SAT-580

COMBINING AMILORIDE AND SPIRONOLACTONE TO MANAGE HYPOKALEMIA AND EDEMA IN PATIENTS WITH CUSHING SYNDROME BEING TREATED WITH MIFEPRISTONE

Jennifer Cheng, DO[1], Rachel L. Bunta, MBA[2], Adnan Haider, MD[3]

[1]Jersey Shore University Medical Center, Neptune, NJ, USA, [2]Corcept Therapeutics, Menlo Park, CA, USA, [3]West Virginia University, Morgantown, WV, USA.

INTRODUCTION

- Hypokalemia, edema, and hypertension are prevalent in patients with ectopic adrenocorticotropic hormone (ACTH)-secreting (EAS) Cushing syndrome (CS).
- Cortisol, at high concentrations, can act like a mineralocorticoid (aldosterone), leading to worsening hypertension, hypokalemia, and edema.
- Hypercortisolism treatment with mifepristone (Korlym®, Corcept Therapeutics) can increase cortisol levels due to its competitive glucocorticoid receptor antagonism.[1]
 - In an open-label, 24-week, multicenter SEISMIC trial, hypokalemia and edema occurred in 44% and 26% of patients, respectively.[1]
- For patients with hypercortisolism, effective management of the mineralocorticoid-related clinical manifestations is essential for the successful treatment of hypercortisolism.[1]
 - Hypokalemia should be corrected prior to initiating mifepristone and monitored frequently during treatment.[2]
 - Potassium should be measured 1 to 2 weeks after starting or increasing the dose of mifepristone and monitored periodically thereafter.
 - Hypokalemia should be treated with potassium supplementation, and a mineralocorticoid antagonist (eg, spironolactone) can be added if needed.
- Given the abnormally elevated levels of cortisol in patients with EAS, potassium supplementation and spironolactone might not be enough; an additional potassium-sparing diuretic may be required.

CASE PRESENTATION

PATIENT HISTORY AND PRESENTATION

- A 32-year-old male patient reported gaining 50 lbs in 5 months and presented with violaceous abdominal striae (**Figure 1B**), lower extremity edema (**Figure 1C**), and muscle weakness. **Figure 1A** shows this patient 10 months prior to diagnosis.
- The patient had a history of hypokalemia and was treated with potassium supplementation (40 mEq BID of potassium chloride).
- His hypertension remained uncontrolled (BP 140/90) despite metoprolol 50 mg daily, amlodipine 10 mg daily, lisinopril 20 mg daily, furosemide 40 mg daily, and hydrochlorothiazide 12.5 mg daily.
- His glycated hemoglobin (HbA1c) level was 9.9% despite taking metformin 500 mg BID, insulin aspart 135 units/day, and insulin glargine 110 units/day.
- He had osteoporotic compression fractures in the lumbar spine, a rib fracture, and severe chronic back pain.
- The patient reported depression and had suicidal ideations.
- See **Table 1** for baseline laboratory values.

Figure 1. Patient Presentation

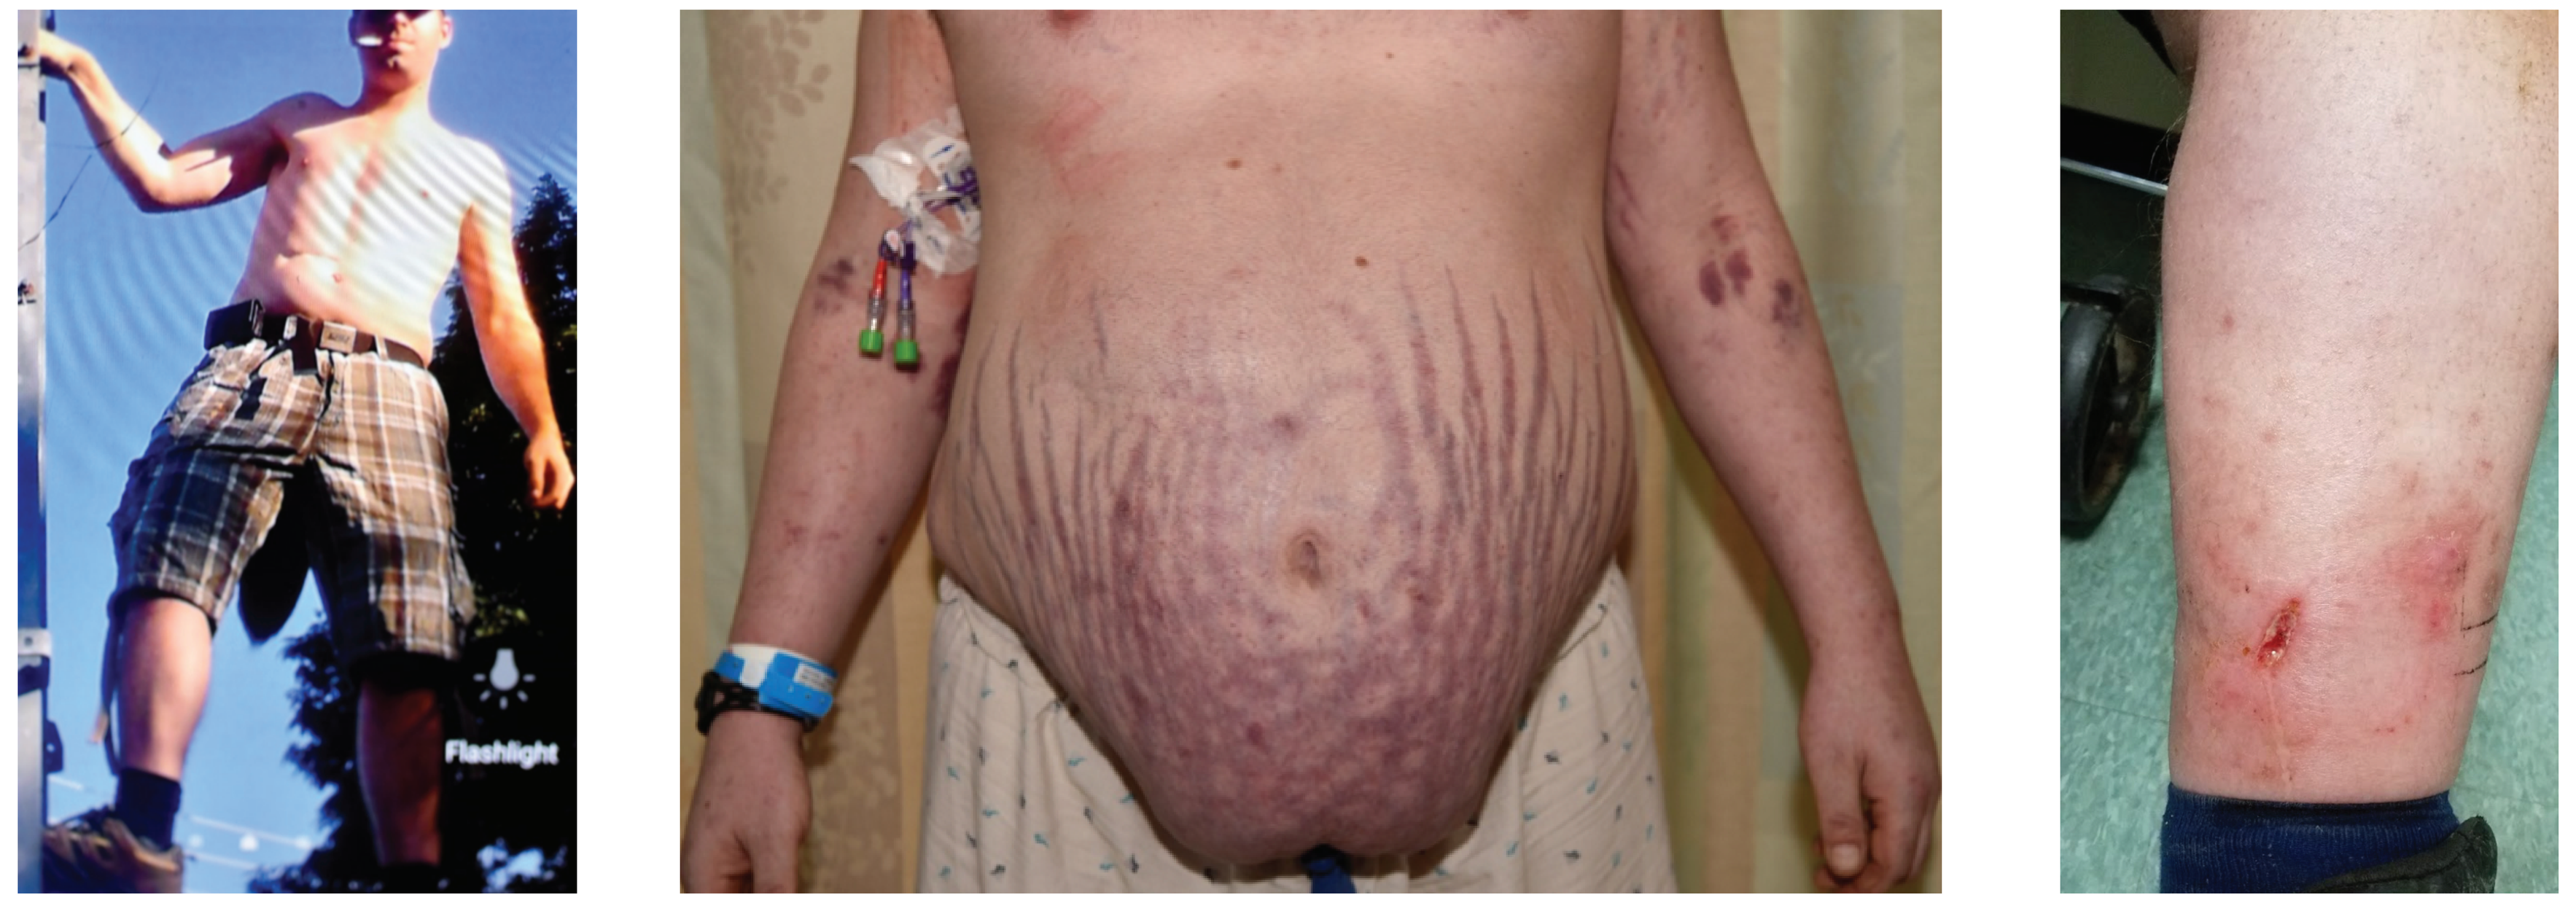
Flashlight

A. Patient 10 Months Prior to Cushing Syndrome Diagnosis

B. Abdomen with Striae (Before Mifepristone Treatment)

C. Persistent Edema with Poor Wound Healing (Before Mifepristone Treatment)

DIAGNOSTIC WORKUP

- Baseline laboratory assessments confirmed ACTH-dependent CS.
- An octreotide scan could not localize the tumor.

Table 1. Baseline Laboratory Values

	Laboratory Value	Normal Range
Fasting glucose, mg/dL	200-240	65-99
HbA1c, %	9.9	<5.7
Potassium, mEq/L	3.9	3.3-4.9
Sodium, mEq/L	139	134-142
BUN, mg/dL	26	8-26
Creatinine, mg/dL	0.72	0.6-1.2
AM cortisol, µg/dL	28.7	7.0-25
UFC, µg/dL	4995, 1570	4-50
LNSC, nmol/L	4.49	0.3-4.3
ACTH, pg/mL	170	6-50

HbA1c, glycated hemoglobin; ACTH, adrenocorticotropic hormone; BUN, blood urea nitrogen; LNSC, late night salivary cortisol; UFC, urinary free cortisol.

TREATMENT SELECTION

- Mifepristone 300 mg was initiated and eventually titrated to 1200 mg to treat the patient's hypercortisolism.

RESULTS

- After being on mifepristone for 1 month and having reduced the potassium supplementation 20 mEq/day, he developed hypokalemia.
- Spironolactone 100 mg BID was added; furosemide and hydrochlorothiazide were discontinued (**Table 2**).
- The patient subsequently developed edema. He gained 10 lbs and had decreased urination; his dosage of spironolactone was increased to 300 mg QD, and amiloride 10 mg BID was added.
- After 1 month of the combination diuretics, the patient lost 9 lbs and his edema improved (**Figure 2**).
- Potassium supplementation was discontinued (**Table 2**).
- Three months after mifepristone therapy, the patient reported improvements in psychiatric symptoms: depression and anxiety lessened; he was more hopeful and no longer had suicidal ideations.
- At 5 months, BP improved to 116/68 even with the discontinuation of metoprolol and lisinopril (**Table 2**).
- At 8 months, HbA1c improved from 9.9% to 6.6%.
 - Metformin and insulin aspart were discontinued; daily insulin glargine requirements were reduced 30 units/day (**Table 3**).
- Muscle strength improved.
- Abdominal girth decreased and striae improved.
- Potassium levels remained stable while on spironolactone and amiloride. The patient's edema decreased and remained stable until he underwent surgery for a lung carcinoid that was later discovered.

Table 2. Medication List

Baseline	After Mifepristone Treatment	After Surgery
Potassium chloride 40 mEq BID	Discontinued	--
Metoprolol 50 mg QD	Discontinued	--
Amlodipine 10 mg QD		Discontinued
Lisinopril 20 mg QD	Discontinued	--
Hydrochlorothiazide 12.5 mg QD	Discontinued	
Furosemide 40 mg QD	Discontinued	--
Metformin 500 mg BID	Discontinued	--
Insulin aspart 135 U/day (given in 3 divided doses with meals at breakfast, lunch, dinner)	Discontinued	--
Insulin glargine 110 U/day	Reduced to 30 U/day	Discontinued

Table 3. Changes in HbA1c Levels

	Baseline	1 Mo	4 Mo	8 Mo
HbA1c, %	9.90	6.90	5.50	6.60
Insulin aspart dose, units	135	105	20	Discontinued
Insulin glargine dose, units	110	100	60	30
Metformin dose	500 mg BID	500 mg BID	Discontinued	Discontinued
Mifepristone dose	300 mg QD x 2 weeks, then 600 mg QD	900 mg QD	1200 mg QD	1200 mg QD

Figure 2. Weight Change

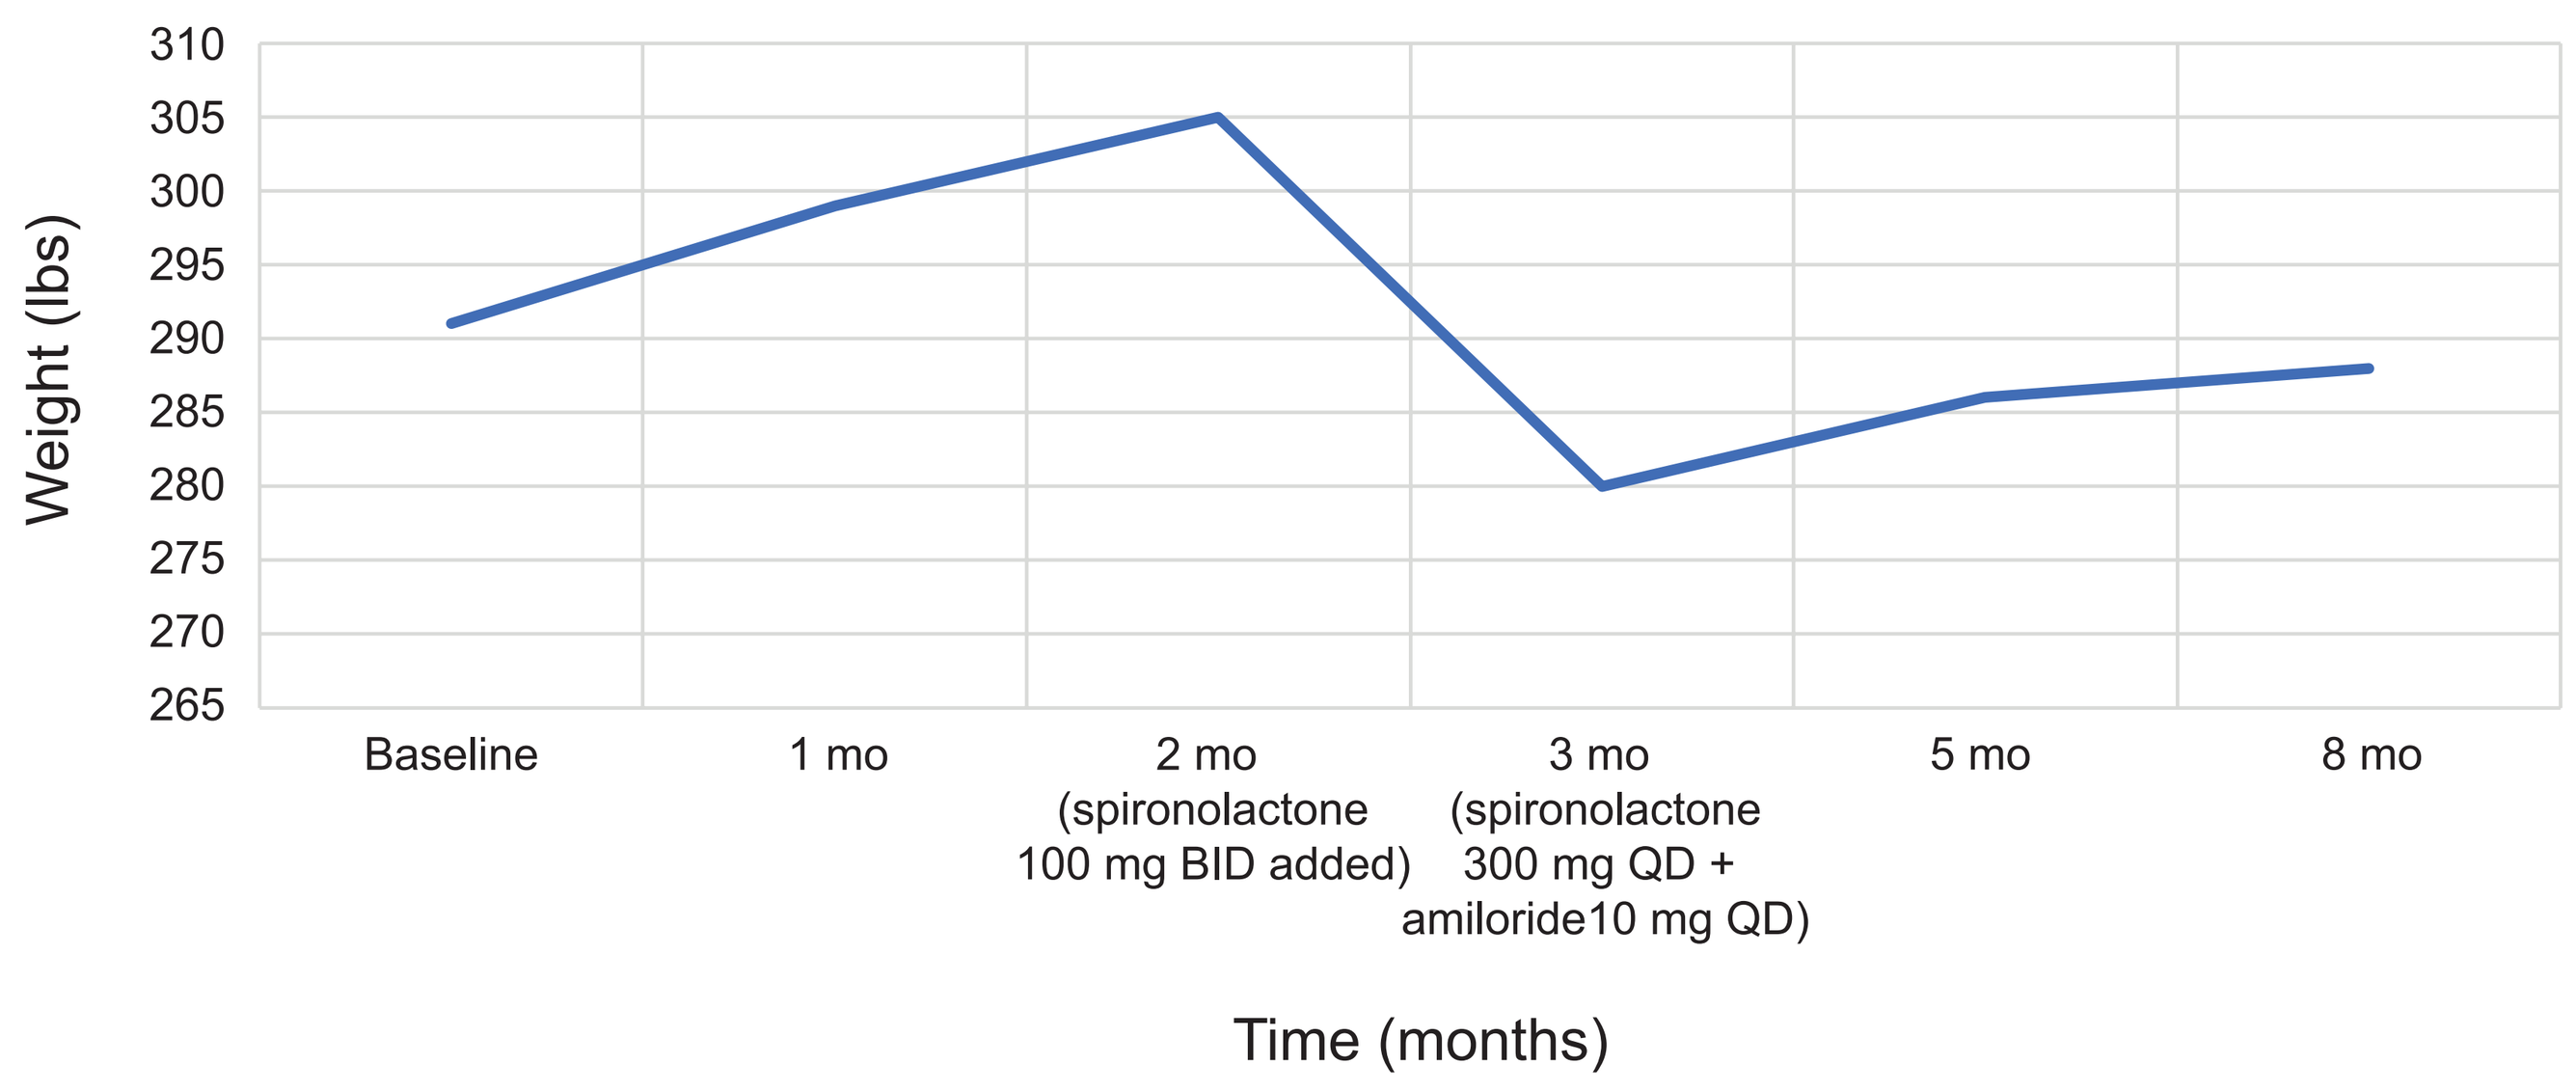
310
305
300
295
290
285
280
275
270
265
Weight (lbs)
Baseline
1 mo
2 mo (spironolactone 100 mg BID added)
3 mo (spironolactone 300 mg QD + amiloride10 mg QD)
5 mo
8 mo
Time (months)

SURGICAL INTERVENTION

- After 8 months of treatment, a lung carcinoid (2 cm x 1.8 cm) was ultimately discovered; surgery was selected and mifepristone discontinued.
- Insulin, spironolactone, and amiloride were discontinued following successful surgery.

SUMMARY

- Cortisol at high levels acts like a mineralocorticoid (aldosterone), stimulating absorption of sodium with osmotic reabsorption of water, and excretion of potassium at the renal collecting tubules with resulting hypernatremia, volume expansion and edema, and hypokalemia (serum potassium concentration <3.5 mEq/L [3.5 mmol/L]).
- Hypokalemia should be corrected prior to initiating mifepristone and monitored frequently during treatment.
 - However, hypokalemia can occur at any time during mifepristone treatment. The addition of potassium supplementation and/or mineralocorticoid antagonist therapy (eg, spironolactone) may be necessary to treat hypokalemia.
- In the present case of a patient with ectopic Cushing syndrome, additional diuretic was necessary to manage his hypokalemia and edema.
- Amiloride and spironolactone are both potassium-sparing diuretics.
 - Spironolactone is an aldosterone antagonist and acts as a diuretic and antihypertensive. It works on the distal convoluted renal tubule.
 - Amiloride decreases renal potassium excretion and increases diuresis. It works on the sodium channels in the distal tubules and collecting ducts in the kidney, preventing absorption of sodium and increasing excretion of sodium causing natriuresis.
- Combination therapy with spironolactone and amiloride helped to control BP, reduced potassium supplementation, prevented hypokalemia, and reduced edema in this patient.
- Managing the patient's hypokalemia and edema allowed mifepristone to control the patient's weight and improved his metabolic parameters while localization of the tumor continued.

REFERENCES

1. Fleseriu M, et al. *J Clin Endocrinol Metab.* 2012;97(6):2039-2049.
2. Korlym® (mifepristone) 300 mg tablets [Prescribing Information]. Menlo Park, CA: Corcept Therapeutics, Inc; 2017.

DISCLOSURES

JC: Nothing to disclose

RLB: Employee, Corcept Therapeutics.

AH: Nothing to disclose

ACKNOWLEDGMENTS

Editorial support was provided by Mark R. Vogel of Corcept Therapeutics. Funding for design and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster and provided approval of the final version.

Presented at the Endocrine Society Annual Meeting (ENDO 2018), March 17-20, 2018, Chicago, IL