FRI-474

Short-Term Mifepristone Treatment Improved Preoperative Clinical Status in a High-Risk Patient With Unexpected Normalization of Liver Enzymes

Gaja Andzel, MD[1]; Hanh Ngoc Thi Lam[2]; Precious J. Lim, PhD[3]

[1]Beaver Medical Group, Yucaipa, CA; [2]University of California, Riverside, Riverside, CA; [3]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Management of preoperative blood pressure (BP) is essential in reducing perioperative risk of hypertensive crisis. This can be complicated in patients with Cushing's Syndrome (CS), as it is associated with increased mortality and multisystem morbidity.

We report a patient with a unilateral cortisol-secreting adenoma treated with mifepristone (MIFE, Korlym®, Corcept Therapeutics), a glucocorticoid receptor antagonist, prior to undergoing surgery.

Clinical Case: A 30 y/o Caucasian woman with a history of PCOS presented with secondary amenorrhea, new-onset edema, anxiety, fatigue, and insomnia. Despite lifestyle modifications, her BMI was 35.87. Physical exam showed abdominal striae, supraclavicular fat pad, easy bruising and moon facies.

History included resistant hypertension (182/112 mmHg with weekly ER visits for SBP spikes of 200 mmHg; on lisinopril, clonidine, and atenolol), non-alcoholic fatty liver disease (NAFLD) with elevated liver enzymes (AST 46 U/L [10-30 U/L] and ALT 94 U/L [6-29 U/L]), and dyslipidemia (Tchol 205 mg/dL, TG 203 mg/dL, HDL 44 mg/dL, LDL 120 mg/dL).

Worsening of her symptoms prompted hormonal testing for CS: elevated UFC 712.4 µg/d (4-50 µg/d), random cortisol of 26.4 µg/dL, 1-mg DST 20.6 µg/dL (<2 µg/dL), undetectable ACTH <5 pg/mL (6-50 pg/mL), low PRA 0.56 ng/mL/h (0.25-5.82 ng/nL/h), aldosterone <1ng/dL (≤ 28 ng/dL), K+ 4 mmol/L, low DHEA-S 14 µg/dL (18-391 µg/dL), and glucose abnormalities FPG 121 mg/dL (65-99 mg/dL), 2-hr oGTT 279 mg/dL (<140 mg/dL), HbA1C 6% (<5.7%).

CT scan of the abdomen revealed a right adrenal adenoma measuring 3.0 x 2.8 x 2.7 cm, HU 26, washout <50%, atrophic left adrenal gland, and diffuse fatty infiltration of the liver.

Labile BP elevations placed the patient at high risk for unilateral adrenalectomy.

A preoperative optimization plan was initiated to address the effects of CS. MIFE 300mg was initiated and later titrated to 1200mg.

During the 3-month therapy, marked improvements in glucose parameters were observed: FPG 83 mg/dL, 2-hr oGTT 145 mg/dL, HbA1C 5.6%. BP improved (124/90 mmHg) and three out of four BP meds were discontinued. Remarkably, her LFTs, after a transient increase, normalized (AST 22 U/L and ALT 25 U/L). With improvements in cardiometabolic status, she underwent unilateral adrenalectomy. There were no intra- or postoperative complications requiring extended hospitalization. Liver enzymes continued to be normal postoperatively.

Conclusion: Surgical intervention remains the first-line treatment for patients with CS. However, CS patients often present with high-risk comorbidities making them poor surgical candidates. MIFE provides an effective option to address the effects of CS while improving surgical eligibility, risk and outcomes. Additionally, normalization of liver enzymes was an unexpected benefit observed during MIFE therapy in a patient with a past NAFLD diagnosis.

INTRODUCTION

- Cushing's syndrome (CS) is a multisystemic condition resulting from prolonged exposure to cortisol
- Prolonged hypercortisolism is associated with cardiometabolic disorders including hypertension (HTN), obesity, diabetes mellitus (DM), insulin resistance, dyslipidemia, and hepatic steatosis (20% of CS patients)[1,2]
- Many of these disorders are also common in the general population, presenting a diagnostic challenge that results in undiagnosed CS for a protracted period[1,3]
- Surgical resection of the underlying tumor remains the definitive treatment
- Medical management has been used to treat hypercortisolism for improvement of metabolic indices in patients who are at high risk for surgery[4] or are not eligible for surgery[5]
- Mifepristone (Korlym®, Corcept Therapeutics) is a competitive glucocorticoid receptor antagonist approved for patients with all forms of CS who have DM or glucose intolerance and have failed surgery or are not candidates for surgery
 - 87% of patients treated with mifepristone in the 24-week SEISMIC trial had significant clinical improvement. Clinical improvements included significant reductions in hemoglobin A_{1C} (HbA_{1C}), fasting plasma glucose (FPG), weight, and diastolic blood pressure (BP)[6]
- We describe a patient with a history of HTN, morbid obesity, dyslipidemia, and non-alcoholic fatty liver disease (NAFLD) who underwent further examination, revealing a 3-cm adrenal adenoma that was autonomously secreting cortisol, and was treated with mifepristone prior to adrenalectomy

CLINICAL PRESENTATION

- A 30-year-old woman presented with:
 - New-onset edema
 - Secondary amenorrhea and acne (previously diagnosed as polycystic ovarian syndrome)
 - Anxiety
 - Fatigue
 - Insomnia
 - Weight gain despite lifestyle modifications (BMI 35.87)
- History included:
 - Resistant hypertension (182/112 with weekly emergency department visits for systolic BP spikes of 200 mmHg)
 - Dyslipidemia (total cholesterol 205 mg/dL, triglycerides 203 mg/dL, HDL 44 mg/dL, LDL 120 mg/dL)
- Past diagnosis of NAFLD:
 - AST 46 U/L, 1.5-fold greater than upper limit of normal (ULN)
 - ALT 94 U/L, 3.2-fold greater than ULN

Assessments

- Physical examination:
 - Abdominal striae
 - Supraclavicular fat pad
 - Easy bruising
 - Moon facies
- Laboratory parameters:
 - Elevated UFC: 712 µg/day (normal range 4-50 µg/day)
 - Failure to suppress on 1-mg dexamethasone: 20.6 µg/dL (normal level 1.8 µg/dL)
 - Random cortisol: 26.4 µg/dL
 - Suppressed ACTH: <5 pg/mL (normal range 6-50 pg/mL)
 - Low plasma renin activity: 0.56 ng/mL/h (normal range 0.25-5.82 ng/mL/h)
 - Low DHEA-S: 14 µg/dL (normal range 18-391 µg/dL)
 - Glucose abnormalities:
 - FPG: 121 mg/dL (normal range 65-99 mg/dL)
 - 2-hour oGTT: 279 mg/dL (normal level <140 mg/dL)
 - HbA_{1C}: 6% (normal <5.7%)
- Computed tomography (CT) scan of the abdomen (**Figure 1**) revealed:
 - Right adrenal adenoma measuring 3.0 x 2.8 x 2.7 cm, Hounsfield unit 26, washout <50%
 - Atrophic left adrenal gland
 - Diffuse fatty infiltration of the liver

Figure 1. Abdominal CT Scan Images

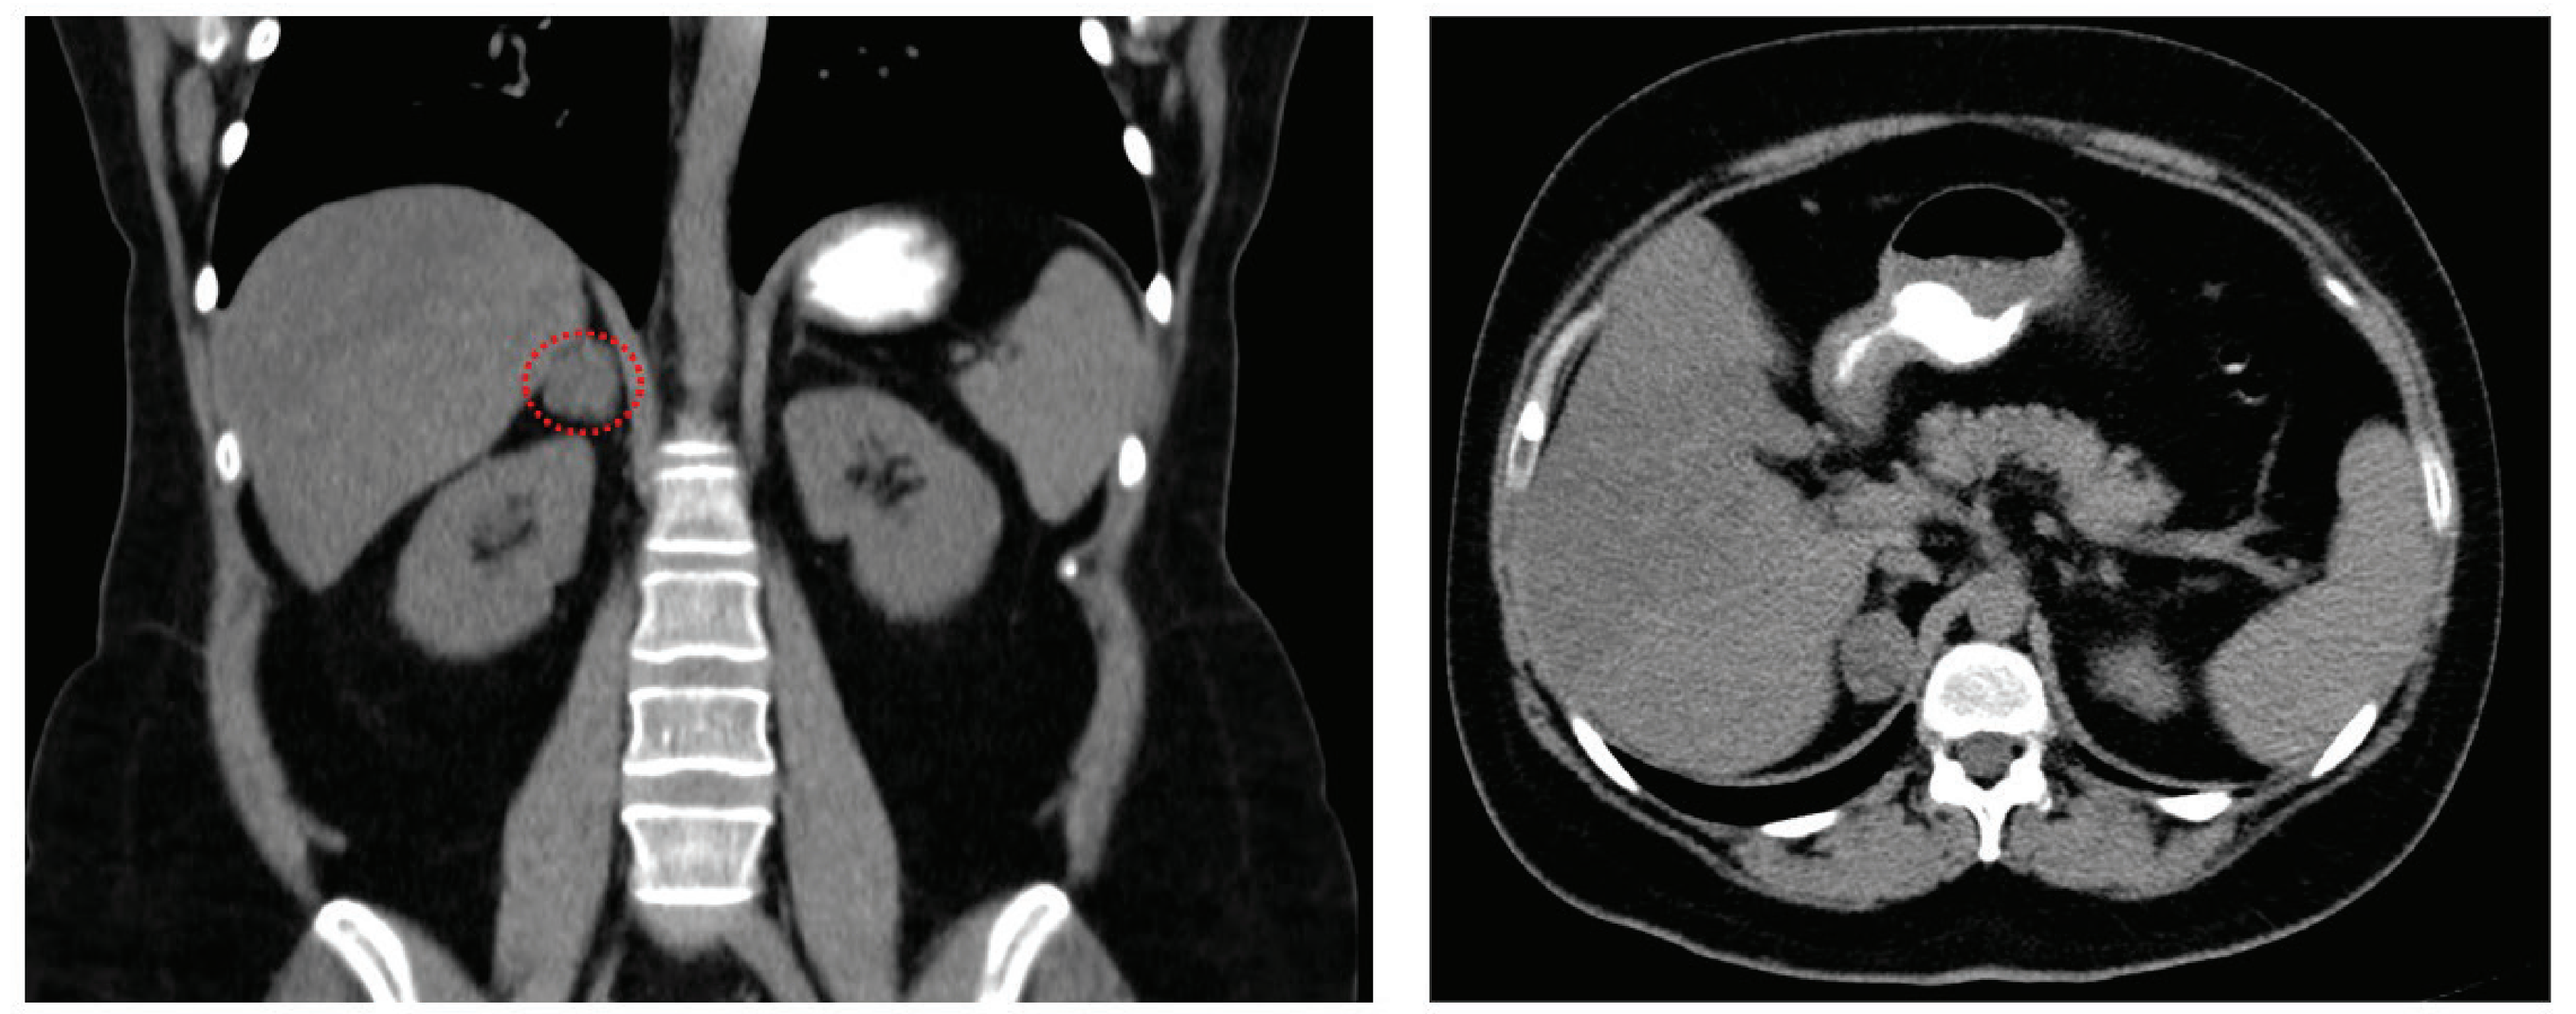

Right adrenal gland: 2.8 x 2.6 x 2.4 cm. Left adrenal gland: Atrophic.

TREATMENT COURSE AND OUTCOMES

- Labile BP elevations placed the patient at high risk for unilateral adrenalectomy

Treatment with Mifepristone

- Mifepristone was initiated at 300 mg daily and increased to 1200 mg daily over 3 months while awaiting definitive surgical treatment
- Within 3 months on mifepristone, improvements in cardiometabolic parameters (**Table**) were noted:
 - Rapid improvement in BP with elimination of systolic BP spikes (**Figure 2**) and most BP medications were discontinued
 - Rapid and marked improvements in glucose parameters (**Figure 3**)
 - Normalization of liver enzymes (**Table**)
- Within the first 2 weeks, the patient reported:
 - Nausea and fatigue that resolved without intervention
 - Increased heart rate that resolved with adjustments in BP medications

Table. Laboratory Parameters in a 3-Month Period Before and During Mifepristone and After Adrenalectomy

Baseline	Before Mifepristone	With Mifepristone	After Adrenalectomy
BP (120/80 mmHg)	182/112	124/90	128/80
FPG (65-99 mg/dL)	121	83	81
2-hour oGTT (<140 mg/dL)	279	145	
HbA_{1C} (<5.7%)	6	5.6	5.5
AST (10-30 U/L)	46	22	26
ALT (6-29 U/L)	94	25	33
PRA (0.25-5.82 ng/mL/h)	0.56	2.79	

ALT, alanine aminotransferase; AST, aspartate aminotransferase; BP, blood pressure; FPG, fasting plasma glucose; HbA_{1C}, hemoglobin A_{1C}; oGTT, oral glucose tolerance test; PRA, plasma renin activity.

Figure 2. Changes in Blood Pressure After Treatment With Mifepristone

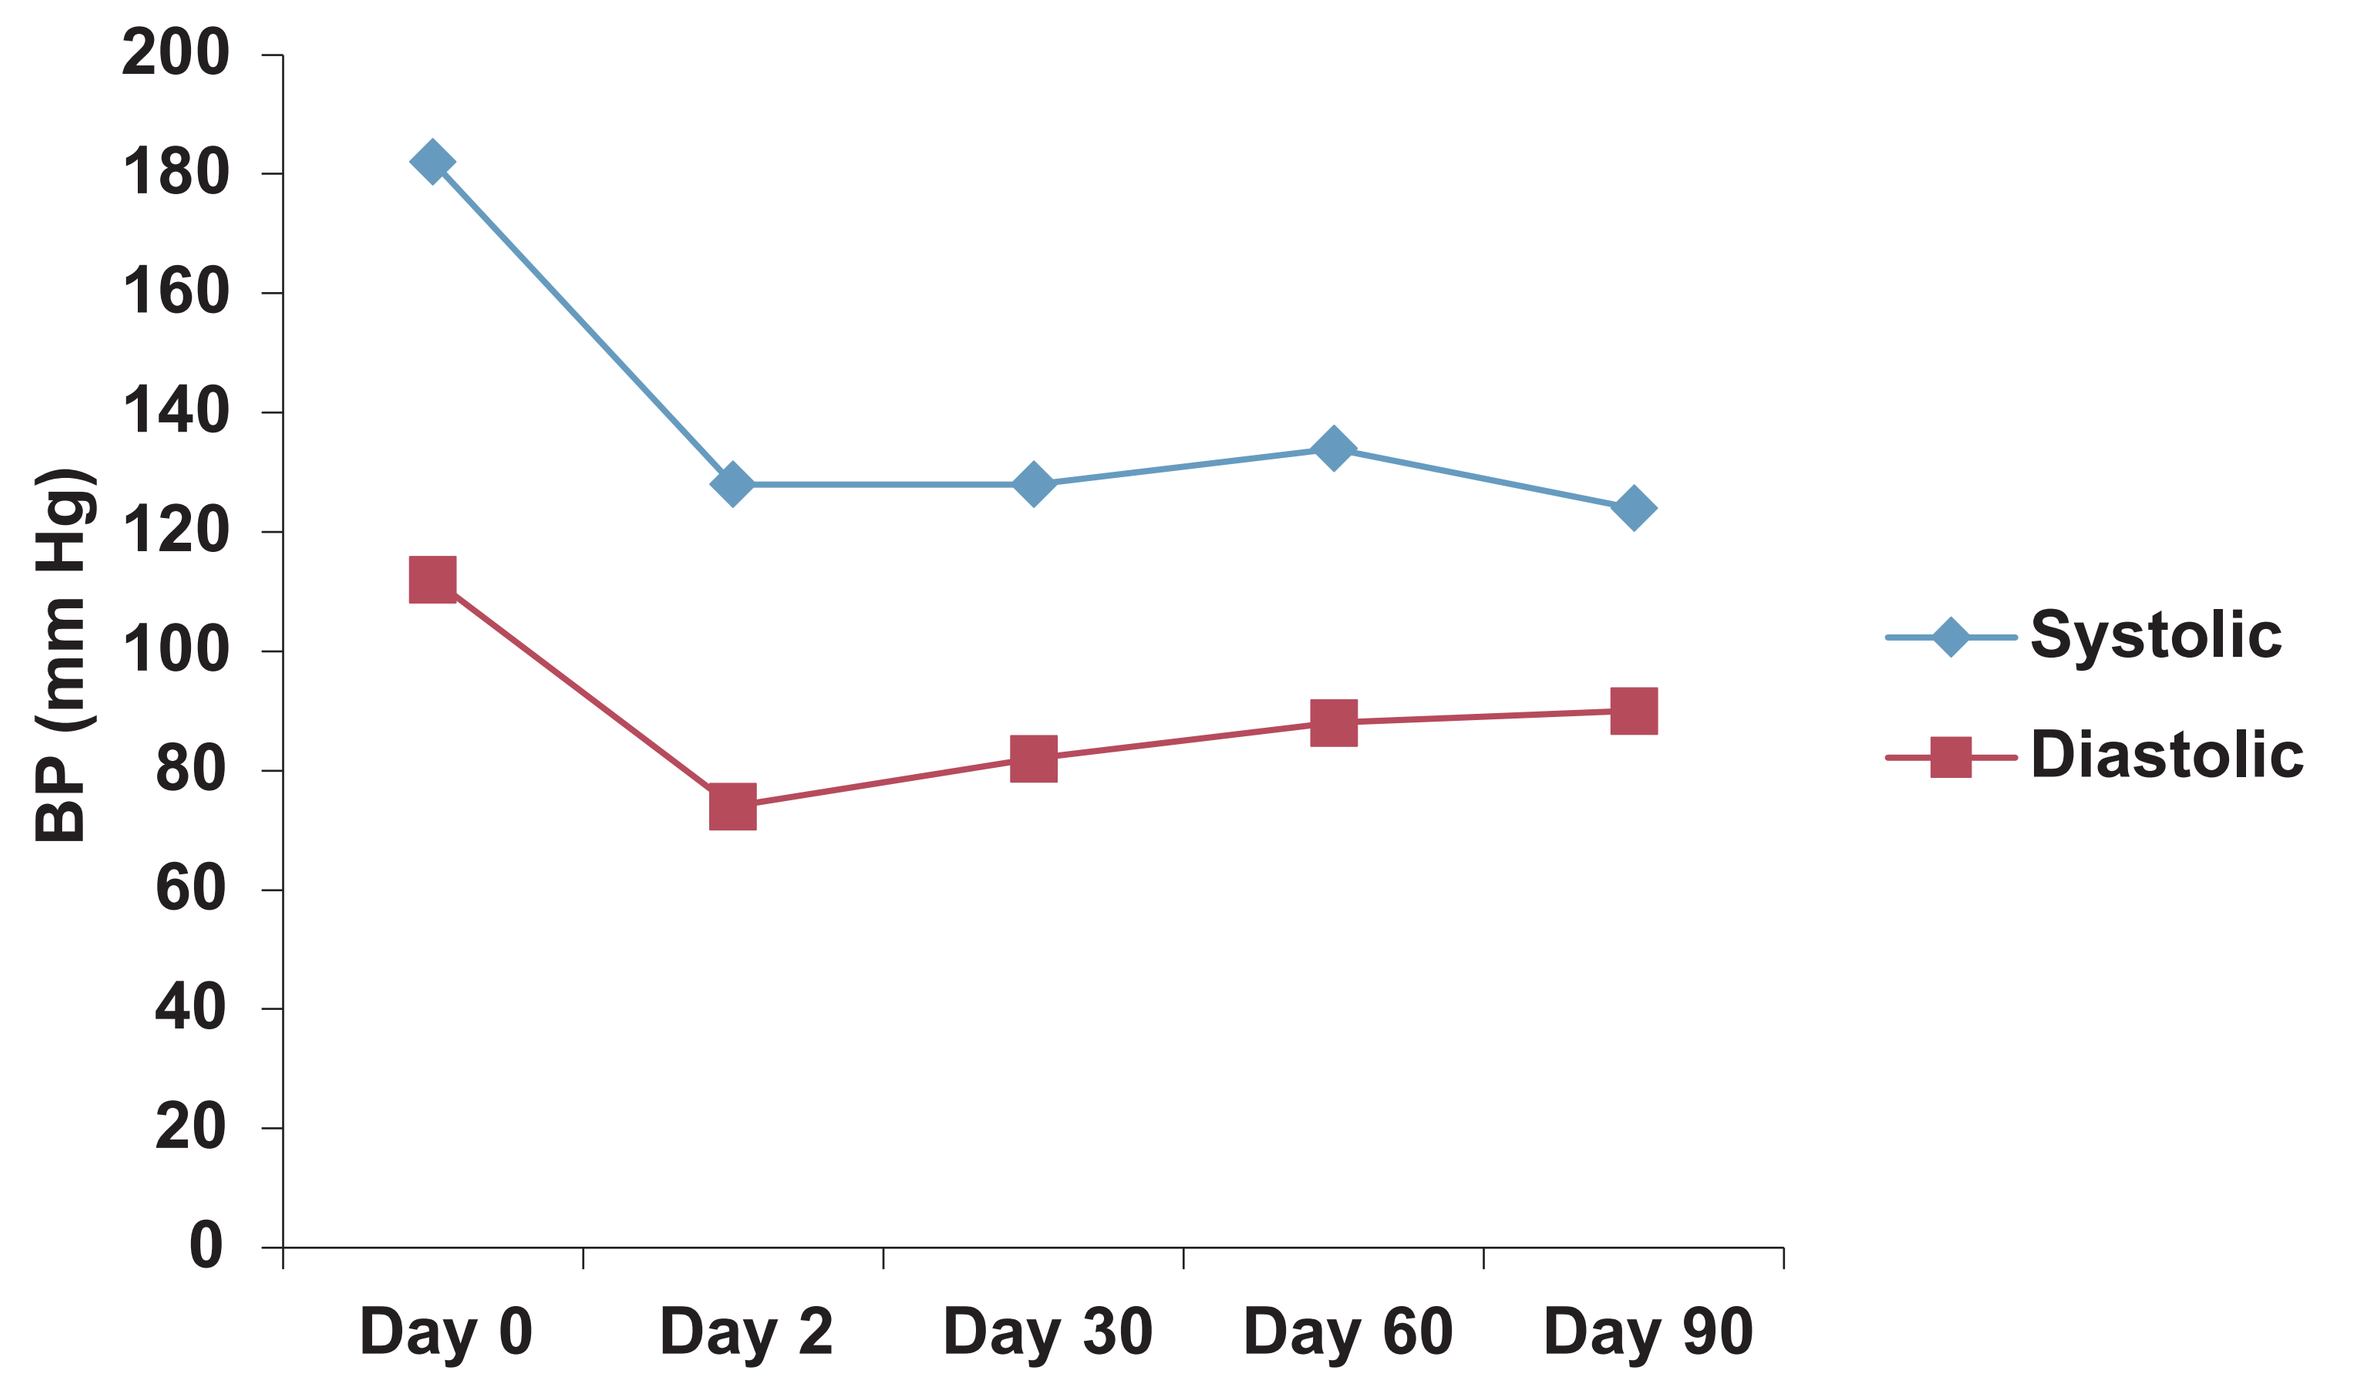
BP (mm Hg)
200
180
160
140
120
100
80
60
40
20
0
Day 0
Day 2
Day 30
Day 60
Day 90
Systolic
Diastolic

Figure 3. Changes in Fasting Plasma Glucose After Treatment With Mifepristone

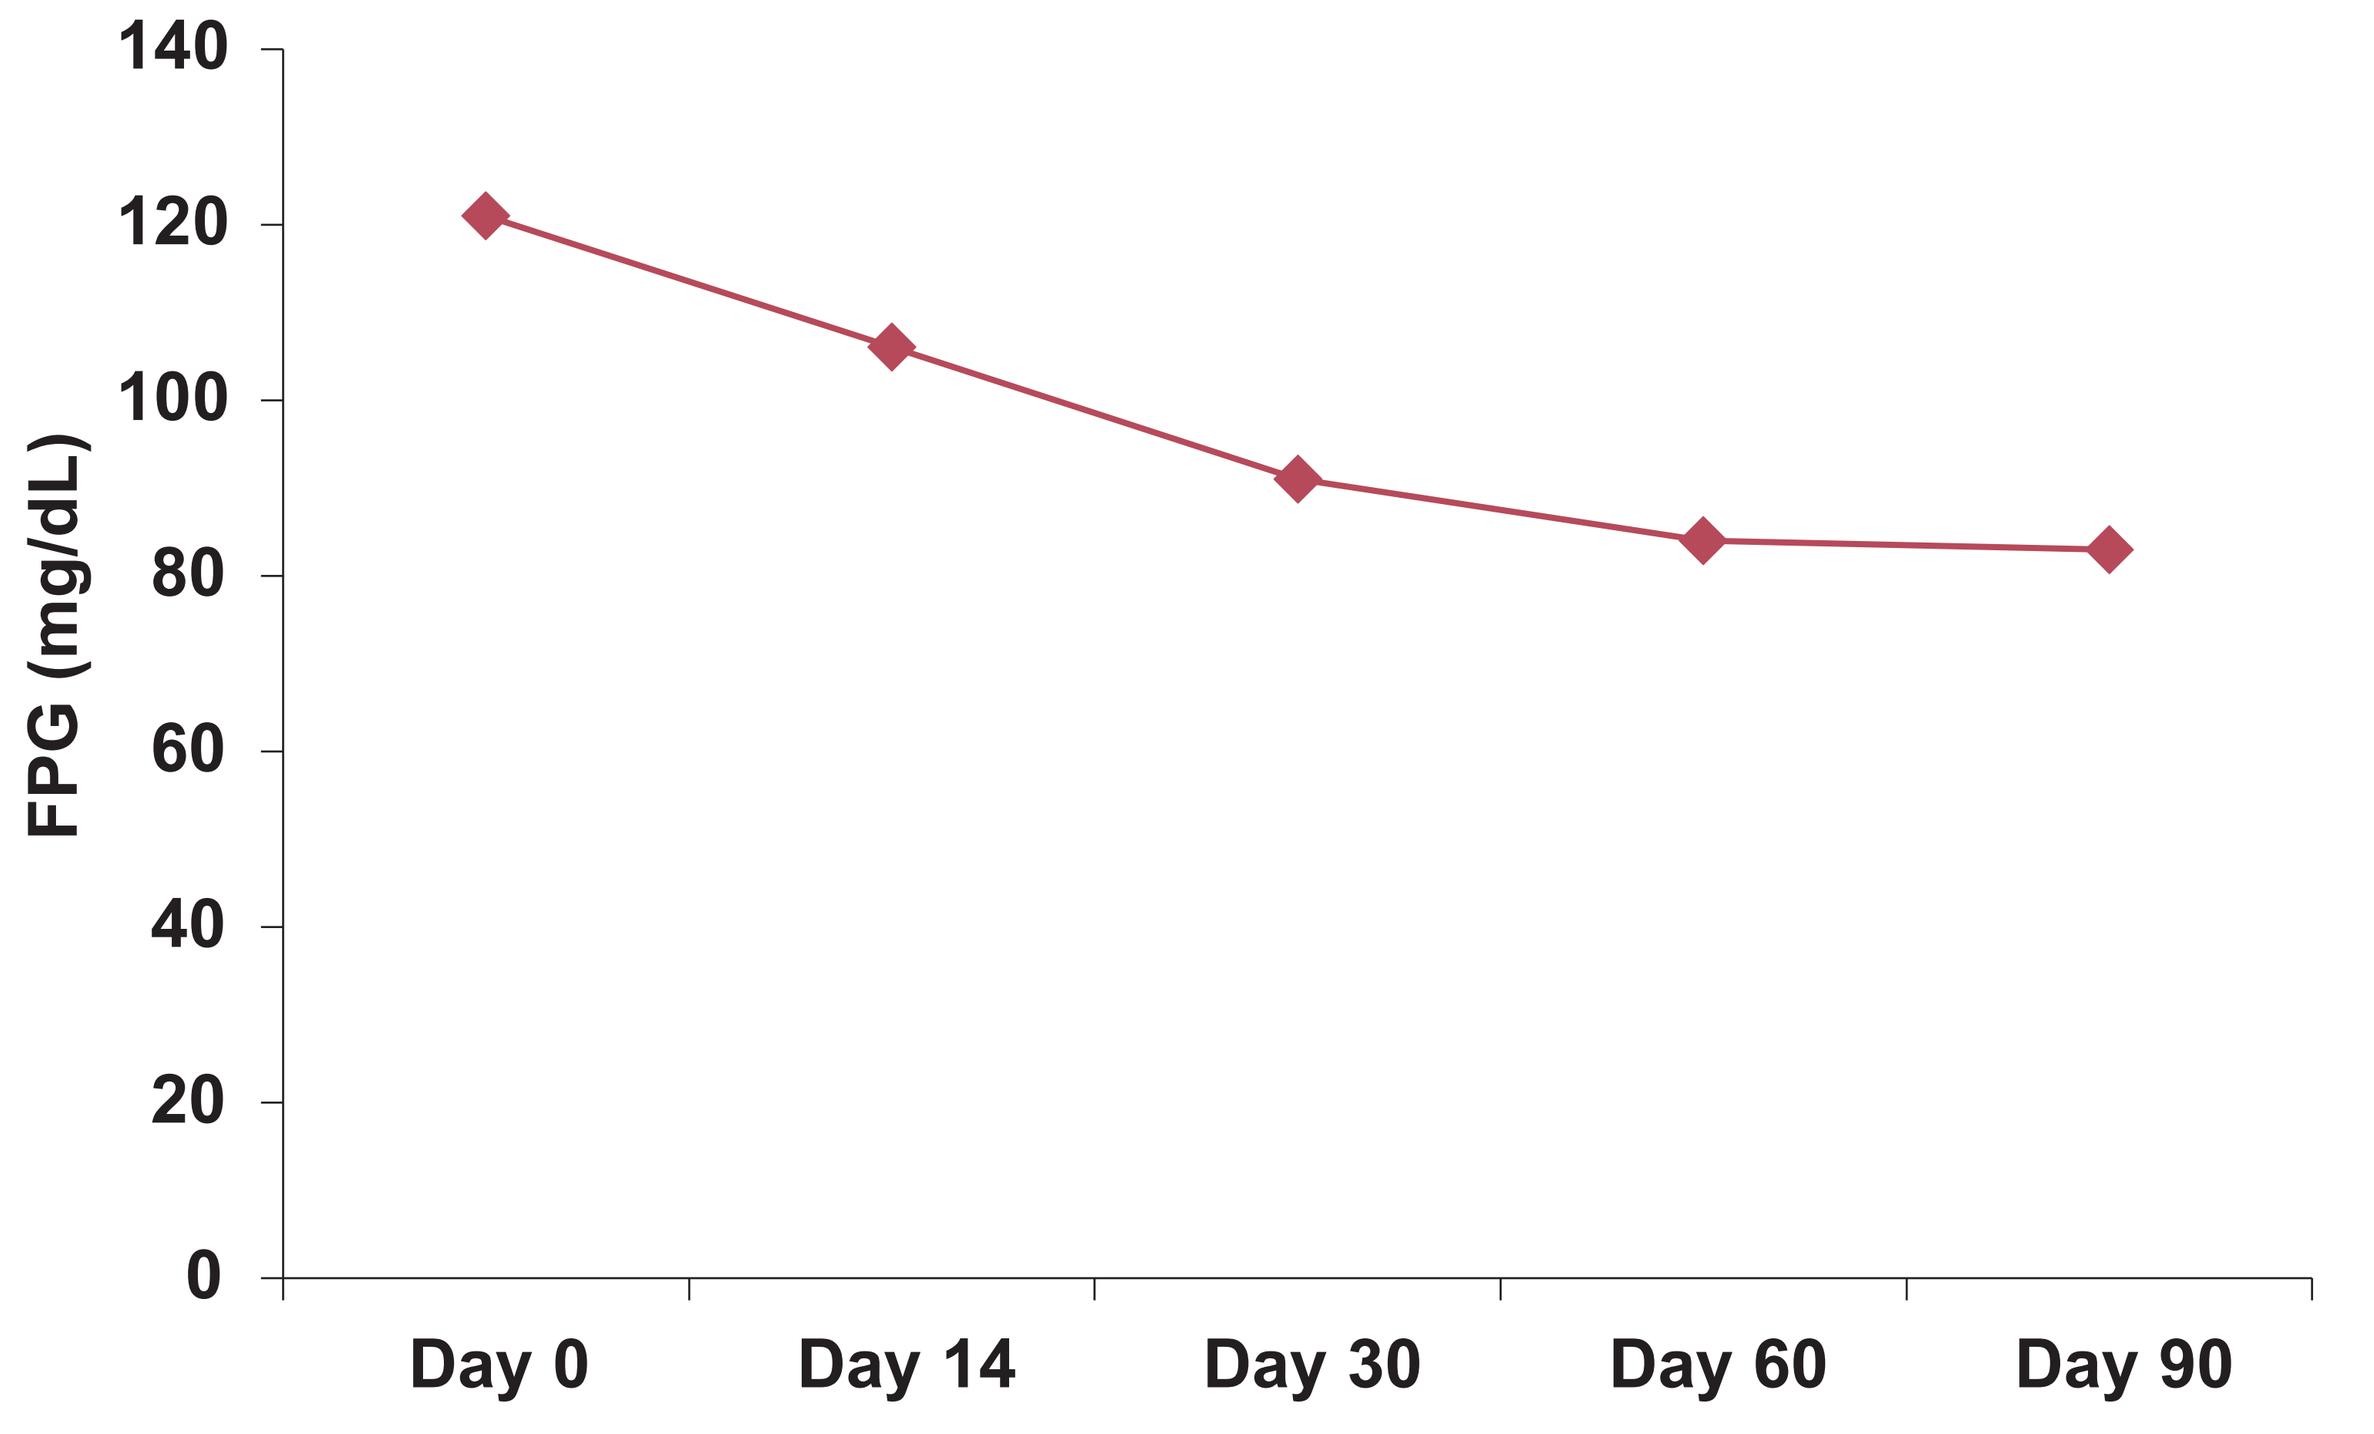
FPG (mg/dL)
140
120
100
80
60
40
20
0
Day 0
Day 14
Day 30
Day 60
Day 90

Adrenalectomy

- Right adrenalectomy was performed via robot-assisted laparoscopic approach
- There were no intraoperative or postoperative complications requiring extended hospitalization
- BP remains improved, and glucose parameters and liver enzymes continue to be normalized postoperatively

DISCUSSION AND CONCLUSIONS

- Screening for CS is warranted when several clinical features are present, including resistant hypertension, unexplained weight gain, dyslipidemia, and NAFLD
- Surgery was postponed in our patient until preoperative clinical status was improved
- Preoperative treatment with mifepristone was well tolerated and resulted in several clinical improvements within 3 months
- Prospective trials are needed to evaluate the effects of mifepristone on surgical outcomes

REFERENCES

1. Nieman LK, et al. *J Clin Endocrinol.* 2008;93:1526-1540.
2. Rockall AG, et al. *Eur J Endocrinol.* 2003;149:543-548.
3. Newell-Price J, et al. *Lancet.* 2006;367:1605-1617.
4. Valassi E, et al. *Clin Endocrinol.* 2012;77:735-742.
5. Moraitis AG, Auchus RJ. *AACE Clin Case Rep.* 2015;1(3):e204-e207.
6. Fleseriu M, et al. *J Clin Endocrinol.* 2012;97:2039-2049.

ACKNOWLEDGMENTS

Funding for development, editorial, design, and production support was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors contributed to the development of the poster, reviewed the material, and provided approval of the final version.

DISCLOSURES

GA, HNTL: Nothing to disclose; PJL: Employee, Corcept Therapeutics

Presented at the Endocrine Society Annual Meeting (ENDO 2016), April 1-4, 2016, Boston, Massachusetts