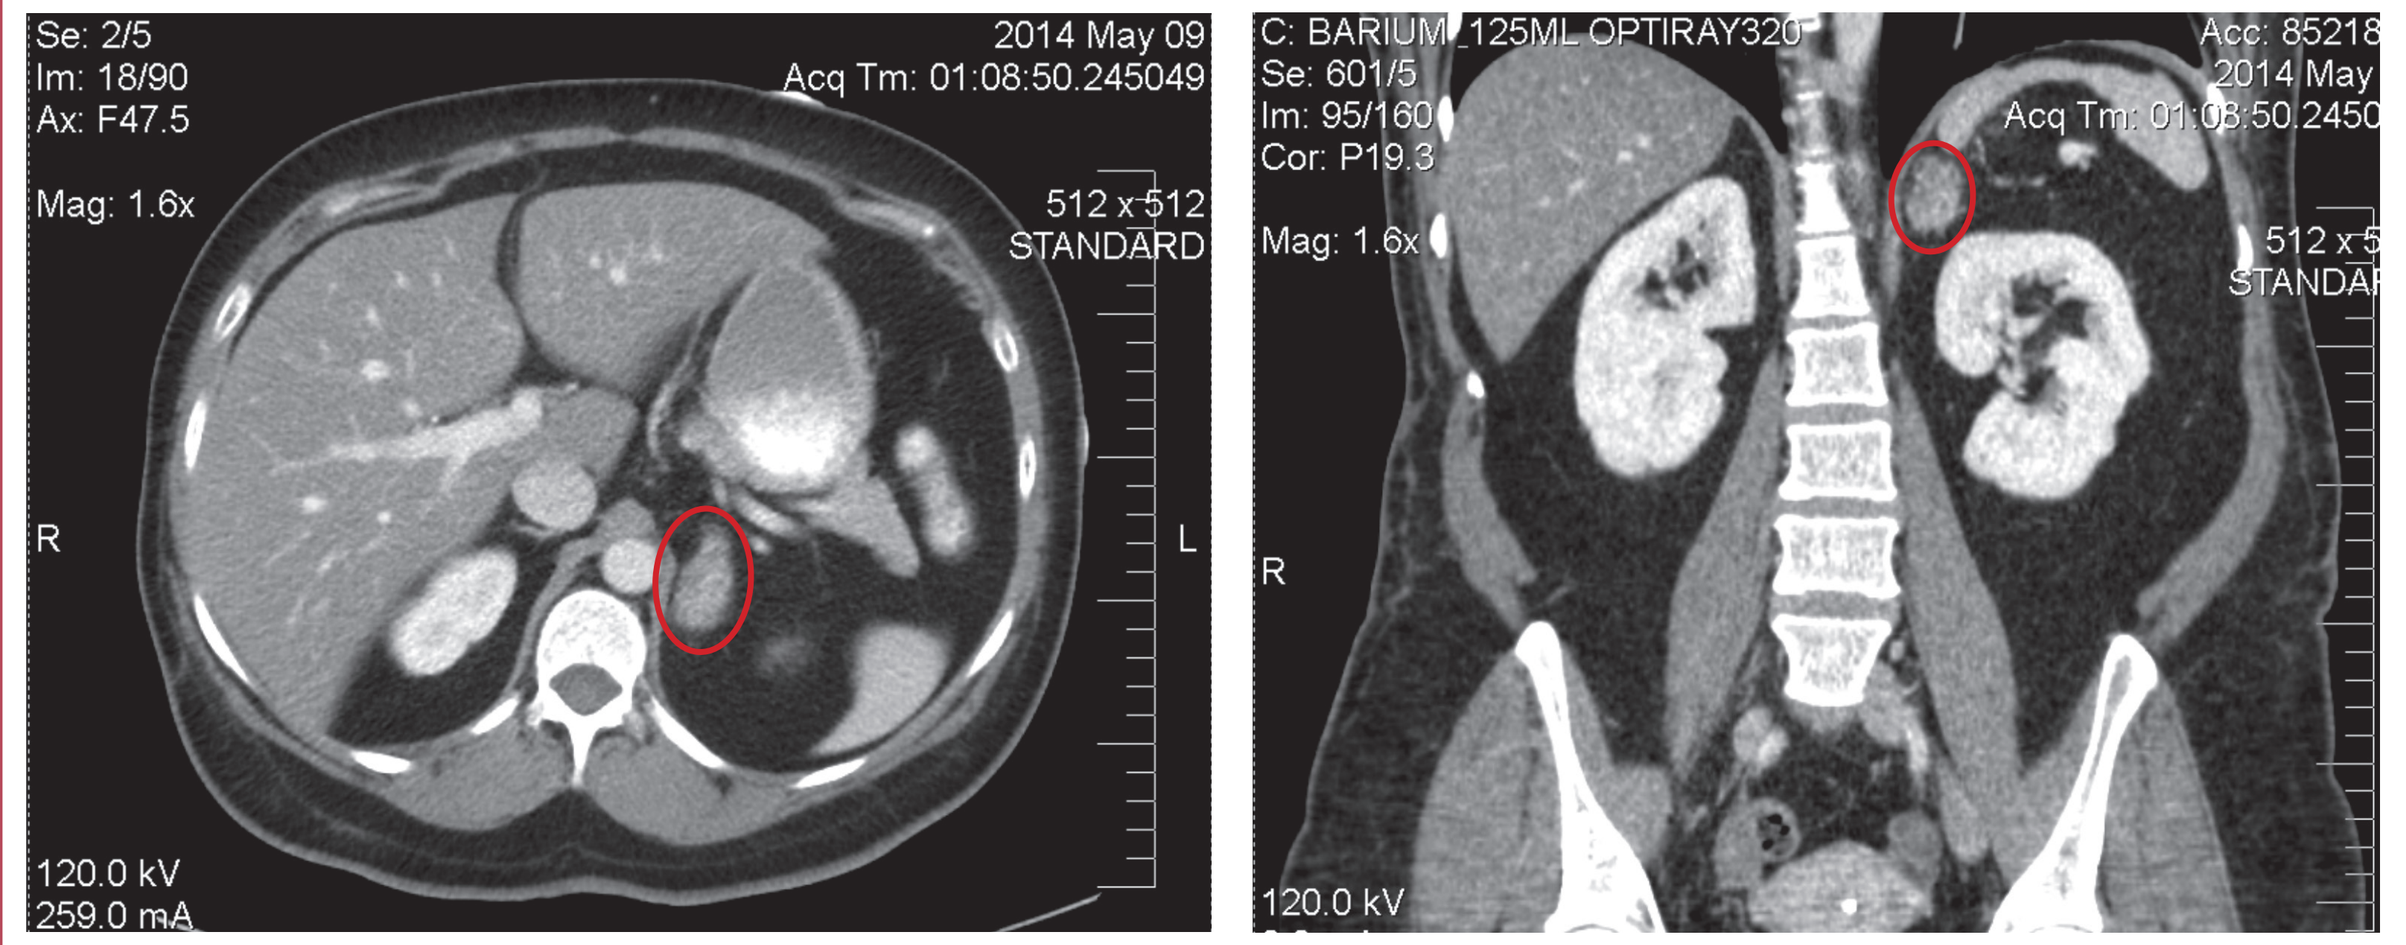
Se: 2/5
Im: 18/90
Ax: F47.5
Mag: 1.6x
2014 May 09
Acq Tm: 01:08:50.245049
512 x 512
STANDARD
R
L
120.0 kV
259.0 mA
C: BARIUM 125ML OPTIRAY320
Se: 601/5
Im: 95/160
Cor: P19.3
Mag: 1.6x
R
120.0 kV

- There were no overt features of CS, but the patient complained of persistent abnormal weight gain (weight 178 pounds, BMI 30.6 kg/m^2) with central obesity
- Biochemical testing was negative for:
 - Pheochromocytoma
 - Primary aldosteronism
- Laboratory parameters
 - Autonomous cortisol secretion showed a mildly elevated urinary free cortisol (UFC) of 62.6 µg/dL (normal range: 4-50 µg /mL)
 - Failure to suppress cortisol on successive 1-mg dexamethasone suppression tests (13.2 µg/dL and 12.5 µg/dL)
 - Elevated midnight salivary cortisol of 0.19 µg/dL (normal range: <0.09 µg/mL)
 - Suppressed ACTH on successive tests (<5 pg/mL; normal range: 5-45 pg/mL)
 - Low dehydroepiandrosterone sulfate (DHEA-S) of 20 µg/dL (normal range: 42-290 µg/dL)
 - Baseline LFTs were elevated
 - ALT was 232 U/L (normal range: 2-60 U/L); 3.9-fold greater than the upper limit of normal (ULN)
 - AST was 110 U/L (normal range: 2-40 U/L); 2.8-fold greater than the ULN

TREATMENT COURSE

- Patient declined adrenalectomy but was agreeable to medication
- Mifepristone was initiated at 300 mg daily and increased to 900 mg daily over 20 weeks

Treatment Outcomes

- Marked improvement in LFTs was observed during mifepristone therapy (**Figure 2**)
 - At week 24:
 - More than a 7-fold reduction of ALT to 32 U/L (normal range: 6-29 U/L)
 - A 5-fold reduction of AST to 22 U/L (normal range: 10-35 U/L)

Figure 2. Change in liver enzymes over time after initiating mifepristone

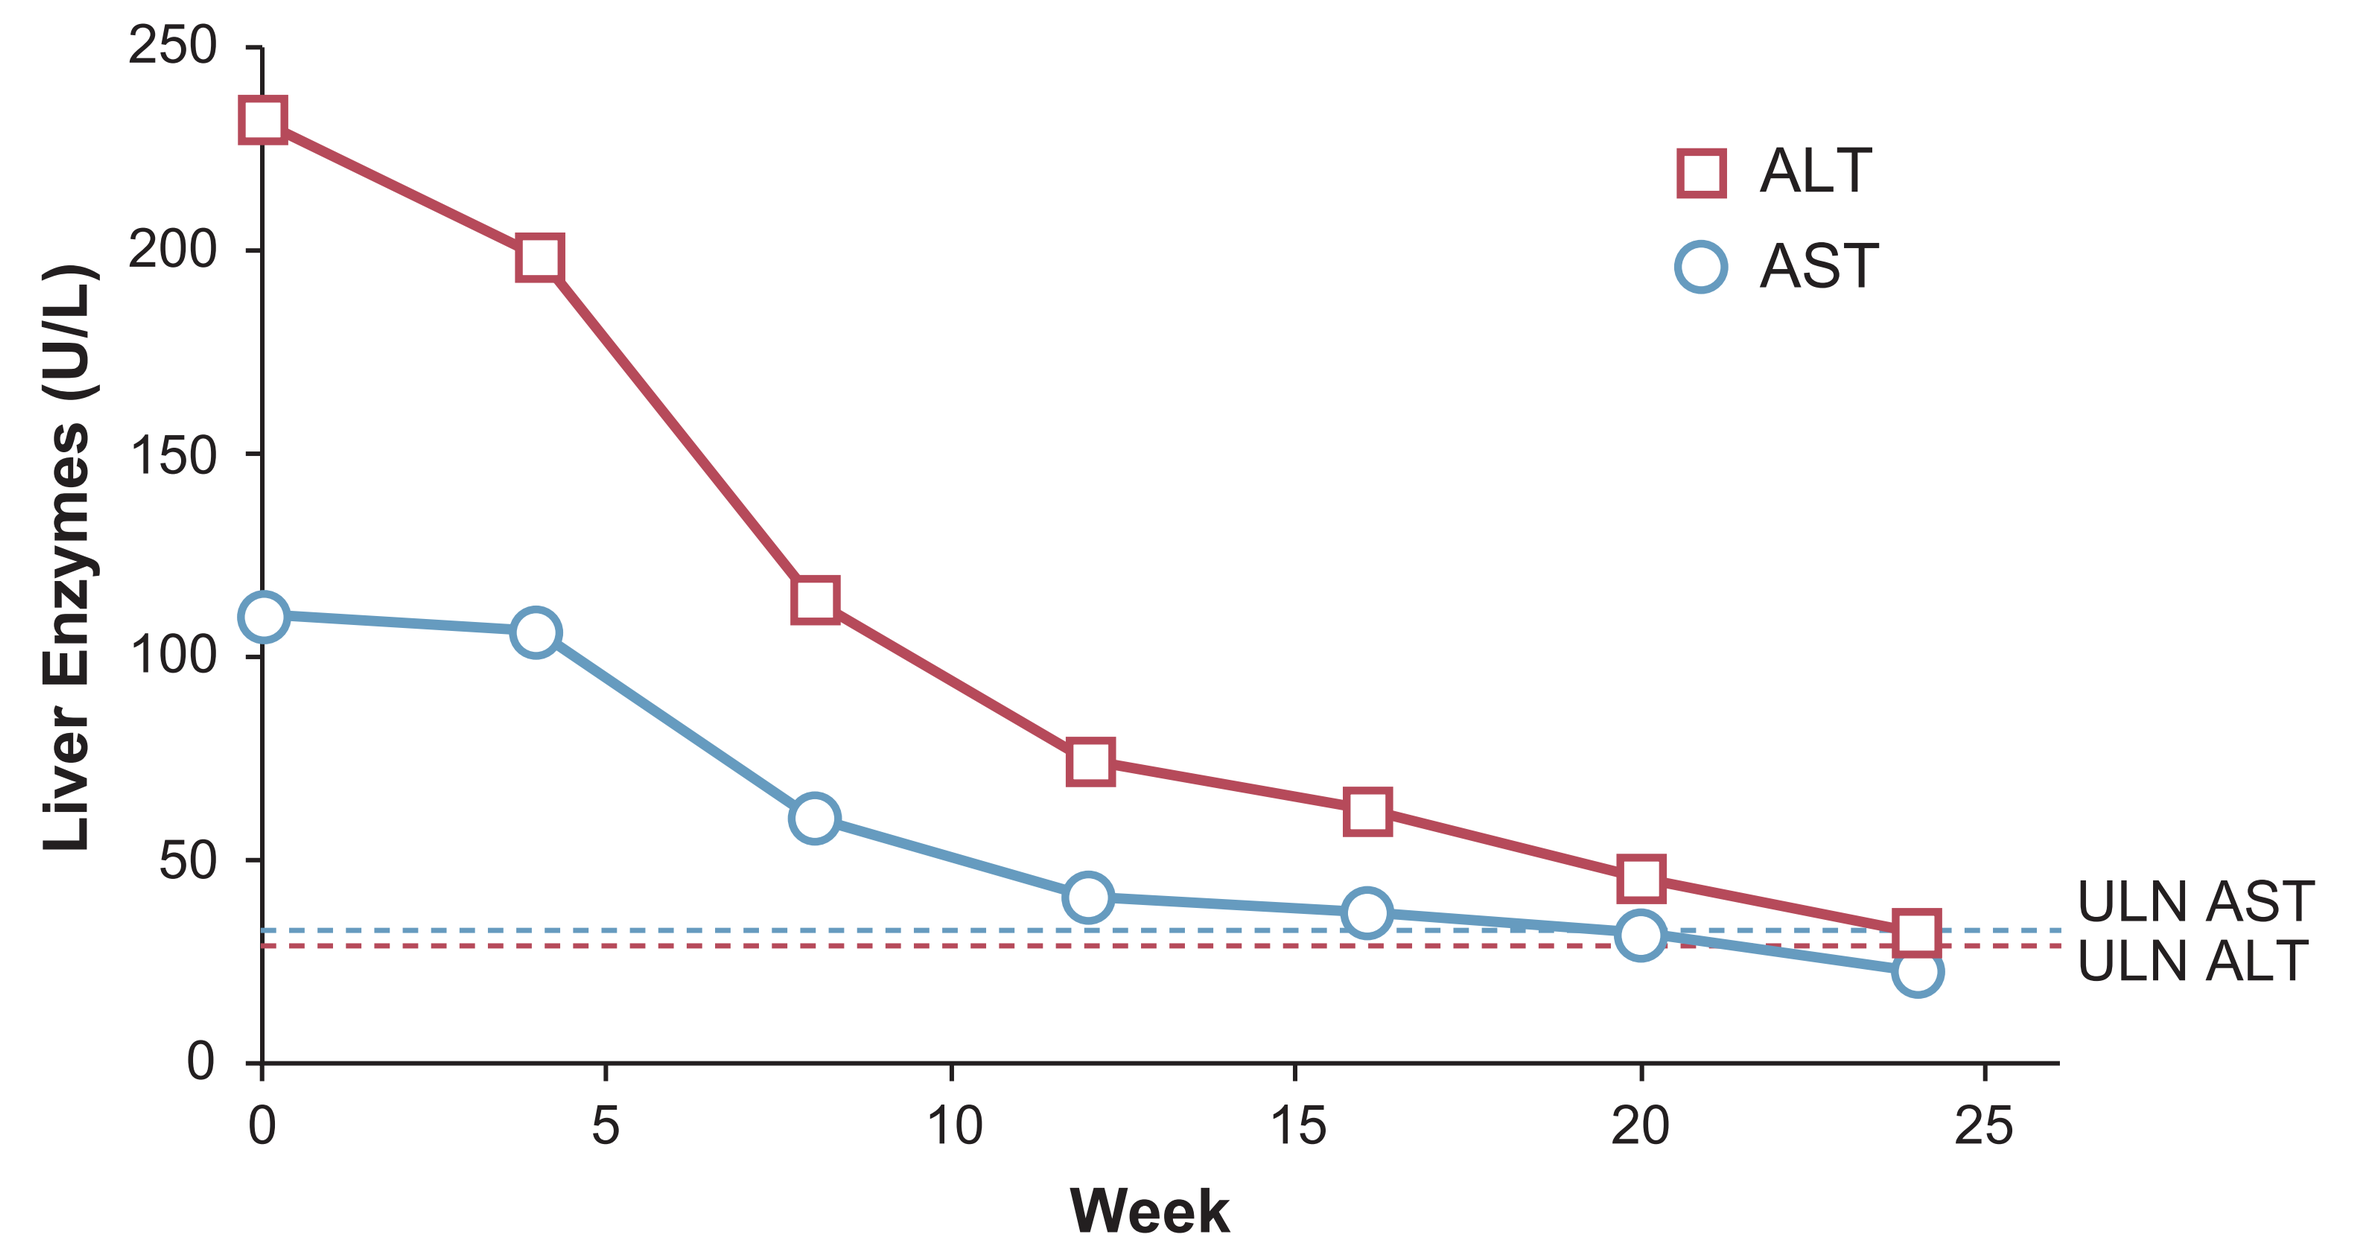
Liver Enzymes (U/L)
250
200
150
100
50
0
0
5
10
15
20
25
Week
ALT
AST
ULN AST
ULN ALT

ULN = upper limit of normal.

- Other associated outcomes
 - At week 4, antihypertensive medications were discontinued
 - At week 20:
 - BP was stable at 120/82 mmHg (**Figure 3**)
 - Weight had decreased from 178 to 160 lbs, corresponding to a BMI change from 30.6 to 27.5 kg/m^2 (**Figure 4**)
 - By week 24, ACTH (**Figure 5**) and DHEA-S returned to normal

Figure 3. Change in blood pressure over time after initiating mifepristone

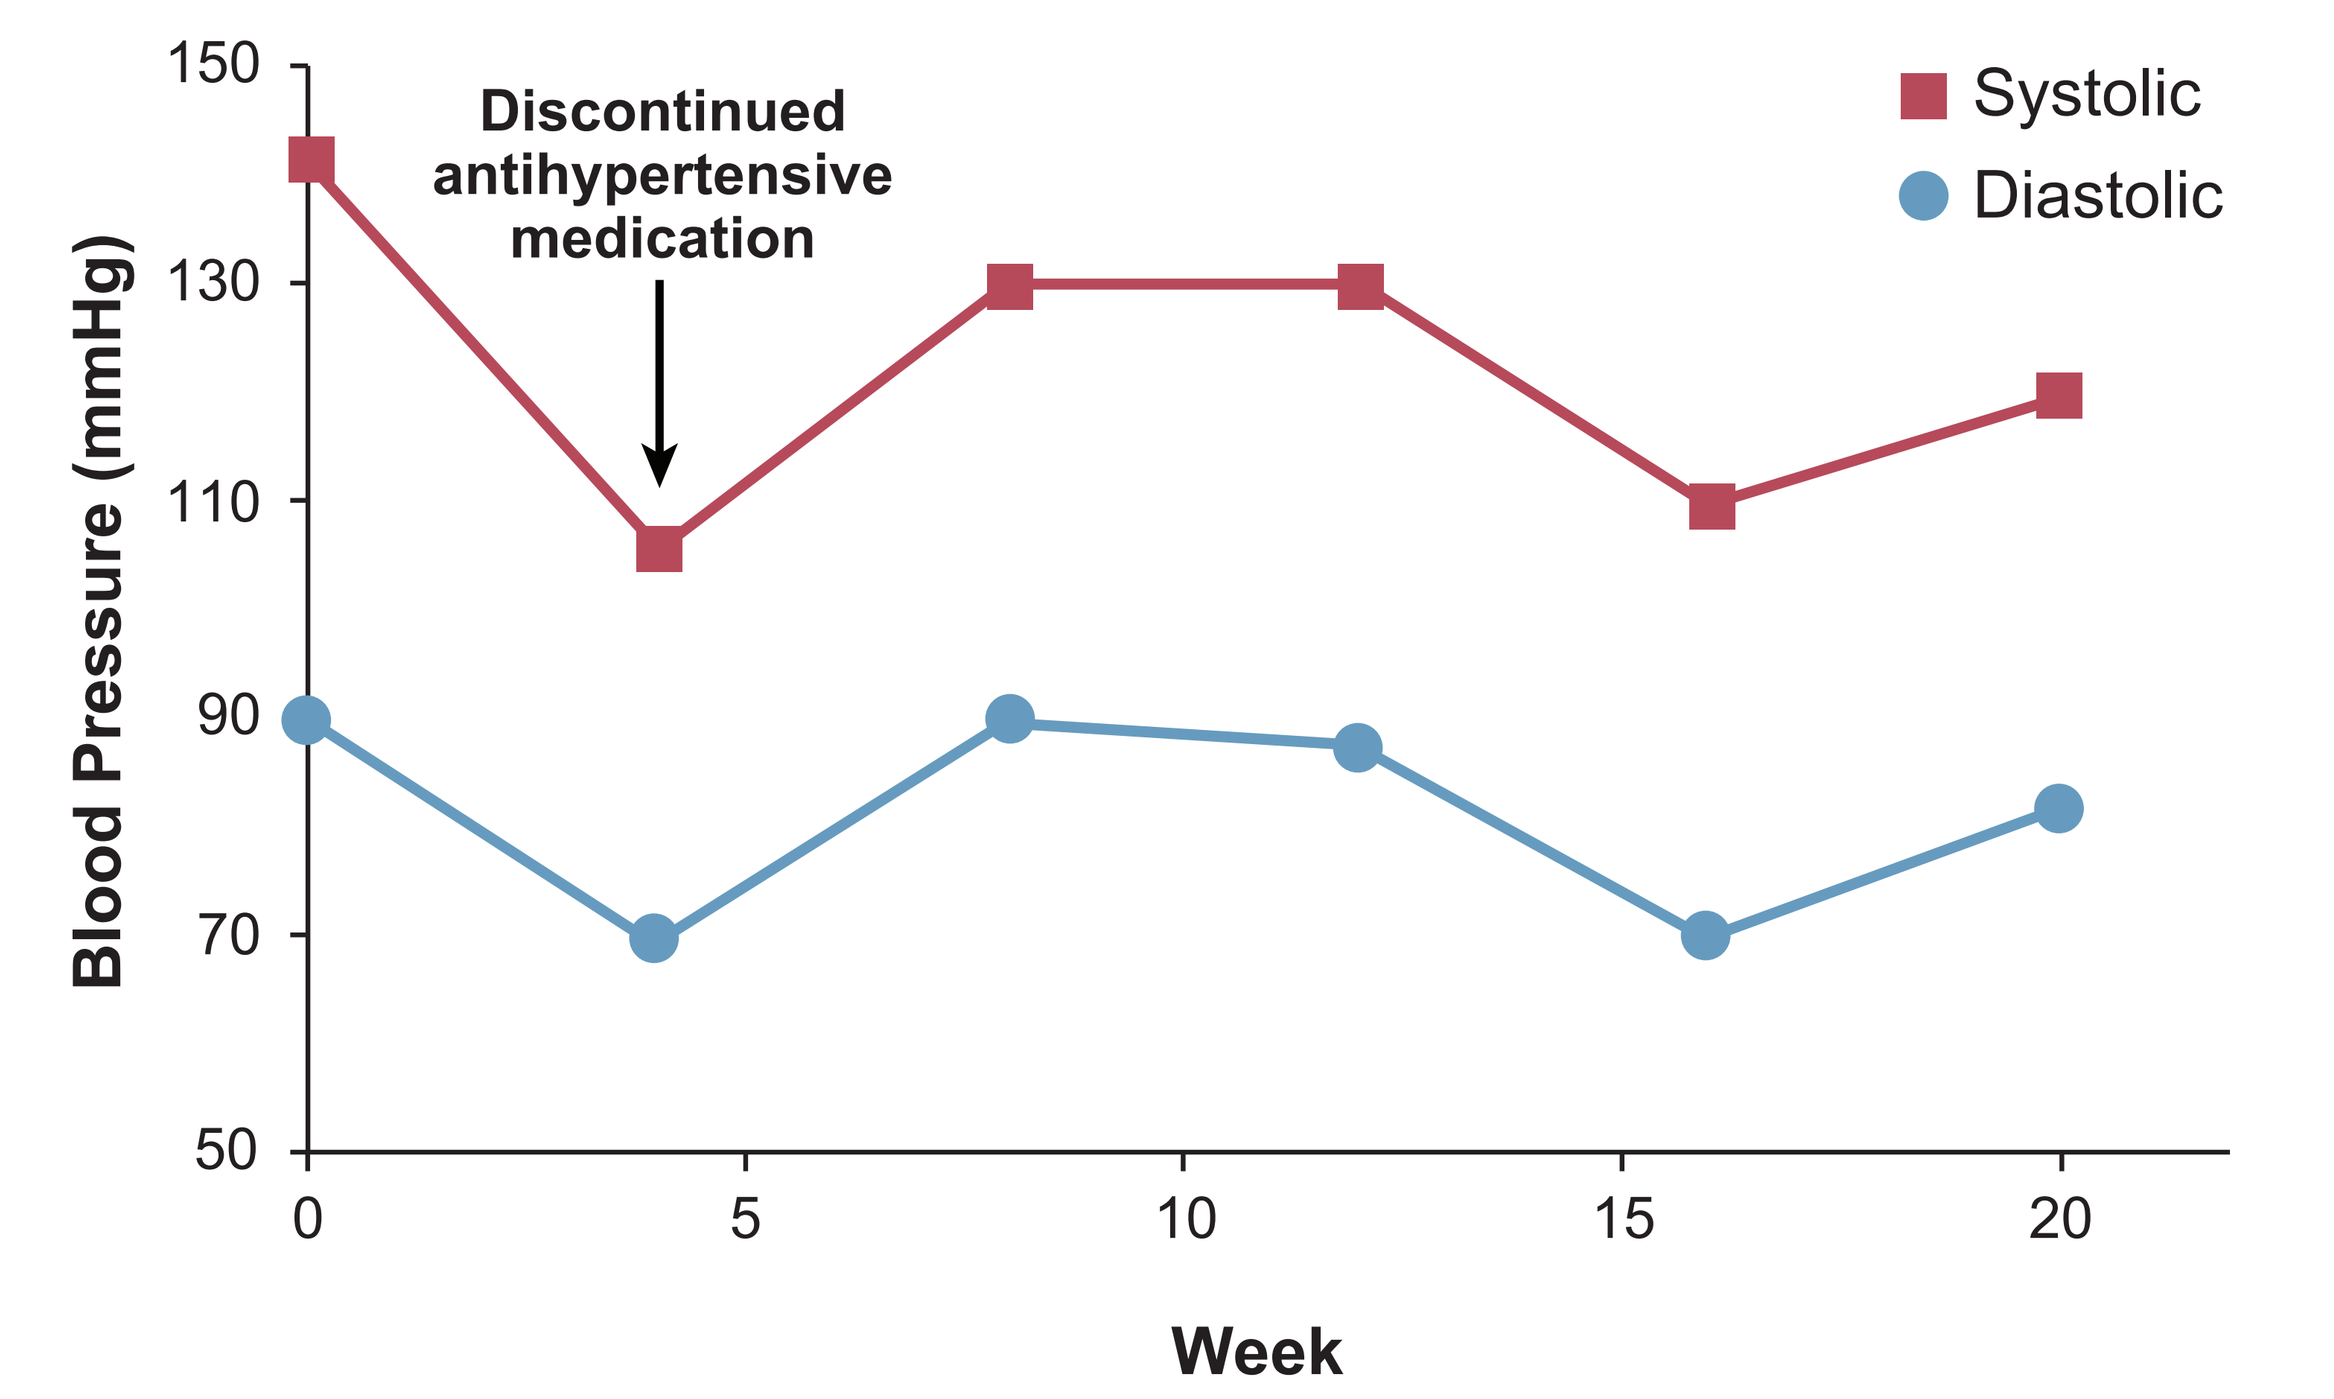
Blood Pressure (mmHg)
150
130
110
90
70
50
0
5
10
15
20
Week
Discontinued antihypertensive medication
Systolic
Diastolic

Figure 4. Change in weight over time after initiating mifepristone

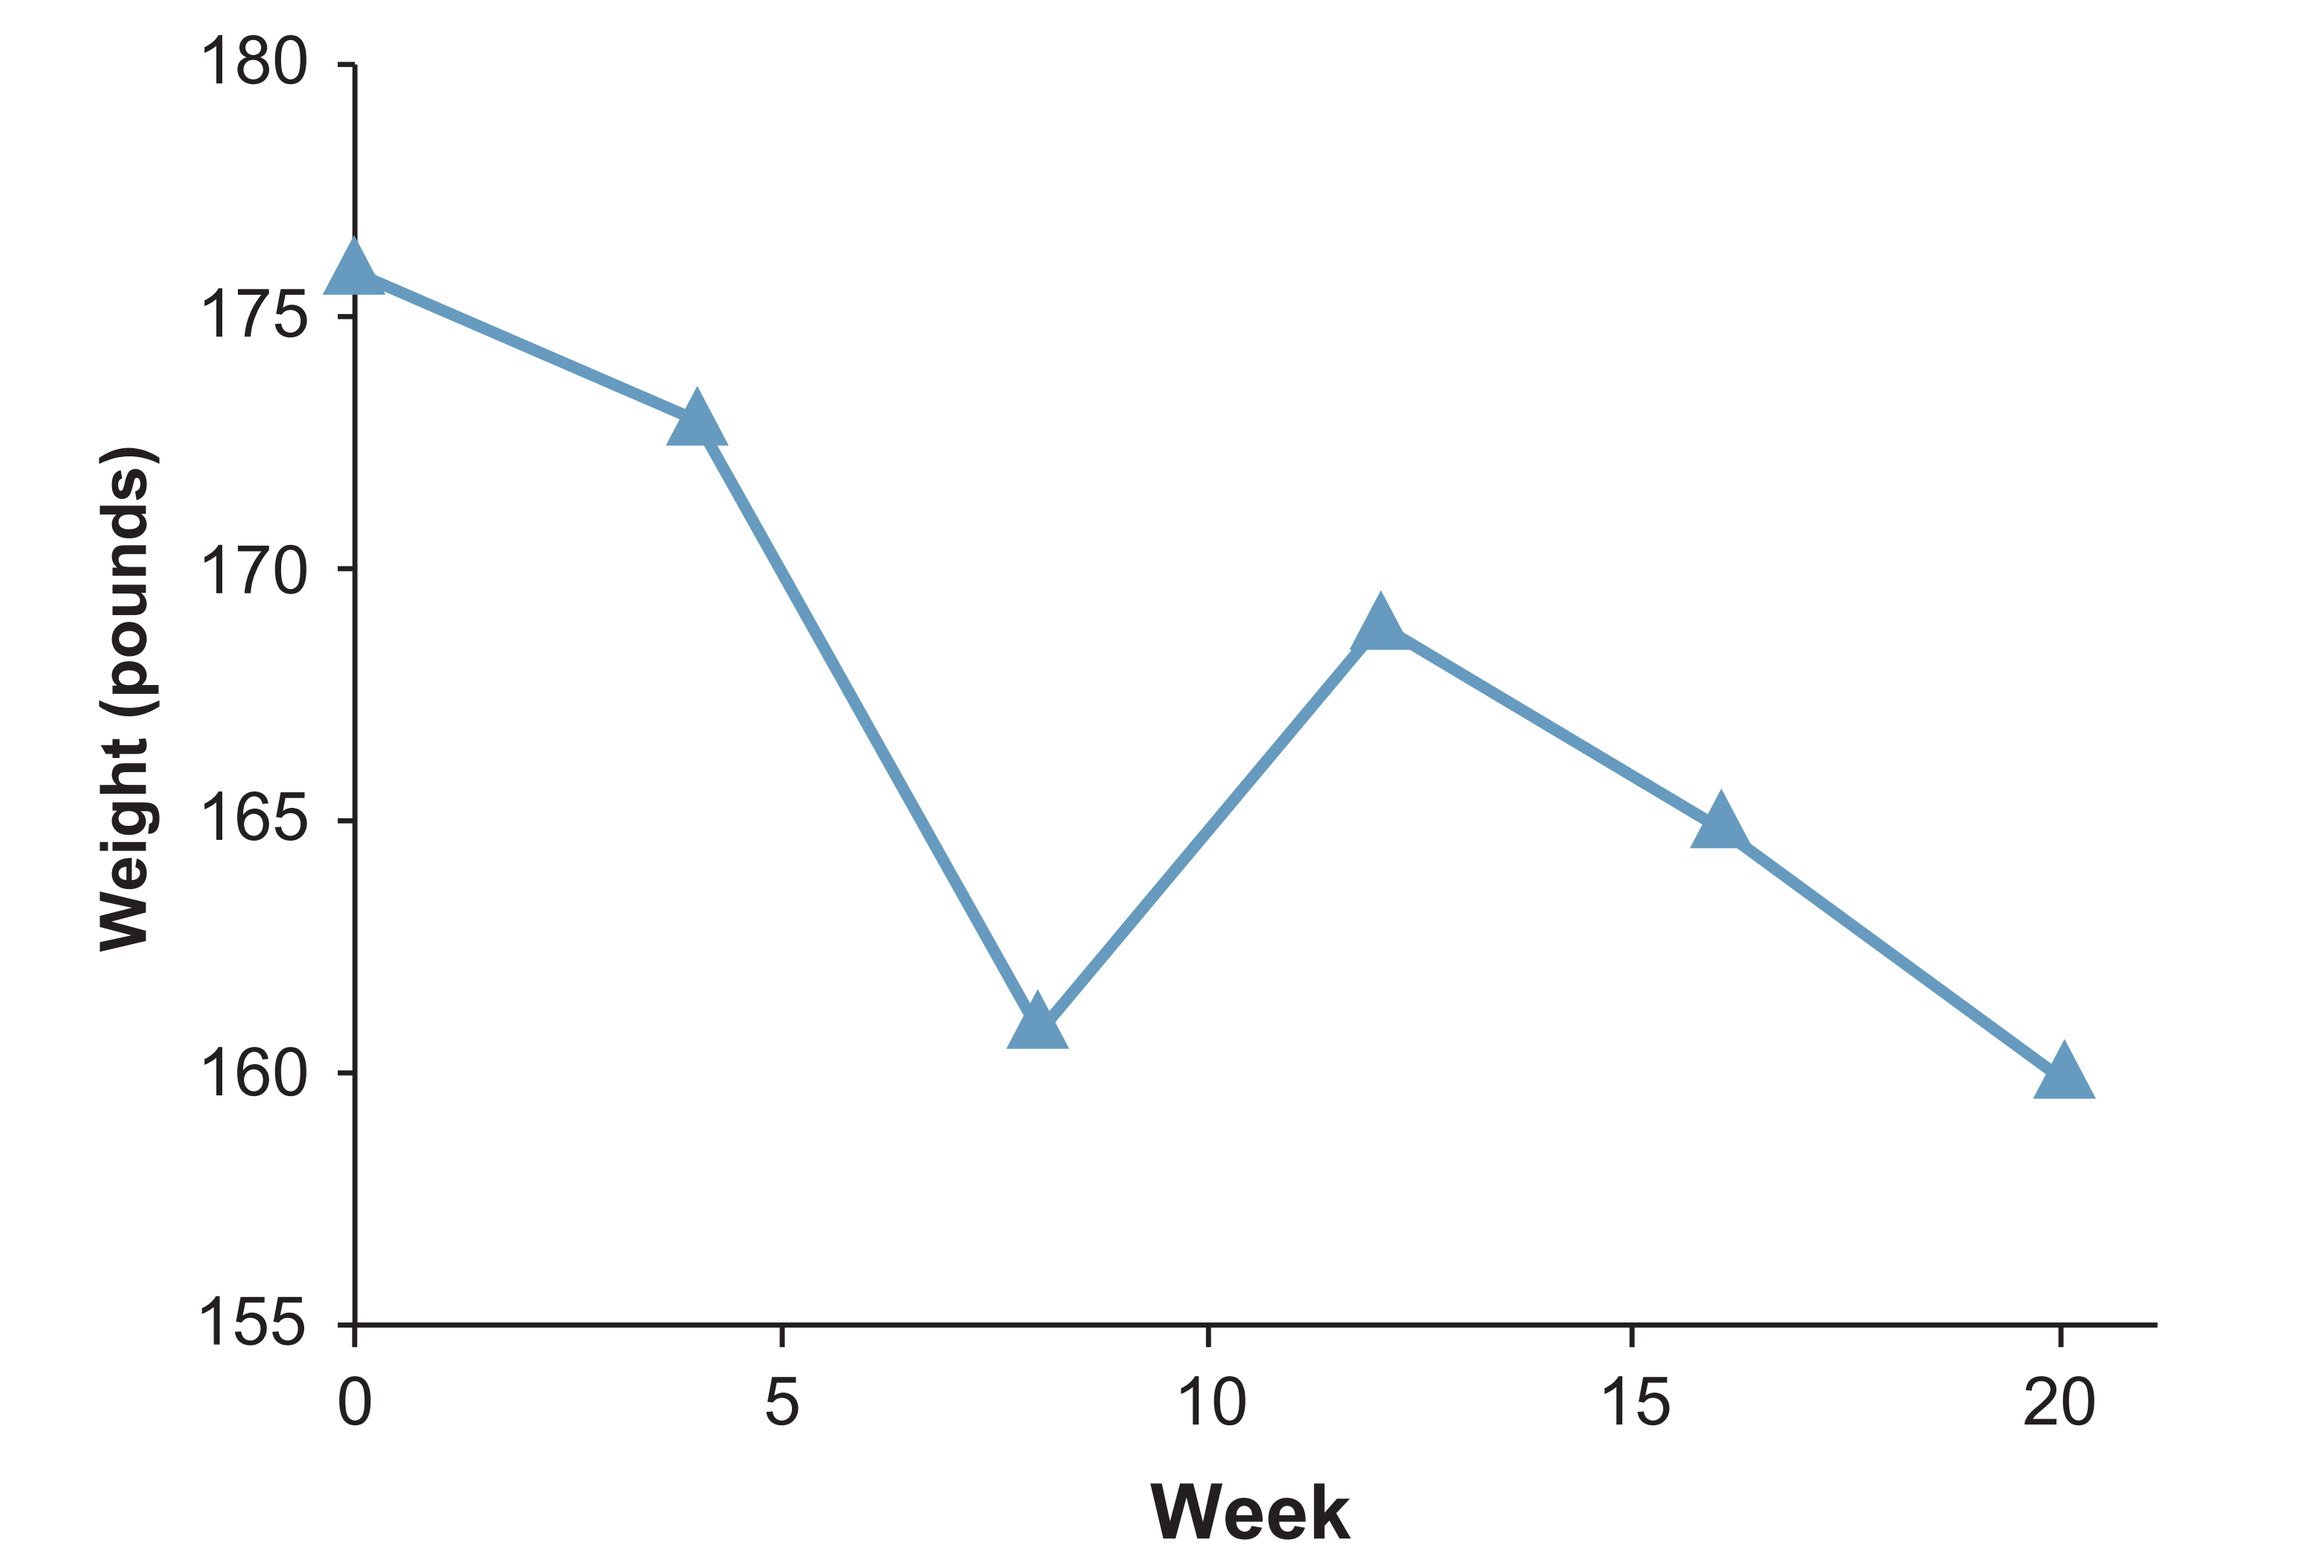
Weight (pounds)
180
175
170
165
160
155
0
5
10
15
20
Week

Figure 5. Change in ACTH over time after initiating mifepristone

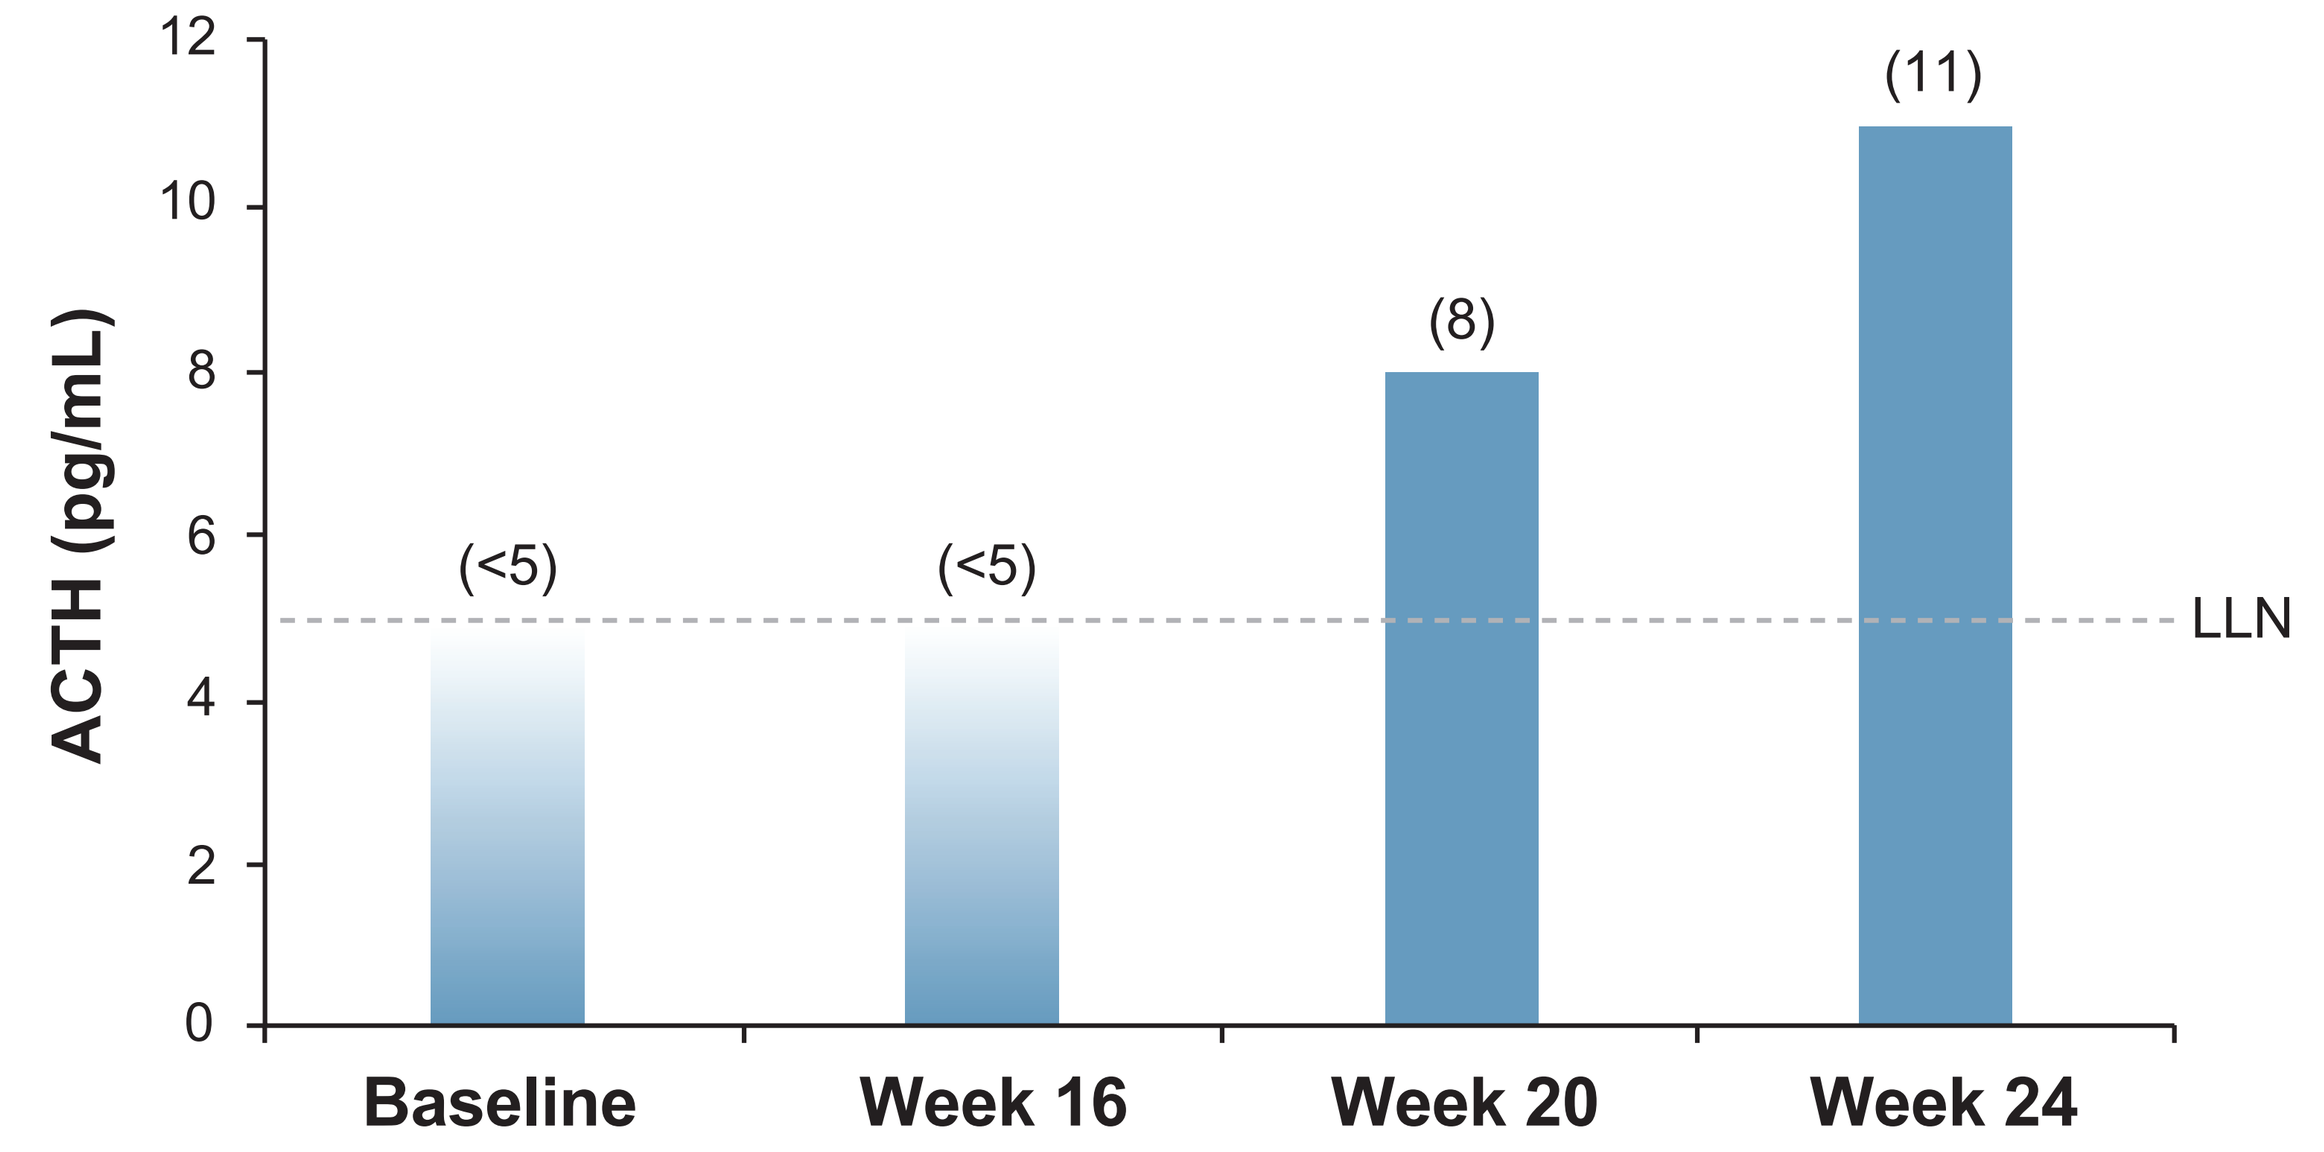
ACTH (pg/mL)
12
10
8
6
4
2
0
(<5)
(<5)
(8)
(11)
LLN
Baseline
Week 16
Week 20
Week 24

ACTH = adrenocorticotropin; LLN = lower limit of normal.

SUMMARY AND CONCLUSIONS

- 20 weeks of treatment with mifepristone led to marked improvement in liver enzymes in a patient with unilateral cortisol-producing adenoma and long-standing NASH
- Reductions in associated co-morbidities related to persistent hypercortisolemia included
 - Reduction in weight
 - Normalized BP with discontinuation of antihypertensive medication
 - Increased ACTH and DHEA-S to normal levels
- In this case, management of CS with mifepristone was successful

REFERENCES

1. Rockall AG, et al. *Eur J Endocrinol.* 2003 149:543-548
2. Fleseriu M, et al. J *Clin Endocrinol Metab.* 2012 97:2039-2049

ACKNOWLEDGEMENT

Funding for development, editorial, design, and production support was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors contributed to the development of the poster, reviewed the material, and provided approval of the final version.

DISCLOSURE

ER: Nothing additional to disclose.

ML: Employee, Corcept Therapeutics.

Presented at ENDO 2015 • San Diego, California • March 5-8, 2015