SAT-579

A CURIOUS CASE OF BIOCHEMICAL AND CLINICAL REMISSION AFTER TREATMENT WITH MIFEPRISTONE, A GLUCOCORTICOID RECEPTOR ANTAGONIST

Pejman Cohan, MD[1], Daniel F. Kelly, MD[2], Precious J. Lim, PhD[3]

[1]Specialized Endocrine Care Center, Beverly Hills, CA, USA, [2]Pacific Neuroscience Institute, Santa Monica, CA, USA, [3]Corcept Therapeutics, Menlo Park, CA, USA

INTRODUCTION

- Selective transsphenoidal adenomectomy (TSS), the current firstline treatment of patients with Cushing disease (CD), is often performed after radiologic confirmation of a pituitary adenoma.
- Surgical success rates of TSS can vary widely across centers, with the possibility of disease recurrence years after apparent disease remission.
 - In a recent multicenter study of 230 patients, only 41% of patients achieved remission after the initial TSS.[1]
 - Repeat orders of TSS have diminishing success rates.
- Patients may also present without a visible adenoma on magnetic resonance imaging (MRI), making the decision to operate difficult. This is why hypercortisolism is diagnosed clinically and biochemically.
 - It has been reported that 40%-50% of MRIs are negative when macroadenomas are absent.[2]
- We describe a case of a 23-year-old patient with recurrent CD after multiple procedures of TSS.
 - The patient exhibited clinical signs and symptoms of recurrent hypercortisolism but did not have biochemical confirmation on urine-free cortisol (UFC) and lacked radiographic evidence of an adenoma.
 - Mifepristone therapy was initiated, which effectively controlled the symptoms of her recurrence.
 - Recent literature supports early therapeutic intervention in patients with recurrent CD despite normal UFC.[3]
- Our case is unique in that she remained in biochemical and clinical remission even after discontinuation of mifepristone.
- Spontaneous remission in CD is rare, with most, if not all, cases likely caused by infarction of an adrenocorticotropic hormone (ACTH)-secreting pituitary adenoma.[4-7]
- We hypothesize that, in this case, mifepristone could have promoted tumor shrinkage and remission of CD, as evidenced clinically and biochemically.

CASE PRESENTATION AND RESULTS

- A 23-year-old woman presented in 2002 with weight gain, hypertension, depression, and other morbidities (**Figures 1** and **2**).
- Biochemical workup and inferior petrosal sinus sampling (IPSS) suggested a pituitary source of hypercortisolism (**Table 1**).
- MRI revealed a 5-mm pituitary lesion.

Figure 1. Clinical Course

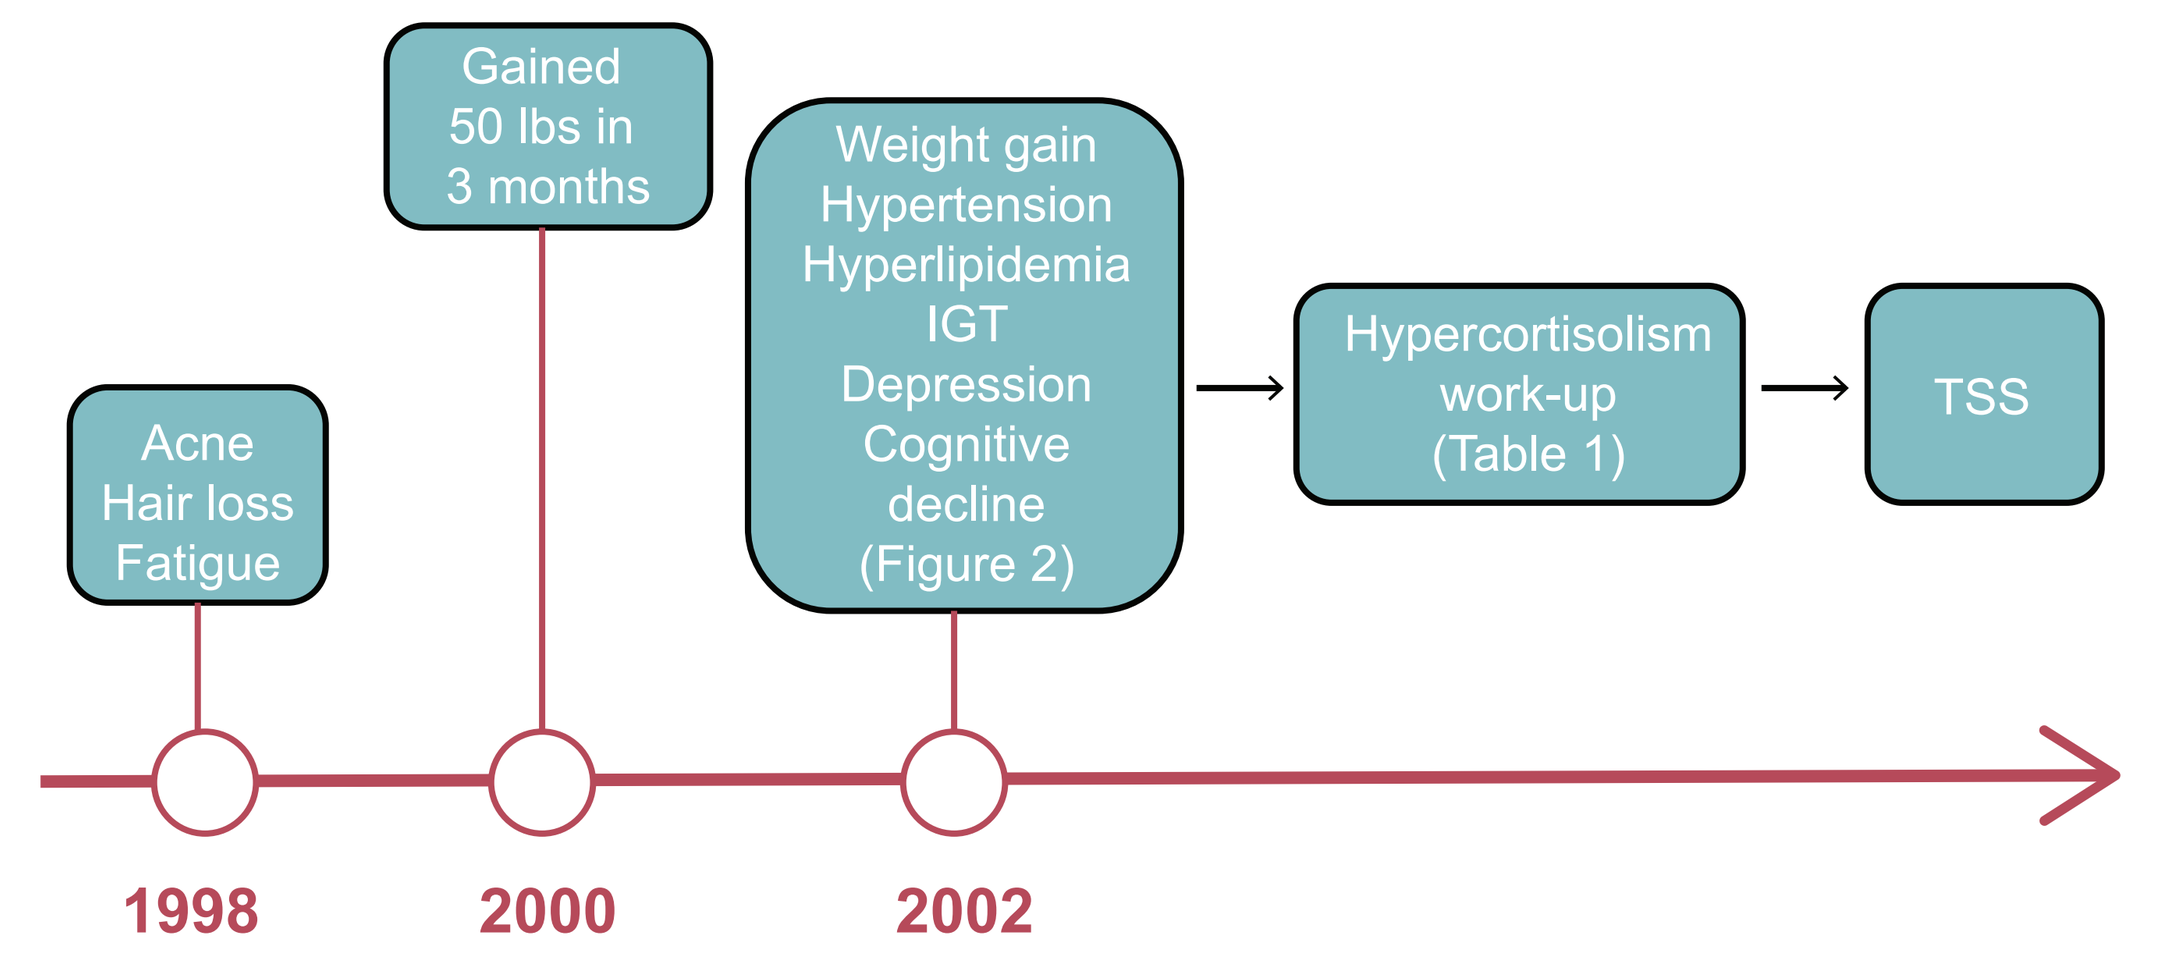
Acne
Hair loss
Fatigue
Gained
50 lbs in
3 months
Weight gain
Hypertension
Hyperlipidemia
IGT
Depression
Cognitive
decline
(Figure 2)
Hypercortisolism
work-up
(Table 1)
TSS
1998
2000
2002

Figure 2. The Patient (A) Before Hypercortisolism; (B) with Clinical Signs and Symptoms of Hypercortisolism; and (C) in Remission After Initial TSS

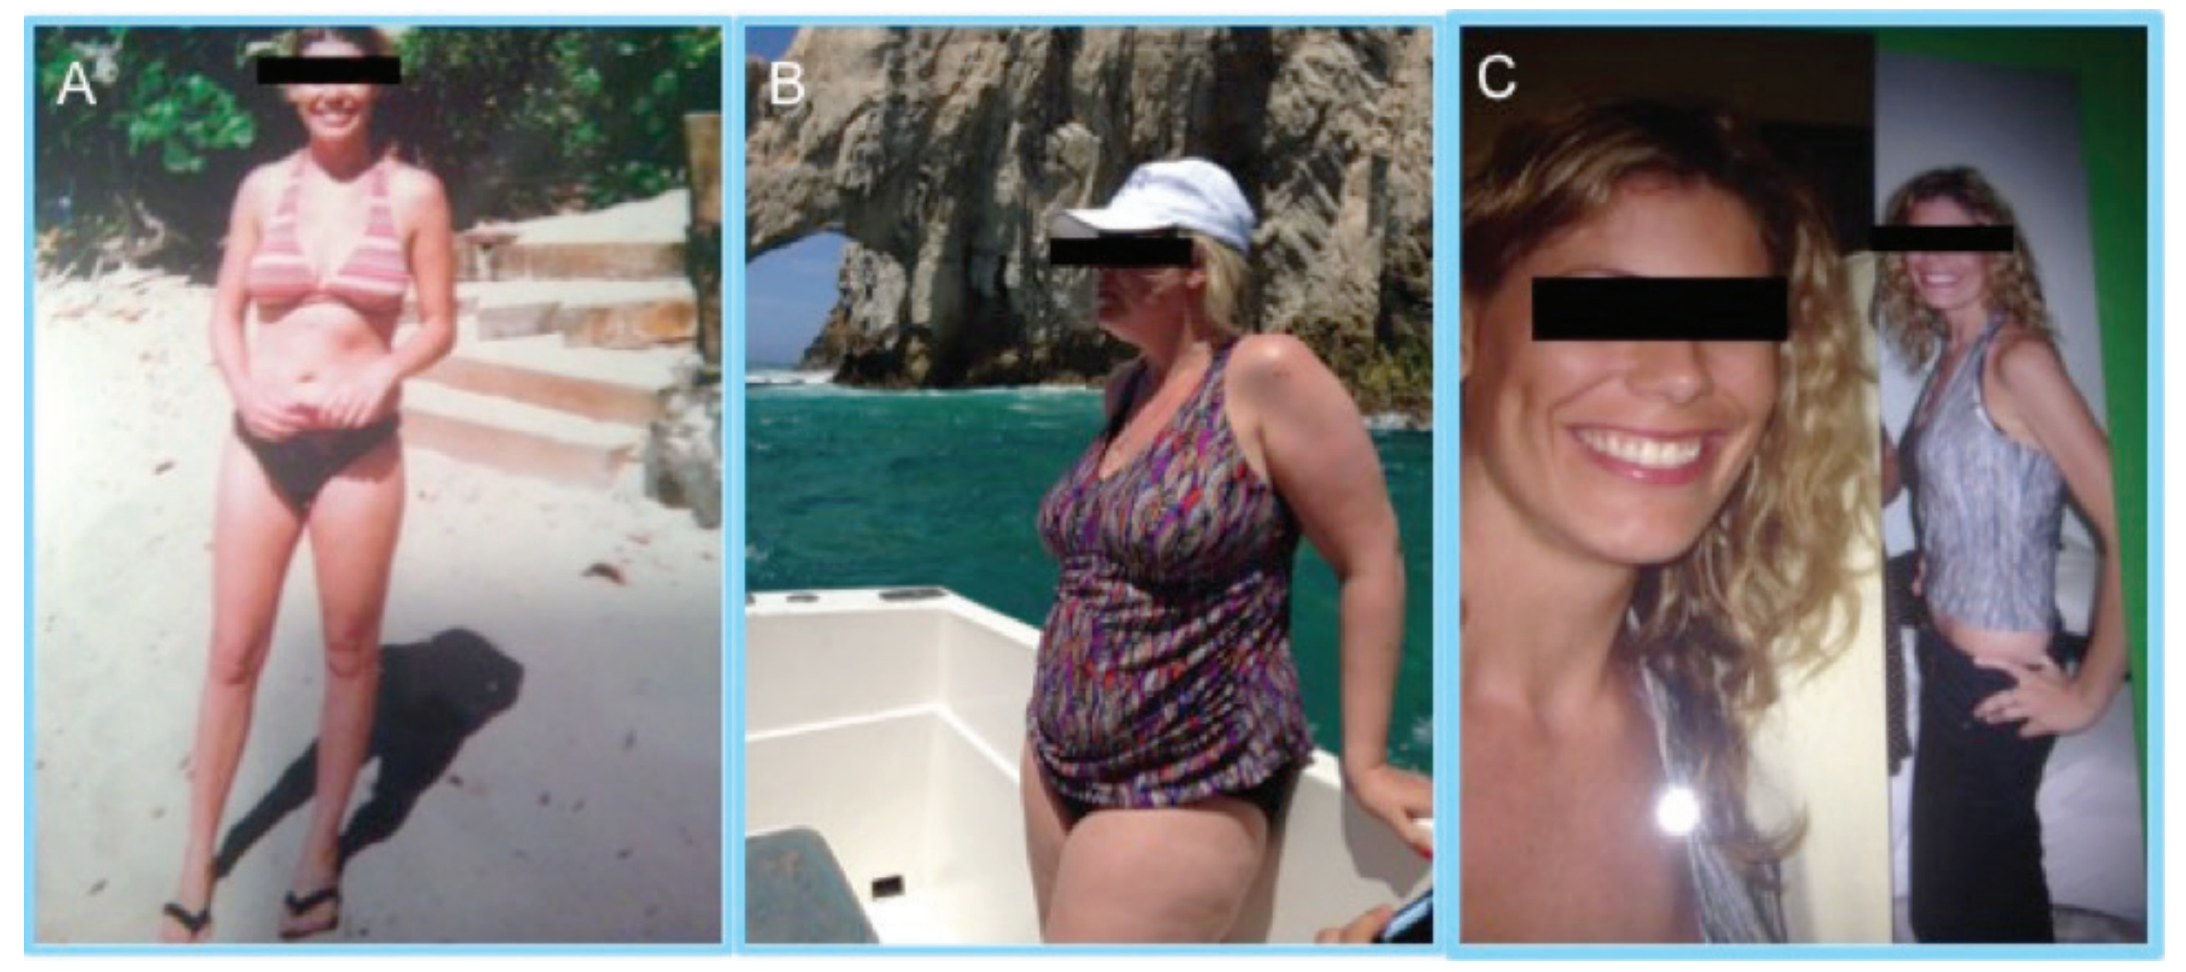
A
B
C

Table 1. Initial Diagnostic Workup

Test	Result	Normal Range
AM serum cortisol	25 μg/dL	10-20 μg/dL
AM ACTH	64 pg/mL	5-27 pg/mL
UFC	160 μg/day	<50 μg/day

ACTH, adrenocorticotropin hormone; UFC, urine-free cortisol.

FIRST TRANSSPHENOIDAL SURGERY

- The patient underwent successful TSS.
- Prolonged postoperative adrenal insufficiency required hydrocortisone replacement.
- After 5 years:
 - Hypothalamic-pituitary-adrenal (HPA) axis recovered.
 - Low insulin-like growth factor 1 (IGF-1), hypothyroidism, normal menses.
 - Weight returned to baseline (118 lbs) (**Figure 2C**).

FIRST RECURRENCE

- Seven years after initial TSS, the disease recurred (**Figure 3** and **Table 2**).
- Two years later, the patient presented with further worsening of biochemical parameters (**Table 3**).
- MRI showed a possible hypoenhancement in the right sella.

Figure 3. Course of First and Second Recurrence

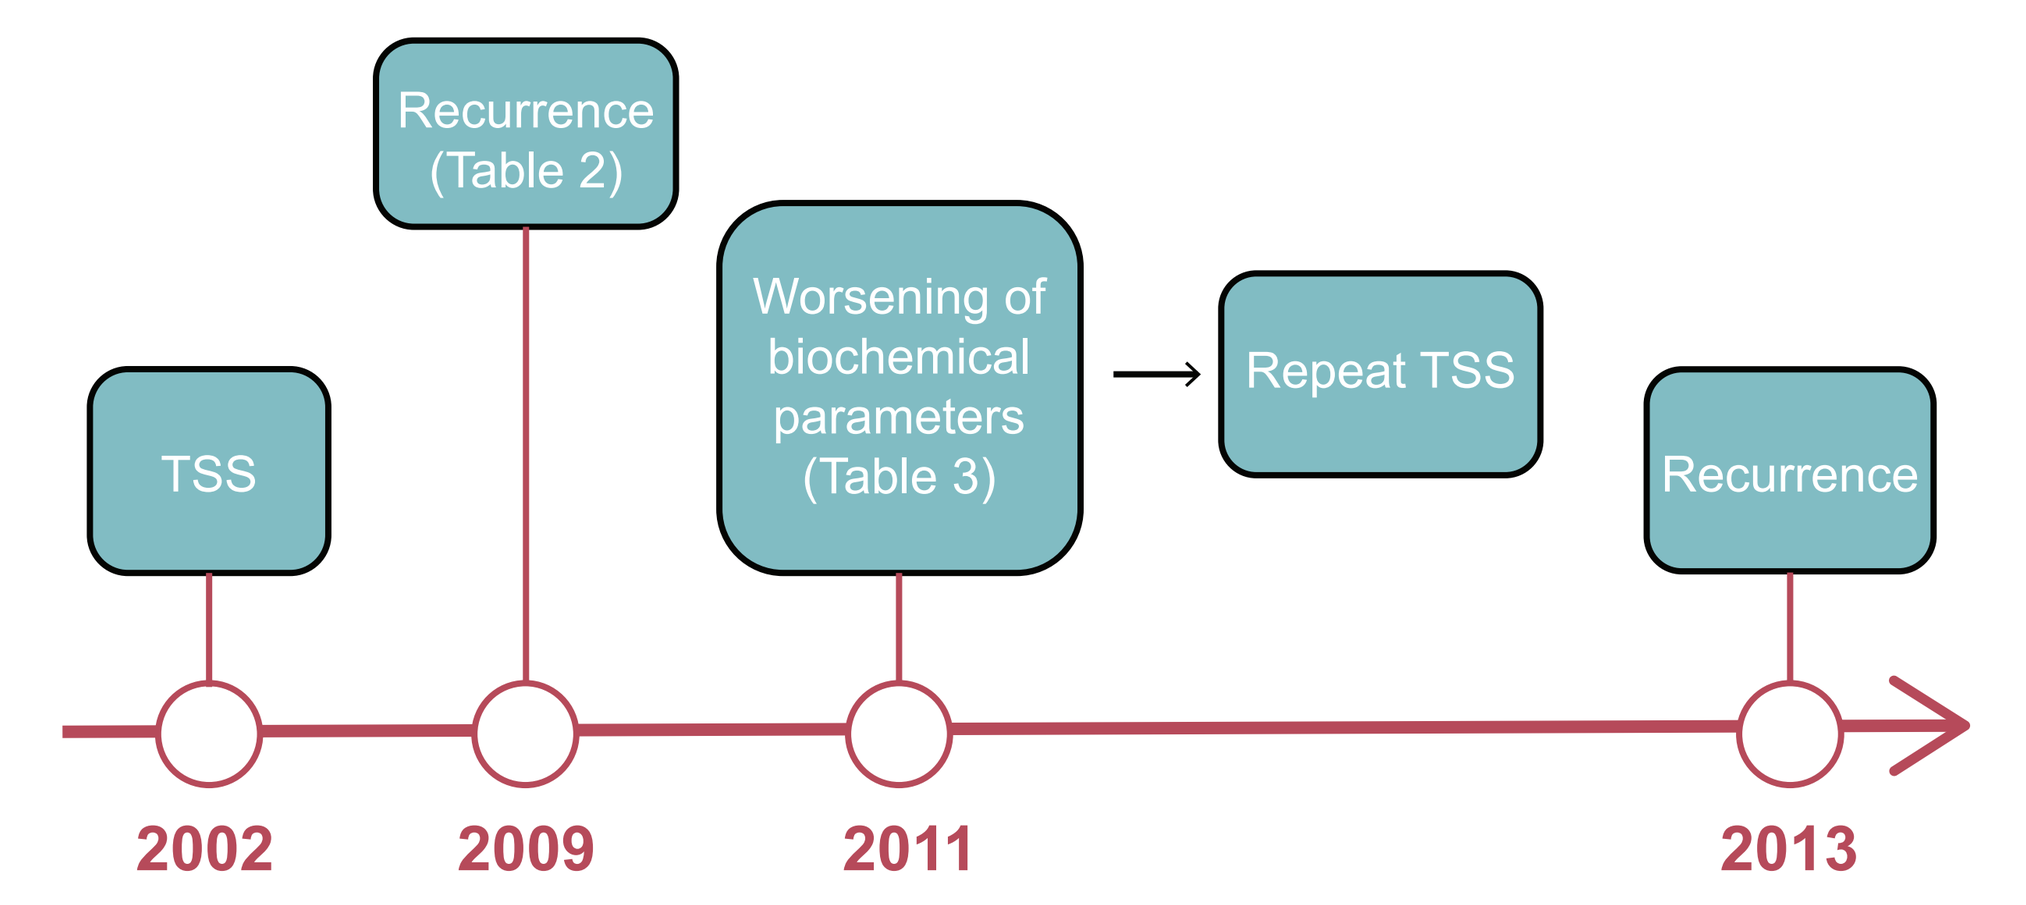
TSS
Recurrence
(Table 2)
Worsening of
biochemical
parameters
(Table 3)
Repeat TSS
Recurrence
2002
2009
2011
2013

Table 2. Biochemical and Clinical Parameters Indicating Recurrence (7 Years After First TSS)

Test	Result	Normal Range
AM ACTH	51 pg/mL	5-27 pg/mL
UFC	64 μg/day	<50 μg/day
Weight (lbs)	157 lbs	N/A

ACTH, adrenocorticotropin hormone; UFC, urine-free cortisol.

Table 3. Worsening of Biochemical and Clinical Parameters After Recurrence (9 Years After First TSS)

Test	Result	Normal Range
AM ACTH	67 pg/mL	5-27 pg/mL
UFC	168 μg/day	<50 μg/day
Weight (lbs)	202 lbs	N/A

ACTH, adrenocorticotropin hormone; UFC, urine-free cortisol.

SECOND TRANSSPHENOIDAL SURGERY

- Repeat TSS was performed with immediate postoperative cortisol of <1 μg/dL.
- The patient started hydrocortisone replacement.
- After 5 months, there was minimal weight loss (9 lbs) and lack of other clinical improvements.
- The HPA axis recovered (cortisol 9.6 μg/dL); therefore, hydrocortisone was stopped.

SECOND RECURRENCE, MIFEPRISTONE TREATMENT INITIATED

- Two years after repeat TSS, UFC rose to 55 μg/day and clinical symptoms of hypercortisolism (weight gain, depression, cognitive decline) returned, but pituitary MRI was negative (**Figure 3**).
- Mifepristone was initiated at 300 mg/day and titrated to 1500 mg/day over the course of 3 months based on clinical response (**Table 4**).
- Mifepristone was discontinued when patient developed amenorrhea (**Table 5**).

Table 4. Mifepristone Treatment Outcomes

Mifepristone dose	1500 mg/day
Clinical improvements	• Weight loss of 20 lbs • Improved mood
Biochemical changes	
AM ACTH (5-27 pg/mL)	85.6 pg/mL
AM serum cortisol (10-20 μg/dL)	37.5 μg/dL

ACTH, adrenocorticotropin hormone.

Table 5. Mifepristone Adverse Events and Interventions

Adverse Event	Intervention
Vaginal bleeding at treatment initiation	Resolved without intervention
Amenorrhea	Mifepristone treatment discontinued

BIOCHEMICAL REMISSION POST-MIFEPRISTONE

- Five months after discontinuation of mifepristone, biochemical parameters were normal, so another MRI was deemed unnecessary (**Table 6**).
- To date (follow-up of 27 months after discontinuing mifepristone), clinical improvements are sustained.
- Repeat biochemical testing deemed unnecessary due to lack of clinical signs and symptoms indicating recurrence.

Table 6. Biochemical Remission After Mifepristone Treatment

AM serum cortisol (10-20 μg/dL)	16.3 μg/dL
AM ACTH (5-27 pg/mL)	19 pg/mL
UFC (ULN <50 μg/day)	30 μg/day

ACTH, adrenocorticotropin hormone; UFC, urine-free cortisol; ULN, upper limit of normal.

DISCUSSION

- Spontaneous and sustained remission is rare in CD.
- Since pituitary apoplexy or infarction is usually the presumed mechanism for nonsurgical remission of hypercortisolism in CD patients, it is possible that this could also be an underlying mechanism in this patient treated with mifepristone.
- We postulate that a nonvisible microadenoma may have been the source of the patient's recurrent hypercortisolism.
- Studies with mifepristone have reported regression of corticotroph tumors in 2 patients, one with a macroadenoma (with prior radiation therapy) and one with a microadenoma (with no radiation therapy).[8]
- Careful follow-up studies are needed to further investigate the exact pathophysiology of spontaneous remission with mifepristone therapy.

REFERENCES

1. Geer EB, et al. *Endocr Pract.* 2017; 23(8): 962-970.
2. Wagner-Bartak NA, et al. *Am J Roentgenol.* 2017;209:19-32.
3. Carroll TB, et al. *Endocr Pract.* 2016; 22(10):1216-1223.
4. Ishibashi M, et al. *Arch Intern Med.* 1993;153(2):251-255.
5. Dickstein G, et al. *Arch Intern Med.* 1991;151(1):185-189.
6. Taylor HC, et al. *Endocr Pract.* 2003;9(2):147-151.
7. Pignatta AB, et al. *Pituitary.* 2004;7(1):45-49.
8. Fleseriu M, et al. *J Clin Endocrinol Metab.* 2014;99(10):3718-3727.

DISCLOSURES

PC: Consultant, Corcept Therapeutics; Speaker, Corcept Therapeutics, Pfizer.
DK: None.
PJL: Employee, Corcept Therapeutics.

ACKNOWLEDGMENTS

Editorial support was provided by Mark R. Vogel of Corcept Therapeutics. Funding for design and production support for this poster was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster and provided approval of the final version.

Presented at the Endocrine Society Annual Meeting (ENDO 2018), March 17-20, 2018, Chicago, IL