FRI-447

Cushing's Syndrome Misdiagnosed as Bipolar Depression

Elena A. Christofides, MD, FACE[1]; Neven Pesa, RN, CPNP[2]; Dat Nguyen, PharmD[2]
[1]Endocrinology Associates, Inc, Columbus, OH; [2]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Cushing's syndrome (CS), resulting from prolonged exposure to excess glucocorticoids, is often associated with numerous physical and mental disturbances. Many psychiatric disorders like depression, mood dysregulation, anxiety, psychosis, and post-traumatic stress disorder (PTSD) are associated with hypercortisolemia and can complicate the diagnostic work-up. We detail a patient with a history of bipolar disorder who ultimately was diagnosed with CS and received treatment with a competitive glucocorticoid receptor antagonist, mifepristone (MIFE, Korlym®, Corcept Therapeutics).

Clinical Case: A 39 y/o woman with a history of hyperlipidemia, impaired glucose tolerance, and hypertension presented with complaints of weight gain (BMI 36 kg/m^2), muscle pain, diaphoresis, skin abnormalities, menstrual irregularities, problems sleeping, and irritability. Medical interview revealed a 10-year history of bipolar disorder treated with lamotrigine 200 mg/d. Clinical and metabolic features (plethora, supraclavicular fat pads, central obesity, hypertriglyceridemia with insulin resistance) led to suspicion of CS. The diagnosis of CS was confirmed by two separate midnight salivary cortisol levels (2.5 and 26.8 nmol/L), a 1-mg overnight dex suppression test (1.9 mcg/dL). ACTH was not suppressed and a pituitary MRI was negative. The patient refused any further testing or surgery and MIFE 300 mg/d was initiated and titrated up to 1200 mg/d within 2 months.

One month after starting MIFE, she reported feeling significantly better, calmer, less agitated, and more "normal." Clinical improvements included decreased plethora, facial puffiness, supraclavicular fullness, and decreased abdominal protrusion. She was weaned off lamotrigine within the next month. Within 4 months, blood pressure stabilized (136/90 mmHg) and her cushingoid appearance improved dramatically. After 10 months, she lost 11.4 kg, her blood glucose normalized (97 mg/dL) and she stated she felt "great."

Adverse events included peripheral edema upon starting MIFE that resolved spontaneously. Menses remained irregular and endometrial thickening was noted. Therapeutic and diagnostic dilation and curettage (D&C) was performed, revealing benign hypertrophy. Due to the known adverse event of endometrial hypertrophy, she elected to have a total hysterectomy without oophorectomy.

Conclusion: When the source of CS is not clearly identifiable, physicians will often treat the comorbidities of CS instead of the hypercortisolemia itself. This hesitation to medically treat CS can lead to poor quality of life and health management. For this patient, MIFE therapy resulted in several beneficial medical, clinical, and psychological outcomes.

INTRODUCTION

- Cushing's syndrome is a complex, multi-system condition resulting from prolonged exposure to cortisol
- It is associated with substantial metabolic dysfunction in addition to psychiatric symptoms (**Table 1**)

Table 1. Clinical Features of Cushing's Syndrome

Nonpsychiatric[1]	% of Patients With Symptom
Obesity/weight gain	70%-95%
Moon face	81%-90%
Fat pads	~50%
Striae	44%-50%
Hirsutism	~75%
Easy bruising	35%-65%
Menstrual irregularity	70%-80%
Muscle weakness	60%-82%
Hypertension	70%-85%
Glucose intolerance	45%-70%

Psychiatric	% of Patients With Symptom
Major depression[2]	50%-70%
Anxiety[3]	12%-79%
Bipolar disorder, including manic and hypomanic[4,5]	~30%

- Clinicians should be aware of the potential role of Cushing's syndrome in the differential diagnosis of psychiatric symptoms
- Treatment of Cushing's syndrome has demonstrated improvements in depression, anxiety, somatic symptoms and neuroticism scores[6]
- The following case highlights how the diagnosis of Cushing's syndrome was made in a patient with persistent psychiatric symptoms

CLINICAL PRESENTATION

- A 39-year-old female patient with a 10-year history of bipolar disorder presented with a set of clinical and metabolic features that prompted a work-up for Cushing's syndrome
- She had been on various psychiatric medications without benefit and was on lamotrigine 200 mg/day at the time of presentation
- Clinical and laboratory assessments are presented in **Table 2**

Table 2. Patient Clinical and Laboratory Assessments

Clinical Manifestations	Value
Central obesity	BMI 36 kg/m^2, weight 217 lbs
Supraclavicular fat pads	
Bipolar depression	
Hypertension	150/92
Plethora	
Muscle pain	
Diaphoresis	
Skin abnormalities	
Menstrual irregularities	
Problems sleeping	
Irritability	

Laboratory Assessments	Value
Fasting glucose	133 mg/dL (normal 65-99)
Late-night salivary cortisol	2.5 nmol/L (normal <0.3-4.3) 26.8 nmol/L (retest 2 days later)
1-mg DST	1.9 µg/dL
ACTH	<1.1 pg/mL (normal 7.2-63.3)
Urinary free cortisol	9 µg/24hr (normal 0-50)
DHEA-S	227.6 µg/dL (normal 57.3-279.2)
Cortisol	18.1 µg/dL (normal 2.3-11.9)

ACTH, adrenocorticotropic hormone; BMI, body mass index; DST, dexamethasone suppression test; DHEA-S, dehydroepiandrosterone sulfate.

Imaging

- Pituitary MRI, abdominal CT, and octreotide scans were all negative

Treatment

- Mifepristone (Korlym®, Corcept Therapeutics) was initiated at a dose of 300 mg/d and titrated up to 1200 mg/d after 2 months
- After 1 month, patient reported feeling significantly better, calmer, less agitated, and more "normal"
- Lamotrigine was reduced gradually and discontinued after 1 month
- Within 4 months, her blood pressure stabilized (136/90 mmHg)
- Clinical improvements included decreased plethora and facial puffiness (**Figure 1**), as well as decreased supraclavicular fullness and decreased abdominal protrusion
- After 10 months, the patient lost 25.1 lbs, her blood glucose normalized (97 mg/dL), her BP decreased to 120/78, and she stated she felt "great"
 - At 12 months, the patient weighed 194 lbs (BMI 32.3 kg/m^2)

Figure 1. Facial Changes During Mifepristone Therapy

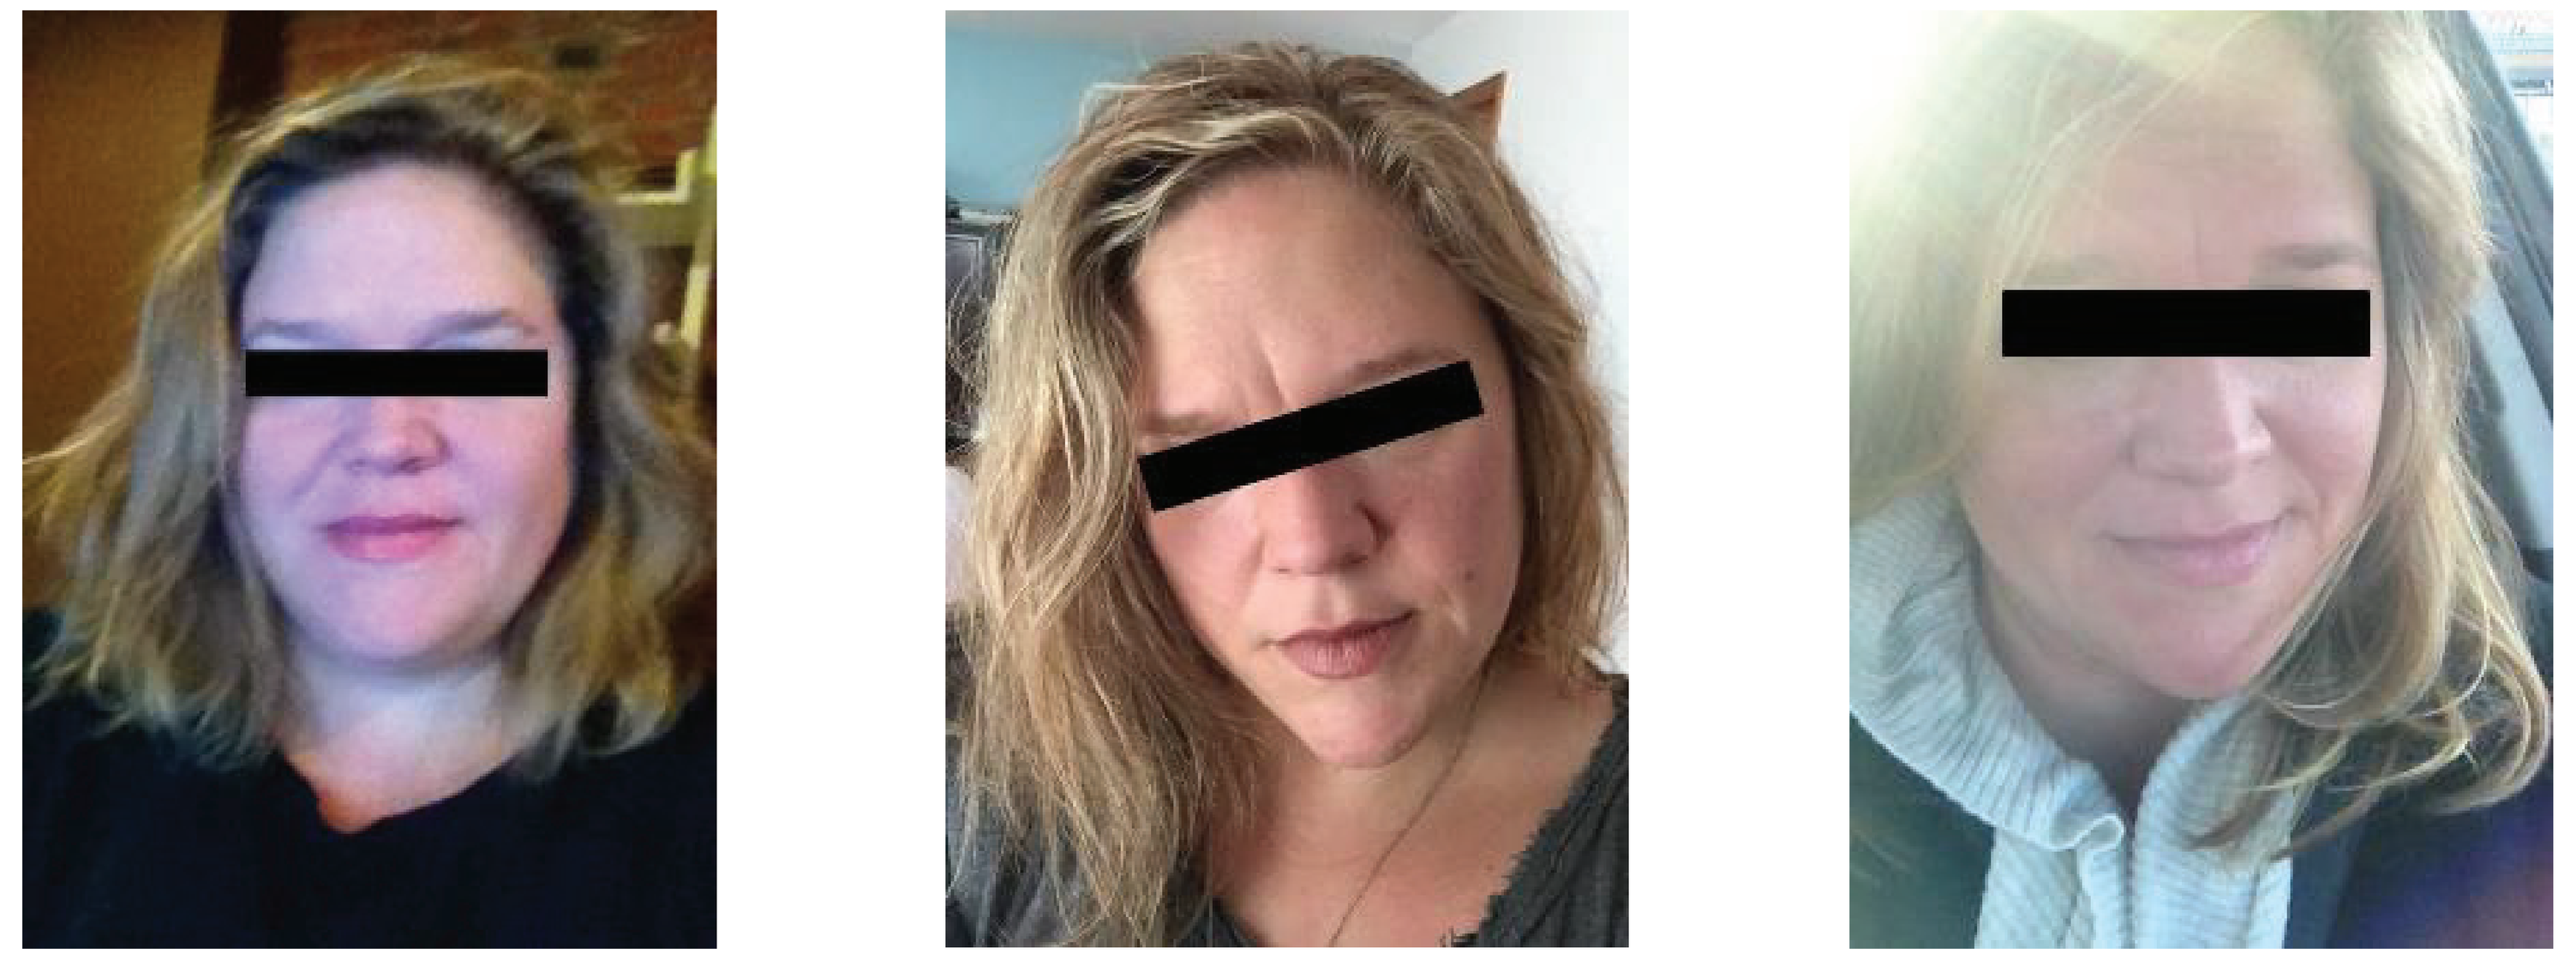
Before Treatment
4 Months of Treatment
>1 Year of Treatment

Safety

- Peripheral edema was observed upon starting mifepristone but resolved spontaneously
- Cessation of menses (which were irregular at baseline) prompted transvaginal ultrasound, revealing endometrial thickening
- Therapeutic and diagnostic dilation and curettage was performed, revealing benign hypertrophy
- Patient elected to have a total hysterectomy without oophorectomy

SUMMARY AND CONCLUSIONS

- Prolonged exposure to cortisol as seen in Cushing's syndrome can lead to psychiatric disease, cognitive impairment, mood disorders, and sleep abnormalities
- Cushing's syndrome is often overlooked during the differential diagnosis of psychiatric symptoms
- Even when suspicion of Cushing's syndrome is high, if the source of the tumor cannot be localized, the underlying cause of the patient's psychiatric symptoms is often not adequately addressed
- This patient had bipolar depression secondary to Cushing's syndrome with a tumor that could not be localized
- The patient was treated with mifepristone, which improved her metabolic, clinical, and psychiatric symptoms

REFERENCES

1. Sharma ST, et al. Cushing's syndrome: epidemiology and developments in disease management. *Clin Epidemiol.* 2015;7:281-93.
2. Sonino N, Fava GA. Psychosomatic aspects of Cushing's disease. *Psychother Psychosom.* 1998;67:140-46.
3. Loosen PT, et al. Psychiatric phenomenology in Cushing's disease. *Pharmacopsychiatry.* 1992;25:192-98.
4. Haskett RF. Diagnostic categorization of psychiatric disturbance in Cushing's syndrome. *Am J Psychiatry.* 1985;142:911-16.
5. Hudson JI, et al. Phenomenology and family history of affective disorder in Cushing's disease. *Am J Psychiatry.* 1987;144:951-53.
6. Kelly WF, et al. A prospective study of psychiatric and psychological aspects of Cushing's syndrome. *Clin Endocrinol.* 1996;45:715-20.

ACKNOWLEDGMENTS

Funding to support the preparation of this poster was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ.

DISCLOSURES

EAC: Nothing to disclose. NP, DN: Employees, Corcept.

Presented at the Endocrine Society Annual Meeting (ENDO 2016), April 1-4, 2016, Boston, Massachusetts