LBSun-22

No Incidence of Adrenal Insufficiency (AI) in Normo-cortisolemic Patients Treated With Mifepristone (MIFE): An Analysis of 5 Placebo-Controlled Clinical Trials

David Cram, PharmD; Andreas Moraitis, MD; Dat Nguyen, PharmD
Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Adrenal insufficiency (AI) is a major concern for all surgical and medical treatments of Cushing's syndrome (CS). We analyzed the incidence of AI in patients with normal cortisol levels treated with MIFE (Korlym®, Corcept Therapeutics), a selective glucocorticoid receptor (GR) antagonist approved for the treatment for CS.

Methods: To analyze the incidence of AI, data from 5 similar placebo-controlled phase 3 studies evaluating MIFE (300 mg-1200 mg/d for 7 d) for the treatment of Psychotic Depression were pooled (MIFE n=833, placebo n=627). The protocols required investigators to specifically assess for AI at each clinic visit and report all adverse events (AEs).

Results: Subjects (avg. age 44.7±11.4 yrs) were treated for 7 d with MIFE and had clinical assessments at baseline, Days 7, 14, 28, 42, and 56. MIFE doses were 300 mg (n=110), 600 mg (n=471), 1200 mg (n=252), and placebo (n=627). At baseline, subjects were normo-cortisolemic, with mean serum cortisol 12.9±5.8 µg/dL (n=600 placebo), 12.3±5.7 µg/dL (n=802, MIFE); mean ACTH 22.5±15.1 pg/mL (n=572 placebo), 19.5±12.5 pg/mL (n=774, MIFE). As expected with MIFE treatment, mean ACTH & serum cortisol increased significantly and were dose-dependent, indicating adequate GR binding. No events of AI or similar terminology (Addisonian crisis, adrenal crisis, or hypoadrenalism) were reported in either treatment groups even though investigators were required to specifically assess for the potential of AI at each clinic visit. Additionally, our analysis of patients with ≥2 concurrent adverse events (dizziness/dizzy, fatigue, weakness, hypoglycemia, hypotension, lethargy, malaise, nausea, orthostatic hypotension, syncope, vomiting) that potentially could be associated with AI revealed no statistically significant differences between placebo and MIFE.

Discussion: The development of AI is a serious concern for clinicians treating CS. Generalized signs & symptoms of cortisol withdrawal, an indicator of successful treatment with MIFE in patients with CS, are often mistaken for AI. Data from this large population of patients with normal ACTH and cortisol production treated with MIFE did not yield any cases of AI nor did it present any combination of symptoms that could be attributed to AI.

INTRODUCTION

- The development of adrenal insufficiency, especially Addisonian crisis, is a major concern for both surgical and medical interventions used in the treatment of Cushing's syndrome
- The clinical signs and symptoms of adrenal insufficiency are often nonspecific, making early detection and diagnosis difficult (**Table 1**)[1]
- Treatment with mifepristone (Korlym®, Corcept Therapeutics), an effective glucocorticoid receptor antagonist, presents an additional challenge because receptor antagonism disrupts the cortisol production feedback mechanism (**Figure 1**)
- The resulting increase in cortisol and adrenocorticotropic hormone (ACTH) makes the measurement of cortisol meaningless in establishing the diagnosis of adrenal insufficiency
- Additionally, signs and symptoms of cortisol withdrawal, indicators of successful mifepristone treatment (**Table 2**),[2] can be mistaken for adrenal insufficiency
- This analysis assesses the impact of mifepristone administration on patients with normal cortisol levels in regard to developing adrenal insufficiency

Table 1. Clinical Manifestations of Chronic Adrenal Insufficiency[1]

	Approx. Frequency (% of patients)
Symptoms	
Weakness/tiredness/fatigue	100
Anorexia	100
Nausea	86
Vomiting	75
Constipation	33
Abdominal pain/discomfort	31
Diarrhea	16
Salt craving	16
Postural dizziness	12
Muscle/joint pain	6-13
Signs	
Weight loss	100
Hyperpigmentation	94
Hypotension	88-94
Lab Abnormalities	
Hyponatremia	88
Hyperkalemia	64
Hypercalcemia	6

Figure 1. Glucocorticoid receptor (GR) antagonism disrupts the cortisol production feedback mechanism. Mifepristone competes with cortisol to bind GR receptors. By antagonizing these receptors in the hypothalamus and pituitary gland, mifepristone interferes with the negative feedback mechanism that regulates cortisol production. Without negative feedback, an increase in adrenocorticotropic hormone (ACTH) stimulates production of more cortisol.

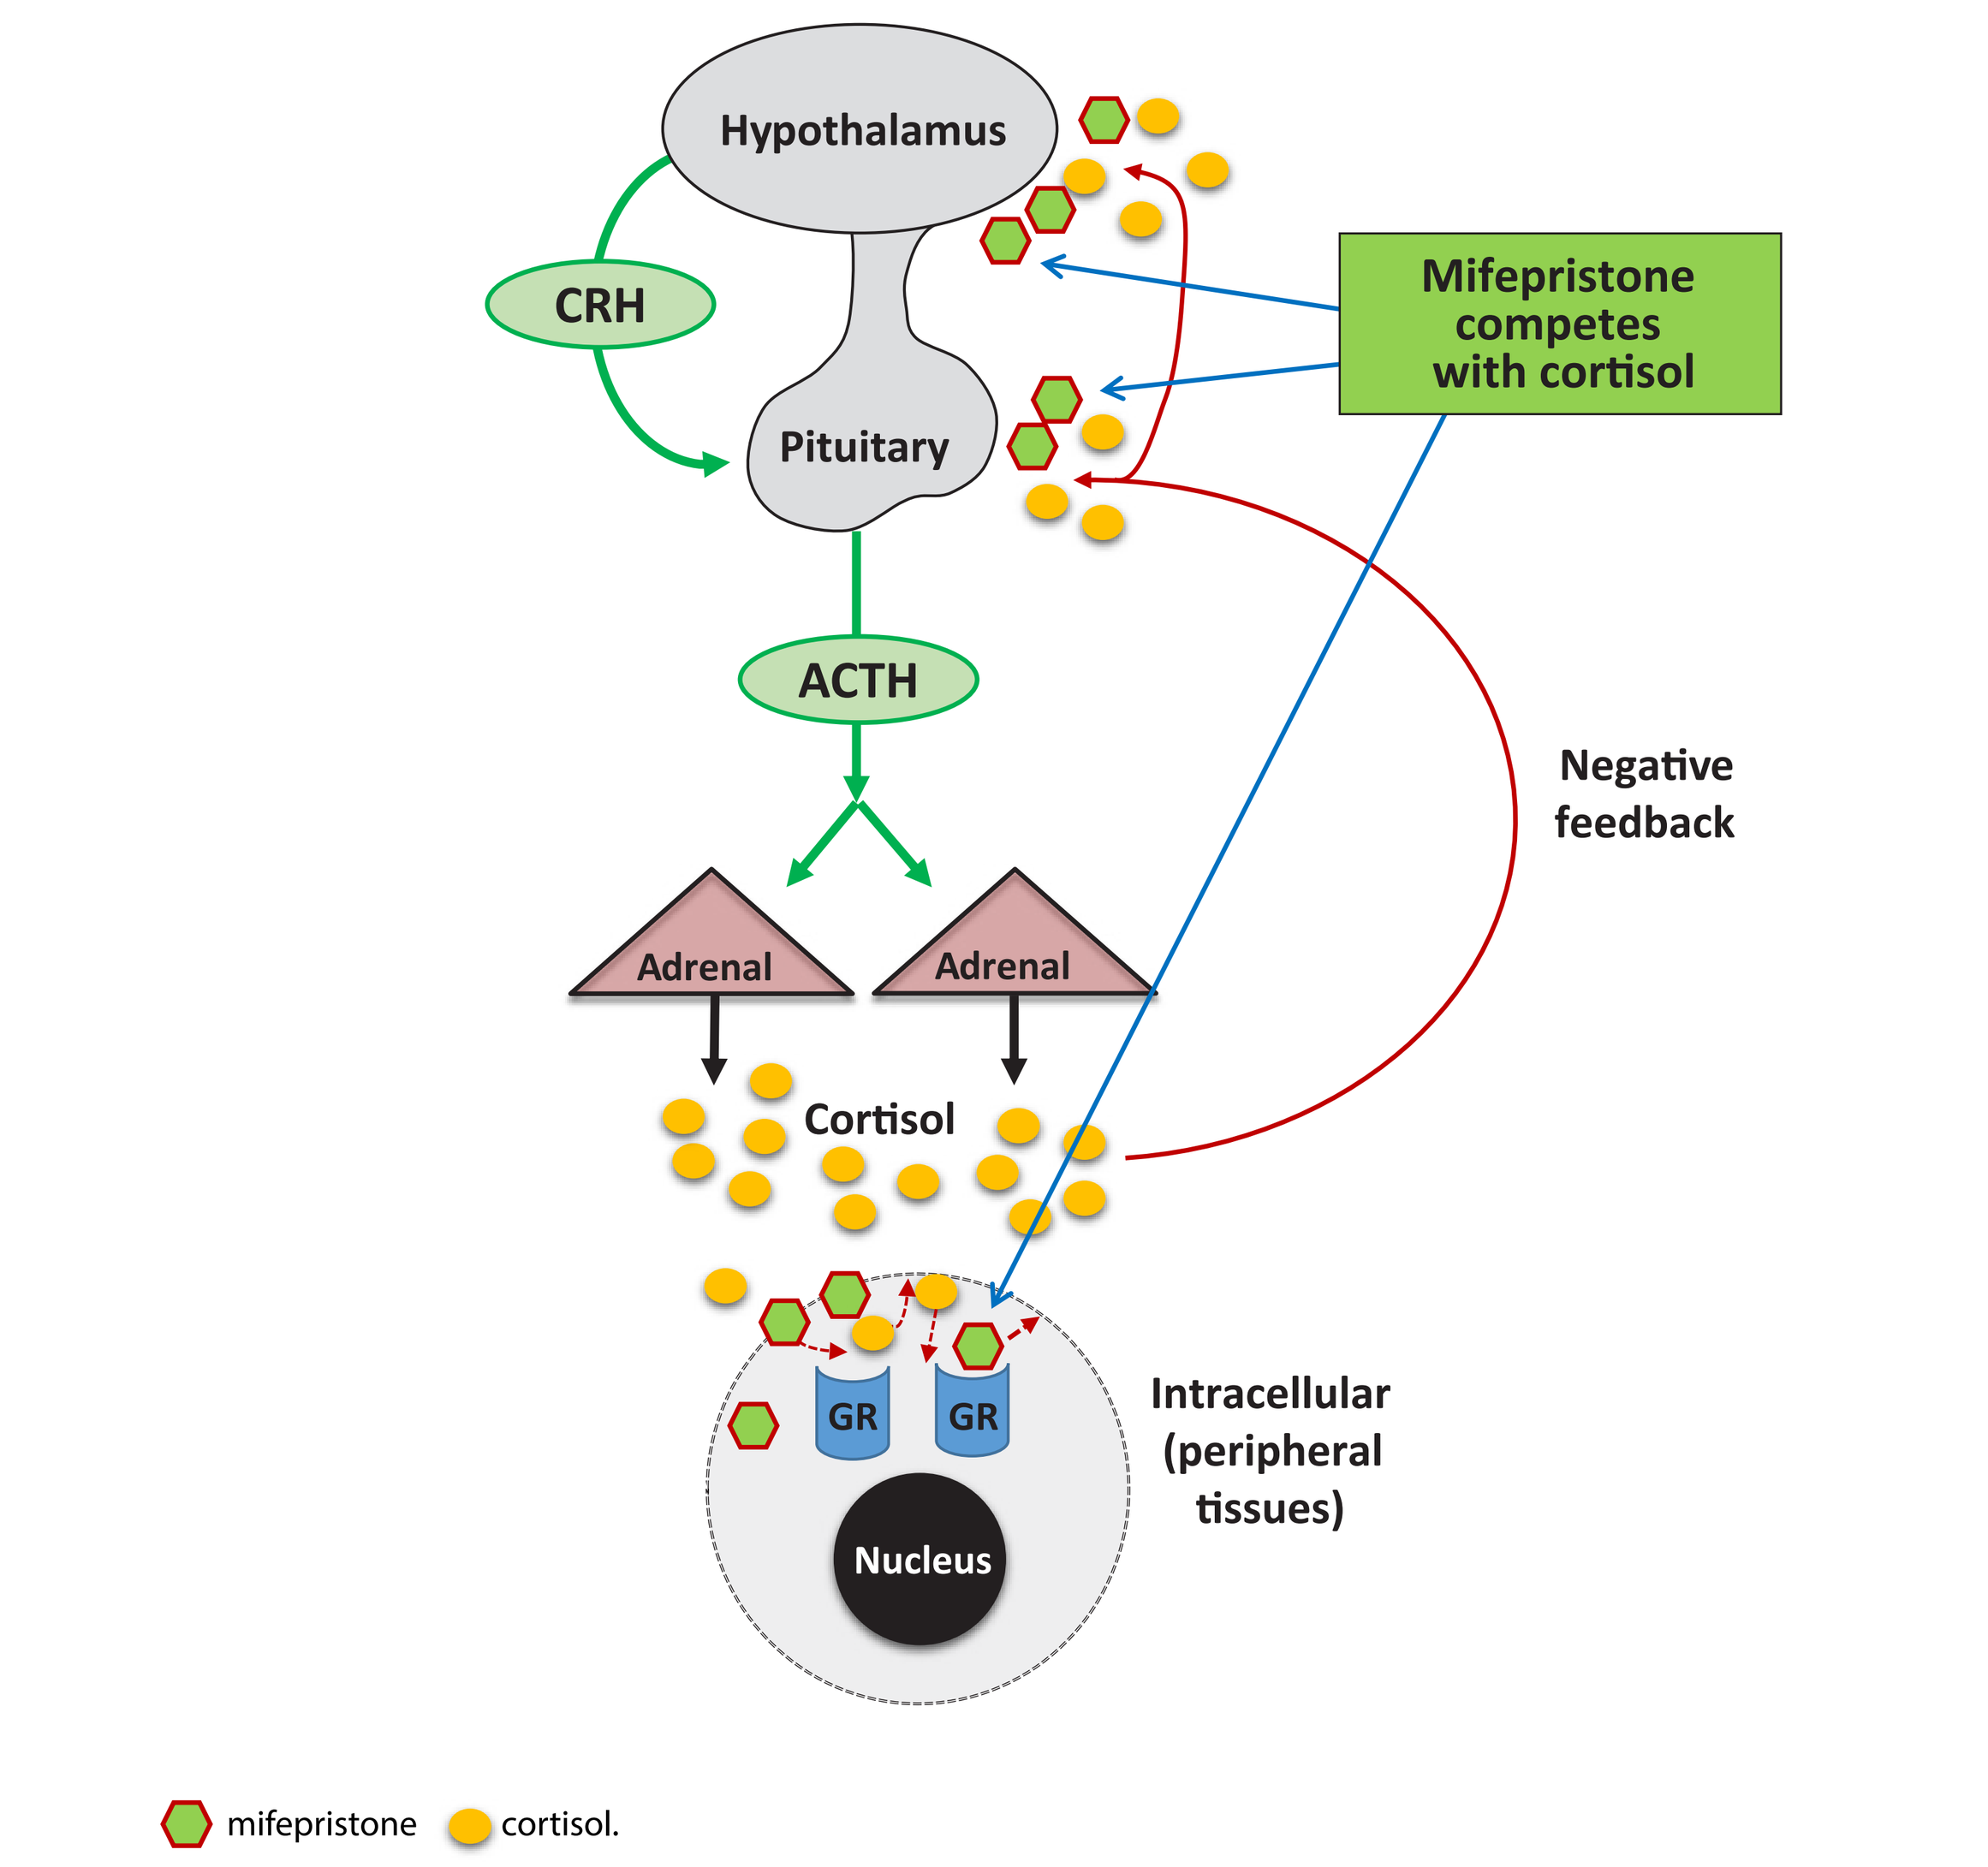

Hypothalamus
CRH
Pituitary
Mifepristone competes with cortisol
Negative feedback
ACTH
Adrenal
Adrenal
Cortisol
GR
GR
Nucleus
Intracellular (peripheral tissues)
mifepristone
cortisol.

Table 2. Treatment-Emergent Adverse Events in Cushing's Syndrome Patients Receiving Mifepristone[2,3]

Adverse Event	Percent (%) of Patients (n=50)
Nausea	48
Fatigue	48
Blood potassium decreased	34
Arthralgia	30
Vomiting	26
Hypertension	24
Dizziness	22
Decreased appetite	20
Dyspnea	16
Myalgia	14
Diarrhea	12
Anorexia	10
Constipation	10
Somnolence	10

METHODS

Study Design

- Data from 5 similar placebo-controlled phase 3 studies evaluating mifepristone for the treatment of patients with psychotic depression were pooled
- Subjects were randomized to 7 days of double-blind treatment with mifepristone or placebo
- Mifepristone doses varied across studies (300 mg, 600 mg, or 1200 mg per day)
- Clinical assessments were made at baseline and at Days 7, 14, 28, 42, and 56
- Investigators were required to specifically assess for adrenal insufficiency at each clinic visit and report all adverse events (AEs)
- Blood samples were collected at baseline and Day 7 for determination of cortisol and ACTH levels

Analysis Plan

- Data from all 5 studies were available for statistical analyses, representing mifepristone doses of 300 mg (n=110), 600 mg (n=471), and 1200 mg (n=252), and placebo (n=627)
- Results are presented as all doses combined (300 mg + 600 mg + 1200 mg, n=833) vs placebo (n=627)
- A systematic search of the safety database was conducted for incidence of adrenal insufficiency:
 - Analysis 1: Search for subjects with AEs due to Addisonian crisis, adrenal crisis, adrenal insufficiency, or hypoadrenalism
 - Analysis 2: Search for subjects with ≥2 concurrent AEs (dizziness/dizzy, fatigue, weakness, hypoglycemia, hypotension, lethargy, malaise, nausea, orthostatic hypotension, syncope, vomiting) that could be associated with adrenal insufficiency

RESULTS

- At baseline, subjects (mean age 44.7±11.4 years) were normo-cortisolemic (**Table 3**)
- After 7 days of mifepristone, mean ACTH and serum cortisol increased significantly and were dose-dependent (**Table 4**)
- Adverse events from all 5 studies (n=1,460) were evaluated by dose group and by all doses combined; treatment-emergent adverse events (TEAEs) were reported in 556/833 mifepristone-treated patients (66.7%) and 386/627 placebo-treated patients (61.6%) (**Table 5**)
- The safety of mifepristone vs placebo was comparable, with generally similar numbers of events noted in the two groups (**Table 5**)
- No events of adrenal insufficiency or similar terminology were reported in either treatment group
- Analysis of subjects with ≥2 concurrent adverse events that potentially could be associated with adrenal insufficiency revealed very few affected patients in either the mifepristone or placebo groups, with no statistically significant differences between them (**Table 6**)
 - Even at the highest dose of 1200 mg, there were no subjects with ≥2 concurrent adverse events

Table 3. Baseline Characteristics of Study Subjects

	Mifepristone (n=833)	Placebo (n=627)	Total (N=1,460)
Age, years (mean±SD) (range, 18-77)	44.7 (11.6)	44.7 (11.2)	44.7 (11.4)
Gender (% female)	57.9	60.5	59.0
ACTH mean (±SD)	19.6±12.5 pg/mL (n=778)	22.5±15.1 pg/mL (n=572)	20.8±13.7 pg/mL (n=1,350)
Serum cortisol (mean±SD)	12.1±5.4 µg/dL (n=807)	12.4±5.4 µg/dL (n=601)	12.2±5.4 µg/dL (n=1,408)

ACTH, adrenocorticotropic hormone.

Table 4. Change in Mean ACTH and Mean Serum Cortisol After 7 Days

	Mifepristone			
	300 mg	600 mg	1200 mg	Placebo
ACTH (pg/mL)				
Baseline	17.8	19.5	20.3	22.5
Day 7	33.0	51.0	65.7	23.4
Change (%)	113.9	216.0	287.9	16.2
Cortisol (µg/dL)				
Baseline	11.9	13.0	10.4	12.4
Day 7	23.1	30.8	30.5	12.7
Change (%)	123.7	167.9	237.4	16.2

ACTH, adrenocorticotropic hormone.

Table 5. Adverse Events (AEs) With Frequency of ≥5% in Mifepristone Studies in Patients with Psychotic Depression (N=1,460)

	Mifepristone				
Adverse Event (%)	300 mg (n=110)	600 mg (n=471)	1200 mg (n=252)	All Doses (n=833)	Placebo (n=627)
Subjects with ≥1 TEAE	66.4	62.2	75.4	66.7	61.6
Nausea	18.2	14.0	17.5	15.6	11.2
Headache	18.2	9.3	23.8	14.9	11.6
Dizziness	8.2	6.6	11.5	8.3	5.4
Dry mouth	4.5	5.1	11.5	7.0	5.4
Diarrhea	9.1	4.0	9.1	6.2	5.1
Constipation	2.7	4.2	9.5	5.6	5.7
Insomnia	3.6	5.7	5.2	5.3	6.2
Fatigue	8.2	3.6	5.6	4.8	3.3
Vomiting	4.5	3.8	7.5	5.0	2.6
Anxiety	5.5	3.0	5.2	4.0	3.7
Dyspepsia	1.8	2.1	9.5	4.3	3.2
Somnolence	7.3	2.8	3.2	3.5	3.8
Back pain	6.4	1.7	2.8	2.6	2.7
Rash	0.9	1.7	5.2	2.6	1.9

Multiple AEs for specific AE preferred term are only counted once for each subject. Listed by highest percentage order in last column. Preferred term is listed if a frequency of at least 5% in any column is calculated. TEAE, treatment-emergent adverse event.

Table 6. Subjects With ≥2 Concurrent Adverse Events (AEs) Potentially Associated With Adrenal Insufficiency

	Mifepristone (n=833)	Placebo (n=627)	*P*-value (Fisher's exact test)
2 Concurrent AEs, n (%)	13 (1.56)	4 (0.64)	0.1392
3 Concurrent AEs	4 (0.4)	0	0.1395
4 Concurrent AEs	0	1 (0.16)	1.0
ALL	17 (2.04)	5 (0.80)	0.0802

DISCUSSION

- In patients with hypercortisolism (Cushing's syndrome), glucocorticoid receptor antagonism with mifepristone can produce signs and symptoms of cortisol withdrawal, which may be mistaken for adrenal insufficiency
- Our analysis of patients with normal cortisol levels treated with mifepristone indicated no evidence of adrenal insufficiency, even at the highest doses

REFERENCES

1. UpToDate. Clinical manifestations of chronic adrenal insufficiency. http://www.uptodate.com/contents/clinical-manifestations-of-adrenal-insufficiency-in-adults?source=search_result&search=adrenal+insufficiency&selectedTitle=1%7E150.
2. Fleseriu M, et al. Mifepristone, a glucocorticoid receptor antagonist, produces clinical and metabolic benefits in patients with Cushing's syndrome. *J Clin Endocrinol Metab*. 2012;97(6):2039-49.
3. Korlym® (mifepristone) [prescribing information]. Corcept Therapeutics, Menlo Park, CA; June 2013.

ACKNOWLEDGMENTS

Funding to support the preparation of this poster was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ.

DISCLOSURES

DC, AM, DN: Employees, Corcept.

Presented at the Endocrine Society Annual Meeting (ENDO 2016), April 1-4, 2016, Boston, Massachusetts