LBS-061

Treatment of Ectopic Cushing's Syndrome With Mifepristone While Pending Tumor Localization

M. Fariba Rahnema, MD[1]; Dolores Chavez, DHA-C, MBA, MT(ASCP)[2]; Dat Nguyen, PharmD[2]

[1]Valley Endocrinology, Las Vegas, NV; [2]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: The recognition and treatment of Cushing's syndrome (CS) can be challenging. Diagnostic testing and tumor localization (especially ectopic) can be a lengthy and complex process. In the interim, management of the manifestations of CS is needed.

We detail the successful treatment with mifepristone (MIFE, Korlym®, Corcept Therapeutics), a competitive glucocorticoid receptor antagonist, in a patient suspected of ectopic CS without localization.

Clinical Case: A 53-yr-old woman with hypogammaglobulinemia on IVIg presented in the ED with new-onset hypokalemia (2.2 mEq/L) and congestive heart failure. She had facial hirsutism, moon facies, lower extremities swelling, skin hyperpigmentation, headache, fatigue, easy bruising, and muscle weakness (requiring a cane) that started 3-4 mos before the ED visit. She also had resistant hypertension (144/90 mmHg), BMI 27.1 (134 lbs) and a recent diagnosis of diabetes (A1c 9%, fasting blood glucose [FBG] >200 mg/dL).

Abdominal CT: bilateral upper lung opacities, bilateral adrenal hyperplasia, multiple liver and ovarian cysts. CT angiography: multiple pulmonary nodules (15 mm right upper lobe (possible lung cancer), 28 mm left upper lobe, 13 mm left mid lung lobe). Brain MRI: diminutive pituitary; empty sella. Labs: abnormal AST 88 U/L, ALT 55 U/L; UFC 2378 µg/24h, AM cortisol 99 µg/dL, PM cortisol 106 µg/dL, successive ACTH values of 115 & 127 pg/mL.

Relevant medication history: levothyroxine 50 mg/d, metformin 100 mg BID, Humalog 4U before dinner, Levemir 10 U qhs. After the ER visit, furosemide and hydralazine were discontinued. Losartan 50 mg/d, aldactone 100 mg BID, and KCl 200 mEq/d were added to amlodipine 10 mg and atenolol 50 mg/d.

The tumor could not be localized but ectopic CS was suspected due rapidly evolving Cushing's with negative MRI/empty sella, severe hypokalemia, edema, high ACTH and cortisol.

MIFE was initiated at 300mg/d and titrated every 2 weeks to the current dose of 1200 mg/d. After 3 months: the patient weighed 121 lbs (BMI 23.9), stable BP (116/77 mmHg), FBG decreased (125 mg/dl), normal A1c (5.7%), normal LFTs (ASTs 16 U/L, 42 U/L) and normal K (4.7 mEq/L).

Several antidiabetic and antihypertensive meds were discontinued: Humalog, Levemir, atenolol, and losartan. KCl was reduced from 200 to 40 mEq/d. She reported experiencing pruritus during therapy.

Although still using a cane, the patient reported feeling more energetic (significant improvement in lower extremity edema), and more engaging with better concentration.

With stabilization, octreotide scan and additional MRIs are scheduled to localize the ectopic tumor.

Conclusion: The addition of mifepristone provided control and improvements in several manifestations of CS (A1c, weight, BP, LFTs, appearance) while eliminating the need for several antidiabetes and antihypertensive medications, representing a viable option for patient whom tumor localization is difficult.

INTRODUCTION

- Cushing's syndrome (CS) is a complex, multisystemic condition resulting from prolonged exposure to cortisol
 - CS is associated with substantial metabolic dysfunction, including weight gain, hypertension, insulin resistance, diabetes mellitus, and dyslipidemia
- The recognition and treatment of CS can be challenging
- Diagnostic testing and tumor localization (especially ectopic) can be a lengthy and complex process
- Therefore, management of the manifestations of CS is needed during the process
- We detail the successful treatment of a patient with suspected ectopic CS using mifepristone (Korlym®, Corcept Therapeutics), a competitive glucocorticoid receptor antagonist approved for patients with all forms of CS who have diabetes or glucose intolerance and have failed or are not candidates for surgery
 - In the 24-week SEISMIC study of patients with CS and type 2 diabetes or hypertension, significant clinical improvement was noted in 87% of patients treated with mifepristone, including significant reductions in hemoglobin A1c (HbA1c), fasting plasma glucose (FPG), weight, and diastolic blood pressure (BP)[1]

CLINICAL PRESENTATION

- In June 2014, a 53-year-old woman with hypogammaglobulinemia treated with intravenous IgG (IVIg) presented in the emergency department (ED) with CS-like symptoms (listed below), new-onset hypokalemia with serum potassium of 2.2 mEq/L (normal range 3.5-5.0 mEq/L), and congestive heart failure

Additional Details

- Facial hirsutism
- Moon facies
- Lower extremity edema
- Skin hyperpigmentation
- Headache
- Fatigue
- Bruising easily
- Muscle weakness (requiring a cane, started 3-4 months before the ED visit)
- Resistant hypertension, 144/90 mmHg
- Body mass index (BMI), 27.1 kg/m^2 (134 lbs)
- Recent diagnosis of diabetes
 - Hemoglobin A1c (HbA1c), 9%
 - Fasting plasma glucose (FPG), >200 mg/dL

Imaging (Figure 1)

- Computed tomography (CT)
 - Bilateral upper lung opacities
 - Bilateral adrenal hyperplasia
 - Multiple liver and ovarian cysts
- CT angiography
 - Multiple pulmonary nodules (15 mm right upper lobe [possible lung cancer], 28 mm left upper lobe, and 13 mm left mid lung lobe)
- Brain magnetic resonance imaging (MRI)
 - Diminutive pituitary
 - Empty sella

Figure 1A-C. Lung, adrenals, and pituitary

A. Lung nodules B. Bilateral hyperplastic adrenals C. Pituitary empty sella

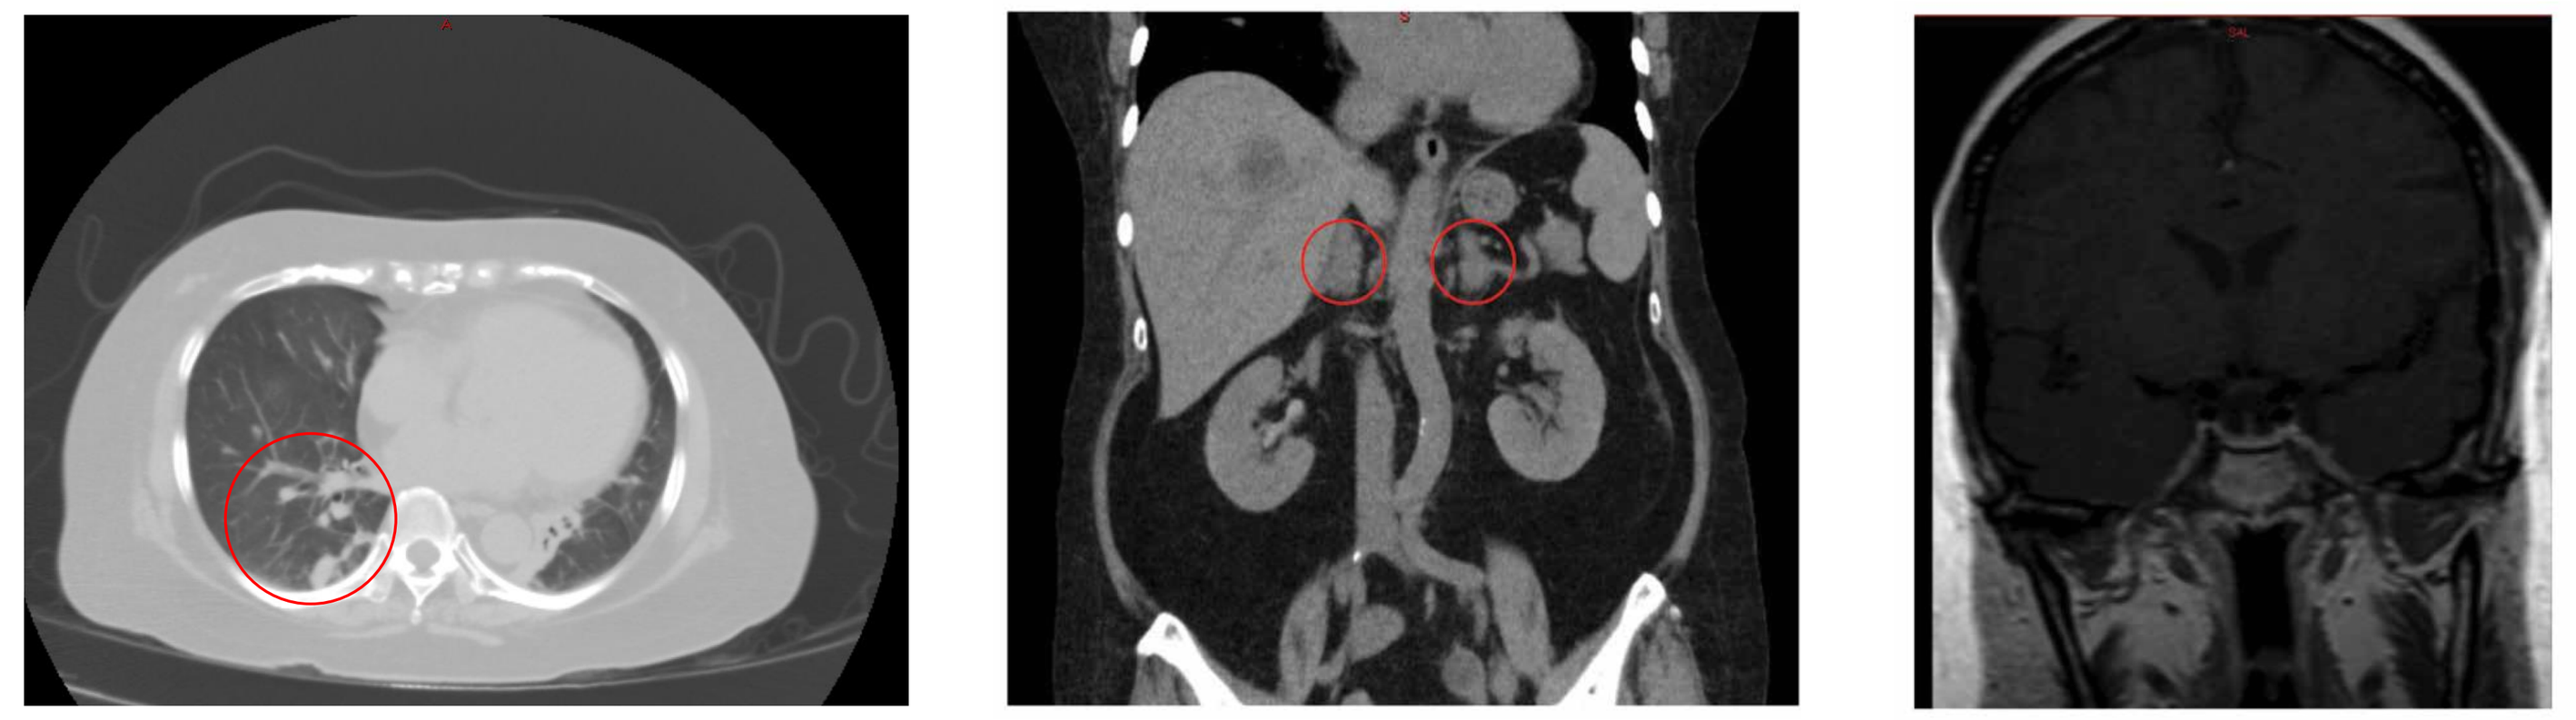

Laboratory Results

- High AST of 88 U/L (normal range, 10-35 U/L)
- High ALT of 55 U/L (normal range, 6-29 U/L)
- High urine free cortisol (UFC) level of 2378 µg/24h (normal range, 4-50 µg/24h determined by tandem mass spectrometry)
- High morning cortisol level of 99 µg/mL (normal range, 4-22 µg/dL)
- High evening cortisol level of 106 µg/mL (normal range, 2-17 µg/dL)
- High successive adrenocorticotropin (ACTH) levels of 115 and 127 pg/mL (normal range, 6-50 pg/mL)
- High testosterone level of 77 ng/dL (normal range, 2-45 ng/dL)
- High dehydroepiandrosterone sulfate (DHEA-S) level of 470 µg/dL (normal female range: 22-266 µg/dL)
- High chromogranin A level of 21.5 ng/mL (normal range: 1.9-15 ng/mL)

Medication

- Relevant medication history (see **Table 1** for full medication history)
 - Levothyroxine 50 µg once daily
 - Metformin 100 mg twice daily
 - Lispro insulin 4 units before dinner
 - Detemir insulin 10 units before bed
- After the ED visit by endocrinologist
 - Discontinued:
 - Furosemide
 - Hydralazine
 - Added:
 - Losartan 50 mg once daily
 - Spironolactone 100 mg twice daily
 - KCl 200 mEq once daily
 - Amlodipine 10 mg daily
 - Atenolol 50 mg once daily

Table 1. Medication use before and after mifepristone[a]

Medication	Dose and frequency	Before mifepristone	After mifepristone
	Anti-diabetes medications		
Metformin	1000 mg twice daily	Yes	Yes
Lispro insulin	4 units before bed	Yes	No
Detemir insulin	10 units before bed	Yes	No
Aspart insulin	Sliding scale	Yes	No
	Anti-hypertensive medications		
Hydralazine	10 mg twice daily	Yes	No
Atenolol	25 mg once daily	Yes	No
Furosemide	40 mg once daily	Yes	No
Losartan	50 mg once daily	Yes	No
Amlodipine	10 mg once daily	Yes	Yes
	Hypokalemia medications		
KCl	200 mEq twice daily then decreased to 40 mEq twice daily	No	Decreased
Spironolactone	100 mg twice daily then decreased to 50 mg twice daily	Yes	Decreased

[a]Mifepristone first 2 weeks, 300 mg once daily; weeks 3 and 4, 600 mg daily; weeks 5 and 6, 900 mg daily; then 1200 mg once daily.

Diagnosis

- The tumor could not be localized but ectopic CS was suspected due to rapidly evolving symptoms, with empty sella, severe hypokalemia, edema, and high levels of ACTH and cortisol

TREATMENT WITH MIFEPRISTONE

- Mifepristone was initiated at 300 mg once daily and titrated every 2 weeks to the current dose of 1200 mg once daily

Clinical Outcomes

- Several laboratory parameters (weight, BMI, BP, HbA1c, FPG, AST, ALT, and potassium) improved (**Table 2**)
- In addition, several antidiabetic and antihypertensive medications were discontinued (**Table 1**):
 - Lispro insulin, detemir insulin, spironolactone (initiated during ED visit), and losartan (initiated during ED visit)
- KCl was reduced from 200 (initiated during ED visit) to 40 mEq/d
- Lower extremity edema improved
- Facial redness and peri-orbital edema improved
- Although still using a cane, the patient reported she was less dependent when rising from a chair (proximal myopathy improving), feeling more energetic and engaging, and less depressed; she also stated she had better concentration
- Patient reported experiencing pruritus during therapy

Table 2. Various laboratory measurements after 3 months of mifepristone therapy[a]

Parameter	Before mifepristone	After mifepristone
Weight, lbs	134	121
BMI, kg/m^2	27.1	23.9
BP, mmHg	140/90	116/77
FPG, mg/dL	>200	125
HbA1c, %	9.0	5.7
AST, U/L	88	16
ALT, U/L	55	42
Potassium, mEq/L	2.2	4.7

[a]Mifepristone first 2 weeks, 300 mg once daily; weeks 3 and 4, 600 mg daily; weeks 5 and 6, 900 mg daily; then 1200 mg once daily.

BMI=body mass index; BP=blood pressure; FPG=fasting plasma glucose; HbA1c=hemoglobin A1c.

Follow-Up

- CT angiogram
 - 15 mm right upper lobe (worrisome for lung cancer), 28 mm left upper lobe, and 13 mm mild lung nodule
 - Repeated chest CT with and without contrast reported thoracic vertebral lesion
 - Metastatic lesion cannot be excluded because lung lesion is smaller in size; did not recommend biopsy
 - Stable liver cysts
- Lumbar spine MRI
 - Chronic L2-L4 compression fracture
 - No high-grade spinal stenosis
- Octreotide scan scheduled to localize the ectopic tumor

SUMMARY AND CONCLUSIONS

- Ectopic CS was suspected in a patient because CS-like symptoms continued to worsen but a tumor could not be localized
- Treatment of the CS symptoms was necessary while the tumor was localized
- After 3 months of treatment with mifepristone, improvements in several manifestations of CS (HbA1c, weight, BP, LFTs, appearance) were observed and several antidiabetes and antihypertensive medications were no longer needed
- Mifepristone represents a viable option for patient where tumor localization is difficult and other characteristics of CS are present

REFERENCE

1. Fleseriu M, et al. *J Clin Endocrinol Metab.* 2012;97:2039-49

ACKNOWLEDGEMENT

Funding for development, editorial, design, and production support was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors contributed to the development of the poster, reviewed the material, and provided approval of the final version.

DISCLOSURES

MFR: Nothing additional to disclose. DC: Employee, Corcept Therapeutics. DN: Employee, Corcept Therapeutics.

Presented at ENDO 2015 • San Diego, California • March 5-8, 2015