SUN 555

Persistent Acromegaly Controlled on Pegvisomant with Co-Existing Non-ACTH Dependent Cushing's Syndrome (CS) Due to Bilateral Adrenal Adenomas with Dramatic Clinical Response to Mifepristone

Murray B. Gordon, MD, FACE[1]; Kellie L. Spiller, MS[1]; Rachel Bunta, MBA[2]; Michael S. Gordon, MD, FACE[3]

[1]Division of Endocrinology, Allegheny Neuroendocrinology Center, Allegheny General Hospital, Pittsburgh, PA; [2]Corcept Therapeutics, Menlo Park, CA; [3]Division of Endocrinology, Diabetes and Hypertension, Brigham and Women's Hospital, Boston, MA

ABSTRACT

Introduction: The combination of both acromegaly and CS is extremely rare, with only 6 cases in the literature. Both diseases are characterized by comorbidities such as diabetes mellitus, hypertension, proximal muscle weakness, menstrual abnormality, sleep apnea, and cardiovascular disease. We present a patient whose acromegaly was controlled but subsequently diagnosed with non-ACTH dependent CS due to bilateral adrenal adenomas.

Case: A 60 y/o woman presented with diaphoresis, headache, and new onset type 2 diabetes. She was diagnosed with acromegaly 20 years ago. She underwent transsphenoidal debulking of a pituitary macroadenoma followed by gamma knife radiotherapy. After failing with bromocriptine and octreotide, her acromegaly was controlled with pegvisomant 30 mg sc QD with little change in glucose metabolism. Central hypothyroidism was treated with levothyroxine 137 mcg QD. After referral to our pituitary center, symptoms of weight gain, centripetal obesity, easy bruisability and severe proximal muscle weakness dating back to her initial diagnosis of acromegaly were noted and CS was suspected. An IGF-1 level was 187 ng/mL (41-279), IGF-1 SDS +1.2. Pituitary MRI revealed empty sella syndrome. An ACTH level was <5 ng/mL (6-50); 11PM salivary cortisol levels ranging between 2.8-6.4 nmol/L (<4.3); 8AM plasma cortisol levels were intermittently elevated, ranging between 18.6-33.9 mcg/dL (7-25); urinary free cortisol levels were consistently normal. Adrenal CT revealed bilateral adrenal lesions: right 1.4 cm and left 2.4 cm (both -4 Hounsfield units). A trial of ketoconazole resulted in no change in symptoms or hypercortisolism. Hba1c 6.7%, FBG 120 mg/dL on metformin 1000 mg BID and glimepiride 4 mg QD, ALT 21 U/L (7-55), AST 14 U/L (8-48), triglycerides 227 mcg/dL, BP 122/78. Mifepristone 300 mg QD (Korlym®, Corcept Therapeutics), a glucocorticoid receptor antagonist, was initiated with spironolactone 100 mg QD.

ACTH level rose to 24 pg/mL at 4 months and 77 pg/mL at 1 year. She lost 70lbs and had marked improvement in both Cushingoid features and proximal muscle weakness. Hba1c 5.8% & FBG 87 mg/dL despite glimepiride discontinuation; triglycerides 117 mg/dL, AST 11 U/L, ALT 10 U/L; IGF-1 dropped to 91 ng/mL, IGF-1 SDS -0.6 and pegvisomant was decreased to 25 mg sc QD with resulting IGF-1 125 ng/mL, IGF-1 SDS 0. She has been on mifepristone for over 2 years and has persistent weight loss, reduced anxiety, and normal muscle strength manifested by a regained ability to walk upstairs normally.

Conclusion: CS can be overlooked when treating an acromegaly patient because their comorbidities overlap. This case illustrates that concomitant CS should be considered when symptoms consistent with CS persist in a patient whose acromegaly appears to be otherwise controlled. Mifepristone was used as a diagnostic and therapeutic tool to confirm a diagnosis of CS by showing a clear biochemical and clinical response to the pharmacological intervention.

INTRODUCTION

- Acromegaly and Cushing's syndrome are rare neuroendocrine diseases with similar multisystem comorbidities, such as diabetes mellitus, hypertension, proximal muscle weakness, and cardiovascular disease
- There are similarities between diagnostic and treatment strategies for both acromegaly and Cushing's syndrome
 - Biochemical screening and tumor localization with appropriate imaging
 - A multi-modal treatment strategy that includes surgery, medical therapy, and/or radiation
 - Goals of therapy include normalization of biochemical variables; improvement in signs, symptoms, and comorbidities; and minimization of long-term mortality risk
- Medical therapy agents can be divided into two categories based on their effects
 - Biochemical control by normalizing elevated hormones
 - Receptor antagonist to block the effects
- We present a patient whose acromegaly was controlled but who was subsequently diagnosed with ACTH-independent Cushing's syndrome due to bilateral adrenal adenomas. This patient was medically managed by three separate receptor antagonists.
 - Pegvisomant – Growth hormone receptor antagonist
 - Mifepristone – Glucocorticoid receptor antagonist
 - Spironolactone – Mineralocorticoid (aldosterone) receptor antagonist

CASE PRESENTATION

- A 60-year-old female patient with a history of acromegaly was referred in 2012
- She presented with symptoms of weight gain, centripetal obesity, easy bruisability, severe proximal muscle weakness, abdominal striae, facial plethora and erythema, and increased supraclavicular fat pads that had been progressively worsening for over 20 years (**Table 1**, **Figure 1**)
- Her acromegaly was controlled based on IGF-1 levels, but she reported diaphoresis, headache, and new-onset type 2 diabetes
- Cushing's syndrome was suspected based on her Cushingoid phenotype (**Figure 1**)

Table 1. Baseline Demographics at Time of Cushing's Syndrome Diagnosis

Age	60 years
BMI	37
Weight	213 lbs
Blood pressure	122/78 mmHg
Fasting glucose	120 mg/dL
A1c	6.7%

Figure 1. Disease Progression and Effects of Treatment

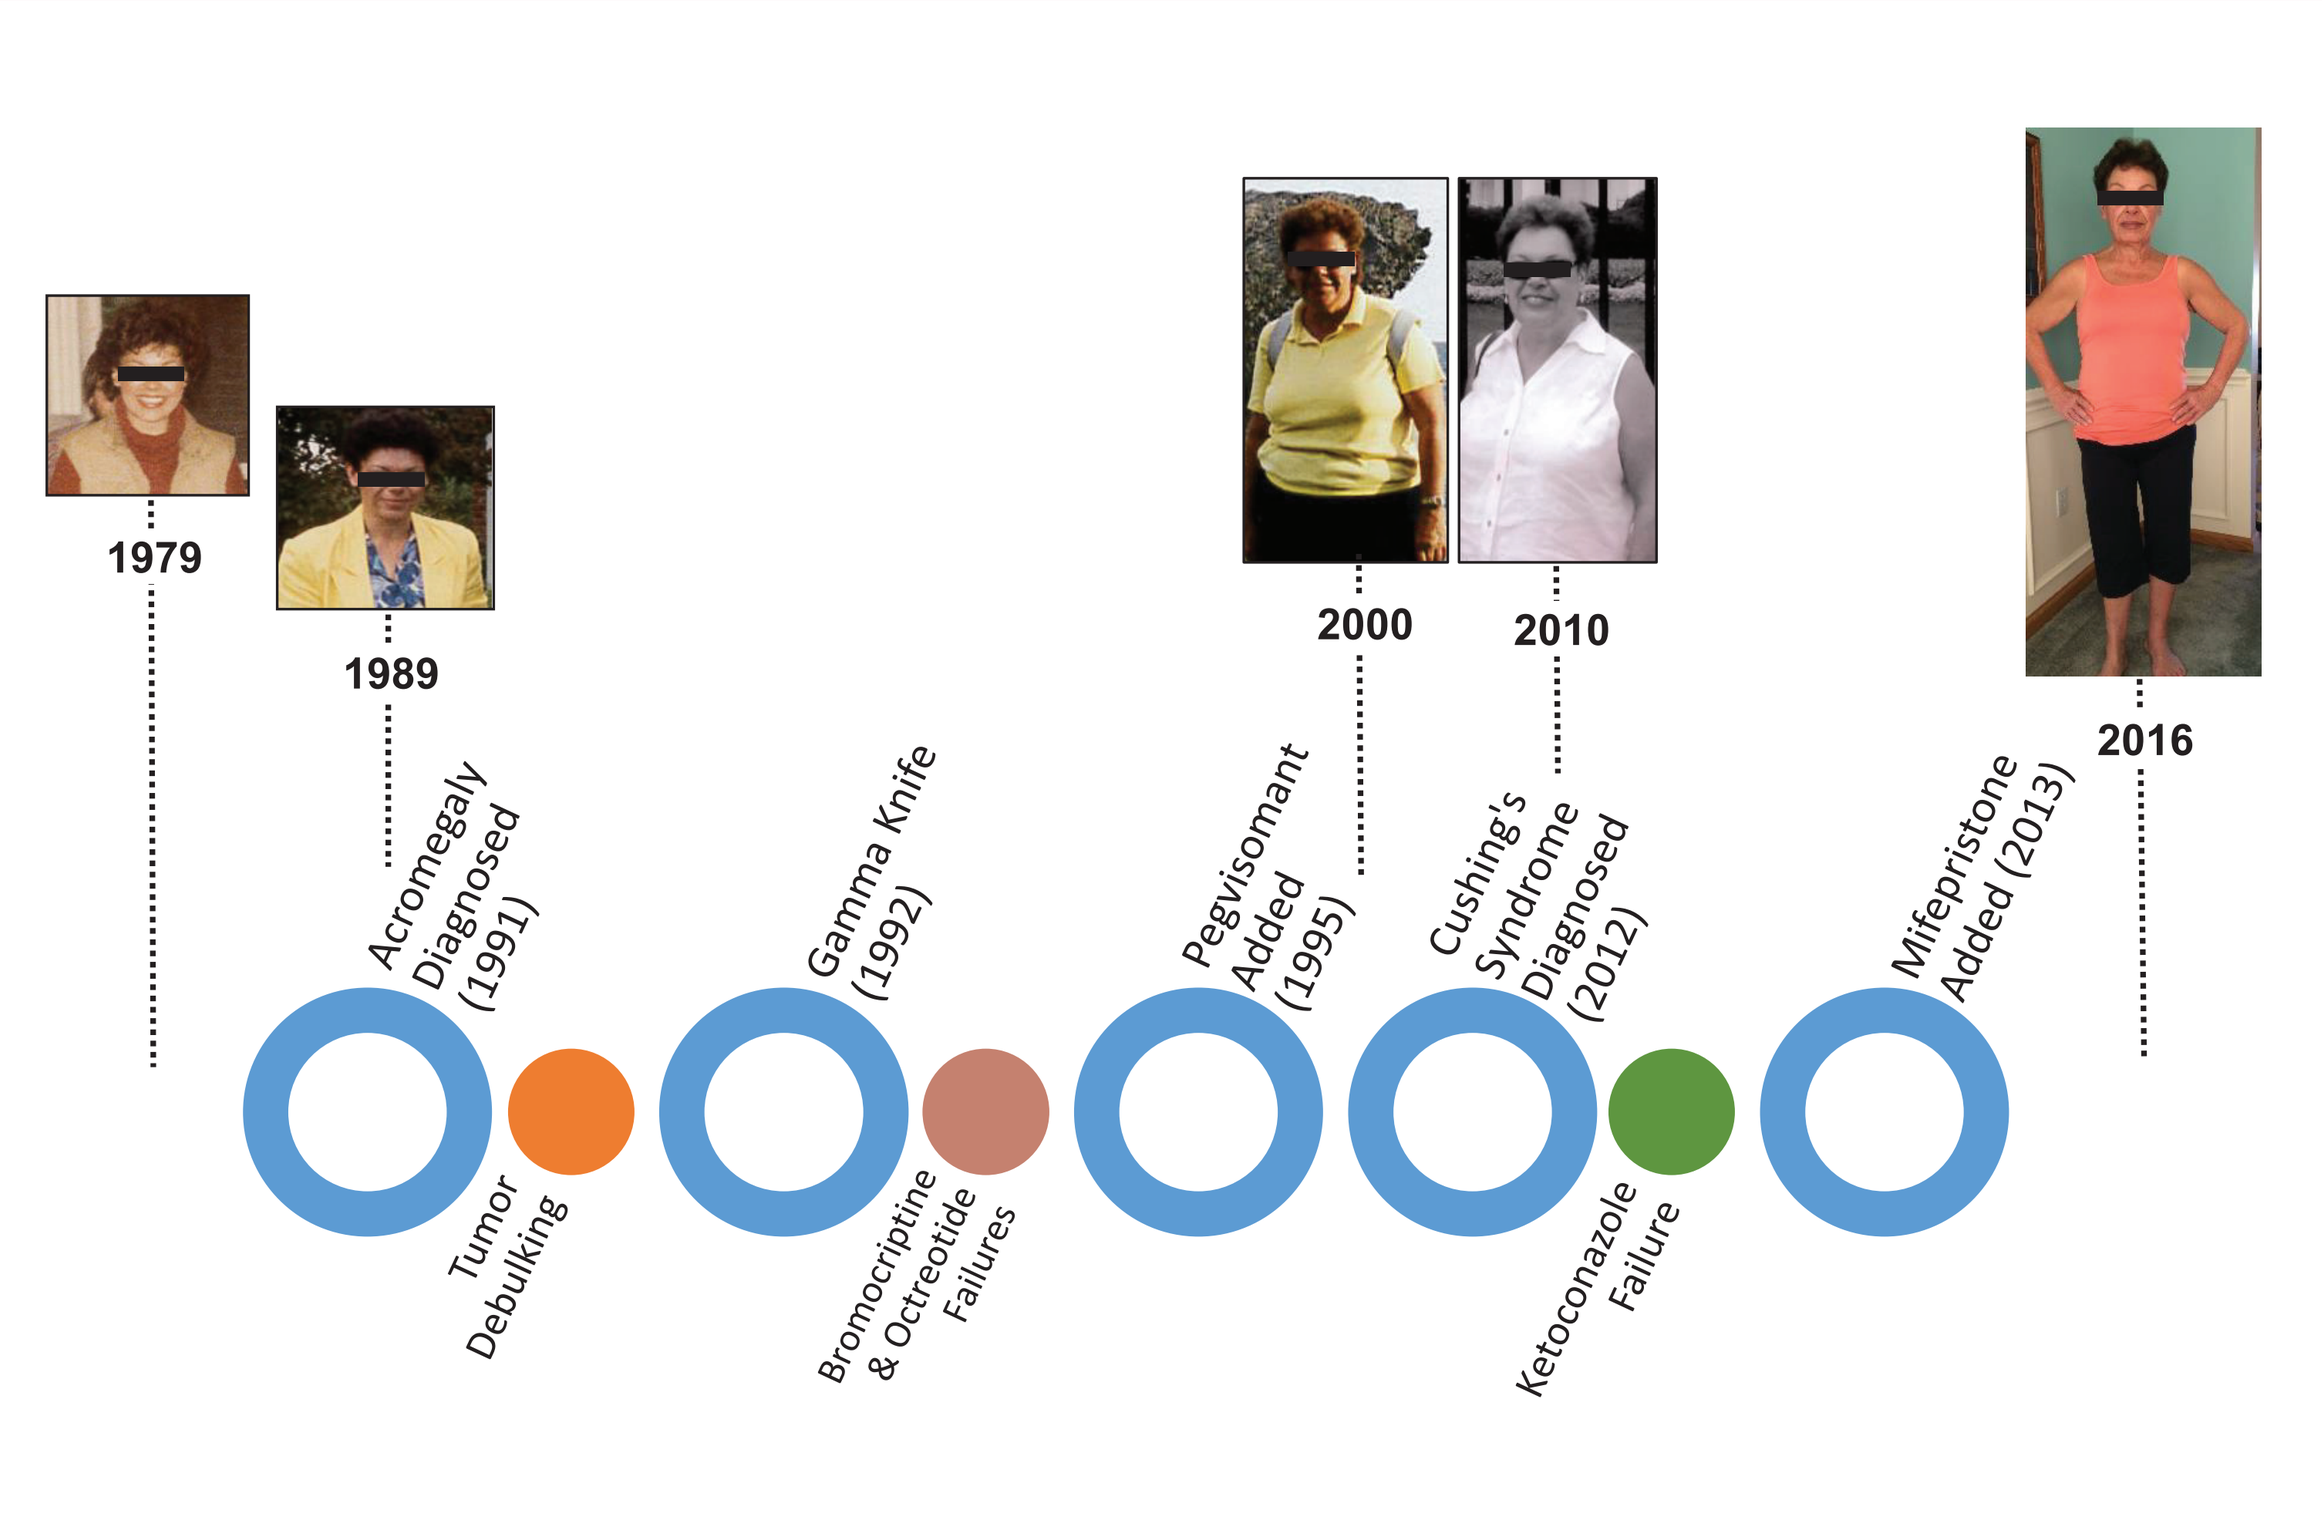
1979
1989
2000
2010
2016
Acromegaly Diagnosed (1991)
Tumor Debulking
Gamma Knife (1992)
Bromocriptine & Octreotide Failures
Pegvisomant Added (1995)
Cushing's Syndrome Diagnosed (2012)
Ketoconazole Failure
Mifepristone Added (2013)

Initial Treatments for Acromegaly

- Upon acromegaly diagnosis
 - The patient underwent transsphenoidal debulking of a pituitary macroadenoma followed by gamma knife radiotherapy (**Figure 1**)
 - Adjuvant medical therapy with bromocriptine and octreotide was unsuccessful
- Acromegaly control was achieved with the addition of pegvisomant 30 mg/day sc (Somavert®, Pfizer), a growth hormone receptor antagonist
 - IGF-1 level was 187 ng/mL (41-279), IGF-1 SDS +1.2, at presentation in 2012
 - Routine liver function tests were normal
- Central hypothyroidism was treated with levothyroxine 137 mcg QD

Diagnosis of Cushing's Syndrome

- Patient reported progressive signs and symptoms of Cushing's syndrome around the time of acromegaly diagnosis
- She was referred to the Allegheny Neuroendocrinology Center (after being managed by two local endocrinologists) due to persistent symptoms despite normalization of IGF-1 levels
- Cushing's syndrome was suspected based on initial workup in 2012 (**Table 2**)
- Pituitary MRI revealed empty sella syndrome and abdomen CT revealed bilateral adenomas (**Figure 2**)
- The patient was negative for Carney's complex stigmata: myxoma (negative echocardiogram), thyroid nodules (negative thyroid sonogram), spotty skin pigmentation, blue nevi, café-au-lait spots, schwannomas
- Multiple endocrine neoplasia type 1 (MEN1) was unlikely because she had normocalcemia, normal pancreas features on abdomen CT, and no family history of endocrine diseases
- Clinical presentation, biochemical testing, and imaging (**Figure 2**) resulted in high suspicion of Cushing's syndrome

Table 2. Cushing's Syndrome Workup and Treatment

PARAMETER	FINDINGS	
Signs and symptoms	Weight gain, centripetal obesity, easy bruisability, new onset diabetes, proximal muscle weakness, abdominal striae, facial plethora & erythema, supraclavicular fat pads	
ACTH	<5 ng/mL	Ref: 6-50 ng/mL
Late-night (11PM) salivary cortisol	2.8 nmol/L to 6.4 nmol/L	Ref: <4.3 nmol/L
Morning (8AM) plasma cortisol	18.6 mcg/dL to 33.9 mcg/dL	Ref: 7-25 mcg/dL
Urinary free cortisol	Consistently in reference range	
Abdomen CT – adrenal	Right adrenal mass, 1.4 cm; left adrenal mass, 2.4 cm; both –4 Hounsfield units	Incidentally discovered in 2003: Right adrenal mass 1.8 cm; left adrenal mass 1.5 cm; both +4 Hounsfield units

Figure 2A-B. Imaging: A. Primary MRI Shows Empty Sella Syndrome B. Adrenal CT Shows Bilateral Adrenal Lesions

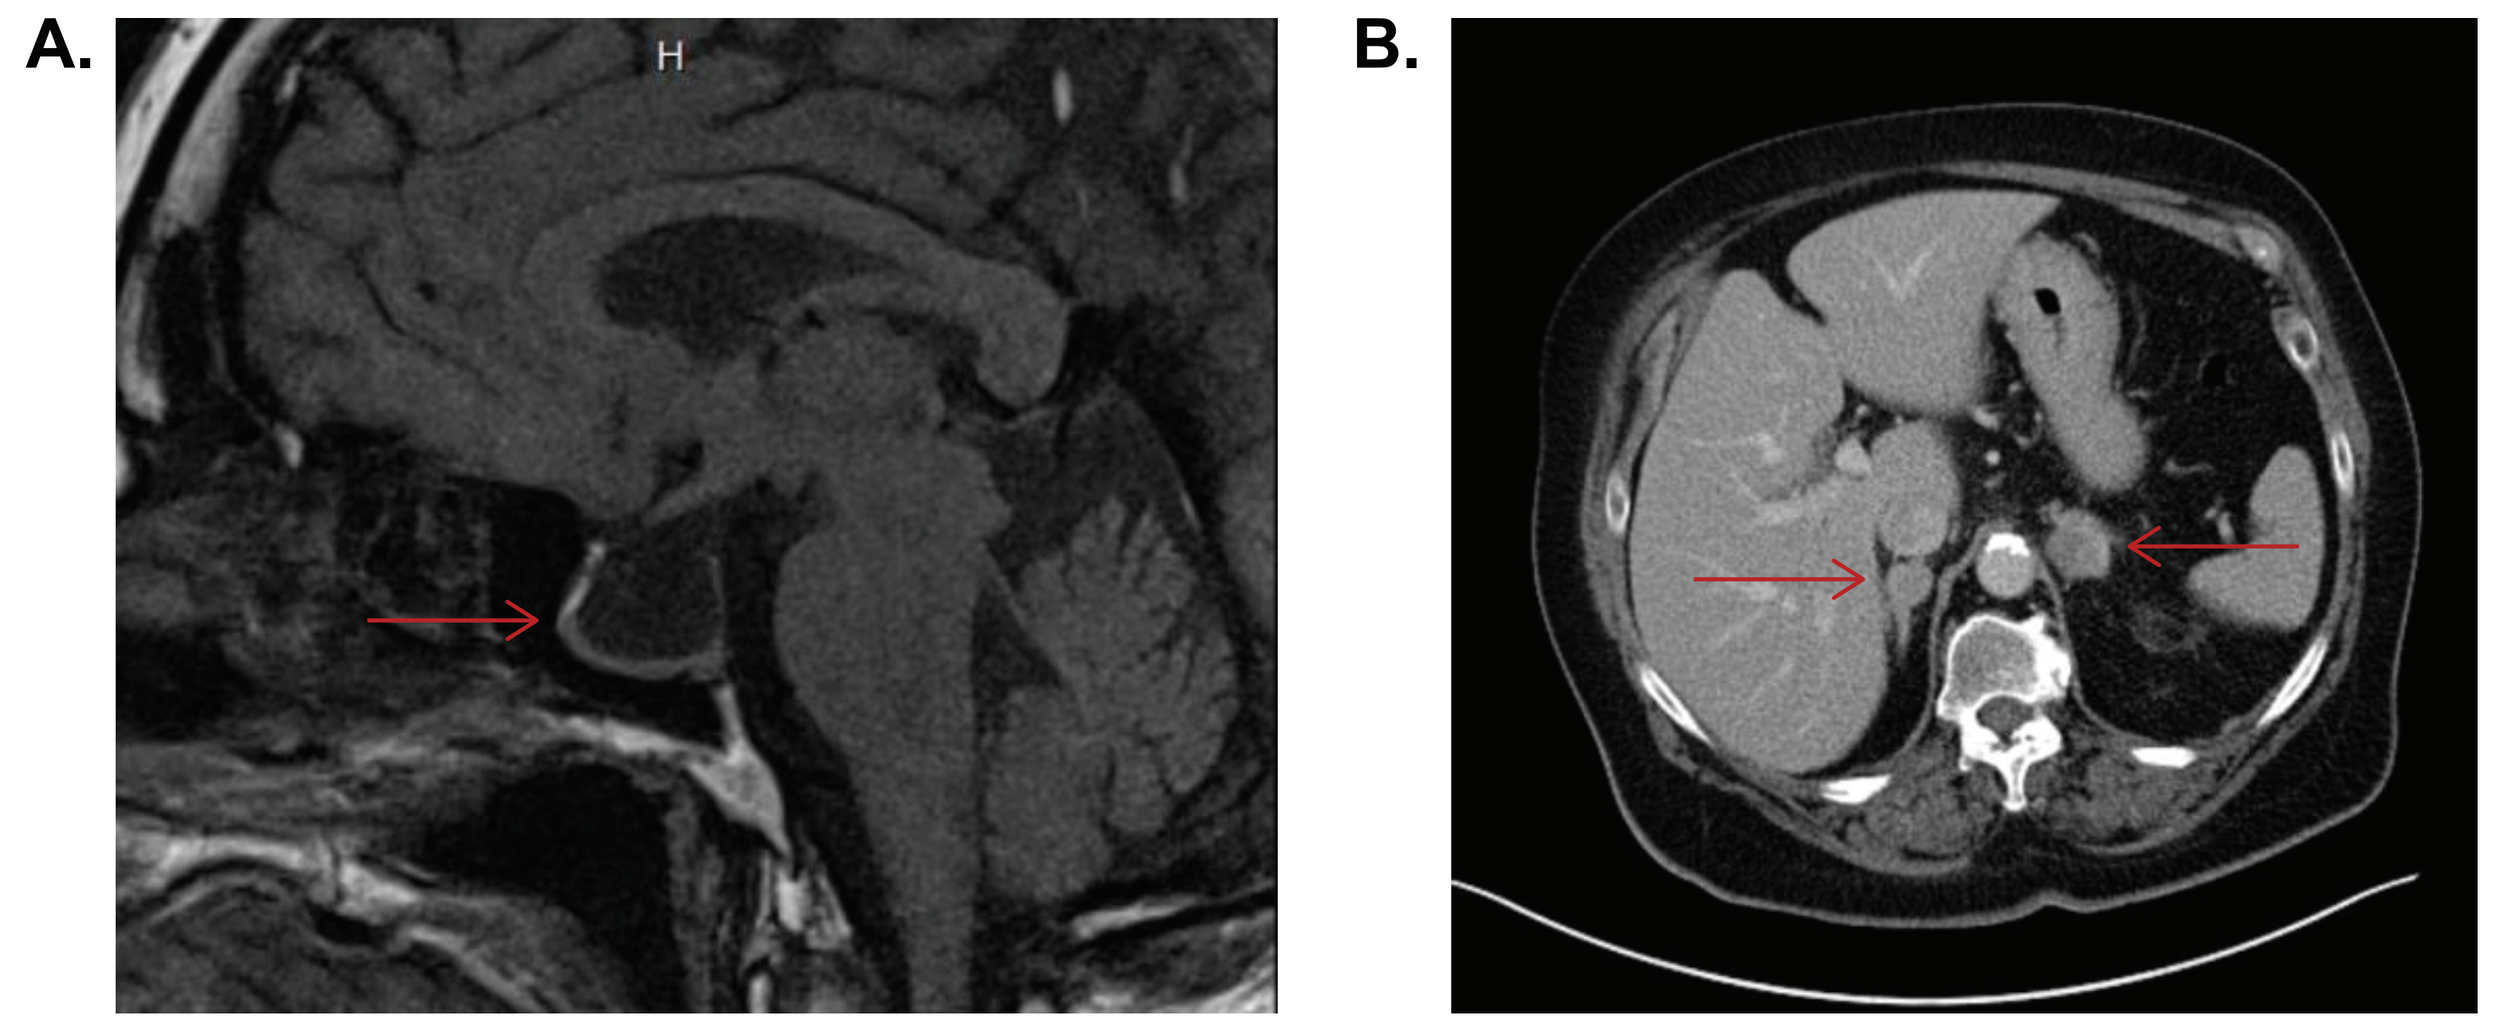
A.
B.

Pituitary MRI was in 2012 with patient on pegvisomant following transsphenoidal debulking and gamma knife therapy for acromegaly. Adrenal CT was in 2012 at time of diagnosis of Cushing's syndrome.

Treatment for Cushing's Syndrome

- A 9-month trial of ketoconazole (max dose 600 mg/day) resulted in no change in symptoms or hypercortisolism and was discontinued
- Mifepristone 300 mg/day (Korlym®, Corcept Therapeutics), a glucocorticoid receptor antagonist, was initiated
- Spironolactone 100 mg/day, an aldosterone receptor antagonist, was added to preempt possible hypokalemia associated with mifepristone

Combination Therapy with Receptor Antagonists: Outcomes

- Three separate receptor antagonists were utilized in this patient
 - Pegvisomant – Growth hormone receptor antagonist
 - Mifepristone – Glucocorticoid receptor antagonist
 - Spironolactone – Mineralocorticoid (aldosterone) receptor antagonist
- The patient displayed marked improvements in her Cushingoid features, achieved persistent weight loss (**Figure 3A**), and regained normal muscle strength
- After ~20 years of pegvisomant, she was able to decrease the dose to 25 mg sc QD when IGF-1 dropped to 91 ng/mL, IGF-1 SDS -0.6
- Fasting blood glucose and A1c decreased despite glimepiride discontinuation and the reduction in pegvisomant dose (**Figure 3B**)
- Blood pressured decreased (**Figure 3C**) and potassium remained within the normal range with no signs of hypokalemia
- Cholesterol and triglycerides improved with a maintenance dose of atorvastatin 40 mg (**Figure 3D**)

Figure 3A-D. Patient Improvements Across 3 Years of Treatment with Mifepristone

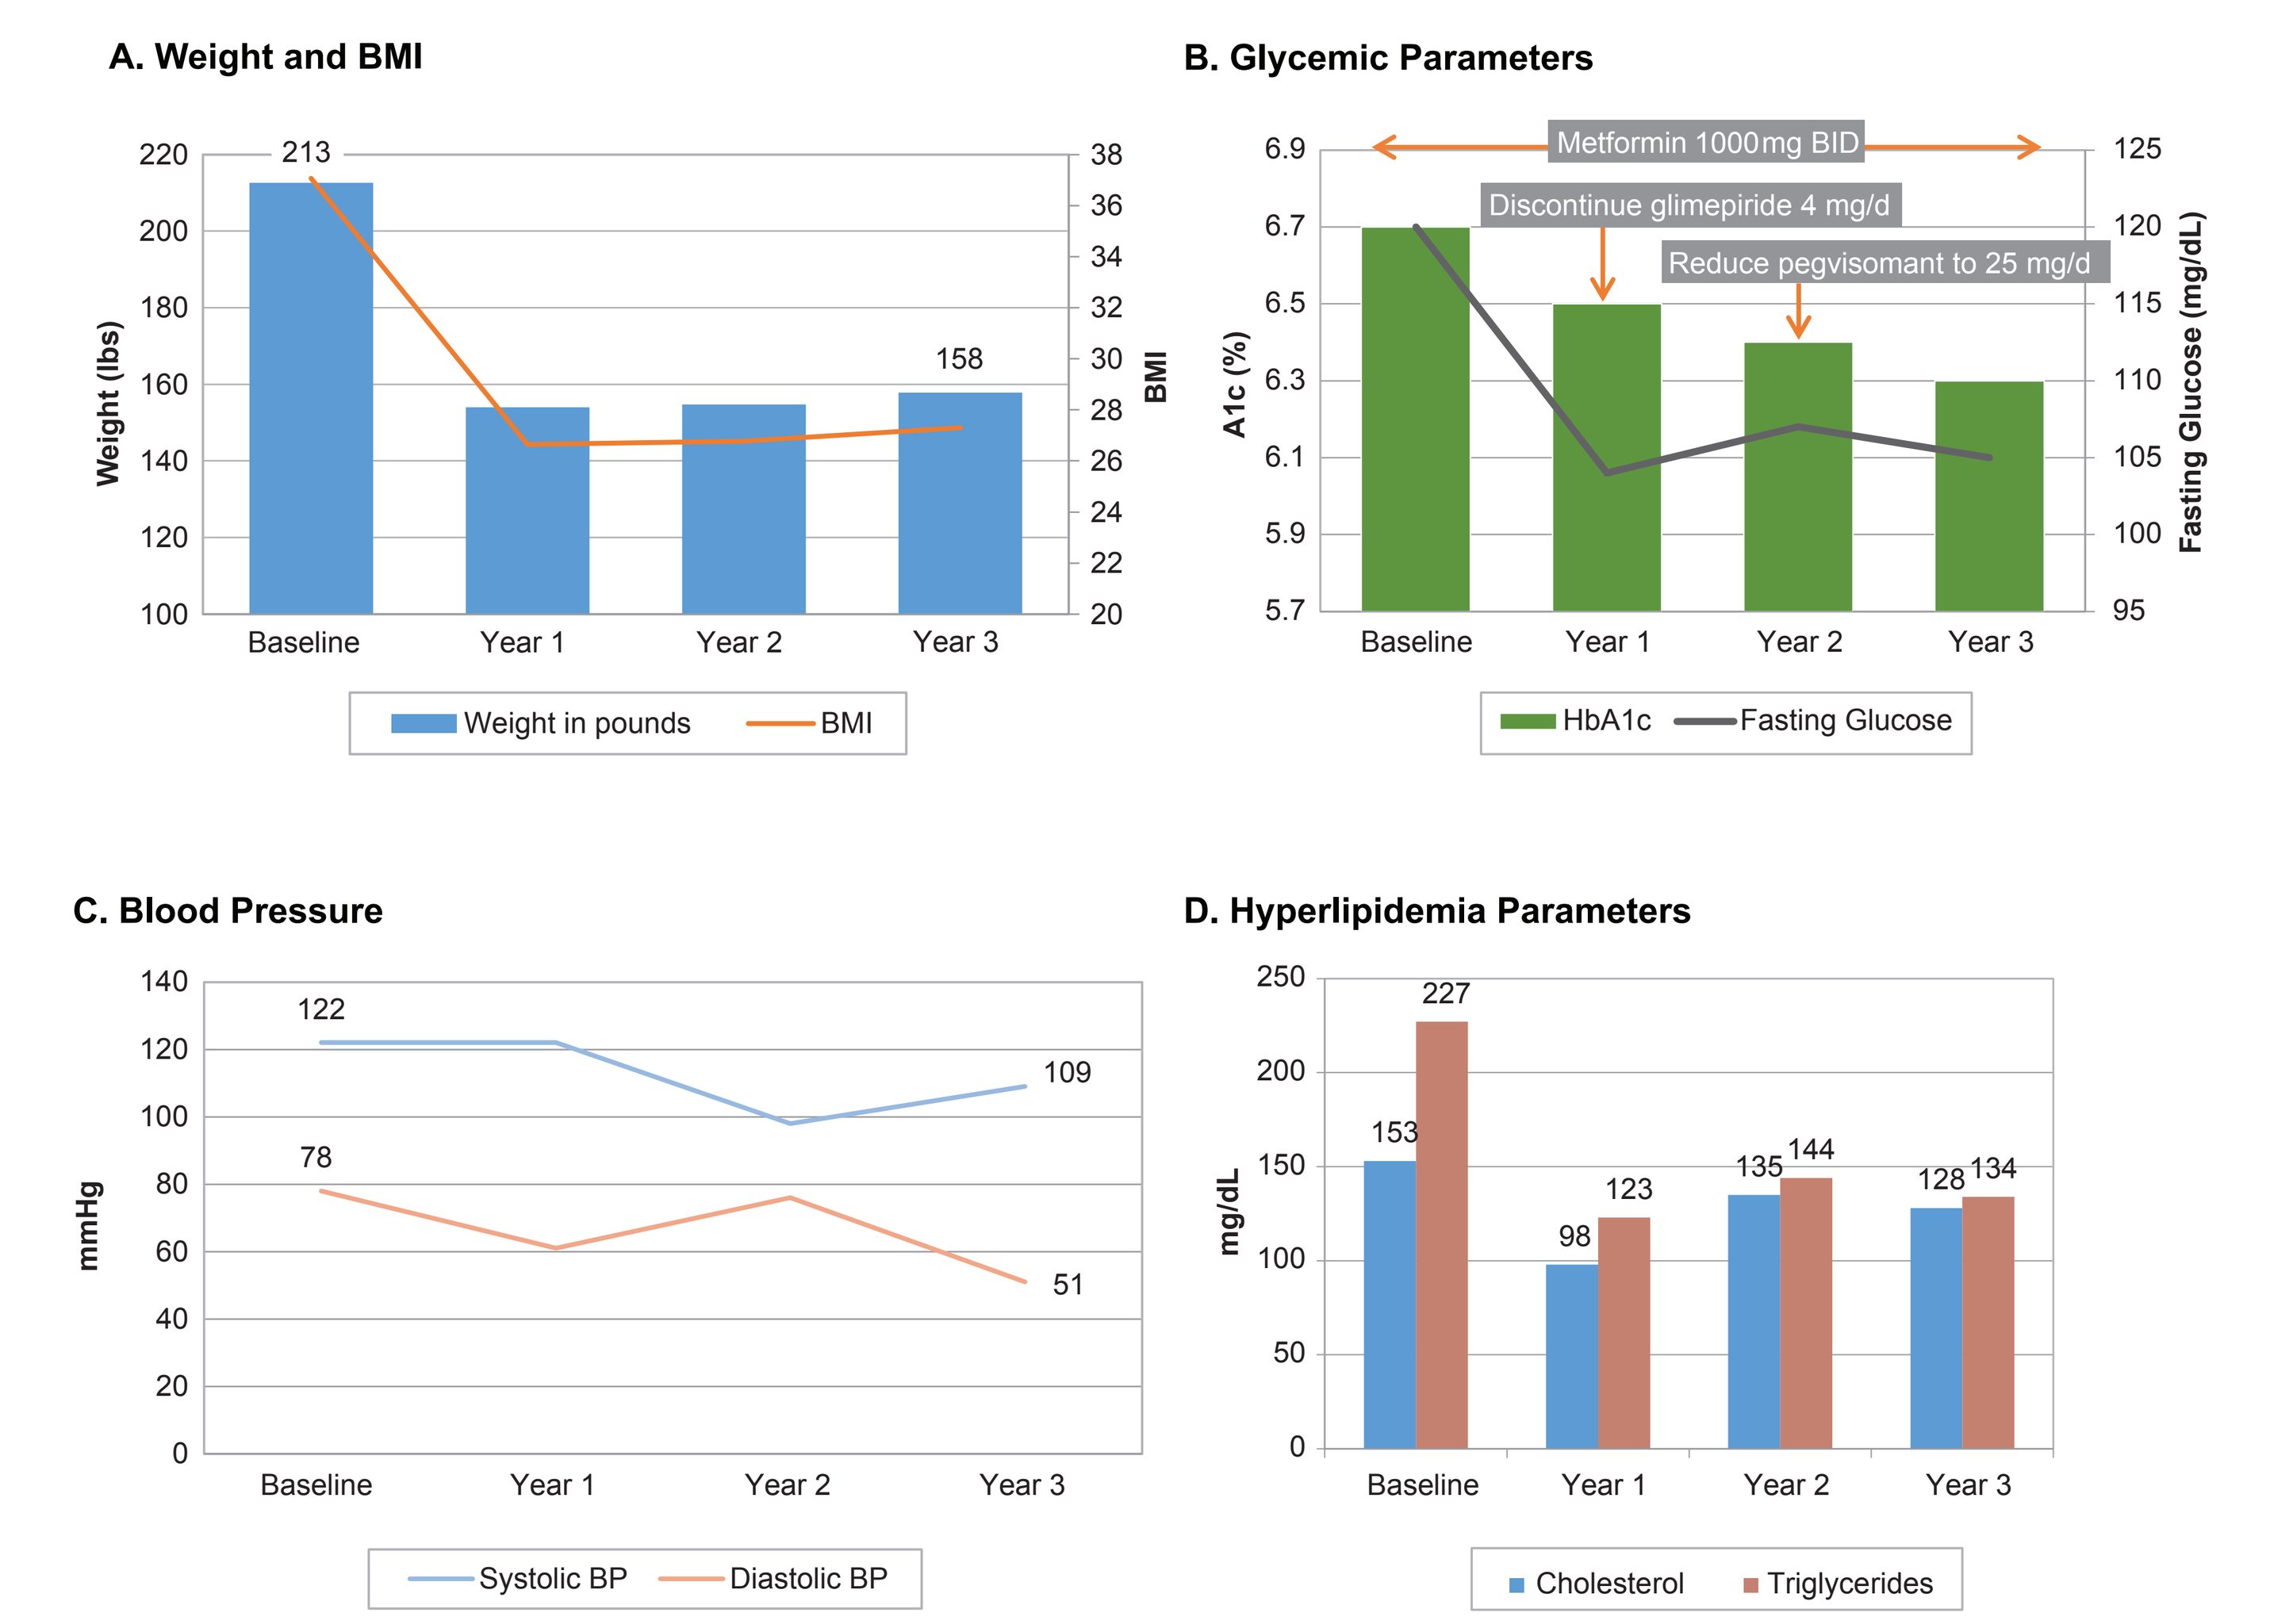
A. Weight and BMI
Weight (lbs)
BMI
213
158
Baseline
Year 1
Year 2
Year 3
Weight in pounds
BMI
B. Glycemic Parameters
A1c (%)
Fasting Glucose (mg/dL)
Metformin 1000mg BID
Discontinue glimepiride 4 mg/d
Reduce pegvisomant to 25 mg/d
HbA1c
Fasting Glucose
C. Blood Pressure
mmHg
122
78
109
51
Systolic BP
Diastolic BP
D. Hyperlipidemia Parameters
mg/dL
153
227
98
123
135
144
128
134
Cholesterol
Triglycerides

Patient was taking glimepiride at baseline, which was discontinued at Year 1. Patient was also taking atorvastatin from baseline through Year 3.

SUMMARY/DISCUSSION

- Monitoring and management of comorbidities associated with acromegaly—such as hypertension, diabetes, and hyperlipidemia—are critically important because cardiovascular and cerebrovascular events are the primary cause of death in acromegaly
- When these comorbidities persist despite adequate control of acromegaly, other endocrine causes should be investigated
- Published case reports[1-3] have demonstrated treatment of ACTH-independent Cushing's syndrome with adrenalectomy can result in improved clinical outcomes and acromegaly control with reduced GH/IGF-1 secretion
- Our case is the first to demonstrate a decrease in dose requirement of pegvisomant after successful treatment of Cushing's syndrome with a glucocorticoid receptor antagonist, mifepristone
- Our case is also the first to report combination therapy with three hormone receptor antagonists
- The synergistic effects of mifepristone and pegvisomant resulted in improved metabolic parameters, glycemic control, and achievement of IGF-1 target with a reduced dose of pegvisomant

REFERENCES

1. Ogo A, et al. A decrease in the dose of pegvisomant was needed for the treatment of acromegaly after adrenalectomy in a patient with coexisting preclinical Cushing's syndrome. *Intern Med.* 2011;50(18): 1987-91.
2. Uchida D, et al. Glucocorticoid-dependency on GH secretion and tumor growth in a GH-producing pituitary adenoma with Cushing's syndrome. *Endocr J.* 2000;47(suppl):S69-S71.
3. Kim SK, et al. Dramatic improvement of diabetes mellitus following the treatment of coexisting acromegaly and Cushing's syndrome. *Intern Med.* 2015;54(19):2471-4.

ACKNOWLEDGMENTS

Medical editorial assistance was provided by MedVal Scientific Information Services, LLC, Skillman, NJ and was sponsored by Corcept Therapeutics.

DISCLOSURES

MBG: Principal Investigator, Corcept, Pfizer. RB: Employee, Corcept. KLS, MSG: Nothing to disclose.

Presented at the Endocrine Society Annual Meeting (ENDO 2016), April 1-4, 2016, Boston, Massachusetts