SAT-405

FKBP5 GENE EXPRESSION BIOMARKER IN ACTH-DEPENDENT CUSHING SYNDROME PATIENTS PRE- AND POSTSURGERY

Irina Bancos, MD[1], Betul Ayse Hatipoglu, MD[2], Kevin Choong Ji Yuen, MD, FRCP (UK), FACE[3], Andreas Moraitis, MD[4]

[1]Mayo Clinic, Rochester, MN, USA, [2]Cleveland Clinic, Beachwood, OH, USA, [3]Swedish Pituitary Center, Swedish Neuroscience Institute, Seattle, WA, USA, [4]Corcept Therapeutics, Menlo Park, CA, USA

ABSTRACT

Background: Current biochemical tests for the diagnosis of Cushing syndrome (CS) using plasma, salivary or urine cortisol levels may not be reliable diagnostic biomarkers, because even when elevated, they do not reflect cellular cortisol activity or correlate with disease severity. This lack of correlation between cortisol level and disease severity may be in part due to glucocorticoid receptor (GR) polymorphisms that either increase or decrease sensitivity to cortisol or other downstream GR abnormalities (eg, GR complex transportation or recycling). A biomarker measuring the activity of cortisol may be beneficial both in diagnosing hypercortisolism and monitoring response to medical or surgical therapy. FK506-binding protein 5 (FKBP5), an essential part of the GR complex, is being investigated as a biomarker of cortisol activity. In the presence of glucocorticoids, FKBP5 mRNA expression is increased and acts within a negative intracellular feedback loop to lower the GR's affinity for cortisol and delays translocation of the GR-cortisol complex into the nucleus. Previous studies in healthy subjects have demonstrated a ~12-fold increase in FKBP5 mRNA expression after a single dose of prednisone administration. Administration of a GR antagonist (mifepristone or relacorilant [formerly referred to as CORT125134]) in combination with prednisone inhibited the prednisone-mediated activation of GR as demonstrated by no change in FKBP5 mRNA levels.

Methods: A multicenter, prospective study investigated the potential of FKBP5 as a biomarker for diagnosis of ACTH-dependent CS, evaluation of surgical success, and detection of relapse after pituitary surgery. The primary study hypothesis is that FKBP5 expression is elevated in participants with CS compared to healthy subjects and this expression decreases after successful surgical treatment. The study enrolled 20 participants with ACTH-dependent CS scheduled for surgery. Biomarker mRNA specimens were collected in conjunction with serum cortisol collections at the following timepoints: presurgical, immediately postsurgical and prior to glucocorticoid replacement, and outpatient follow-up for up to 3 years or until relapse.

Results: Twenty-one evaluable samples are currently available from participants before and after surgery; 12 participants had successful surgery and 9 participants had unsuccessful surgery. Preliminary analysis shows statistically significant differences in FKBP5 expression for presurgical participants with ACTH-dependent CS compared to data from healthy subjects (*P*<.0001) and postsurgical participants with successful surgery.

Conclusion: FKBP5 mRNA expression may potentially be a viable clinical biomarker for diagnosis and monitoring of hypercortisolemic states.

INTRODUCTION

- The diagnosis of Cushing syndrome (CS) remains a major challenge for clinicians.
- Current biochemical tests for the diagnosis of CS using plasma, salivary, or urine cortisol levels may not be reliable diagnostic biomarkers because even when elevated, they do not reflect cortisol activity at the glucocorticoid receptor (GR) level or correlate with disease severity.
- A biomarker measuring cortisol activity may be beneficial both in diagnosing hypercortisolism and monitoring response to medical or surgical therapy.

Figure 1. FKBP5 mRNA Expression[1-3]

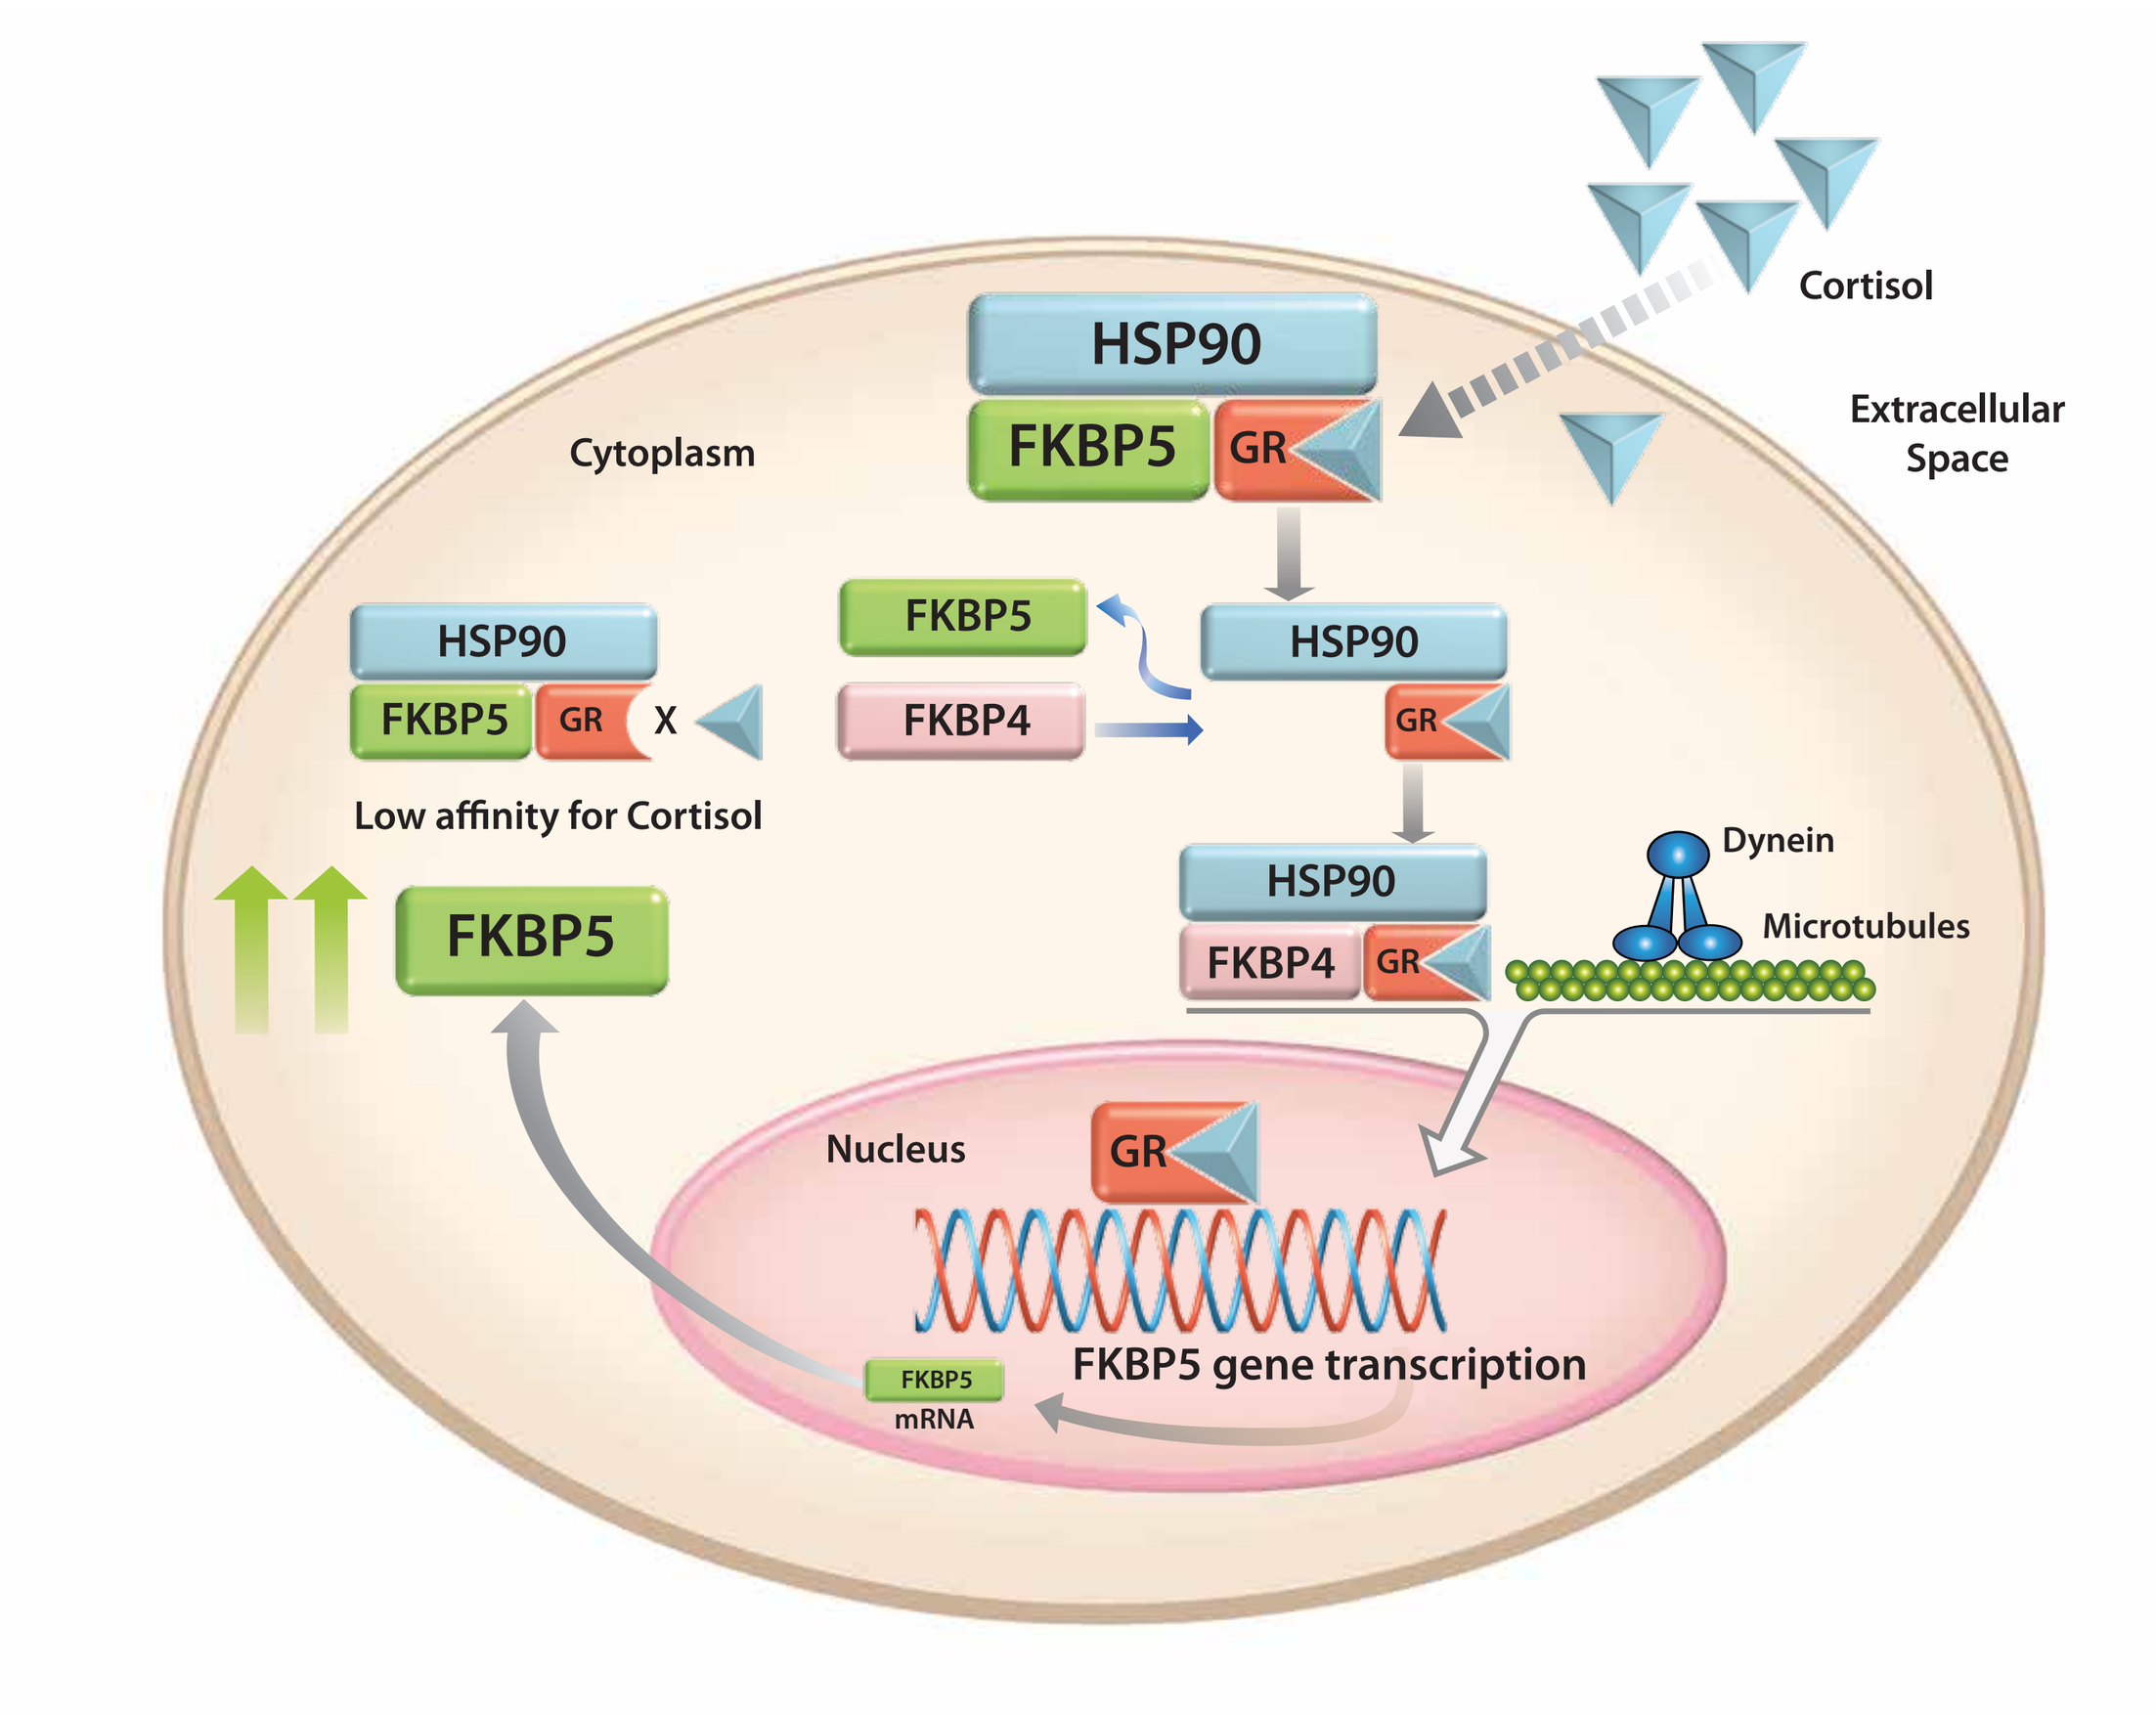
Cortisol
Extracellular Space
Cytoplasm
HSP90
FKBP5
GR
FKBP4
Low affinity for Cortisol
Dynein
Microtubules
Nucleus
FKBP5 mRNA
FKBP5 gene transcription

FKBP5, FK506 binding protein 51; FKBP4, FK506-binding protein 52; HSP90, heat shock protein 90; GR, glucocorticoid receptor.

FKBP5: MARKER OF CORTISOL ACTIVITY

- FK506-binding protein 51 (FKBP5) is a potential cortisol receptor activity biomarker that may be useful in diagnosing hypercortisolism as well as monitoring the response to medical or surgical therapy. It is a co-chaperone of heat shock protein 90 (HSP90) that regulates GR sensitivity. FKBP5 directly inhibits GR activity by interacting with GR to maintain it in an unbound inactive state within the cytoplasm, thus delaying transportation of the GR-cortisol complex into the nucleus (**Figure 1**).[1]
- FK506-binding protein 52 (FKBP4) can only bind to the complex when FKBP5 dissociates, allowing the ligand-activated GR complex to translocate into the nucleus and exert its action as a transcription factor.
- Action of the ligand-activated GR complex on the glucocorticoid response elements (GREs) can then increase the amount of FKBP5 mRNA and ultimately increase FKBP5 protein production.
- Negative feedback loop allows FKBP5 protein to reduce GR activity by lowering the affinity of cortisol for the cytoplasmic GR complex.
- In the presence of glucocorticoids, FKBP5 expression at both the mRNA and protein level is increased as part of an intracellular negative feedback loop for regulating GR activity.[1]
- A study of healthy subjects (n=10) demonstrated a time-dependent increase in FKBP5 mRNA expression after oral administration of prednisone.
 - FKBP5 mRNA increased to a mean peak levels of ~12-fold compared with unstimulated levels within 4 hours of prednisone administration and returned to baseline levels within 24 hours.
- By contrast, oral co-administration of prednisone with mifepristone (Korlym®, Corcept Therapeutics) or another selective GR antagonist (relacorilant, currently in development by Corcept) inhibited the prednisone-mediated activation of the GR and resulted in a reduction of the FKBP5 mRNA expression levels.

STUDY OBJECTIVES

- The effects of elevated cortisol are largely mediated by the GR.
- Current approaches using plasma, salivary, or urine cortisol levels to assess the severity of CS, or determine the response to medical or surgical therapy, are suboptimal because they measure circulating levels of cortisol, not cortisol activity, necessitating the development of a marker that integrates the degree of hypercortisolism as well as the glucocorticoid cellular responsiveness.
- FKBP5 acts as a functional negative regulator of GR sensitivity by reducing binding affinity and restricting nuclear translocation.[4]
- FKBP5 mRNA expression has been shown to be regulated by glucocorticoids via GREs, creating an ultrashort negative feedback loop (**Figure 1**).
- The current study aims to characterize the mRNA expression level of FKBP5 in participants with CS as an initial step in the evaluation of a biomarker for aiding the diagnosis and monitoring of this disease.
- The impact of curative surgery and glucocorticoid replacement therapy on mRNA expression levels of FKBP5 will be evaluated as well as the difference in expression levels between participants with CS and healthy controls.

HYPOTHESIS

- FKBP5 mRNA expression is increased in participants with CS, will decrease to normal after successful surgical treatment, and remain normal during disease remission.

Figure 2. Sample Collection Timeline

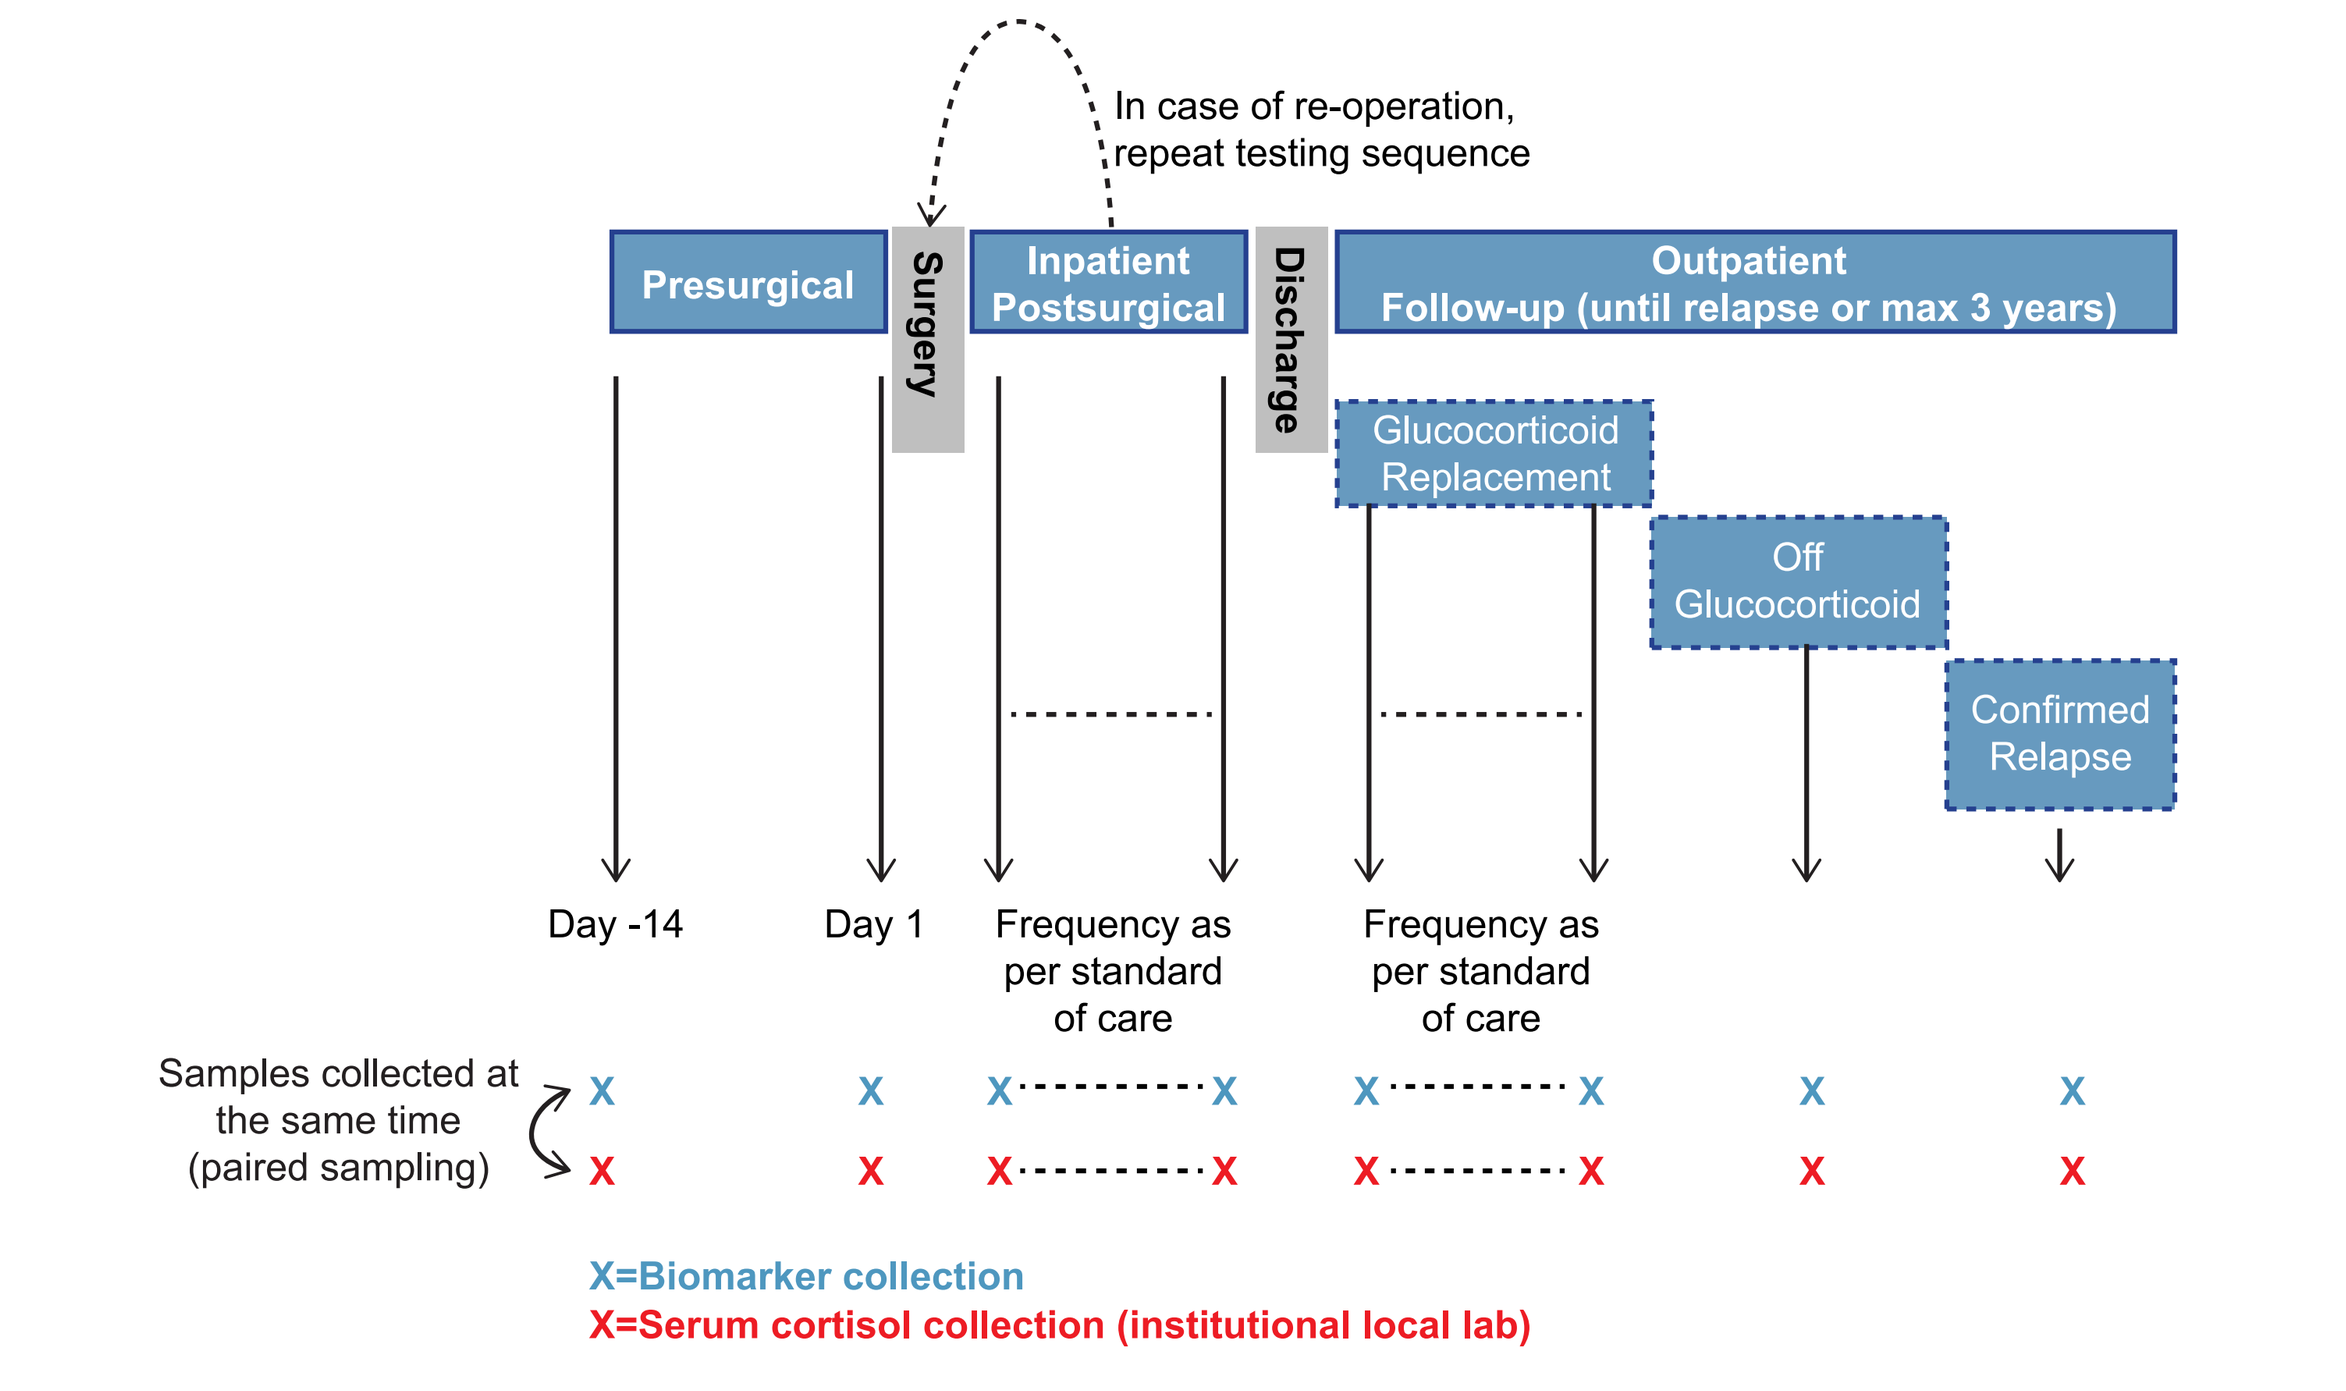
In case of re-operation, repeat testing sequence
Presurgical
Surgery
Inpatient Postsurgical
Discharge
Outpatient Follow-up (until relapse or max 3 years)
Glucocorticoid Replacement
Off Glucocorticoid
Confirmed Relapse
Day -14
Day 1
Frequency as per standard of care
Frequency as per standard of care
Samples collected at the same time (paired sampling)
X=Biomarker collection
X=Serum cortisol collection (institutional local lab)

METHODS

STUDY DESIGN

- A multicenter, prospective, biomarker collection study in participants with ACTH-dependent CS scheduled for surgery.
- mRNA specimens were collected in conjunction with serum cortisol from enrolled participants at the following timepoints (**Figure 2**):
 - Presurgical
 - Immediately postsurgical (and every time serum cortisol was measured per institution protocol) and prior to glucocorticoid replacement in participants who developed postsurgical hypocortisolemia
 - Outpatient follow-up for up to 3 years or until relapse

ELIGIBLE PARTICIPANTS

Inclusion

- Adult participants (≥18 years old) with documented diagnosis of ACTH-dependent, endogenous CS (ectopic or CS) who were scheduled for curative surgery.

Exclusion

- Planned use of preoperative and/or intraoperative glucocorticoid.
- Use of medical therapies for the treatment of CS (adrenostatic, somatostatin analogs, neuromodulators, mifepristone).
- Use of medications that could potentially stimulate the expression of FKBP5 (eg, hormonal therapies).
- Participants with ACTH-independent adrenal CS were not eligible due to the requirement for perioperative glucocorticoids.

SAMPLE COLLECTION (FIGURE 2)

- Paired serum cortisol and FKBP5 biomarker whole blood samples were collected from participants at defined time points:
 - Presurgical Visit (Day -14 to Day 1)
 - Surgical (Day 1)
 - Postsurgical Inpatient (Day 1 to Day 5) and prior to initiation of glucocorticoid replacement therapy for some participants
 - Outpatient Follow-up (postsurgical visits up to 3 years postsurgery)
- Specimens were collected in the morning when possible.
- Frequency of serum cortisol specimen collection was based on local standard of care (eg, every 6 hours until nadir levels achieved).
- PAXgene™ blood RNA tubes were used to collect FKBP5 biomarker specimen and were sent to NeoGenomics (Houston, TX) for analysis using NanoString nCounter® technology (NanoString Technologies).
- Historical samples from 26 healthy subjects were used as controls.

Figure 3. FKBP5 mRNA Expression Pre- and Postsurgery Compared to Healthy Controls

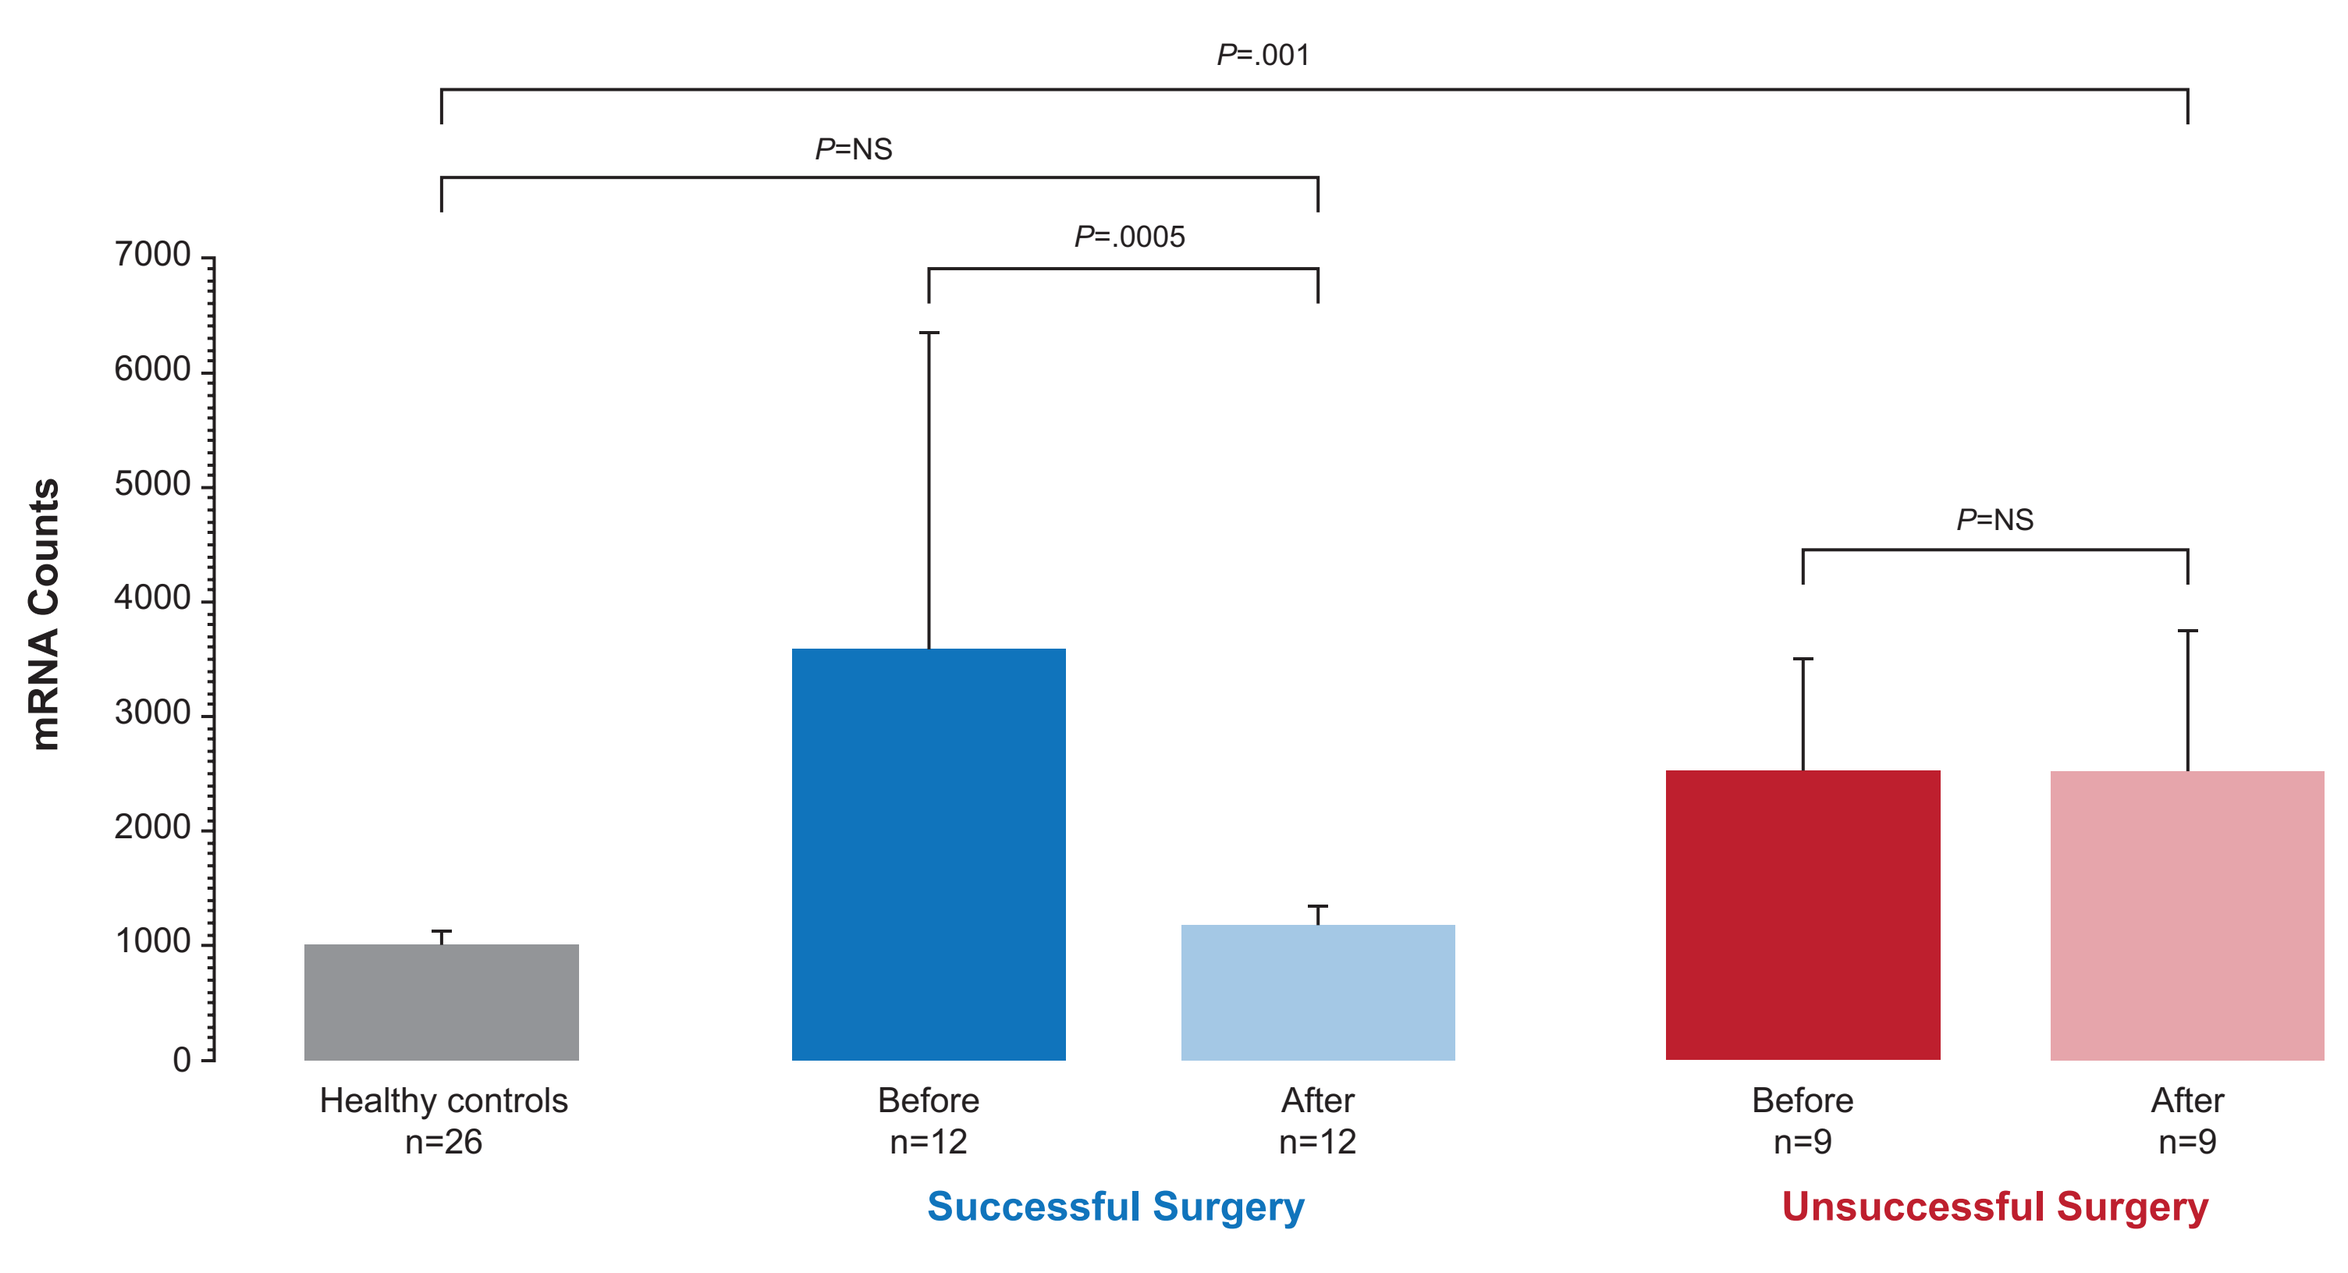
P=.001
P=NS
P=.0005
P=NS
mRNA Counts
7000
6000
5000
4000
3000
2000
1000
0
Healthy controls n=26
Before n=12
After n=12
Successful Surgery
Before n=9
After n=9
Unsuccessful Surgery

NANOSTRING AND RNA ISOLATION METHODOLOGIES

- RNA was isolated from the frozen whole blood samples collected using the PAXgene Blood RNA Kit.
- RNA quality and quantity were assessed using NanoDrop and Agilent Bioanalyzer.
- All samples were tested at an RNA input of 200 ng using the Corcept FKBP5 NanoString panel.
- Gene expression was measured using the nCounter and was run and read on an nCounter Analysis System.
- The assay performance was verified and demonstrated 100% concordance and high correlation between NanoString and real-time quantitative reverse transcription polymerase chain reaction (qRT-PCR).
- Housekeeping genes were selected from publicly available databases based on stability and detectable expression levels across the biomarker of interest.

RESULTS

Table 1. Participant Demographics and Biochemical Characteristics (N=21)

Age, mean (min-max)	41.6 (19-63) years
Sex	17 female; 4 male
Etiology	20 pituitary; 1 ectopic
ACTH (n=15), mean (range)	75 (16-203) pg/mL

- Twenty-six participants were enrolled from 4 research sites from across the United States. Twenty-one had evaluable samples (**Table 1**).
- Analysis shows statistically significant differences in FKBP5 expression for presurgical participants with ACTH-dependent CS compared to data from healthy subjects (*P*≤.0001).
- Participants with successful surgical outcome have comparable FKBP5 mRNA expression to healthy subjects (**Figure 3**).
- Participants with failed surgery have FKBP5 mRNA expression that remained elevated and unchanged from baseline and distinct from heathy subjects (*P*≤.001) (**Figure 3**).

CONCLUSIONS

- FKBP5 mRNA expression is statistically distinct in participants with CS compared to levels in normal healthy subjects.
- FKBP5 mRNA expression in participants with successful surgery is statistically different from those that failed surgery and similar to levels expressed in healthy subjects.
- FKBP5 mRNA expression shows promise as a clinical biomarker for diagnosis and monitoring of patients with hypercortisolism.

REFERENCES

1. Bali U, et al. *J Clin Endocrinol Metab.* 2016;101:4305-4312.
2. Binder EB. *Psychoneuroendocrinology.* 2009;34 Suppl 1:S186-S195.
3. Ellsworth KA, Wang L. http://atlasgeneticsoncology.org/Genes/FKBP5ID40578ch6p21.html. Accessed January 19, 2018.
4. Galigniana NM, et al. *J Neurochem.* 2012;122:4-18.

DISCLOSURES

IB: Research investigator, Corcept Therapeutics.
BAH: Research investigator, Corcept Therapeutics.
KCJY: Advisory board member, research investigator, and speaker, Corcept Therapeutics.
AM: Employee, Corcept Therapeutics.

ACKNOWLEDGMENTS

Editorial support was provided by Dat Nguyen and Mark R. Vogel of Corcept Therapeutics. Funding for design and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster and provided approval of the final version. ClinicalTrials.gov Identifier: NCT02922257.

Presented at the Endocrine Society Annual Meeting (ENDO 2018), March 17-20, 2018, Chicago, IL