MON-403

A Case for Avoiding Unwarranted Surgery: A Patient with Primary Macronodular Adrenal Hyperplasia (PMAH) Initially Presenting as a Unilateral Adrenal Incidentaloma

Pejman Cohan, MD[1]; Precious J. Lim, PhD[2]; Anne L. Peters, MD[3]

[1]Specialized Endocrine Care Center, Beverly Hills, CA; [2]Corcept Therapeutics, Menlo Park, CA; [3]Division of Endocrinology, Keck School of Medicine of the University of Southern California, Los Angeles, CA

ABSTRACT

Background: "Subclinical" Cushing Syndrome (SCS), controversial in name, definition and management, is often described as autonomous cortisol secretion associated with an adrenal adenoma without overt signs of cortisol excess. This can lead to unrecognized long-term exposure to cortisol excess with potentially deleterious metabolic, cardiovascular and skeletal consequences. Historically, there have been few reports of medical treatment for this condition.

We describe a patient with Primary Macronodular Adrenal Hyperplasia (PMAH) who likely had longstanding undiagnosed cortisol excess and ultimately underwent unnecessary surgery. She was then effectively treated with mifepristone (MIFE), a glucocorticoid receptor (GR) competitive antagonist.

Case: A 71 yo woman with a hx of nephrolithiasis, T2DM, easy bruising, and osteoporosis underwent an abdominal CT scan for hematuria, disclosing a 1.7 cm left adrenal nodule. She did not have a family history of T2DM, her BMI was 18.8 kg/m^2 at diagnosis, and her A1C was 6% on metformin.

Physical examination was notable only for lower extremities bruising.

Hormonal labs: UFCs (116, 32, 69, 74, 57, 77 µg/day, ULN <50 µg/day), salivary cortisol (14, 6.7 nmol/L, ULN <4.3 nmol/L), 1-mg DST (18 µg/dL, ULN <1.8 µg/dL, dexamethasone 321 ng/dL nml:180-550 ng/dL), AM serum cortisol (19.1 µg/dL), AM ACTH (13 pg/mL), and DHEA-S (82 µg/dL ULN<111 µg/dL). A1C worsened to 7.9% at presentation.

Patient opted for a second opinion at a tertiary center where serial HPA axis tests were equivocal.

MIFE (300 mg/day) was started but stopped after 1 wk due to fatigue, dizziness, low K+ 3.4 mEq/L [3.5-5.0 mEq/L], and bilateral pedal edema.

Patient subsequently elected to undergo left laparoscopic adrenalectomy, which was performed w/o complications. Pathology revealed macronodular adrenal hyperplasia with a predominant nodule.

Postoperatively, she developed fatigue and dizziness, and hydrocortisone replacement was initiated.

After 4 wks, no improvement in AM cortisol levels was observed and hydrocortisone was halted.

After 6 mos, A1C worsened to 8.1% with no improvement in excessive bruising.

MIFE was restarted at 300 mg/week and gradually increased to 300 mg three times a week (TIW). She developed fatigue (resolved w/o intervention) and TSH elevation (normalized on levothyroxine). After 8 mos, A1C decreased to 6.7% w/o dieting and she remains on MIFE 300 mg TIW.

Conclusions: Clinically relevant autonomous cortisol secretion can occur even without global symptoms of cortisol excess. In this patient, GR antagonism with low dose MIFE was well tolerated and elicited metabolic benefits. Medically modulating the underlying cortisol excess may be an option for patients w/ SCS in whom surgical benefit is uncertain, particularly those who potentially have bilateral adrenal disease.

INTRODUCTION

- "Subclinical" Cushing Syndrome (SCS) is associated with increased risk of cardiovascular events and mortality,[1] as well as type 2 diabetes mellitus (T2DM), hypertension, dyslipidemia, and obesity.[2,3]
- The relatively high prevalence and potential for associated complications underscore the clinical importance of SCS.
- Diagnosing SCS remains a controversial topic in clinical endocrinology.
- Although most commonly described as altered hypothalamic-pituitary-adrenal (HPA) axis secretion associated with an adrenal adenoma without overt signs of cortisol excess, uncertainties in the diagnosis and surgical management[4,5] of SCS delay treatment decisions while comorbidities develop or progress.
- Conversely, some patients undergo unnecessary surgery as evidenced by lack of improvement in metabolic parameters[4] and absence of postoperative adrenal insufficiency.[5]
- Until the challenges in the diagnosis and management of SCS are solved, many patients are subjected to multiple tests and exposed to therapies that fail to address the underlying cause, all while risking progression toward a potentially avoidable significant event.
- Here, we describe a patient who likely had long-standing undiagnosed hypercortisolism and underwent surgery to remove an adrenal nodule; symptoms and pathology subsequently confirmed Primary Macronodular Adrenal Hyperplasia (PMAH).
- She was later effectively treated with mifepristone, an approved medical therapy that modulates the effects of cortisol by acting as a competitive antagonist at the glucocorticoid receptor.

PAST MEDICAL HISTORY

- A 71-year-old woman presented for evaluation of hypercortisolism.
- Past medical history included (**Figure 1**):
 - Kidney stones at age 41.
 - T2DM at age 58.
 - No family history of T2DM.
 - BMI at T2DM diagnosis: 18.8 kg/m^2.
 - HbA1c controlled in 6% range with metformin.
 - Long-standing history of bruising, which worsened at age 66.
 - Osteoporosis T-score of -2.6 at left femoral neck.

Figure 1. Timeline of Past Medical History and Diagnosis

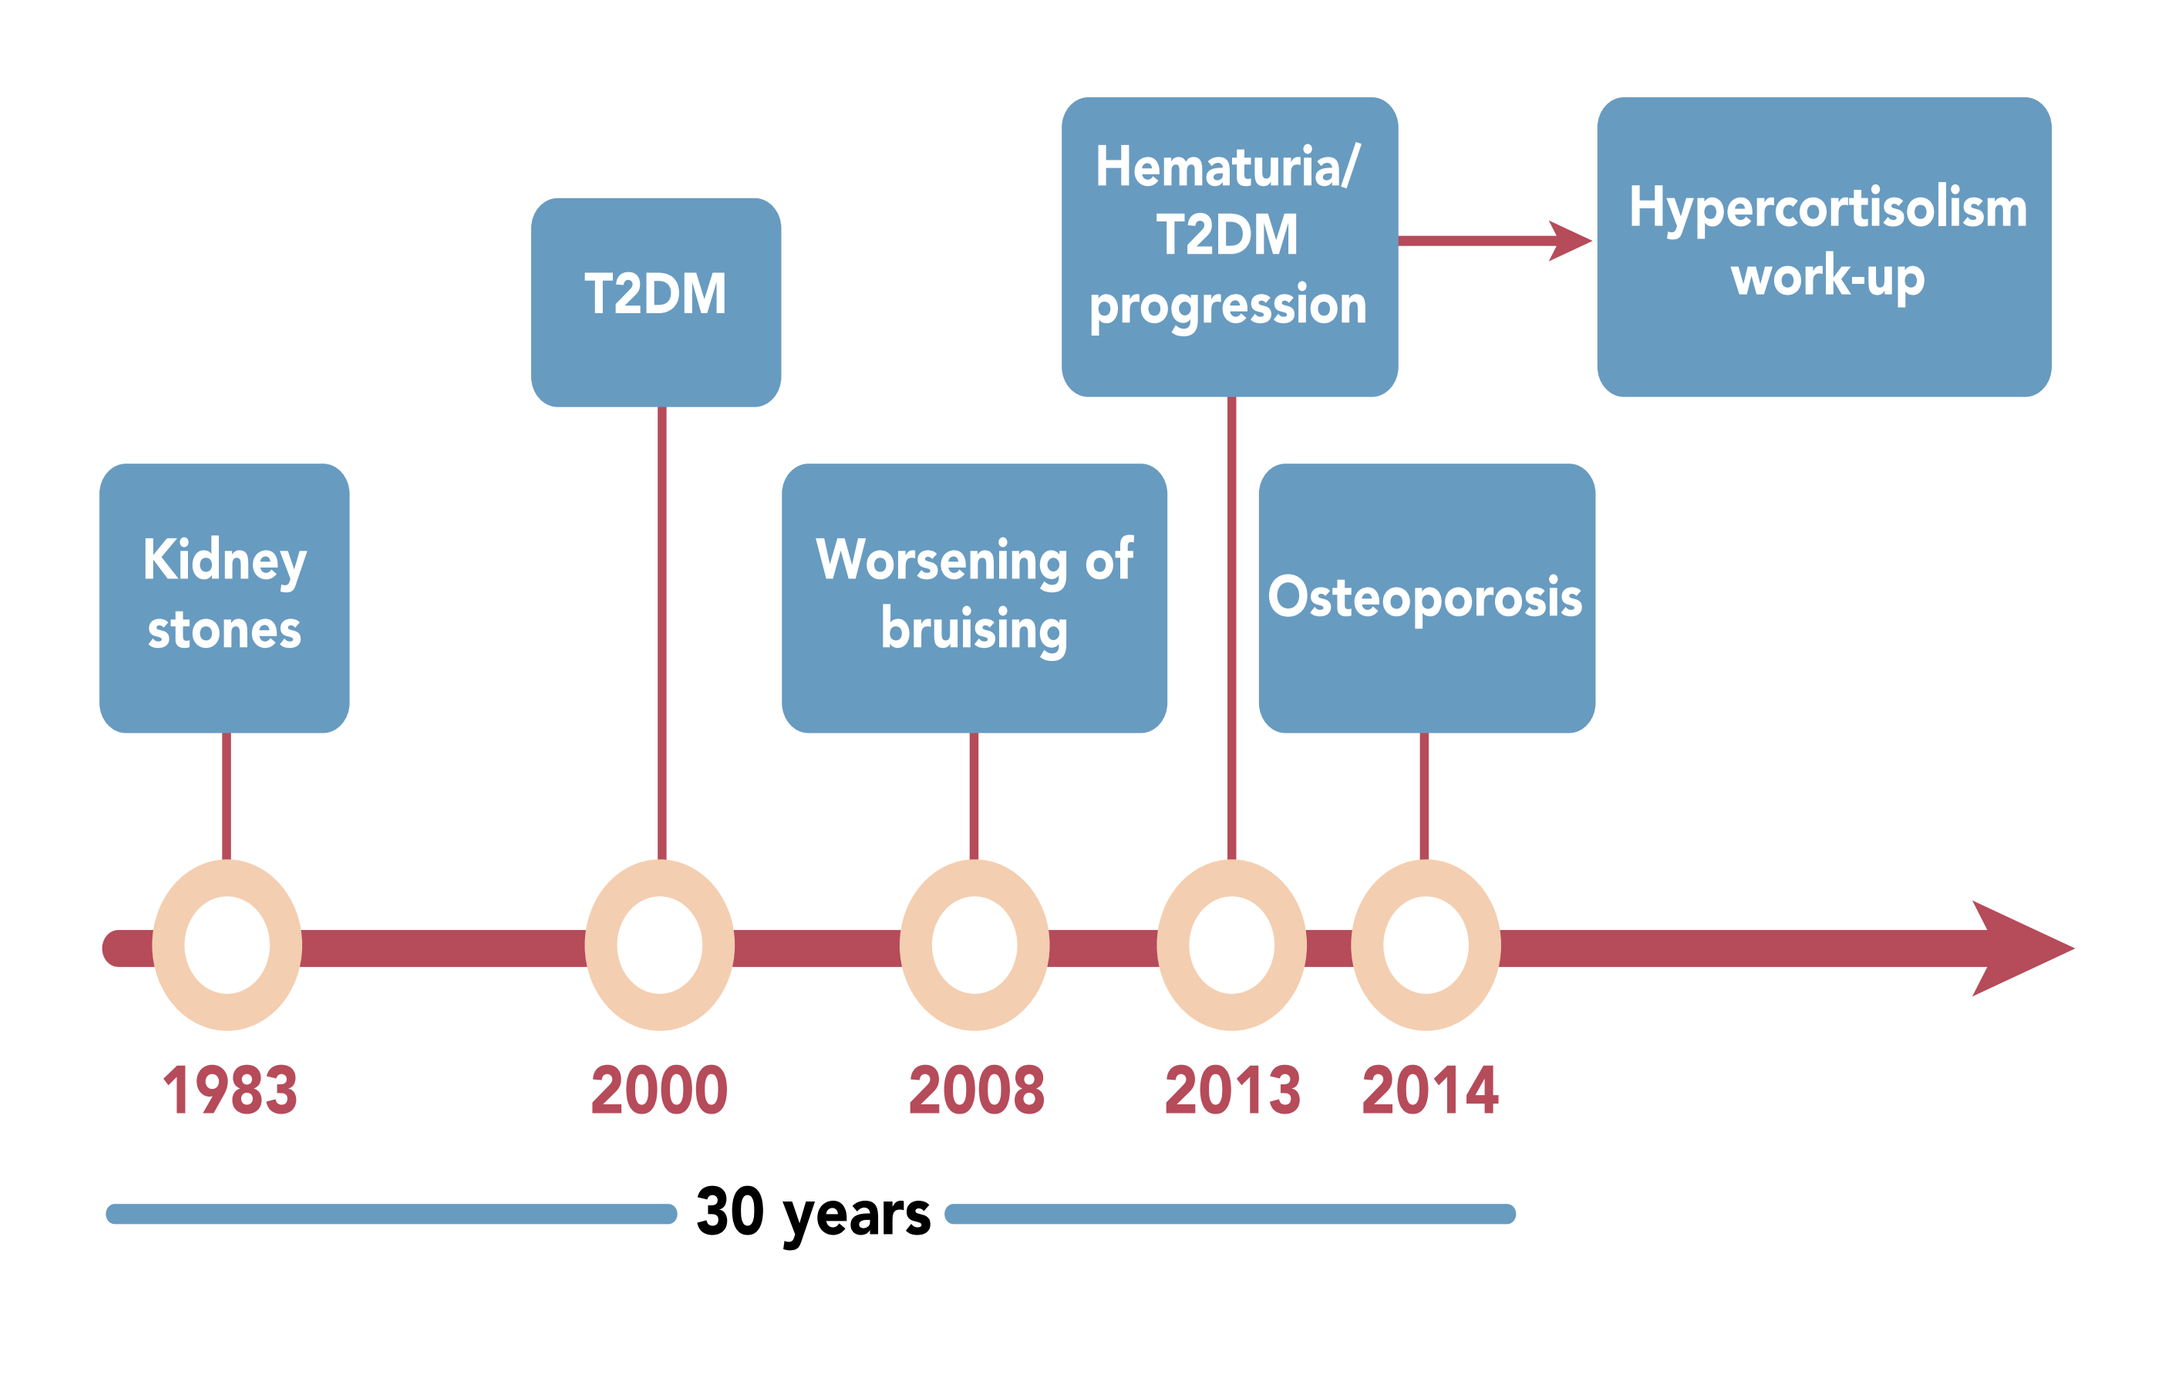
Kidney stones
T2DM
Worsening of bruising
Hematuria/ T2DM progression
Osteoporosis
Hypercortisolism work-up
1983
2000
2008
2013
2014
30 years

Figure 2. Absence of Signs and Symptoms of Overt Hypercortisolism

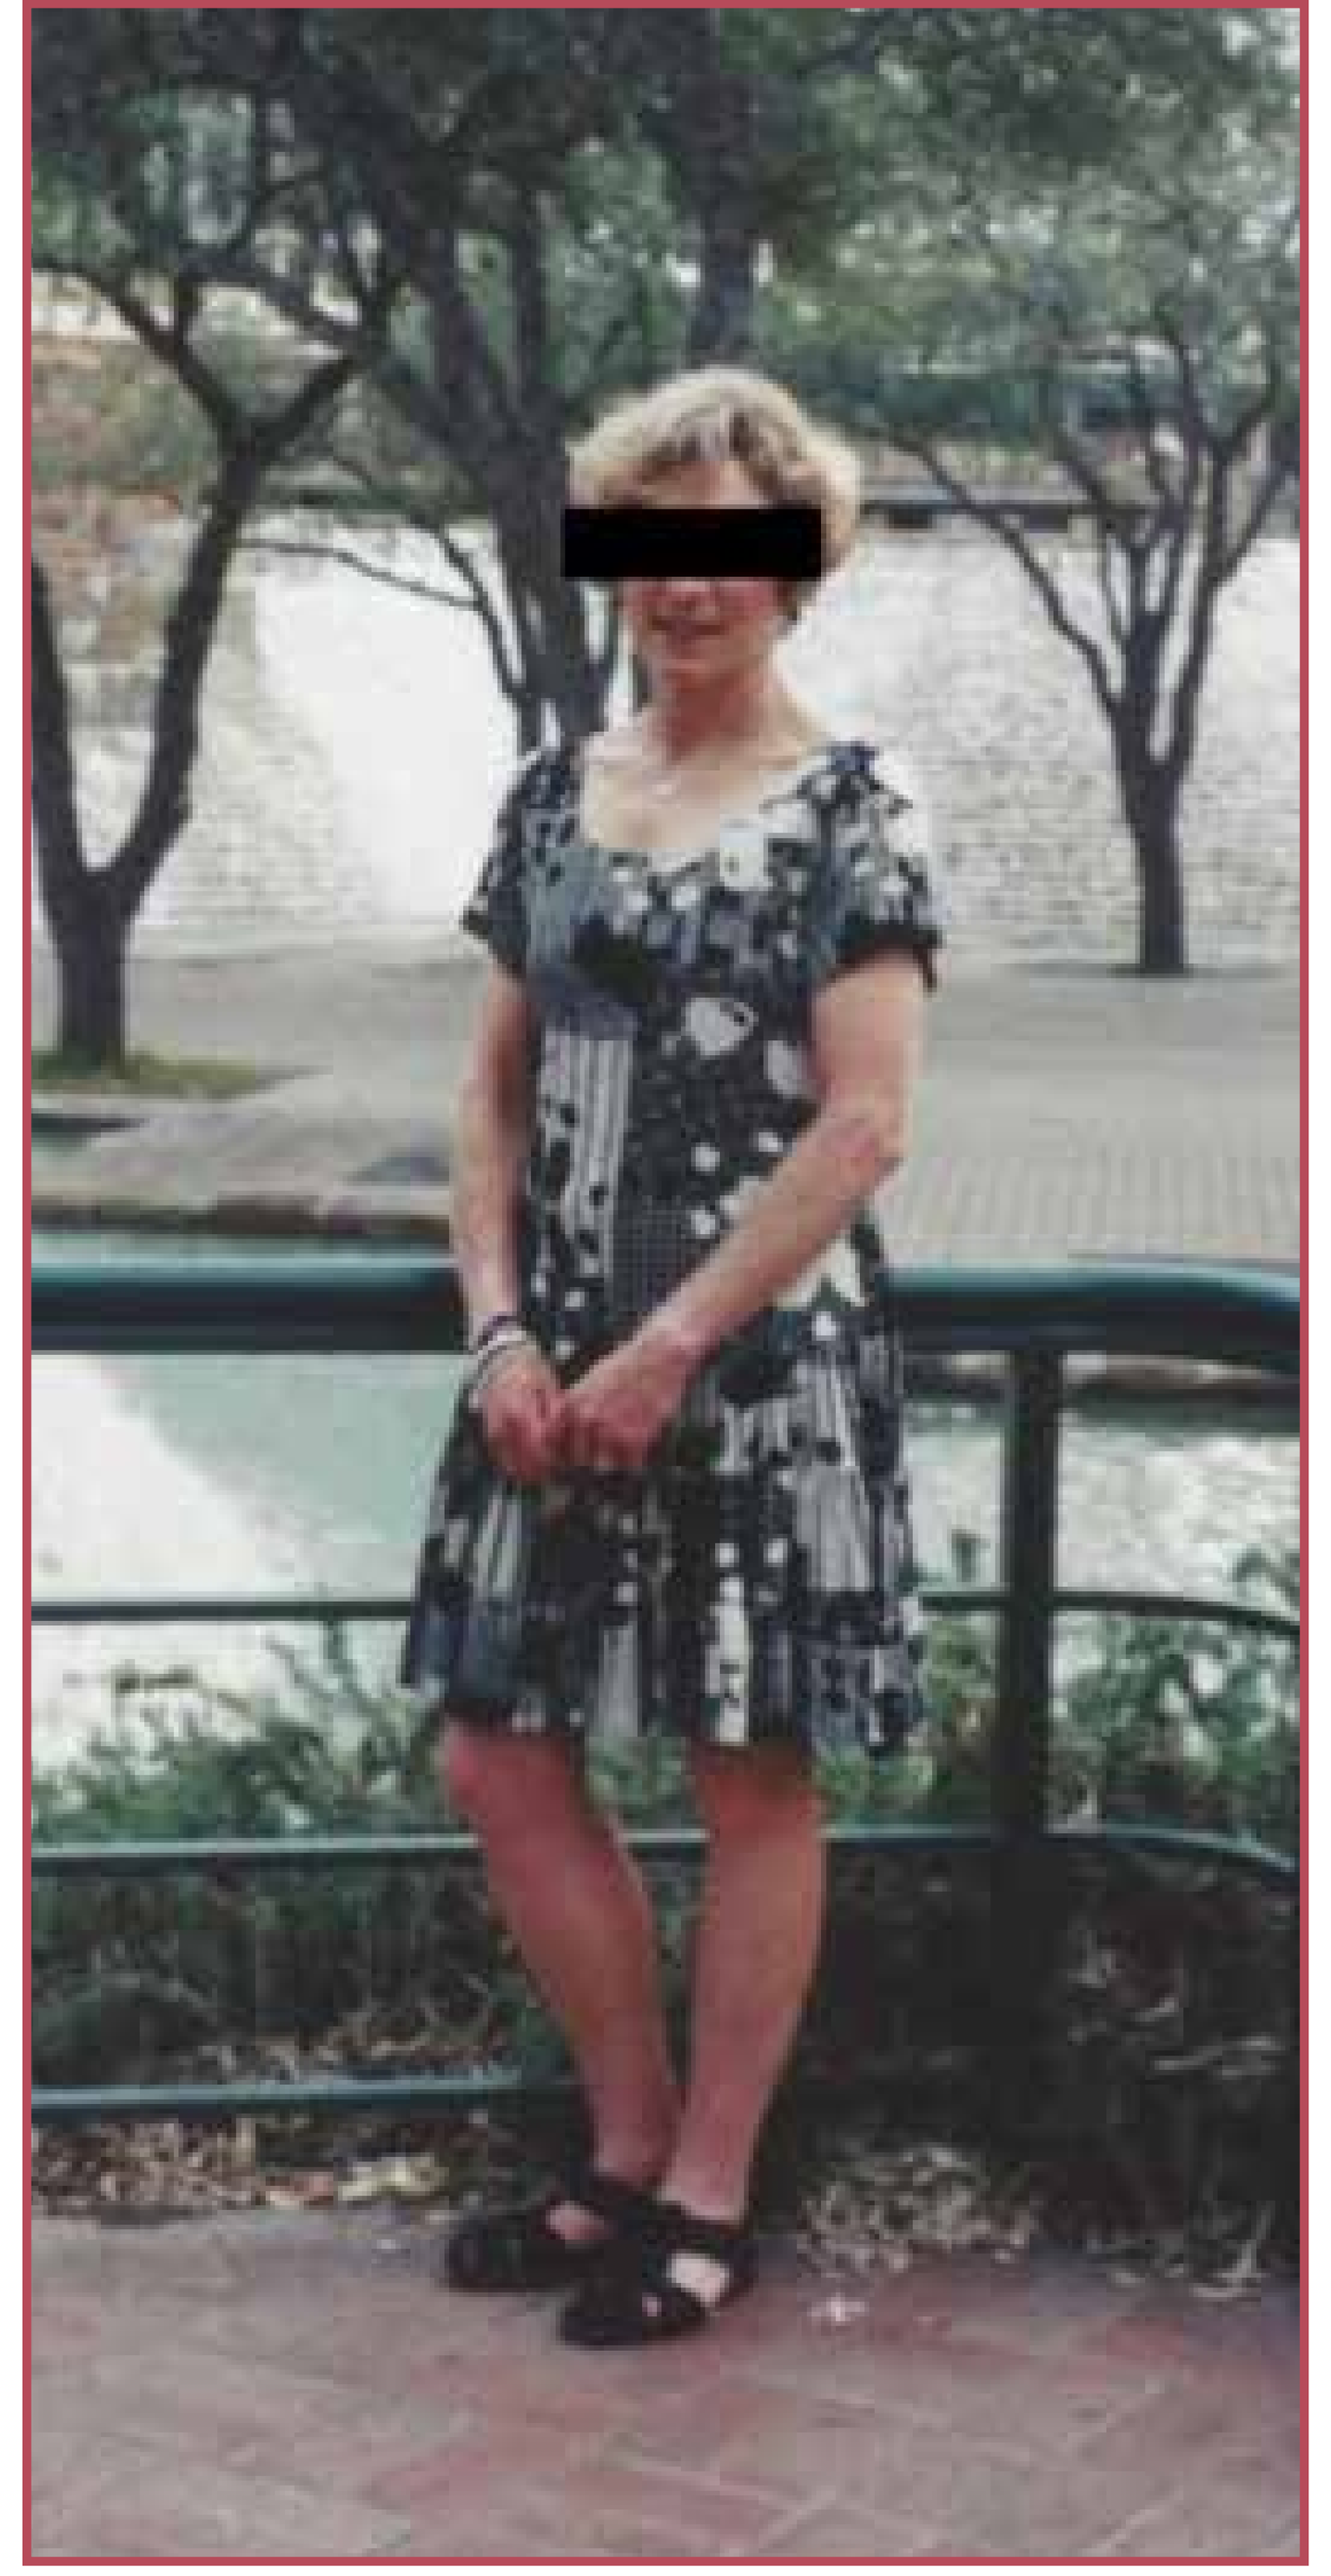

CLINICAL PRESENTATION

- Physical examination was notable only for bruising on lower extremities.
- Abdominal CT scan (**Figure 3**) at age 71 for hematuria, which revealed:
 - Left renal calculus.
 - 1.7 cm left adrenal nodule.
- HbA1c progressively increased to 7.9%.

Figure 3. Images From Abdominal CT Scan Showing Diffuse Nodular Hyperplasia (red) and an Enlarged Gland (blue)

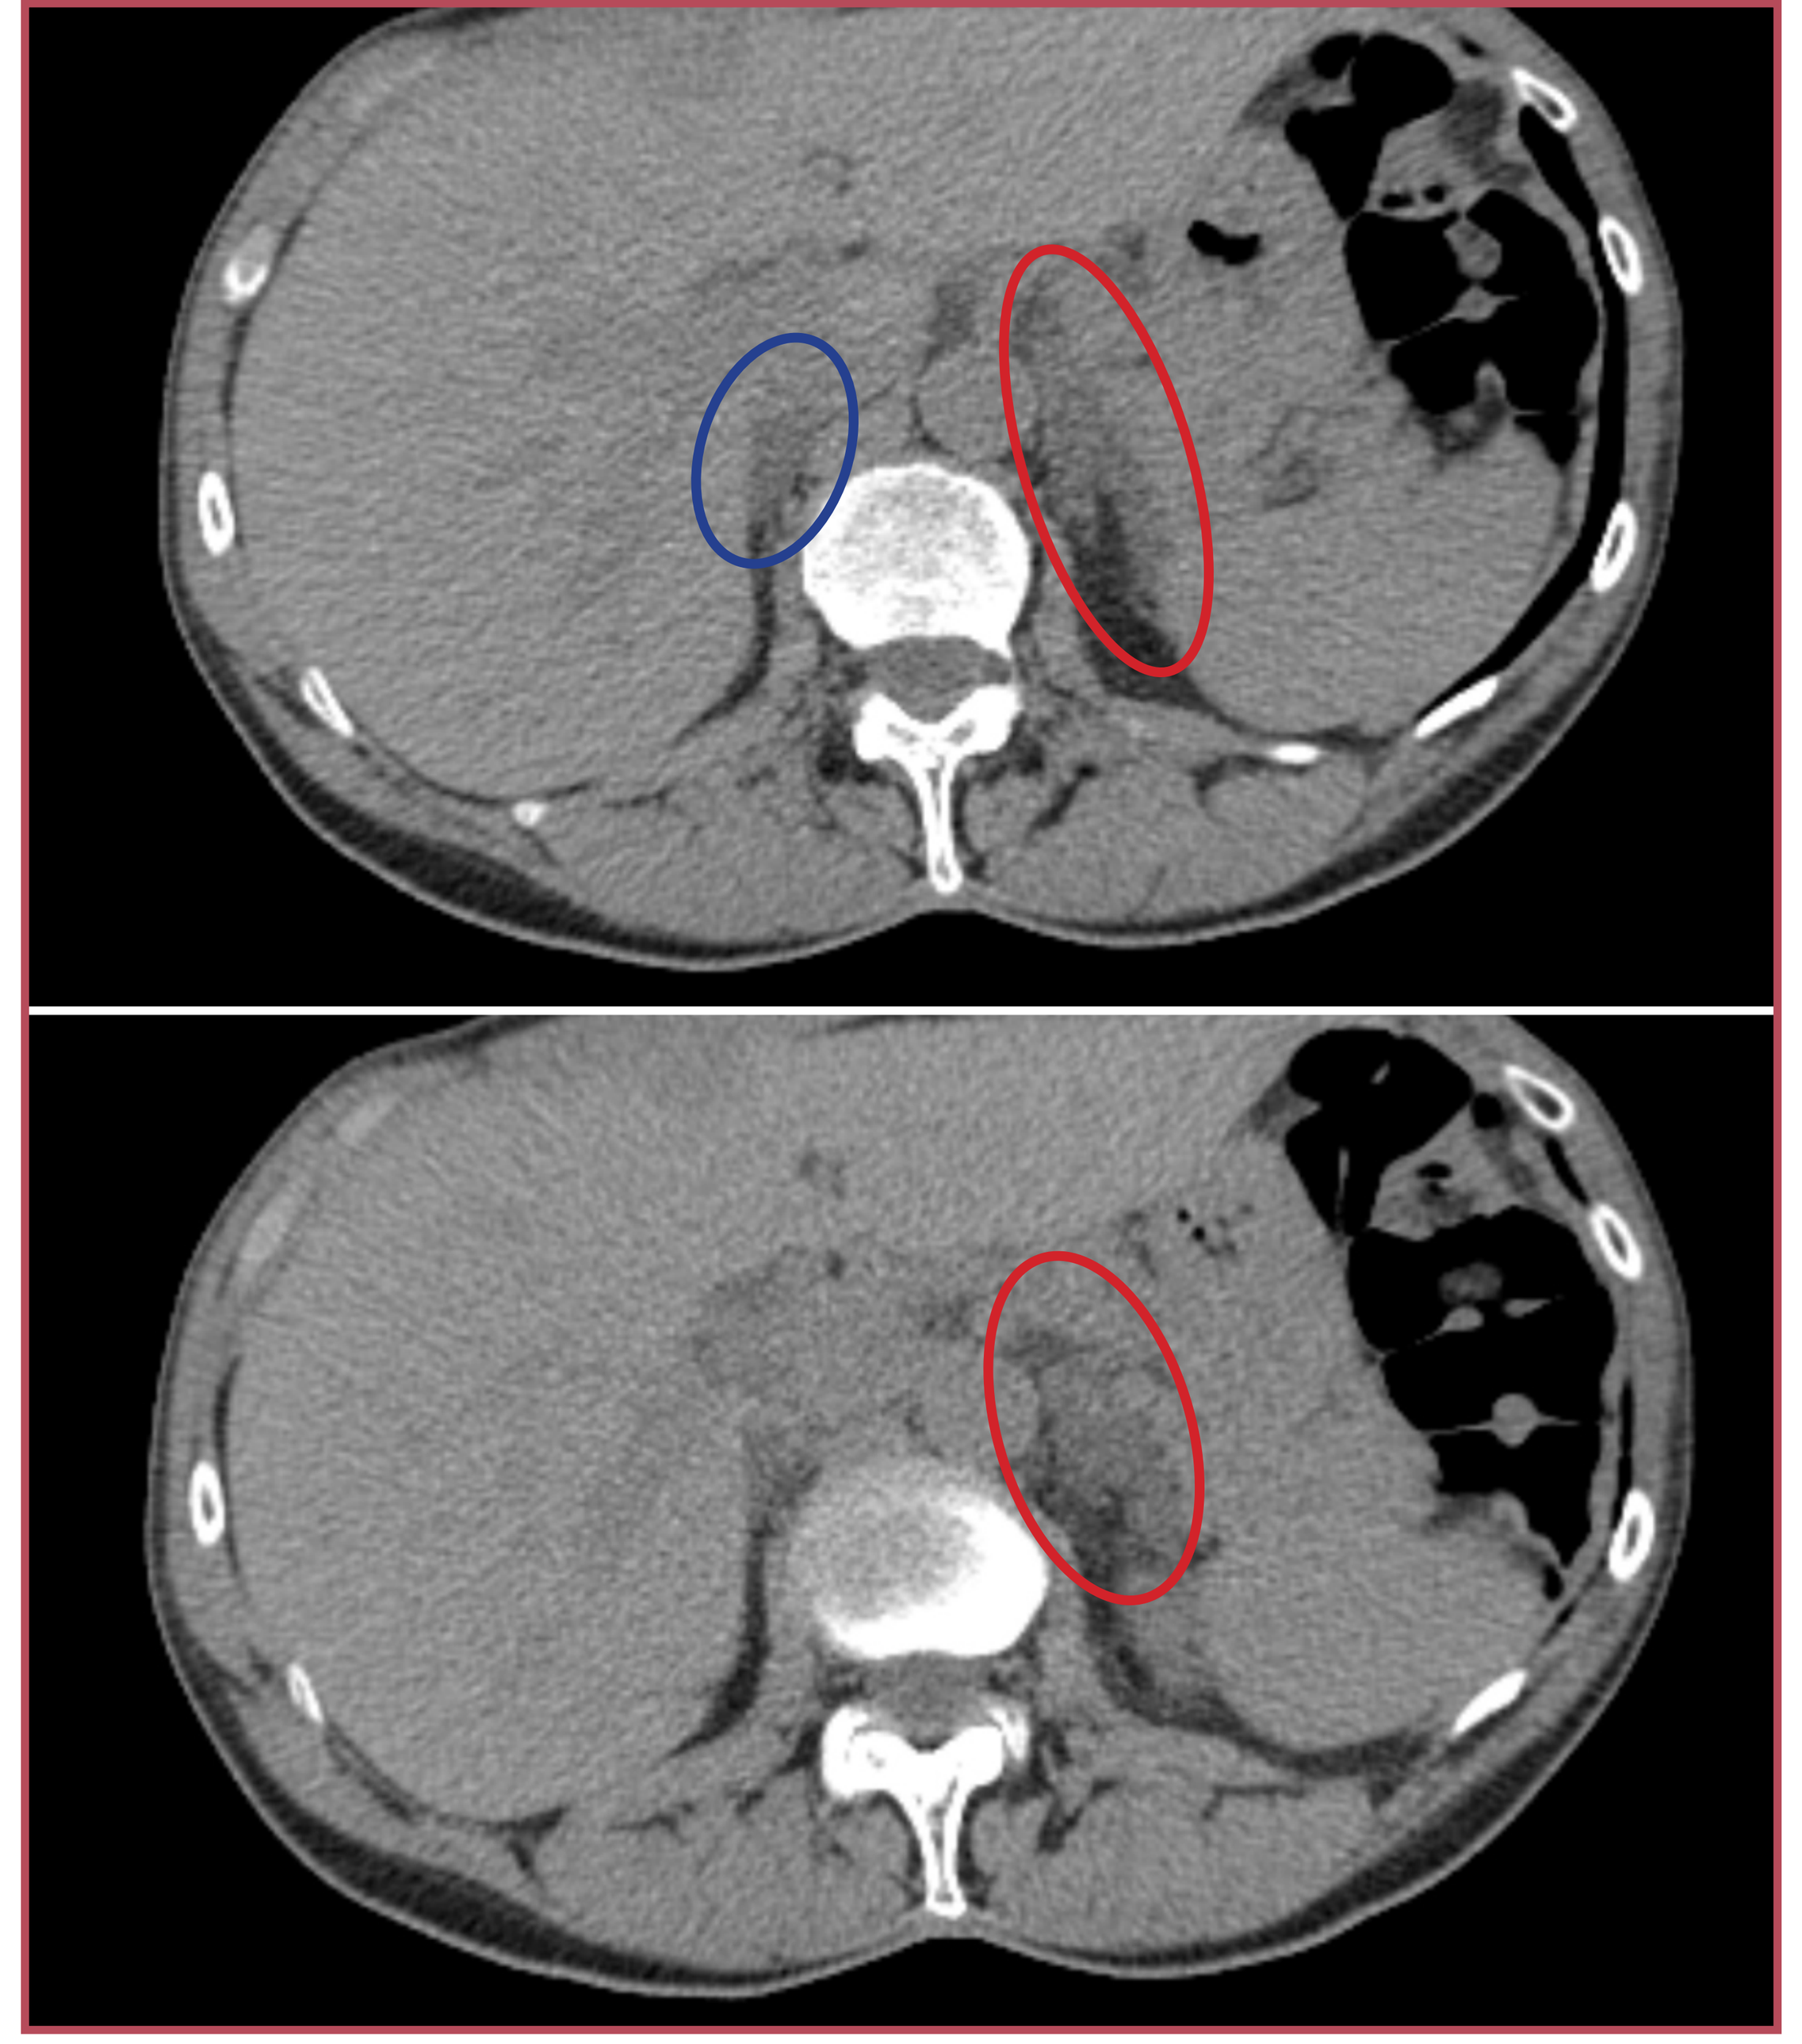

- Initial hormonal work-up (**Table 1**) revealed inconsistent results:
 - AM adrenocorticotropic hormone (ACTH) was in the indeterminate range.
 - Cortisol level after 1-mg dexamethasone suppression test (1-mg DST) was inconsistent with her physical manifestations.

Table 1. HPA Axis Testing Results

Measure (reference)	Result 1	Result 2	Result 3	Result 4	Result 5	Result 6
AM serum cortisol (4-22 µg/dL)	19.1 µg/dL					
AM ACTH (6-50 pg/mL)	13 pg/mL					
DHEA-S (ULN<111 µg/dL)	82 µg/dL					
1-mg DST:						
Cortisol (ULN <1.8 µg/dL)	18					
ACTH (pg/mL)	10					
Dexamethasone (180-550 ng/dL)	321					
UFC (ULN <50 µg/day)	116	32	69	74	57	77
LNSC (ULN <4.3 nmol/L)	14	6.7				

ACTH, adrenocorticotropic hormone; AM, morning; DHEA-S, dehydroepiandrosterone sulfate; DST, dexamethasone suppression test; HPA, hypothalamic-pituitary-adrenal; LNSC, late-night salivary cortisol; UFC, urinary free cortisol.

- The patient opted for a second opinion at a tertiary center (**Table 2**), where HPA axis tests also showed inconsistent results:
 - Late-night salivary cortisol (LNSC) revealed both normal and abnormal results.
 - AM ACTH levels fluctuated.
 - Cortisol level after dexamethasone administration as part of the Dex-CRH test was borderline.

Table 2. HPA Axis Testing Results (Alternate Center)

Measure (reference)	Result 1	Result 2	Result 3
UFC (ULN <45 µg/day)	62.6	50.9	
Serum cortisol, late-night (ULN <7.5 µg/dL)	7		
LNSC (ULN <2.76 nmol/L)	2.29	5.24	
Dex-CRH:			
0-min cortisol (ULN <1.8 µg/dL)	1.8		
15-min cortisol (ULN <1.4 µg/dL)	2		
All ACTH in this series (pg/mL)	<5		
Serial AM ACTH (pg/mL)	<5	8.1	14.6
Serial PM ACTH (pg/mL)	9.9	7.5	6.9

ACTH, adrenocorticotropic hormone; AM, morning; Dex-CRH, combined dexamethasone suppression/corticotropin-releasing hormone test; HPA, hypothalamic-pituitary-adrenal; LNSC, late-night salivary cortisol; PM, evening; UFC, urinary free cortisol.

TREATMENT COURSES AND OUTCOMES

INITIAL MIFEPRISTONE TREATMENT

- Mifepristone was initiated at 300 mg/day.
 - One week later, the patient reported fatigue, dizziness, and leg swelling.
 - Further examination revealed:
 - +1 bilateral pedal edema.
 - Potassium: 3.4 mEq/L (normal range: 3.5-5.0 mEq/L).
- Although advised of these expected side effects, she elected to undergo left laparoscopic adrenalectomy at another tertiary center, which was performed without complications.

FAILED SURGERY

- She developed fatigue and dizziness postoperatively and was placed on hydrocortisone replacement despite urinary free cortisol (UFC) levels of 65 µg/day (ULN <50 µg/day).
- Pathology revealed macronodular adrenal hyperplasia with a predominant nodule.
- At postoperative week 4, cortisol had not declined, and hydrocortisone was discontinued.
- She was monitored for 6 months without improvement in her primary manifestations:
 - Excessive bruising continued.
 - HbA1c worsened to 8.1% (**Table 3**).

Table 3. Changes in HbA1c

	Presentation	6 mo after surgery	3 mo after mifepristone	4 mo after mifepristone
HbA1c (%)	7.9	8.1	7.4	6.7

MIFEPRISTONE RESTART

- Mifepristone was restarted at 300 mg/week, which was gradually increased to 300 mg three times a week (TIW):
 - Three months later, her HbA1c improved to 7.4%.
 - She reported fatigue, which resolved without intervention.
 - She also developed thyroid-stimulating hormone (TSH) elevation (normal Free T4 and Free T3, negative anti-thyroid antibodies) with neuro-cognitive impairment and depression, which normalized on levothyroxine.
- One month later, her HbA1c further improved to 6.7% despite complete liberalization of her diet.

CLINICAL LESSONS

- We describe a case of mild hypercortisolism due to PMAH in an elderly patient.
- She likely had hypercortisolism even well before the formation of kidney stones at age 41.
- Susceptibility to nephrolithiasis is high in hypercortisolism, and indeed, kidney stones are reported in about 15% of CS patients[6] with prevalence remaining high even after remission.[7]
- Although T2DM is frequently associated with hypercortisolism, routine testing in patients with T2DM is performed infrequently.
- In the patient we describe, low body mass index (BMI) confounded clinical suspicion.
- Easy bruising and osteoporosis are also common features of hypercortisolism,[8] and should prompt evaluation.
- Bone tissue is extremely sensitive to cortisol, with several studies suggesting that mild hypercortisolism is associated with accelerated bone loss.[9]
- Osteoporosis and osteopenia may be the presenting symptoms in an otherwise asymptomatic patient.
- Although HPA axis tests suggested mild hypercortisolism, it was unclear whether her T2DM and osteoporosis would improve with unilateral adrenalectomy.
- Moreover, based on the size of her adenoma, surgery was not clearly recommended.[10]
- Unilateral adrenalectomy should be avoided unless benefit is clear, and especially so in patients who potentially have bilateral adrenal disease.
- Aggressive management of T2DM, osteoporosis, and cardiovascular risk is one treatment strategy.
- A reversible medical therapy to target hypercortisolism, the possible underlying cause of all her comorbid conditions, may be the best treatment strategy.

CONCLUSIONS

- Clinically relevant autonomous cortisol secretion can occur even without global symptoms of hypercortisolism.
- In this patient, GR antagonism with low-dose mifepristone was well tolerated and elicited substantial metabolic benefits.
- Medically modulating the underlying cortisol excess may be an option for patients with SCS in whom surgical benefit is uncertain, particularly those who potentially have bilateral adrenal disease.

REFERENCES

1. Di Dalmazi G, et al. *Lancet Diabetes Endocrinol.* 2014; 2(5):396-405.
2. Tauchmanovà L, et al. *J Clin Endocrinol Metab.* 2002;87(11):4872-78.
3. Debono M, Newell-Price J. *Curr Opin Endocrinol Diabetes Obes.* 2015; 22(3):185-92.
4. Toniato A, et al. *Ann Surg.* 2009;249(3):388-91.
5. Di Dalmazi G, et al. *J Clin Endocrinol Metab.* 2014; 99(8):2637-45.
6. Ross EJ, et al. *Q J Med.* 1966; 35(138):149-92.
7. Faggiano A, et al. *J Clin Endocrinol Metab.* 2003; 88(5):2076-80.
8. Pecori Giraldi F, et al. *Clin Endocrinol(Oxf).* 2007; 66(2):251-57.
9. Chiodini I, et al. *J Endocrinol Invest.* 2008; 31(3):267-76.
10. Morelli V, et al. *J Clin Endocrinol Metab.* 2014; 99(3):827-34.

ACKNOWLEDGMENTS

Funding for editorial, design, and production support for this poster was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Skillman, NJ. The authors developed and revised the poster and provided approval of the final version.

DISCLOSURES

PC: Speaker: Corcept, Pfizer; **PJL:** Employee: Corcept Therapeutics; **ALP:** Speaker/Advisory board: Abbott, Beckton Dickinson, Bigfoot Biomedical, Boehringer Ingelheim, Eli Lilly, Astra-Zeneca, Intarcia, Janssen, Lexicon, Medtronic-Minimed, Merck, Novo Nordisk, Omada Health, Optum Rx, Sanofi, United Healthcare; Research grant: Dexcom; Editor: Medscape.

Presented at the Endocrine Society (ENDO) 2017 Annual Meeting, April 1-4, 2017, Orlando, FL