SAT-578

MIFEPRISTONE THERAPY PRIOR TO PITUITARY SURGERY IN CUSHING DISEASE PREVENTED NEED FOR LONG-TERM GLUCOCORTICOID REPLACEMENT

Paul A. Sack, MD[1]; James J. Smith, PhD[2]

[1]MedStar Union Memorial Hospital, Baltimore, MD, USA; [2]Corcept Therapeutics, Menlo Park, CA, USA

INTRODUCTION

- In Cushing disease (CD), elevated adrenocorticotropic hormone (ACTH) released from pituitary adenomas causes excessive production of cortisol by the adrenal glands.[1]
- Hypercortisolism then leads to hypothalamic-pituitary-adrenal (HPA) axis dysfunction through disruption of the negative cortisol receptor feedback mechanisms on normal (non-adenoma) pituitary corticotrophs[1,2] (**Figure 1**).
- Successful transsphenoidal surgery (TSS) to remove the pituitary adenoma can be curative in removing the source of excess ACTH production, but often leads to secondary adrenal insufficiency because ACTH produced by the remaining pituitary corticotrophs is suppressed.[3]
- In contrast, with non-CD pituitary adenomas that have an intact HPA cortisol axis prior to surgery, cortisol levels rise quickly after TSS and frequently do not show secondary adrenal insufficiency and do not require postoperative steroids.[4]
- The resulting secondary adrenal insufficiency requires glucocorticoid (GC) replacement therapy that is nonphysiologic and is frequently used long-term (months to years) until the HPA axis recovers and resumes normal function.[3]
- Mifepristone (Korlym®, Corcept Therapeutics) is a competitive GR antagonist shown to stimulate HPA axis activity in patients with CD via inhibition of receptor-mediated negative feedback mechanisms.[5]
- Thus, mifepristone therapy holds the potential to restore HPA axis function in normal corticotrophs prior to TSS, similar to that observed with treatment prior to unilateral adrenalectomy for cortisol-secreting adrenal adenomas.[6]

Figure 1. HPA Axis Function (A) and Dysfunction (B) in CD With Hypercortisolism

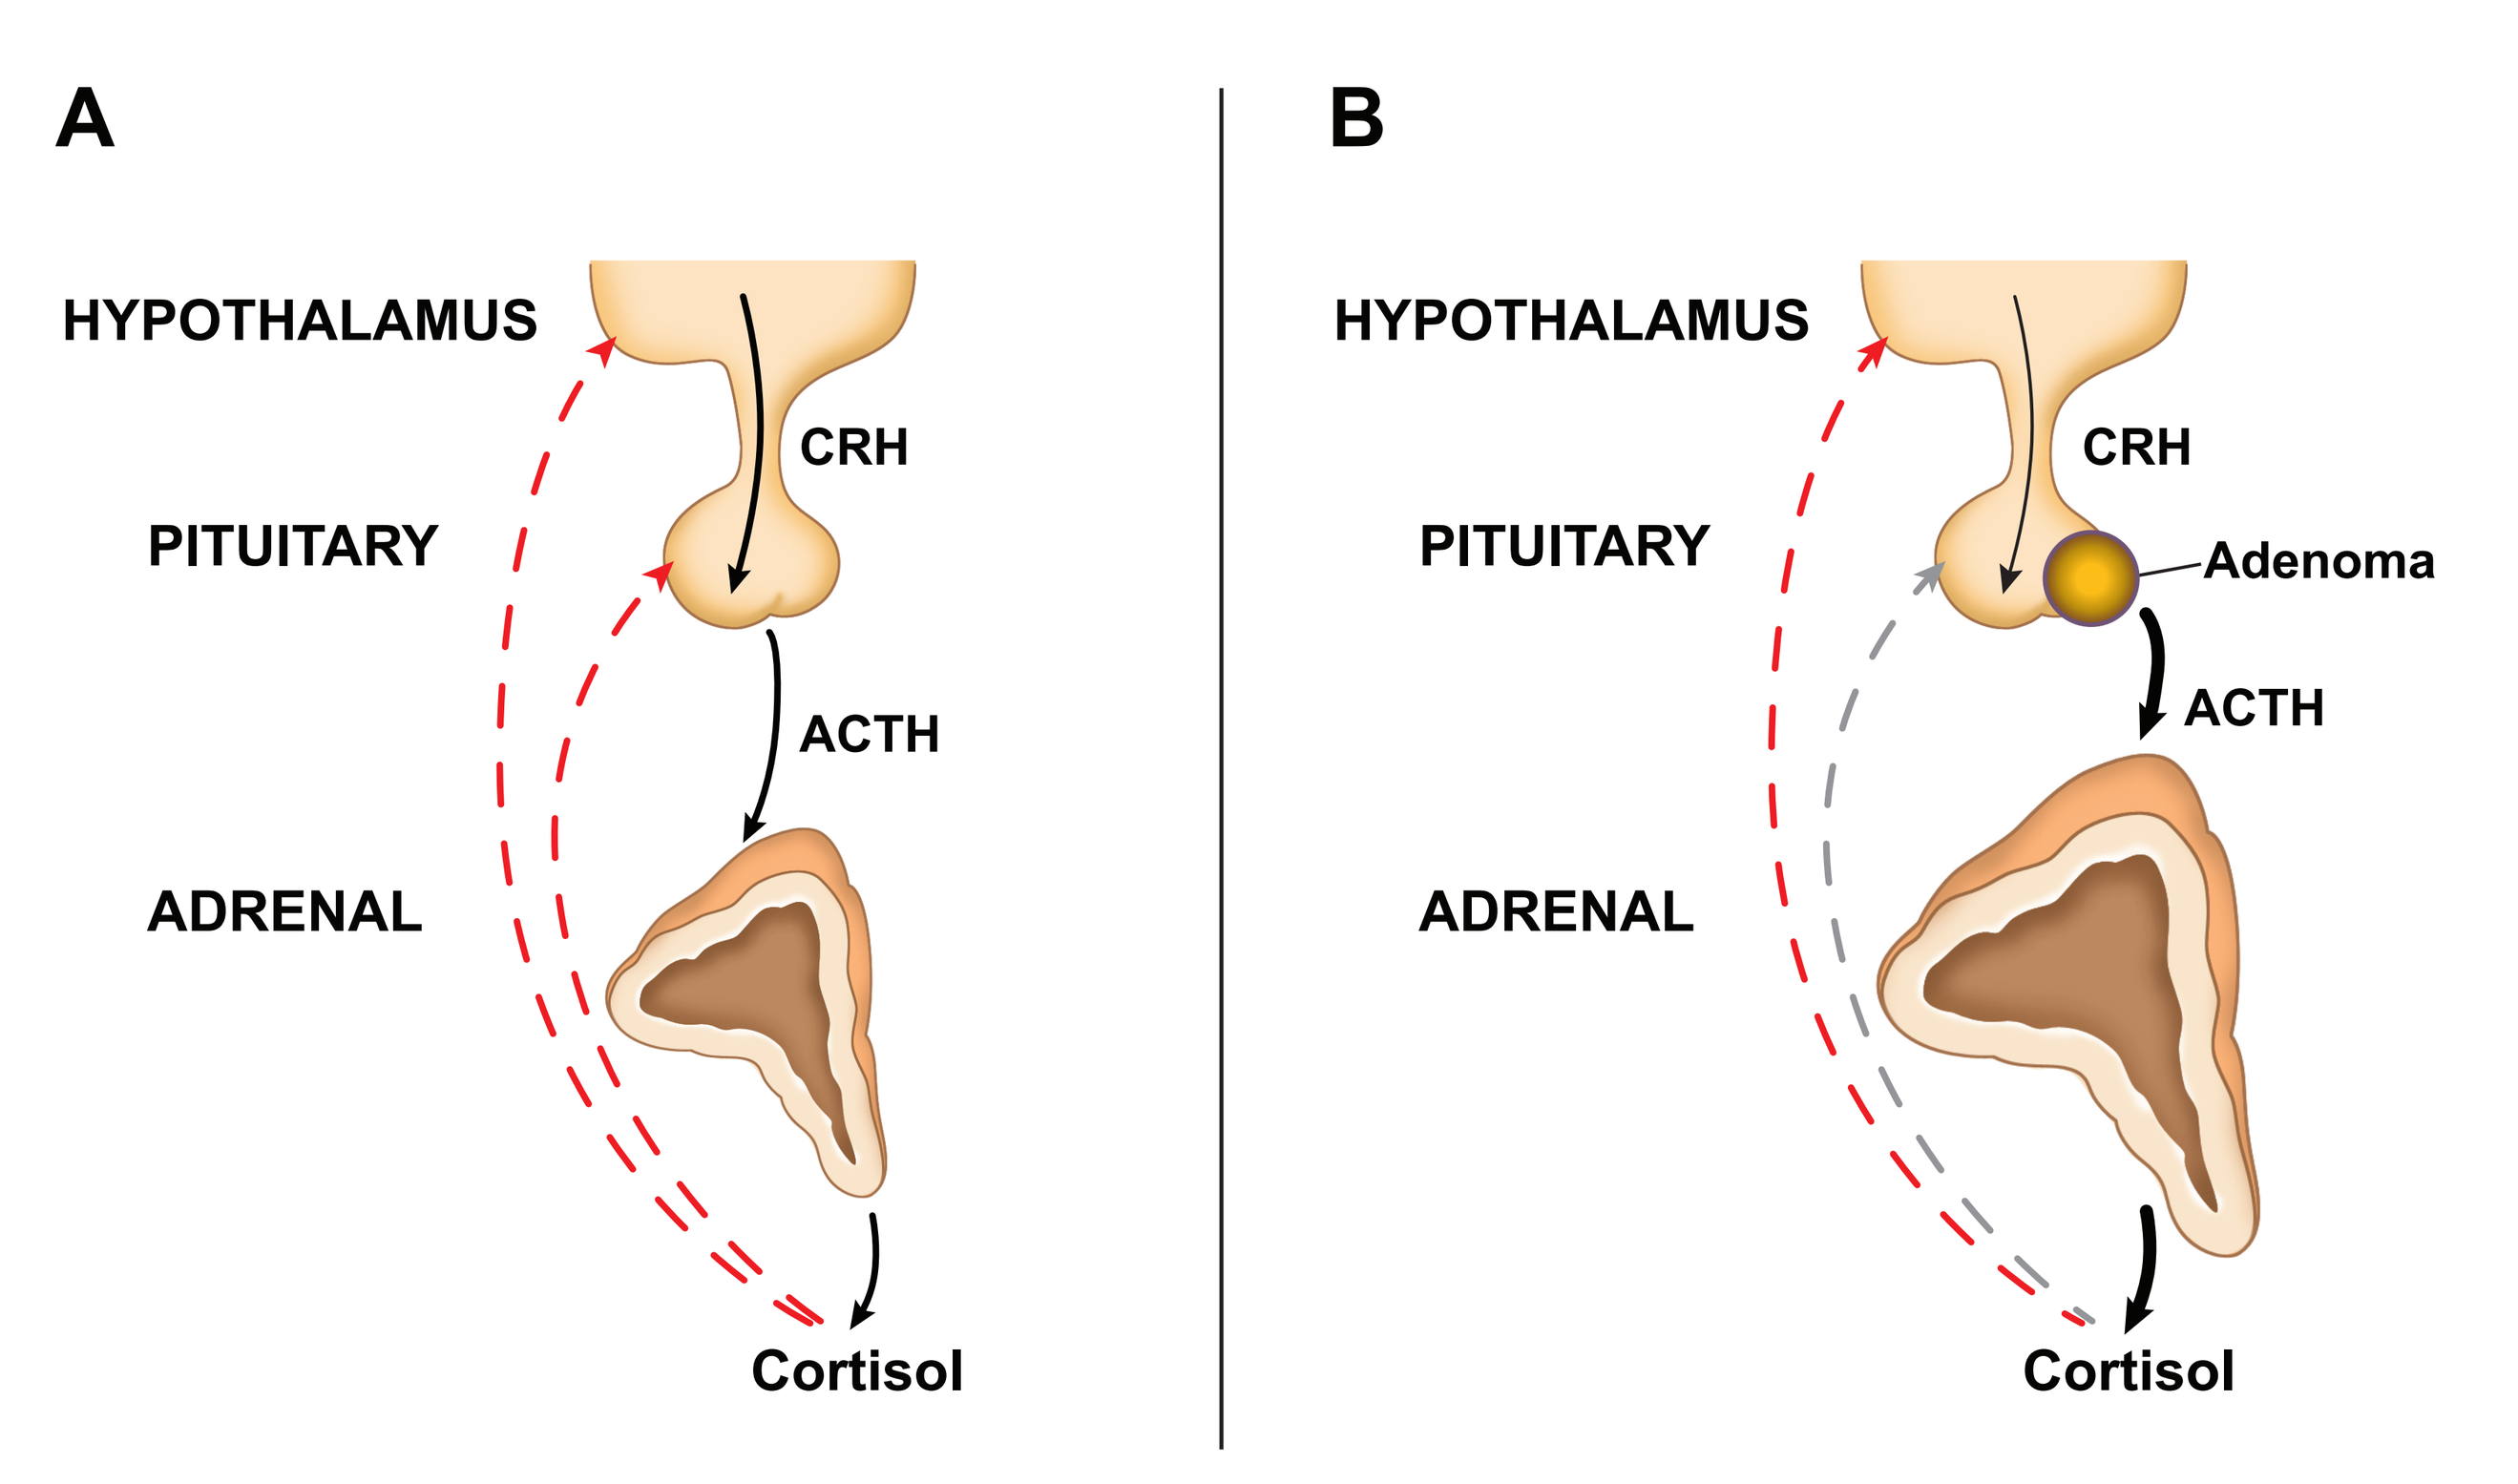
A
HYPOTHALAMUS
CRH
PITUITARY
ACTH
ADRENAL
Cortisol
B
HYPOTHALAMUS
CRH
PITUITARY
Adenoma
ACTH
ADRENAL
Cortisol

Dotted red arrows indicate negative feedback and dotted gray arrow indicates loss of negative feedback. Thicker arrows in (B) denote increased activity and thinner arrow denotes decreased CRH activity. Larger adrenal gland indicates adrenal hyperplasia.

ACTH, adrenocorticotropic hormone; CD, Cushing disease; CRH, corticotropin-releasing hormone; HPA, hypothalamic-pituitary-adrenal.

CASE PRESENTATION AND RESULTS

PATIENT HISTORY AND PRESENTATION

- A 43-year-old woman with a cushingoid appearance presented with type 2 diabetes mellitus (T2DM) (HbA1c of 10.2%).
- Her medications included acarbose (100 mg BID), repaglinide (2 mg TID), metformin (1000 mg BID), glipizide (10 mg QD), and empagliflozin (25 mg QD), as well as prior use of an insulin pump (concentrated U-500 insulin) that had been discontinued.
- Pre-treatment diagnostic workup indicated hypercortisolism (**Table 1**).

Table 1. Diagnostic Workup (6 months prior to TSS)

Diagnostic Assay	Value	Normal Range
Urinary free cortisol	57 μg/24h	≤45 μg/24h
Late-night salivary test	0.259 μg/dL	≤0.145 μg/dL
Dexamethasone suppression test	2.1 μg/dL	<1.8 μg/dL

- Imaging showed a 6 x 8 x 6 mm pituitary microadenoma (**Figure 2**) and normal adrenal glands.
- Patient was treated with mifepristone for 6 months (300 mg QD for 2 weeks and 300 mg BID thereafter).

Figure 2. MRI Image Pre-TSS Showing Pituitary Microadenoma

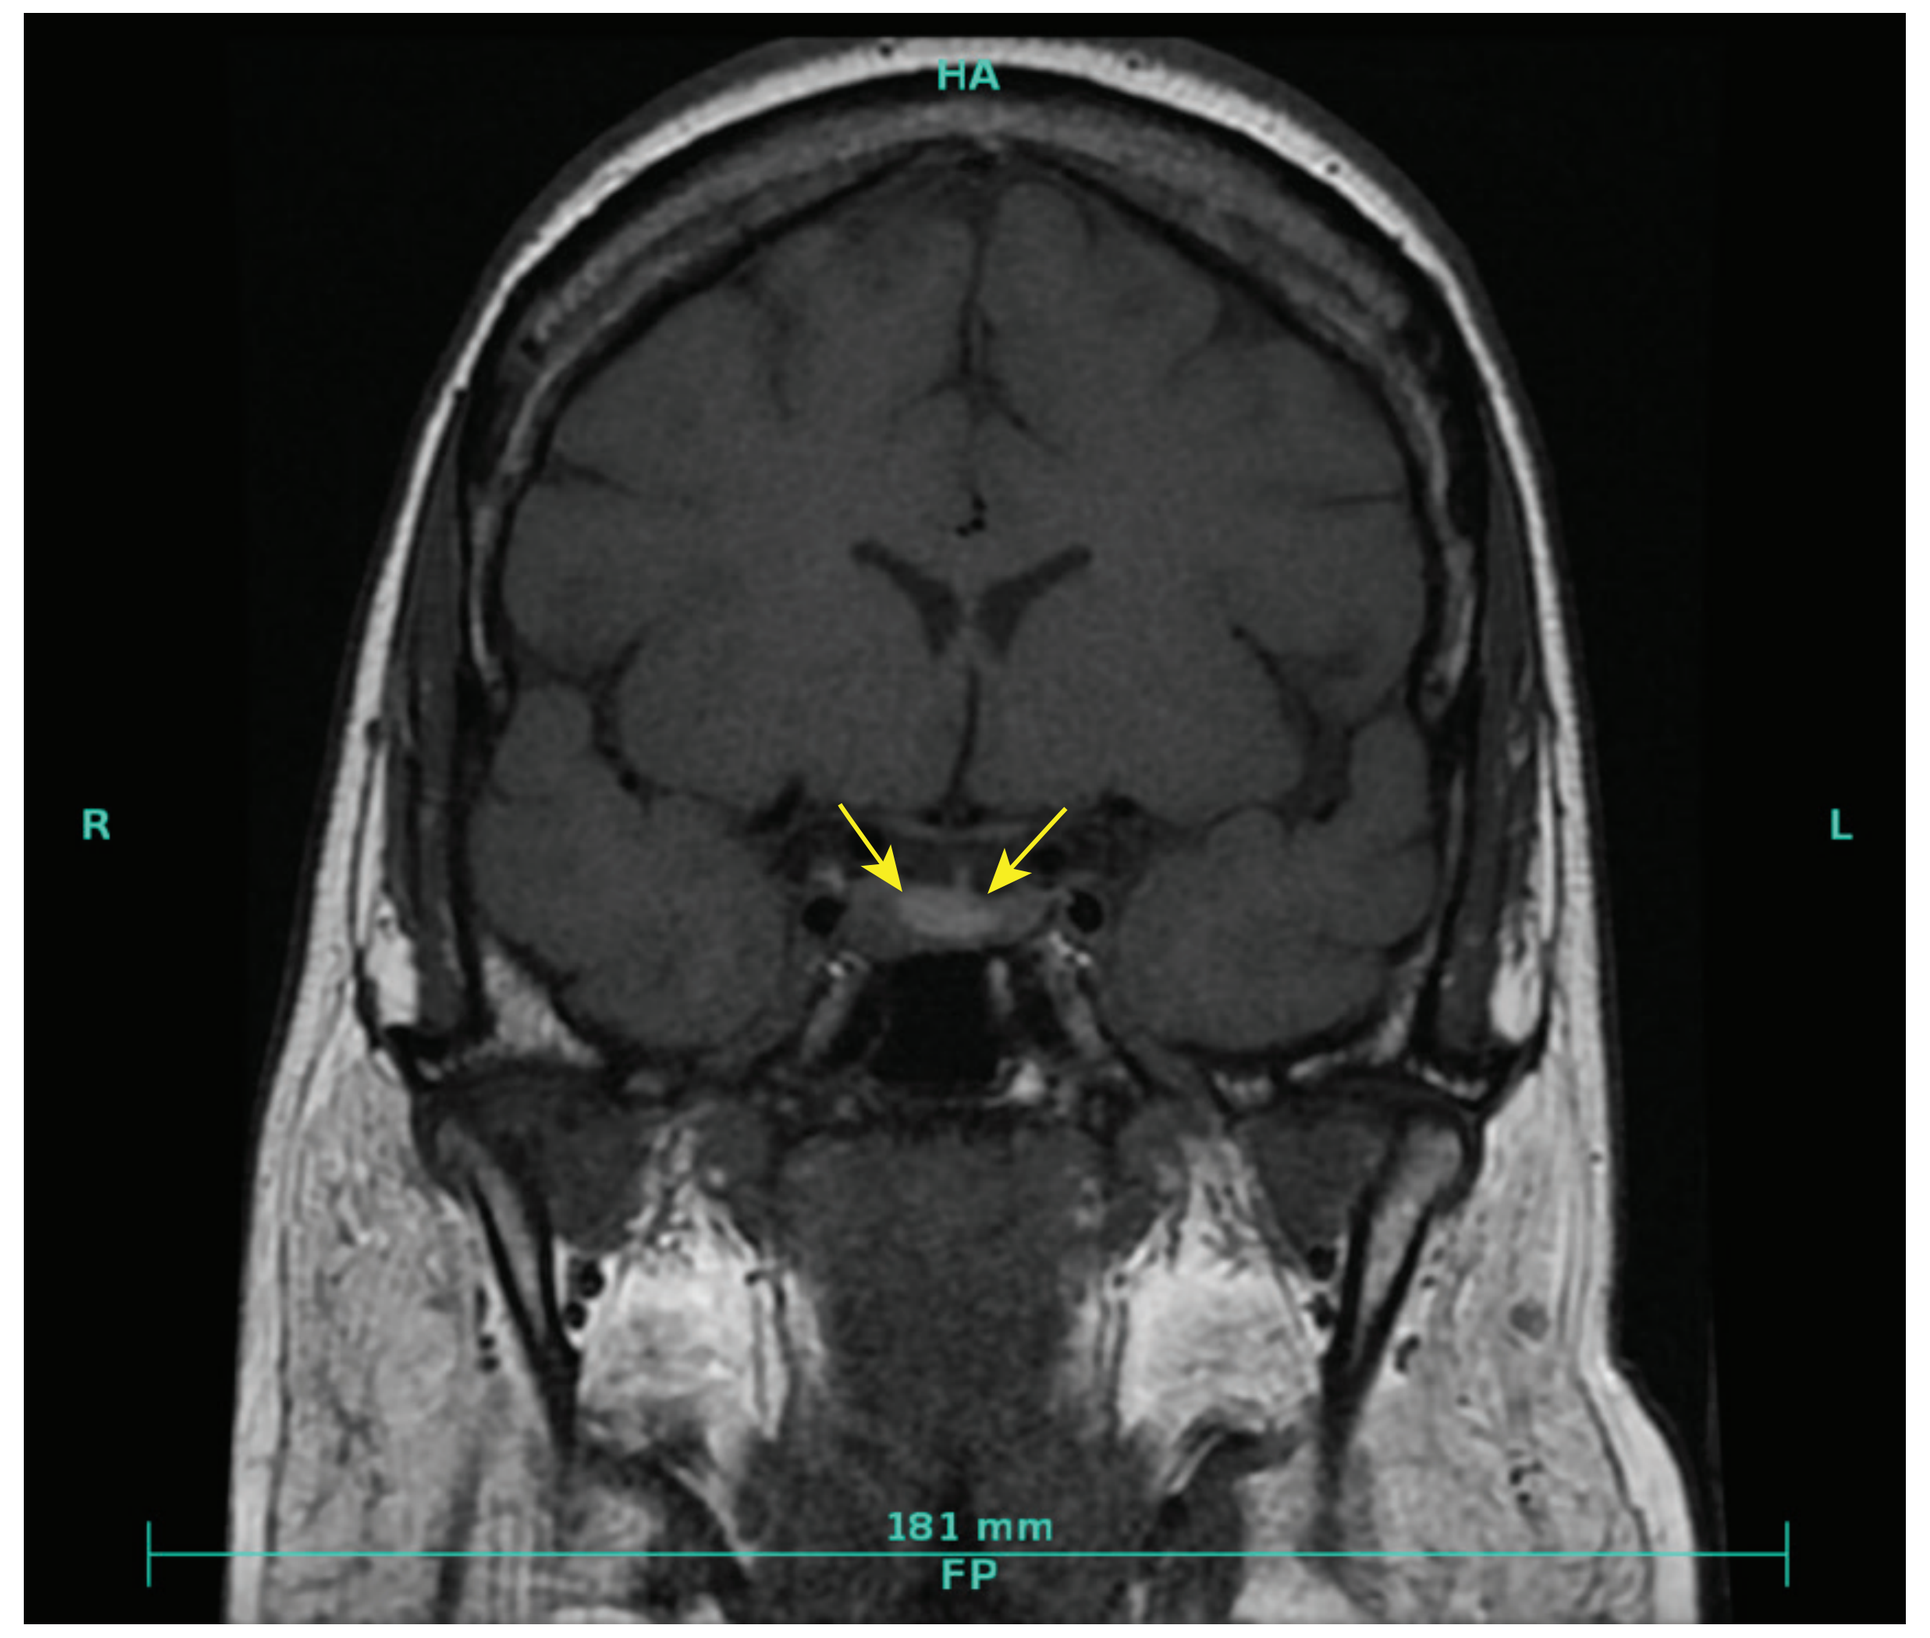
HA
R
L
181 mm
FP

MRI brain without contrast. Findings: Hypoenhancing lesion (approximately 6 x 8 x 6 mm) in the inferior and right lateral aspect of the pituitary (arrows) consistent with a pituitary microadenoma. The lesion was seen on the prior study of July 10, 2009 but better visualized on this study (September 23, 2016). No definite cavernous sinus invasion. No evidence for suprasellar extension or involvement of the optic chiasm. Unremarkable MRI of the brain parenchyma.

- **Table 2** shows cortisol and ACTH laboratory values pre- and post-treatment.

Table 2. Cortisol and ACTH Testing

Time Point	AM Cortisol (μg/dL)	UFC (μg/24h)	DST Cortisol (μg/dL)	AM ACTH (pg/mL)
	Normal: 7-28	Normal: <45	Normal: <1.8	Normal: 10-50
Baseline[a]	11.2	57	2.1	27
1 day post-TSS (dexamethasone added for 3 days)	0.6	X	X	X
2 months post-TSS	8.5	X	X	31.2
3 months post-TSS	9.1	X	X	X
5 months post-TSS	X	X	0.7	X
11 months post-TSS	8.1	X	X	X

[a]Mifepristone therapy for 6 months (2 weeks 300 mg QD and 300 mg BID thereafter).

UFC was measured only at baseline, DST was measured only at baseline and at 5-month follow-up, and ACTH was measured before mifepristone and twice post-TSS.

ACTH, adrenocorticotropic hormone; DST, dexamethasone suppression test; TSS, transsphenoidal surgery; UFC, urinary free cortisol.

RESULTS ATTRIBUTED TO MIFEPRISTONE THERAPY

- HbA1c improved (10.2% to 8.1 %) with mifepristone and further improved to 6.2 % within 11 months post-TSS.
- TSS was successful as indicated by low AM cortisol (0.6 μg/dL, considered adrenally insufficient[4]).
- Patient developed diabetes insipidus postoperatively (which is common in TSS) and was given desmopressin for 3 weeks.
- Post-TSS pathology confirmed a corticotroph adenoma (positive: ACTH; negative: thyroid-stimulating hormone, luteinizing hormone, follicle-stimulating hormone, human growth hormone, and prolactin).

SUMMARY

- Low cortisol levels after tumor resection in endogenous CD indicate successful surgery but usually result in adrenal insufficiency.
 - Often requires GC replacement therapy.
 - Only a 58% probability of recovering adrenal function within 5 years (median time for recovery, 1.4 years).[3]
- This patient with CD had a very low postoperative cortisol, indicating both a successful TSS and secondary adrenal insufficiency.
 - Within 2 months, testing revealed normal HPA axis function concomitant with clinical improvements in glucose regulation and weight reduction.
 - Eleven months post-TSS, the patient's HPA appears normal, along with additional clinical improvements.

CONCLUSIONS

- A recent analysis of patients undergoing TSS for non–ACTH-secreting pituitary adenomas concluded that an intact HPA axis prior to surgery with rising cortisol after surgery precludes the need for postoperative steroid treatment.[7]
- While theoretical, these results suggest that preoperative mifepristone therapy for CD holds the potential to normalize HPA axis function prior to TSS and reduce the need for long-term GC replacement.
- This HPA axis normalization with elimination of postoperative steroids has also been reported with preoperative mifepristone in patients with adrenal Cushing syndrome.[6]

REFERENCES

1. Raff H, Sharma ST, Nieman LK. *Compr Physiol.* 2014;4(2):739-769.
2. Raff H, Carroll T. *J Physiol.* 2015; 593(3):493-506.
3. Berr CM, Di Dalmazi G, Osswald A, et al. *J Clin Endocrinol Metab.* 2015;100(4):1300-1308.
4. Tohti M, Li J, Zhou Y, et al. *PLoS ONE.* 2015;10(3):e0119621.
5. Fleseriu M, Biller BM, Findling JW, et al. *J Clin Endocrinol Metab.*2012;97(6):2039-2049.
6. Saroka RM, Kane MP, Robinson L, Busch RS. *Clin Med Insights Endocrinol Diabetes.* 2016;26(9):31-36.
7. Woodmansee WW, Carmichael J, Kelly D, et al. *Endocr Pract.* 2015;21(7):832-838.

DISCLOSURES

PAS: None.
JJS: Employee, Corcept Therapeutics.

ACKNOWLEDGMENTS

Editorial support was provided by Mark R. Vogel of Corcept Therapeutics. Funding for design and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster and provided approval of the final version.

Presented at the Endocrine Society Annual Meeting (ENDO 2018), March 17-20, 2018, Chicago, IL