MON-402

Mifepristone Treatment Effectively Improved Clinical Symptoms of Severe Hypercortisolism in a Patient With Primary Macronodular Adrenal Hyperplasia (PMAH) Allowing Surgical Treatment and Subsequent Conception

Jennifer U. Mercado, ARNP[1]; Kelley Moloney, ARNP[1]; Precious J. Lim, PhD[2]; Kevin C.J. Yuen, MD, FRCP (UK), FACE[1]

[1]Swedish Pituitary Center, Departments of Neuroendocrinology and Neurosurgery, Swedish Neuroscience Institute, Seattle, WA; [2]Corcept Therapeutics, Menlo Park, CA

ABSTRACT

Background: Endocrine Society Guidelines recommend surgical resection of the causal lesion/s as the first-line treatment for Cushing's syndrome.[1] However, chronic severe hypercortisolism renders the patient high-risk for adverse surgical outcomes, and medical therapy could be considered for preoperative risk optimization. We present a patient with PMAH effectively treated with mifepristone (MIFE), a competitive glucocorticoid receptor antagonist, prior to undergoing bilateral adrenalectomy (BLA).

Case: A 27 yo woman presented with severe hypercortisolism: new onset hypertension (156/104 mmHg), pre-diabetes (A1C 6.3%), weight gain (30 kg in 2 yrs), secondary amenorrhea, dorsocervical and supraclavicular fat pads, abdominal striae, insomnia, depression, anxiety, proximal myopathy, easy bruising, hirsutism, acne, and facial plethora.

Hormonal testing showed multiple elevated 24hr UFC (729.3, 444.2, 292.3, 231.7 μg/24hr, ULN <50 μg/24hr), late night salivary cortisol (5.7, 11.2, 11.8, 13.6, 14.9, 15.4, 15.5, 28.3 nmol/L, ULN <4.1 nmol/L), and suppressed AM ACTH (<1.1 pg/mL, normal 7.2-63.3 pg/mL). Abdominal CT revealed symmetrically enlarged adrenal glands with nodules measuring 1.5-2.9 cm.

Her comorbidities precluded immediate BLA, and MIFE was initiated to optimize the patient's condition in preparation for surgery. After 2 weeks of MIFE (300 mg/d), improvements in blood pressure (122/94 mmHg), hyperglycemia and weight loss (10.83 kg) were noted. After 6 months of MIFE (900 mg/d), hypertension and pre-diabetes resolved, and BMI dropped from 32.43 to 25.26 kg/m^2. Patient reported lightening of striae, reduction of supraclavicular and dorsocervical fat, and improved sleep, energy, headaches, cognitive function and mood. While on MIFE, the patient experienced transient cortisol withdrawal symptoms (nausea, fatigue), which resolved without intervention.

The patient subsequently underwent BLA. Given the substantial improvement in her symptoms post-BLA, the patient was treated on low starting dose hydrocortisone (20 mg BID) and did not report cortisol withdrawal symptoms. Her menstrual periods returned spontaneously within 1 month and she is currently 9 months post-BLA with no significant symptoms, detectable ACTH levels, and was able to conceive.

Conclusion: Short-term MIFE therapy rapidly improved clinical symptoms and cardiometabolic derangements of a young woman with severe hypercortisolism due to PMAH, allowing successful BLA, and subsequent conception.

INTRODUCTION

- Severe hypercortisolism is a life-threatening condition that induces systemic complications, including pulmonary embolism, acute respiratory distress due to opportunistic infections, hypokalemia, psychoses, and uncontrolled diabetes and hypertension.
- Severe hypercortisolism is also associated with amenorrhea, anovulation, and infertility, which are troubling complications in young women desiring pregnancy.
- While Endocrine Society guidelines recommend resection of the causal lesion(s),[1] immediate surgery in patients with severe hypercortisolism can have adverse outcomes.[2]
- Medical therapy is indicated in patients in whom surgery has to be delayed due to prohibitive comorbidities.
- The aim of medical therapy in this setting is to achieve rapid control of hypercortisolism and its metabolic ramifications to minimize operative risk.
- We present an example of treatment of severe hypercortisolism due to primary macronodular adrenal hyperplasia (PMAH) with mifepristone that resulted in rapid resolution of comorbidities, thereby allowing surgery and subsequent conception in a nulliparous woman.

CLINICAL PRESENTATION

- A 27-year-old African woman presented for evaluation of possible Cushing syndrome (CS).
- She was healthy until age 24, when she began experiencing headaches, menstrual irregularities, unexplained weight gain, insomnia, depression, anxiety, nausea, dizziness, and poor memory and concentration.
- At presentation, she was found to have:
 - New-onset hypertension (156/104 mmHg)
 - Pre-diabetes (A1C 6.3%)
 - Precipitous weight gain (30 kg in 2 years)
 - Secondary amenorrhea
- She displayed dorsocervical and supraclavicular fat pads, abdominal striae, excessive body and facial hair, skin bruises, acne, and facial plethora (**Figure 1**).

Figure 1. Patient Pre- and Post-treatment

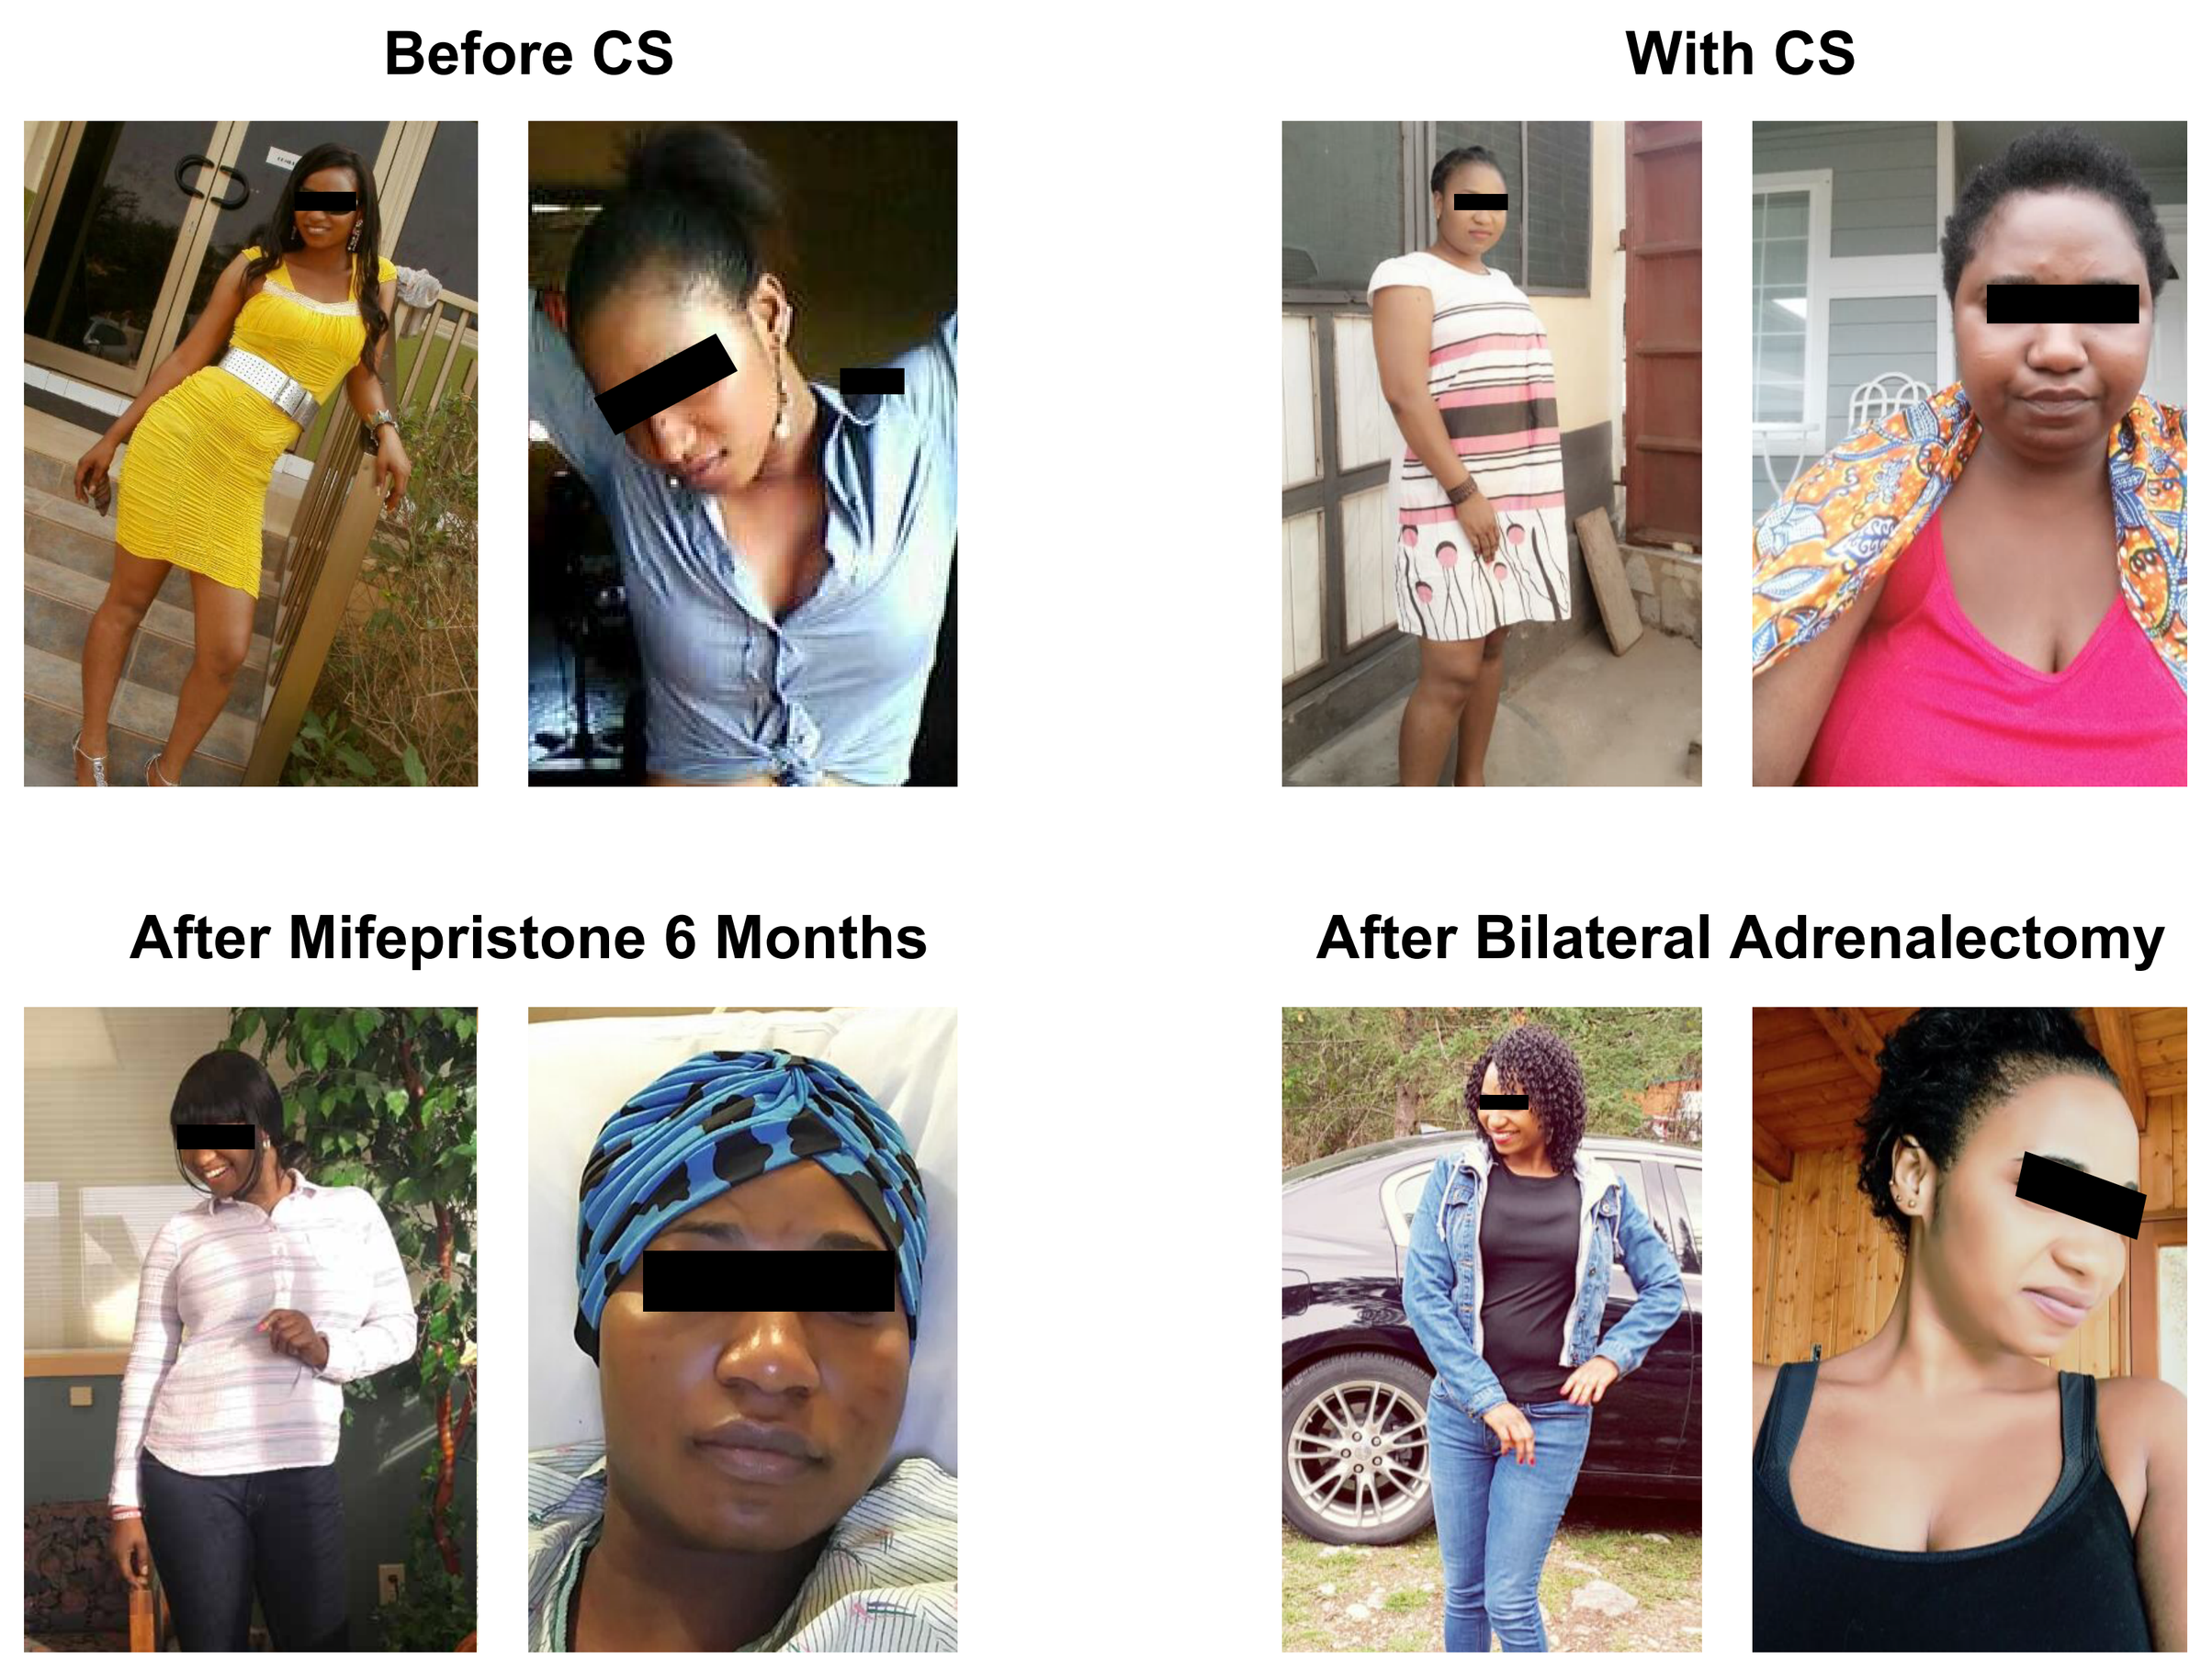
Before CS
With CS
After Mifepristone 6 Months
After Bilateral Adrenalectomy

CS, Cushing syndrome.

- Hormonal testing indicated ACTH-independent CS (**Table 1**).

Table 1. Hormonal Testing Results

Measure (reference)	Result 1	Result 2	Result 3	Result 4	Result 5	Result 6	Result 7
UFC (ULN <50 μg/24hr)	444.2	292.3	231.7				
LNSC (ULN <4.1 nmol/L)	11.2	11.8	13.6	14.9	15.4	15.5	28.3
ACTH (7.2-63.3 pg/mL)	<1.1	<1.1	<1.1				

ACTH, adrenocorticotropic hormone; LNSC, late-night salivary cortisol; UFC, urinary free cortisol.

- Abdominal CT revealed symmetrically enlarged adrenal glands with nodules measuring 1.5-2.9 cm (**Figure 2**).

Figure 2. Sagittal and Coronal Views of Abdominal CT Images Showing PMAH

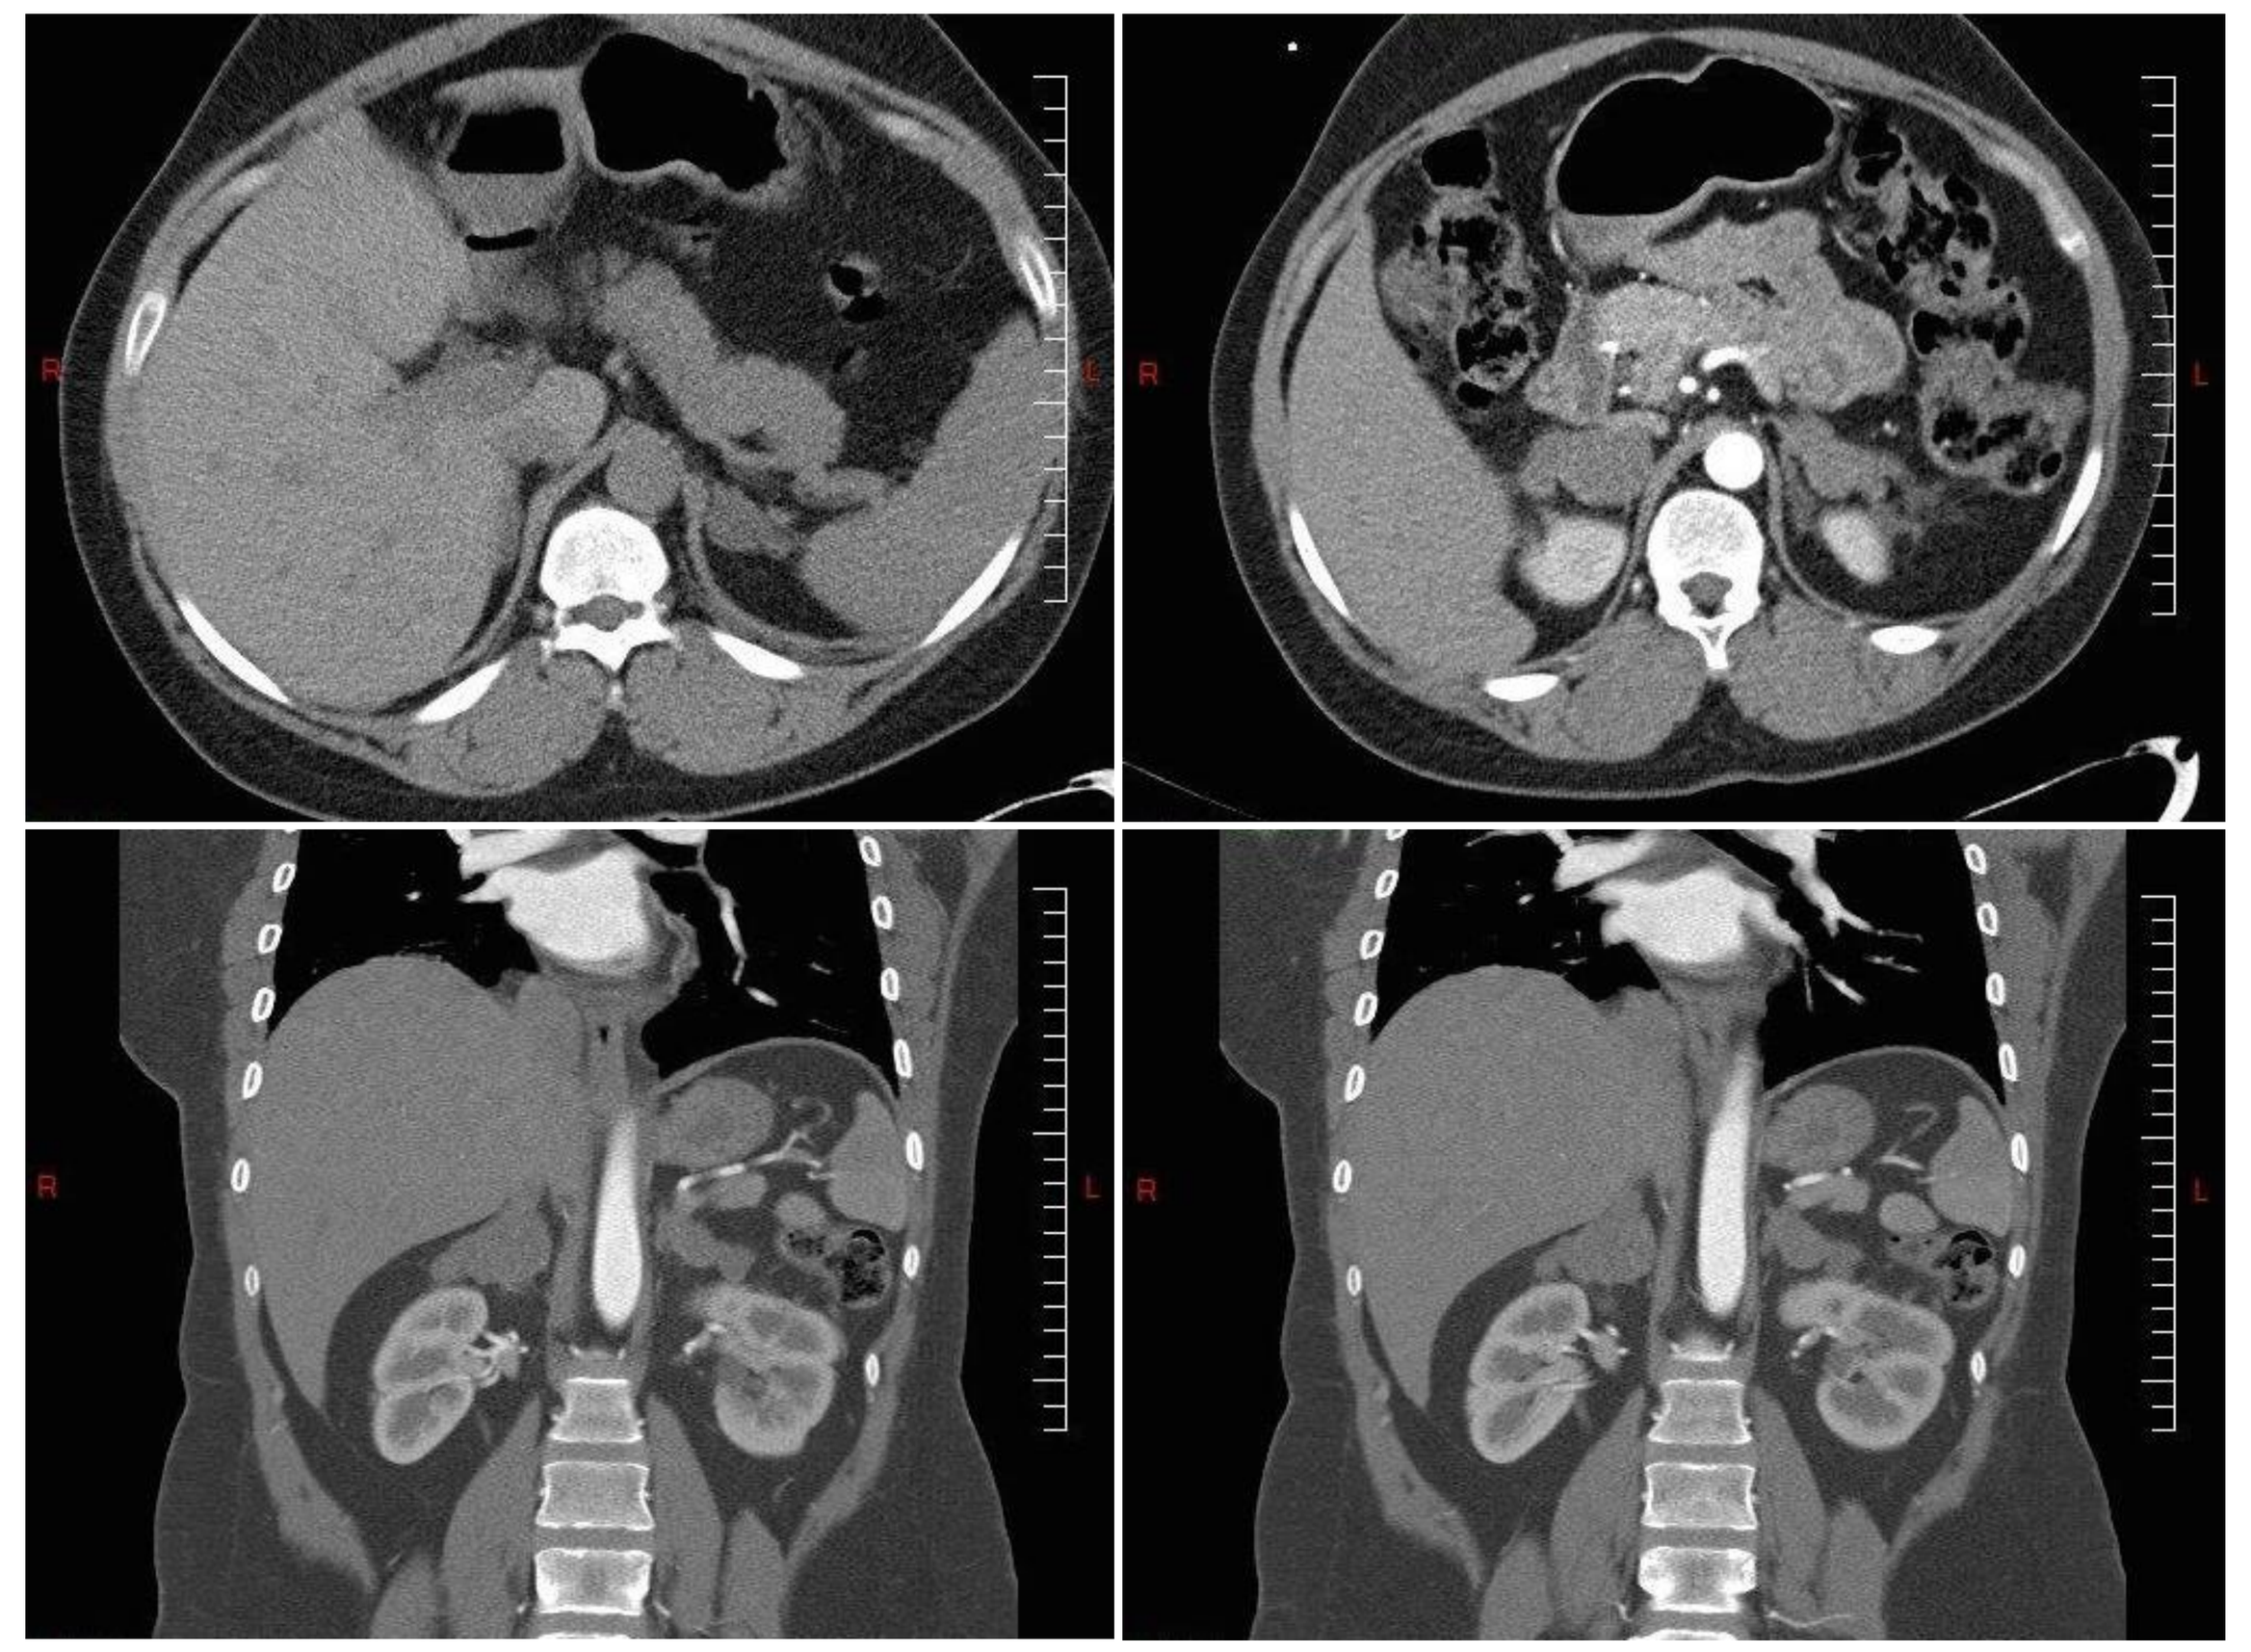

CT, computed tomography; PMAH, primary macronodular adrenal hyperplasia.

TREATMENT COURSE AND OUTCOMES

- Severe hypercortisolism placed the patient at risk for thrombo-embolic events[2] and opportunistic infections,[2] making her ineligible for immediate bilateral adrenalectomy (BLA).
- Mifepristone (Korlym®, Corcept Therapeutics Inc., Menlo Park, CA) was initiated at 300 mg/day to optimize her condition for a future BLA.
- Given mifepristone's mechanism of action, spironolactone was prophylactically administered to prevent hypokalemia (K+ baseline 3.5 mEq/L).
- After 2 weeks, improvements in blood pressure, hyperglycemia, and weight were noted (**Table 2**; **Figures 3** and **4**).
- Mifepristone was titrated to 900 mg/day over 6 months. Hypertension and diabetes resolved and BMI decreased from 32.43 to 25.26 kg/m^2 (**Table 2**; **Figures 3** and **4**).
- Palliation in CS signs and symptoms were noted: lightening of striae, reduction of supraclavicular and dorsocervical fat, decreased frequency of headaches, and improved sleep, energy, cognitive function, and mood.
- While on mifepristone, she experienced transient cortisol withdrawal symptoms that resolved without intervention.
- BLA was performed without complications 1 week after discontinuing mifepristone.
- She did not complain of symptoms of adrenal insufficiency post-BLA, and required a low peri-operative dose of hydrocortisone (20 mg BID).
- Within 1 month of BLA, her menstrual periods returned spontaneously.
- She is currently 9 months post-BLA with no significant symptoms and she has successfully conceived.

Table 2. Laboratory Parameters During Mifepristone Treatment and After Bilateral Adrenalectomy

Measure (reference)	Baseline	2 Weeks With MIFE	6 Months With MIFE	9 Months Post-BLA
BP (120/80 mmHg)	156/104	122/94	98/62	100/68
Glucose parameters				
Fasting plasma glucose (65-99 mg/dL)	115	91	85	82
A1C (<5.7%)	6.3	6.2	–	–
BMI (<30 kg/m^2)	34.28	32.43	25.26	21.94

A1C, glycated hemoglobin; BMI, body mass index; BP, blood pressure; MIFE, mifepristone.

Figure 3. Changes in BP After Treatment With Mifepristone

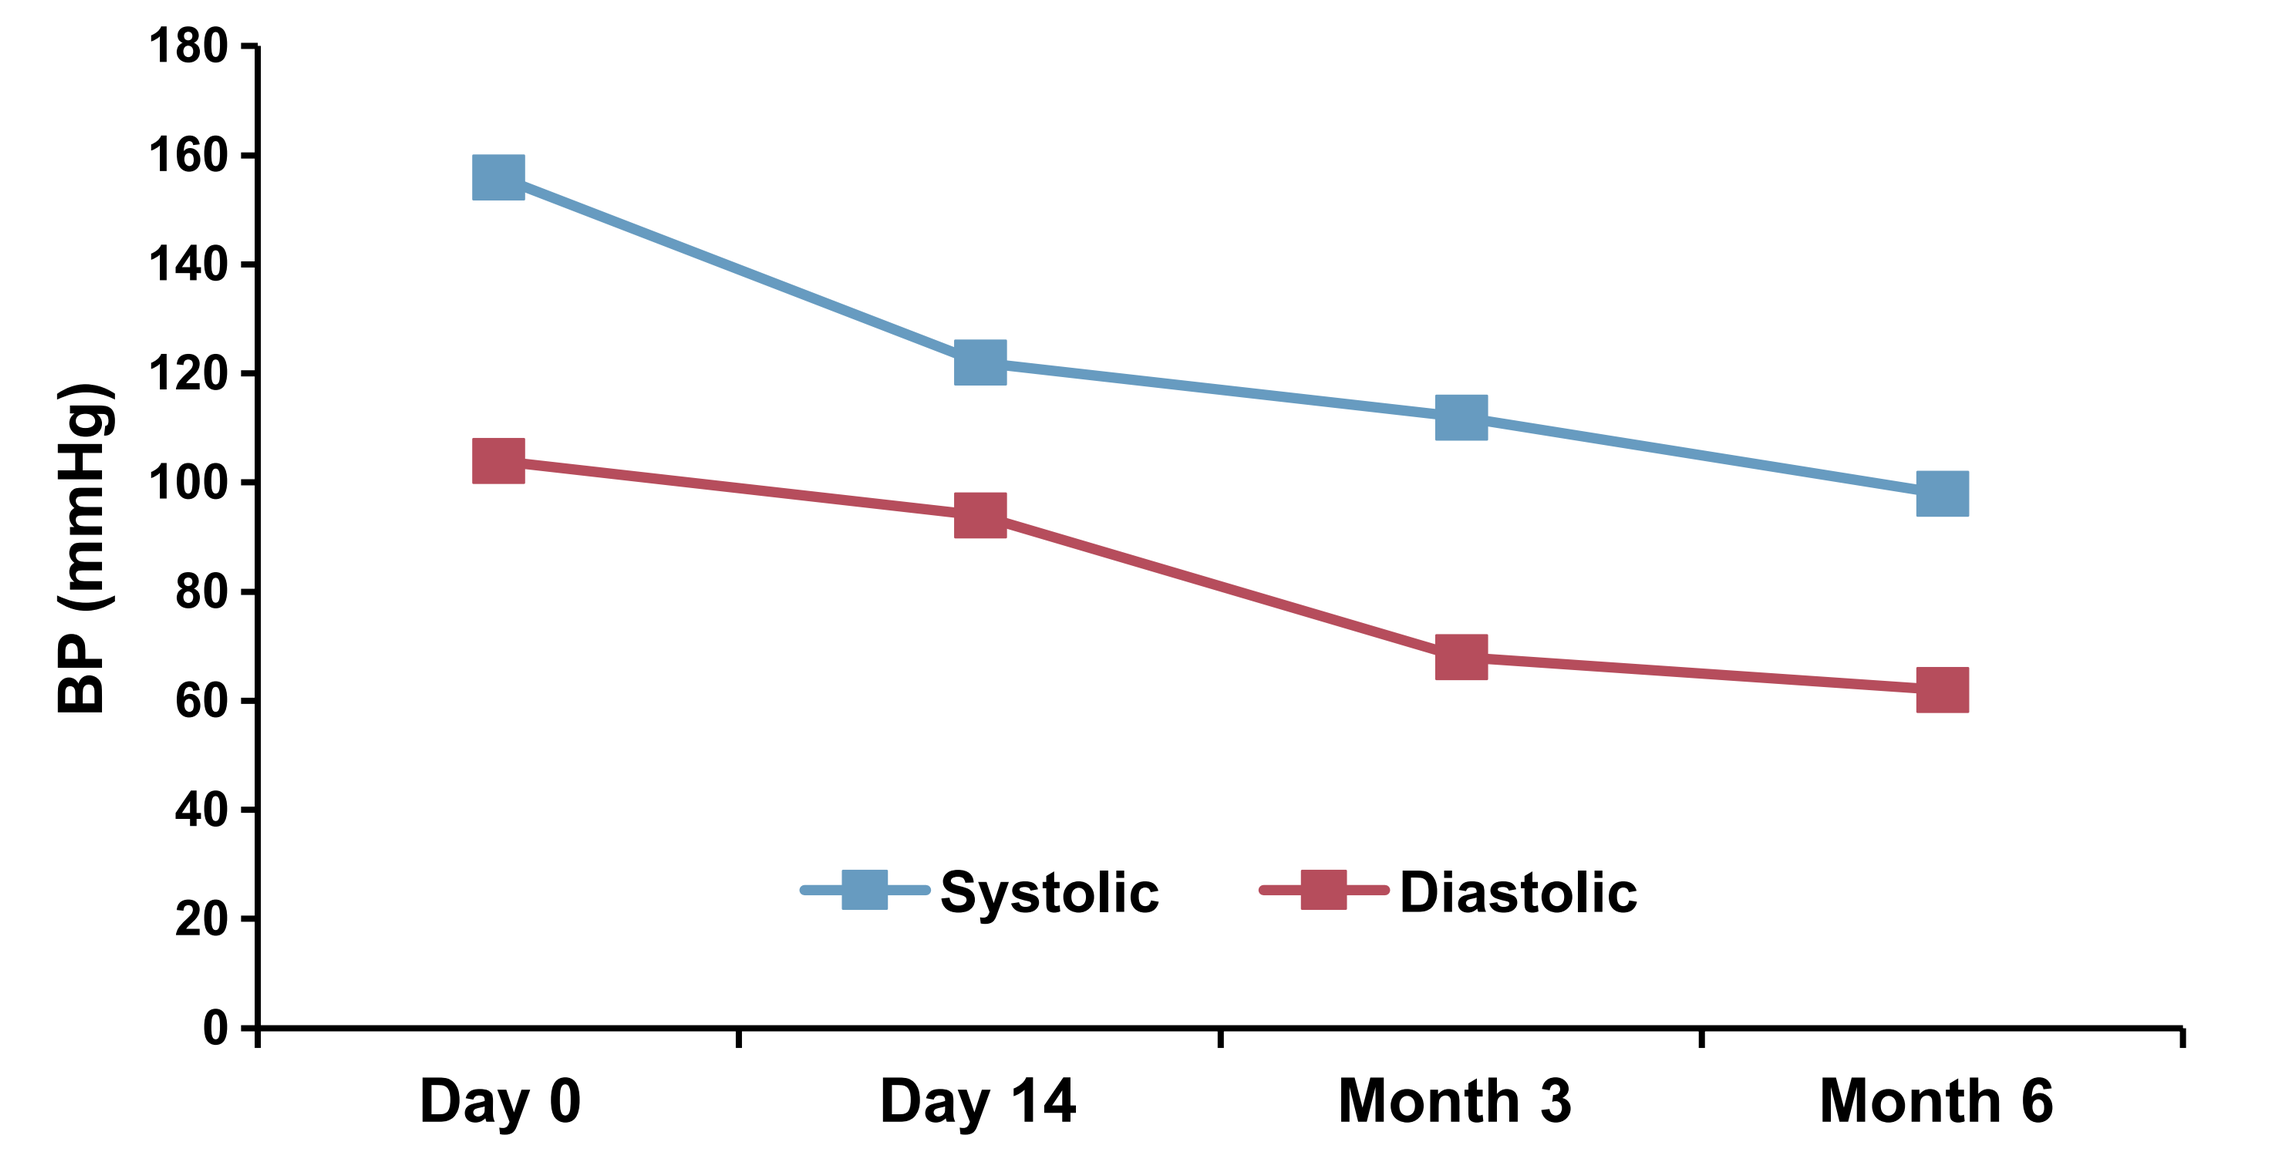
180
160
140
120
100
80
60
40
20
0
BP (mmHg)
Systolic
Diastolic
Day 0
Day 14
Month 3
Month 6

BP, blood pressure.

Figure 4. Changes in BMI After Treatment With Mifepristone

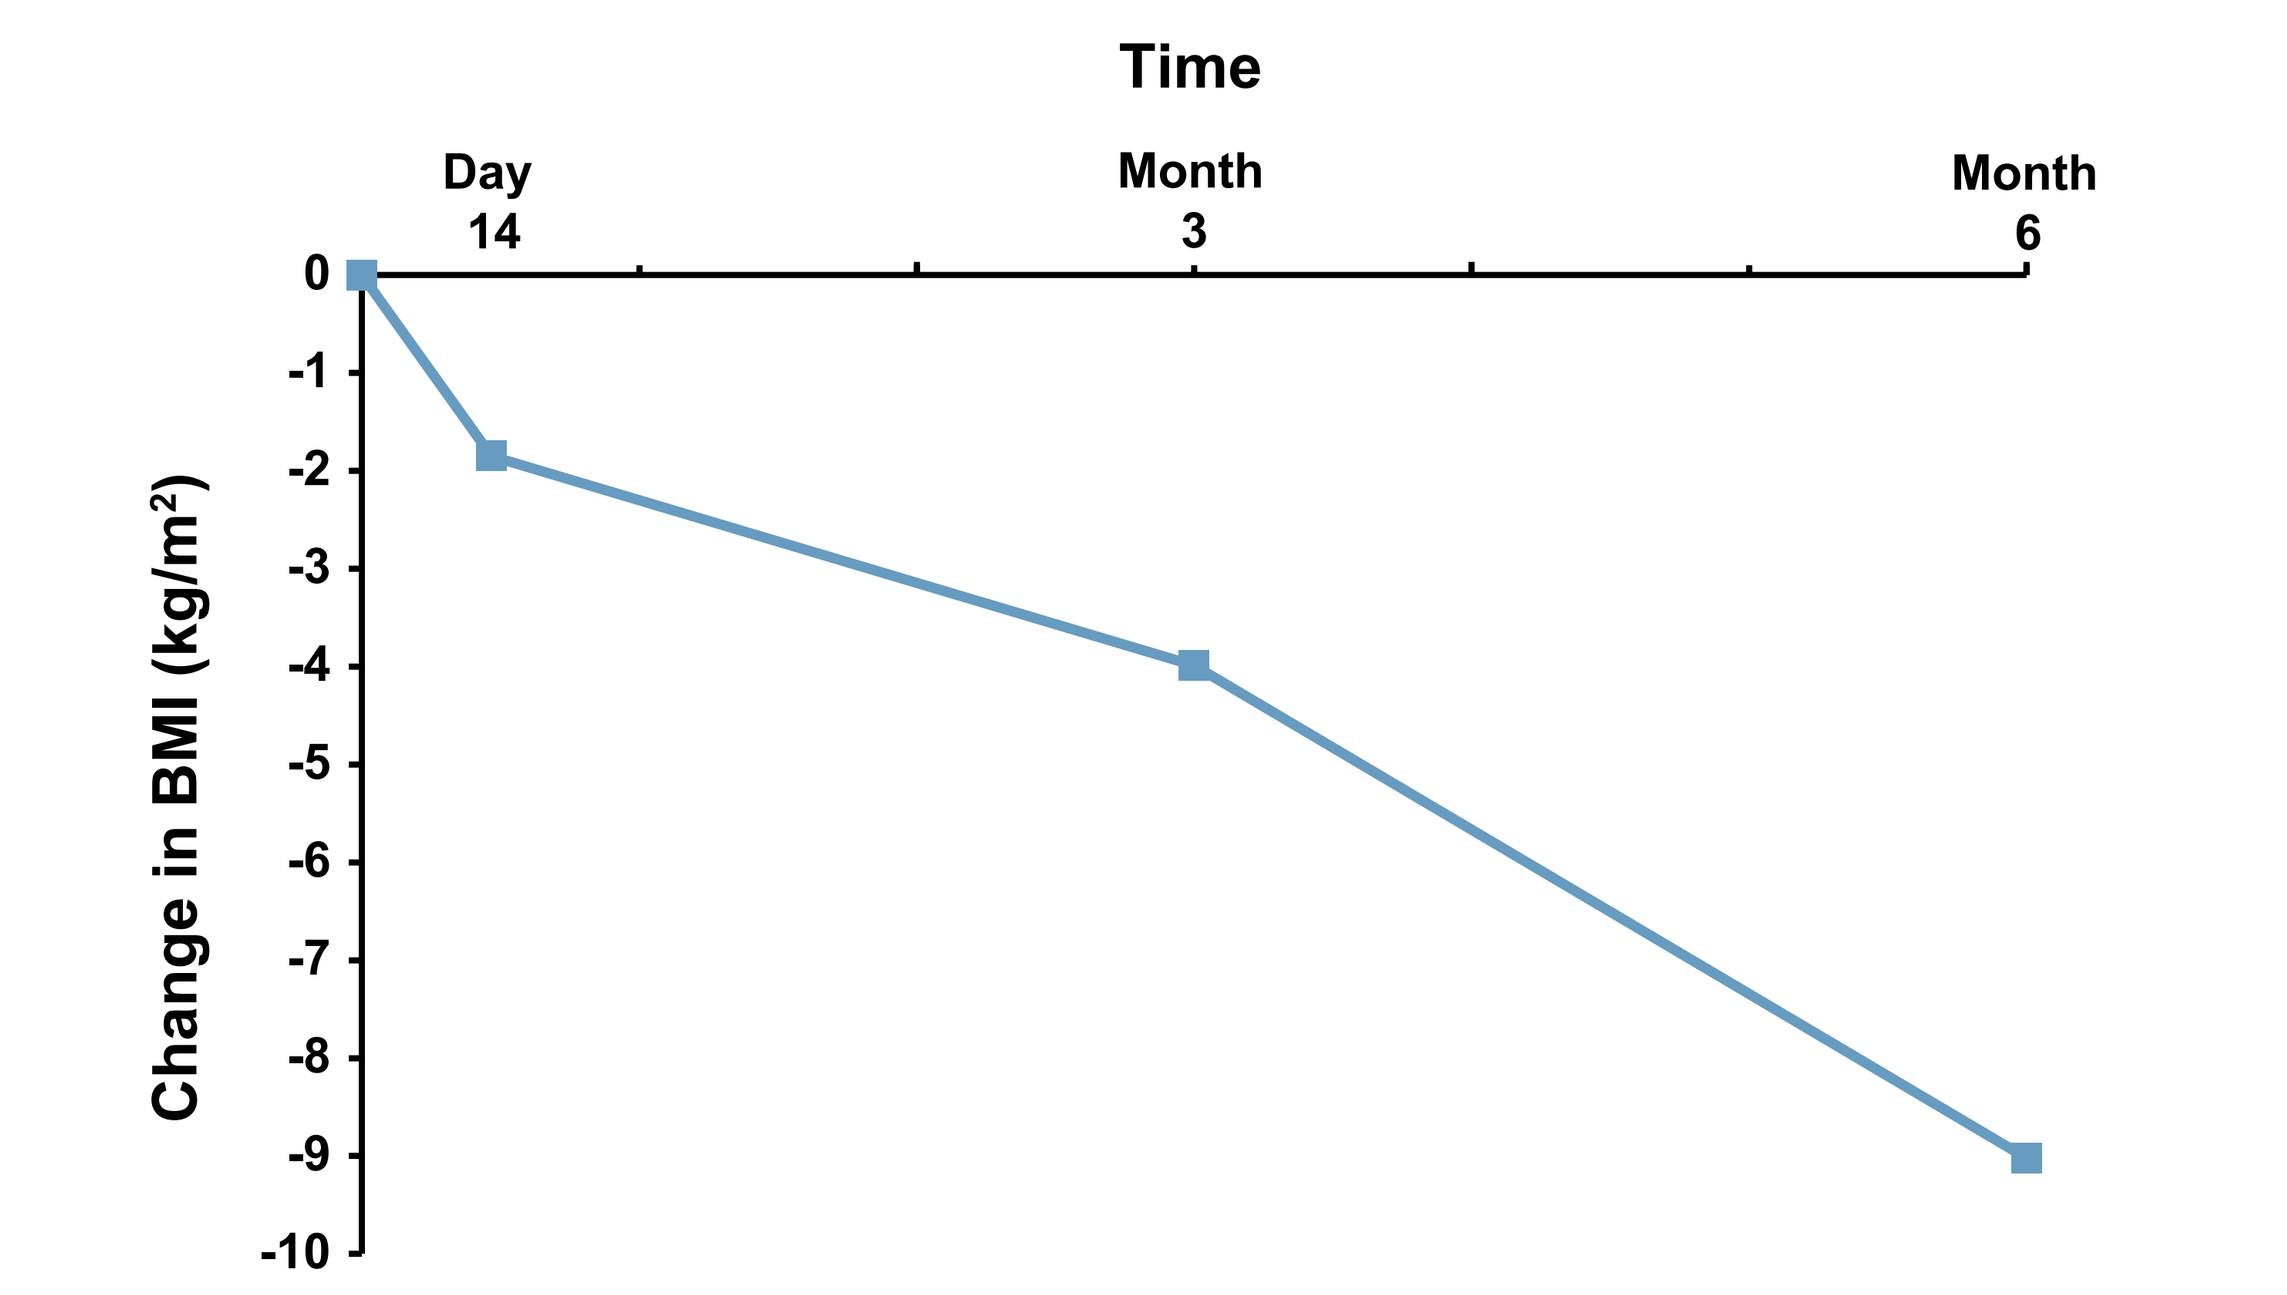
Time
Day 14
Month 3
Month 6
0
-1
-2
-3
-4
-5
-6
-7
-8
-9
-10
Change in BMI (kg/m²)

BMI, body mass index.

DISCUSSION

- We describe a case of severe hypercortisolism due to PMAH in a young nulliparous woman.
- Although successful treatment of PMAH with unilateral adrenalectomy has been reported,[3] BLA was performed to prevent recurrence.
- Her condition precluded immediate BLA, and medical therapy was employed to control her hypercortisolism.
- Mifepristone, a competitive glucocorticoid receptor antagonist, was used based on its published efficacy and tolerability profile.[4,5]
- After addition of mifepristone, the patient showed resolution of hypertension, hyperglycemia, and obesity, which allowed for an uneventful BLA.
- The patient did not complain of symptoms of adrenal insufficiency post-BLA.
- She was started on low-dose hydrocortisone peri-operatively and resumed a normal reproductive cycle within a month post-BLA, allowing for subsequent conception.

CONCLUSION

- This case demonstrates that mifepristone therapy provided rapid improvements in cardiometabolic derangements and other symptoms of severe hypercortisolism, mitigating surgical risk factors in a young woman with severe hypercortisolism due to PMAH, allowing successful BLA and subsequent conception.

REFERENCES

1. Nieman LK, et al. *J Clin Endocrinol Metab.* 2015;100(8):2807-31.
2. Coelho MC, et al. *Eur J Endocrinol.* 2015;173(4):M11-21.
3. Debillion E, et al. *J Clin Endocrinol Metab.* 2015;100(12):4417-24.
4. Fleseriu M, et al. *J Clin Endocrinol Metab.* 2012;97(6):2039-49.
5. Yuen KC, et al. *Endocr Pract.* 2015; 21(10):1087-92.

ACKNOWLEDGEMENTS

Funding for editorial, design, and production support for this poster was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors developed and revised the poster and provided approval of the final version.

DISCLOSURES

JUM, KM: Nothing to disclose.

PJL: Employee: Corcept.

KCJY: Research grants: Novartis, Cortendo; Advisory board: Novartis, Corcept.

Presented at the Endocrine Society (ENDO) 2017 Annual Meeting, April 1-4, 2017, Orlando, FL