SUN-324

ADMINISTRATION OF LOCALIZED PROGESTIN BEFORE MIFEPRISTONE TREATMENT MAY PREVENT ENDOMETRIAL THICKENING AND BREAKTHROUGH BLEEDING

John P. O'Dea, MB, BSc[1]; Precious J. Lim, PhD[2]
[1]O'Dea Medical Hormone Center, Marina Del Rey, CA; [2]Corcept Therapeutics, Menlo Park, CA

INTRODUCTION

- Endometrial thickening is a known potential consequence of mifepristone and other progesterone-receptor modulators.
 - Histological changes associated with endometrial thickening are benign and consistent with progesterone-associated endometrial changes (PAEC) typically observed with progesterone-receptor modulators.
- Dose- and time-dependency for these events cannot be established.
- Thickening of the endometrium often improves when drug is temporarily suspended.
- Oral progesterone and levonorgestrel-releasing intrauterine devices (IUDs) have been tried in response to endometrial thickening, with varying results.
- Data on the proactive use of levonorgestrel-releasing IUDs in patients taking mifepristone are not available.

METHODS

- Charts from 6 premenopausal patients with levonorgestrel-releasing IUDs inserted prior to mifepristone treatment were reviewed for the prevalence of endometrial thickening, spotting, and breakthrough bleeding.

CASE PRESENTATION AND RESULTS

Table 1. Patient Characteristics at Baseline

Patient	Age (yrs)	BMI (kg/m²)	Clinical Diagnoses Prior to Treatment
1	20	39.2	• Polycystic ovary syndrome (PCOS) • Mixed hyperlipidemia • Metabolic syndrome • Major depressive disorder (MDD) • Sleep disorder • Generalized anxiety • Fatty liver • Migraines
2	21	47.2	• PCOS • Obesity • Metabolic syndrome • Hyperandrogenism • MDD • Severe anxiety • Obsessive compulsive disorder (OCD) • Self-injurious behavior • Mixed hyperlipidemia
3	21	19.6	• Hormone-sensitive migraines • Generalized anxiety
4	29	33.2	• MDD • Sleep disorder • Migraine syndrome • Fibromyalgia • Chronic pain • Joint hypermobility
5	38	17.9	• PCOS • OCD • Severe anxiety • Hyperandrogenism
6	42	32.9	• PCOS • Metabolic syndrome • Mixed hyperlipidemia • Hashimoto thyroiditis • Primary hypothyroidism • Fatty liver

Table 2. Patient Outcomes

Patient	Duration of Mifepristone Treatment	Maximum Average Daily Dose	Weight (lbs)		Other Clinical Improvements
			Pre	Post	
1	6 months	300 mg	250	231	• Liver enzymes normalized after 1 month • Normalized blood pressure • Less depressed and general feeling of well-being • Improved cognitive function • Improved mental clarity • Milder headaches
2	2 months	300 mg	250	240	• Normalized blood pressure • Less anxiety • Fewer obsessions
3	4 months	300 mg	118	NA	• Improvement in anxiety
4	6 months	600 mg	206	191	• Normalized blood pressure • Improved MDD • Less frequent and less severe migraines
5	6 months	600 mg	132	NA	• Significant improvements in OCD
6	4 months	450 mg	198	188	• Liver enzymes normalized after 1 month

NA, not available.

Table 3. IUD Product by Patient

Patient	IUD Product	Spotting	Breakthrough Bleeding	Endometrial Thickening
1	Skyla®	Very light menses once a month	None	None
2	Skyla	Slight spotting once	None	None
3	Mirena®	None	None	None
4	Mirena	Mild spotting	None	None
5	Skyla	None	None	None
6	Mirena	Very slight spotting	None	None

IUD, intrauterine device

- Four of 6 premenopausal patients reported light spotting, although none had breakthrough bleeding.
- None had endometrial thickening as per vaginal ultrasound.
- Charts from postmenopausal women were also reviewed, and none reported spotting or breakthrough bleeding.

DISCUSSION

ROLE OF PROGESTERONE OR PROGESTIN IN PREVENTING ENDOMETRIAL THICKENING

- Endometrial thickening develops when estrogen, unopposed by progesterone, stimulates endometrial cell growth.
- Progesterone therapy can thus be used to treat endometrial thickening.
- Progesterone counteracts the effects of estrogen and thins the uterine lining.
- Oral progesterone, DepoProvera® (injection), or an IUD are possible treatment options.

INTRAUTERINE VS OTHER ROUTES OF ADMINISTRATION

- Steady localized release of progestin is preferred over the erratic patterns of oral, systemically active drugs.
- The steady localized release of progestins from levonorgestrel-releasing IUDs minimizes systemic progesterone or progestin in patients.
- Systemic progestins, operating only genomically via nuclear receptors, are distinctly depressogenic.[1]
 - Medroxyprogesterone acetate (MPA) is a distinctly atypical synthetic progestin.
 - MPA is about 30 times stronger than progesterone, with overlapping glucocorticoid agonist effects.
 - MPA fuels major depressive disorder, weight gain, and fatigue.
- Systemic progesterone and progestins may block or oppose some of the surprising benefits of mifepristone and/or estradiol.
 - Migraines are fueled by estrogen withdrawal; hence, the existence of menstrual migraines and menopausal migraines, both of which are the same estradiol-withdrawal migraines.[2]
 - Levonorgestrel-releasing IUDs promote amenorrhea, thus minimizing menstrual migraines in younger patients and hormone therapy-related migraines in menopausal patients.
- Natural progesterone, although less associated with depression than the synthetic progestins, also fuels depression and fatigue via genomic nuclear receptors and via its nongenomic, nonhormonal, neuromodulatory, GABA-agonist effects.
- Levonorgestrel-releasing IUDs reliably prevent endometrial hyperplasia.
 - The Royal College of Obstetricians and Gynaecologists published a guideline in 2016 on the management of endometrial hyperplasia.
 - Levonorgestrel-releasing IUDs are the first-line treatment.
 - Compared to oral progestogens, these IUDs have a higher disease regression rate with a more favorable bleeding profile, and are associated with fewer adverse effects

SPOTTING AND/OR BLEEDING ON LEVONORGESTREL-RELEASING IUD AND MIFEPRISTONE

- Frequent spotting or light bleeding in the first 3 to 6 months are associated with a levonorgestrel-releasing IUD[3] and could explain the light spotting reported by 4 patients.
 - Bleeding may be heavier and more frequent in the first 3 to 6 months of IUD insertion, but none was experienced by any of the patients.
- Bleeding at the start of mifepristone has been commonly reported[4,5] and could also explain the light spotting reported by 4 patients.
- Many patients on mifepristone are administered spironolactone.
 - Spironolactone is an anti-androgenic progestin in addition to its mineralocorticoid antagonist effects.
 - Spironolactone has a distinct tendency to fuel breakthrough bleeding by virtue of its progestagenic properties.

LEARNINGS AND FUTURE RESEARCH

- It could be argued that the patients did not experience bleeding because mifepristone was ineffective.
- Clinical improvements demonstrate the efficacy of mifepristone; thus the absence of bleeding could possibly be attributed to the levonorgestrel-releasing IUD.
- Proactive use of these IUDs is preferred before the endometrium has thickened, which can make insertion more challenging.
- However, with a small number of patients, it is difficult to definitively conclude that these IUDs prevented the endometrial thickening and bleeding associated with mifepristone.

CONCLUSIONS

- In patients who received a levonorgestrel-releasing IUD prior to initiating mifepristone, none experienced endometrial thickening or breakthrough bleeding.
- The use of localized progestin before mifepristone treatment may be an effective method to prevent endometrial thickening and breakthrough bleeding as a result of the progesterone antagonist activity of mifepristone.
- Future studies should target premenopausal women who require mifepristone treatment and compare the efficacy of IUDs vs oral progestins administered before initiating mifepristone therapy.

REFERENCES

1. Kukami J. *Expert Opin Drug Saf.* 2007;6(4):371-374.
2. O'Dea JP, Davis EH. *Neurology.* 1990;40:1470-1471.
3. Mirena® (levonorgestrel-releasing intrauterine system) [prescribing information]. Whippany, NJ: Bayer Corporation; June 2017.
4. Hapangama DK, et al. *J Clin Endocrinol Metab.* 2002;87(11):5229-5234.
5. Schaison G, et al. *J Clin Endocrinol Metab.* 1985;61(3):484-489.

DISCLOSURES

JPO: None.

PJL: Employee, Corcept Therapeutics.

ACKNOWLEDGMENTS

Editorial support was provided by Mark R. Vogel of Corcept Therapeutics. Funding for design and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster and provided approval of the final version.

Presented at the Endocrine Society Annual Meeting (ENDO 2018), March 17-20, 2018, Chicago, IL