SUN-383

Sustained Clinical Improvement After Discontinuation of Mifepristone: Unexpected Remission of Hypercortisolism

Robin Anderson, FNP-C[1]; Neven Pesa, PNP[2]; Amer Kassar, MD[1]
[1]GN Endocrinology, Munster, IN, USA; [2]Corcept Therapeutics, Menlo Park, CA, USA

INTRODUCTION

- Endogenous hypercortisolism (Cushing syndrome) is associated with increased mortality and substantial morbidity[1-5]
- The clinical signs and symptoms of hypercortisolism are often nonspecific and overlap with commonly encountered conditions, including impaired glucose tolerance/diabetes, obesity, hypertension, psychiatric disorders, and osteoporosis[1]
- Knowing when to suspect hypercortisolism is especially challenging in patients without overt cushingoid features (eg, proximal muscle weakness, bruising, plethora, purple striae), particularly when the source is unknown
- Mifepristone (Korlym®, Corcept Therapeutics, Menlo Park, CA) is a competitive glucocorticoid receptor antagonist approved to treat endogenous hypercortisolism of all etiologies
- In patients treated with mifepristone, symptoms of hypercortisolism (eg, hyperglycemia, weight gain, or psychiatric symptoms) can return within several weeks of discontinuation
- Here we describe a patient who experienced clinical improvement in symptoms attributed to excess cortisol after treatment with mifepristone and whose symptoms did not recur after discontinuation of mifepristone

CASE HISTORY AND PRESENTATION

- A 39-year-old female patient presented with persistent weight gain 20 months after diagnosis and successful treatment of thyroid cancer (total thyroidectomy and iodine ablation)
 - Her thyroid function was currently normal with administration of Synthroid 137 μg and Cytomel 10 μg daily
- The patient's chief complaint was "stress" resulting from an inability to lose weight or sleep through the night, fatigue, and recent onset of migraine with vomiting
- No significant weight loss was noted despite diet and exercise; phentermine 15 mg prescribed by her healthcare provider was increased to 37.5 mg
- She had a history of regular but heavy menses and displayed no overt cushingoid features at clinical presentation
- Despite diet, exercise, and phentermine, she lost only 8 lbs in 2 months

WORK-UP FOR SUSPECTED HYPERCORTISOLISM

- Given her difficulty losing weight, poor sleep, migraines, and history of "stress," hypercortisolism was suspected
- Lab results indicated abnormal fasting blood glucose (FBG), low serum potassium (K+) levels, and an abnormal AM cortisol and dexamethasone suppression test (DST), as shown in **Table 1**

Table 1. Baseline Biochemical and Clinical Examination

Parameter	Value
AM cortisol (μg/dL)	29.3 (high)
DST (μg/dL)	2.6 (high)
ACTH (pg/mL)	19
K+ (mEq/L)	3.4 (low), 4.3[a]
FBG (mg/dL)	119, 118[a]
Blood pressure (mm Hg)	130/84, 126/90[a]
Weight (lbs)	200, 192[a]
BMI (kg/m²)	33.2, 31.9[a]

[a]Repeat assessment 2 months later.
ACTH, adrenocorticotropic hormone; BMI, body mass index; DST, dexamethasone suppression test; FBG, fasting blood glucose.

- Imaging of pituitary and adrenals was ordered but the patient declined these tests

MEDICAL THERAPY

- Because of the abnormal biochemical results, a trial of mifepristone 300 mg and spironolactone 25 mg was initiated; at Month 2, the mifepristone dose was increased to 600 mg (**Figure 1**)

Figure 1. Timeline of Treatment and Patient Response

Synthroid 137 μg + Cytomel 10 μg (doses reduced to 112 μg + 7.5 μg at Month 6)
Phentermine 37.5 mg
Mifepristone 300 mg (dose increased to 600 mg at Month 2)
Spironolactone 25 mg (except Months 2-4)
K+ 40 mEq twice-daily supplementation

Patient History	Mifepristone Start	Month 2	Month 4	Month 6	Off Mifepristone/ Follow-up
• Persistent weight gain 20 months after successful treatment for thyroid cancer • Poor sleep • Migraines with vomiting • History of "stress"	• DST: 2.6 μg/dL • FBG: 119 mg/dL • Patient declines imaging tests for suspected hypercortisolism • Trial of mifepristone and spironolactone initiated	• Sleep improves • Edema develops • Blood pressure elevated • Amenorrhea develops • Patient not taking spironolactone	• Low K+ • Elevated blood pressure • Patient resumes spironolactone	• Migraines, edema, and fatigue resolved • Mifepristone, phentermine, spironolactone, K+ stopped	• Repeat DST: 0.4 μg/dL • Thyroid function tests normal • Weight and FBG remain normal • Sleep remains improved • Regular menses resume

DST, dexamethasone suppression test; FBG, fasting blood glucose.

RESULTS WITH MIFEPRISTONE

- A diagnosis of hypercortisolism was supported by the improvement in FBG and weight (**Figure 2**)

Figure 2. Patient Weight and FBG During the Observation Period

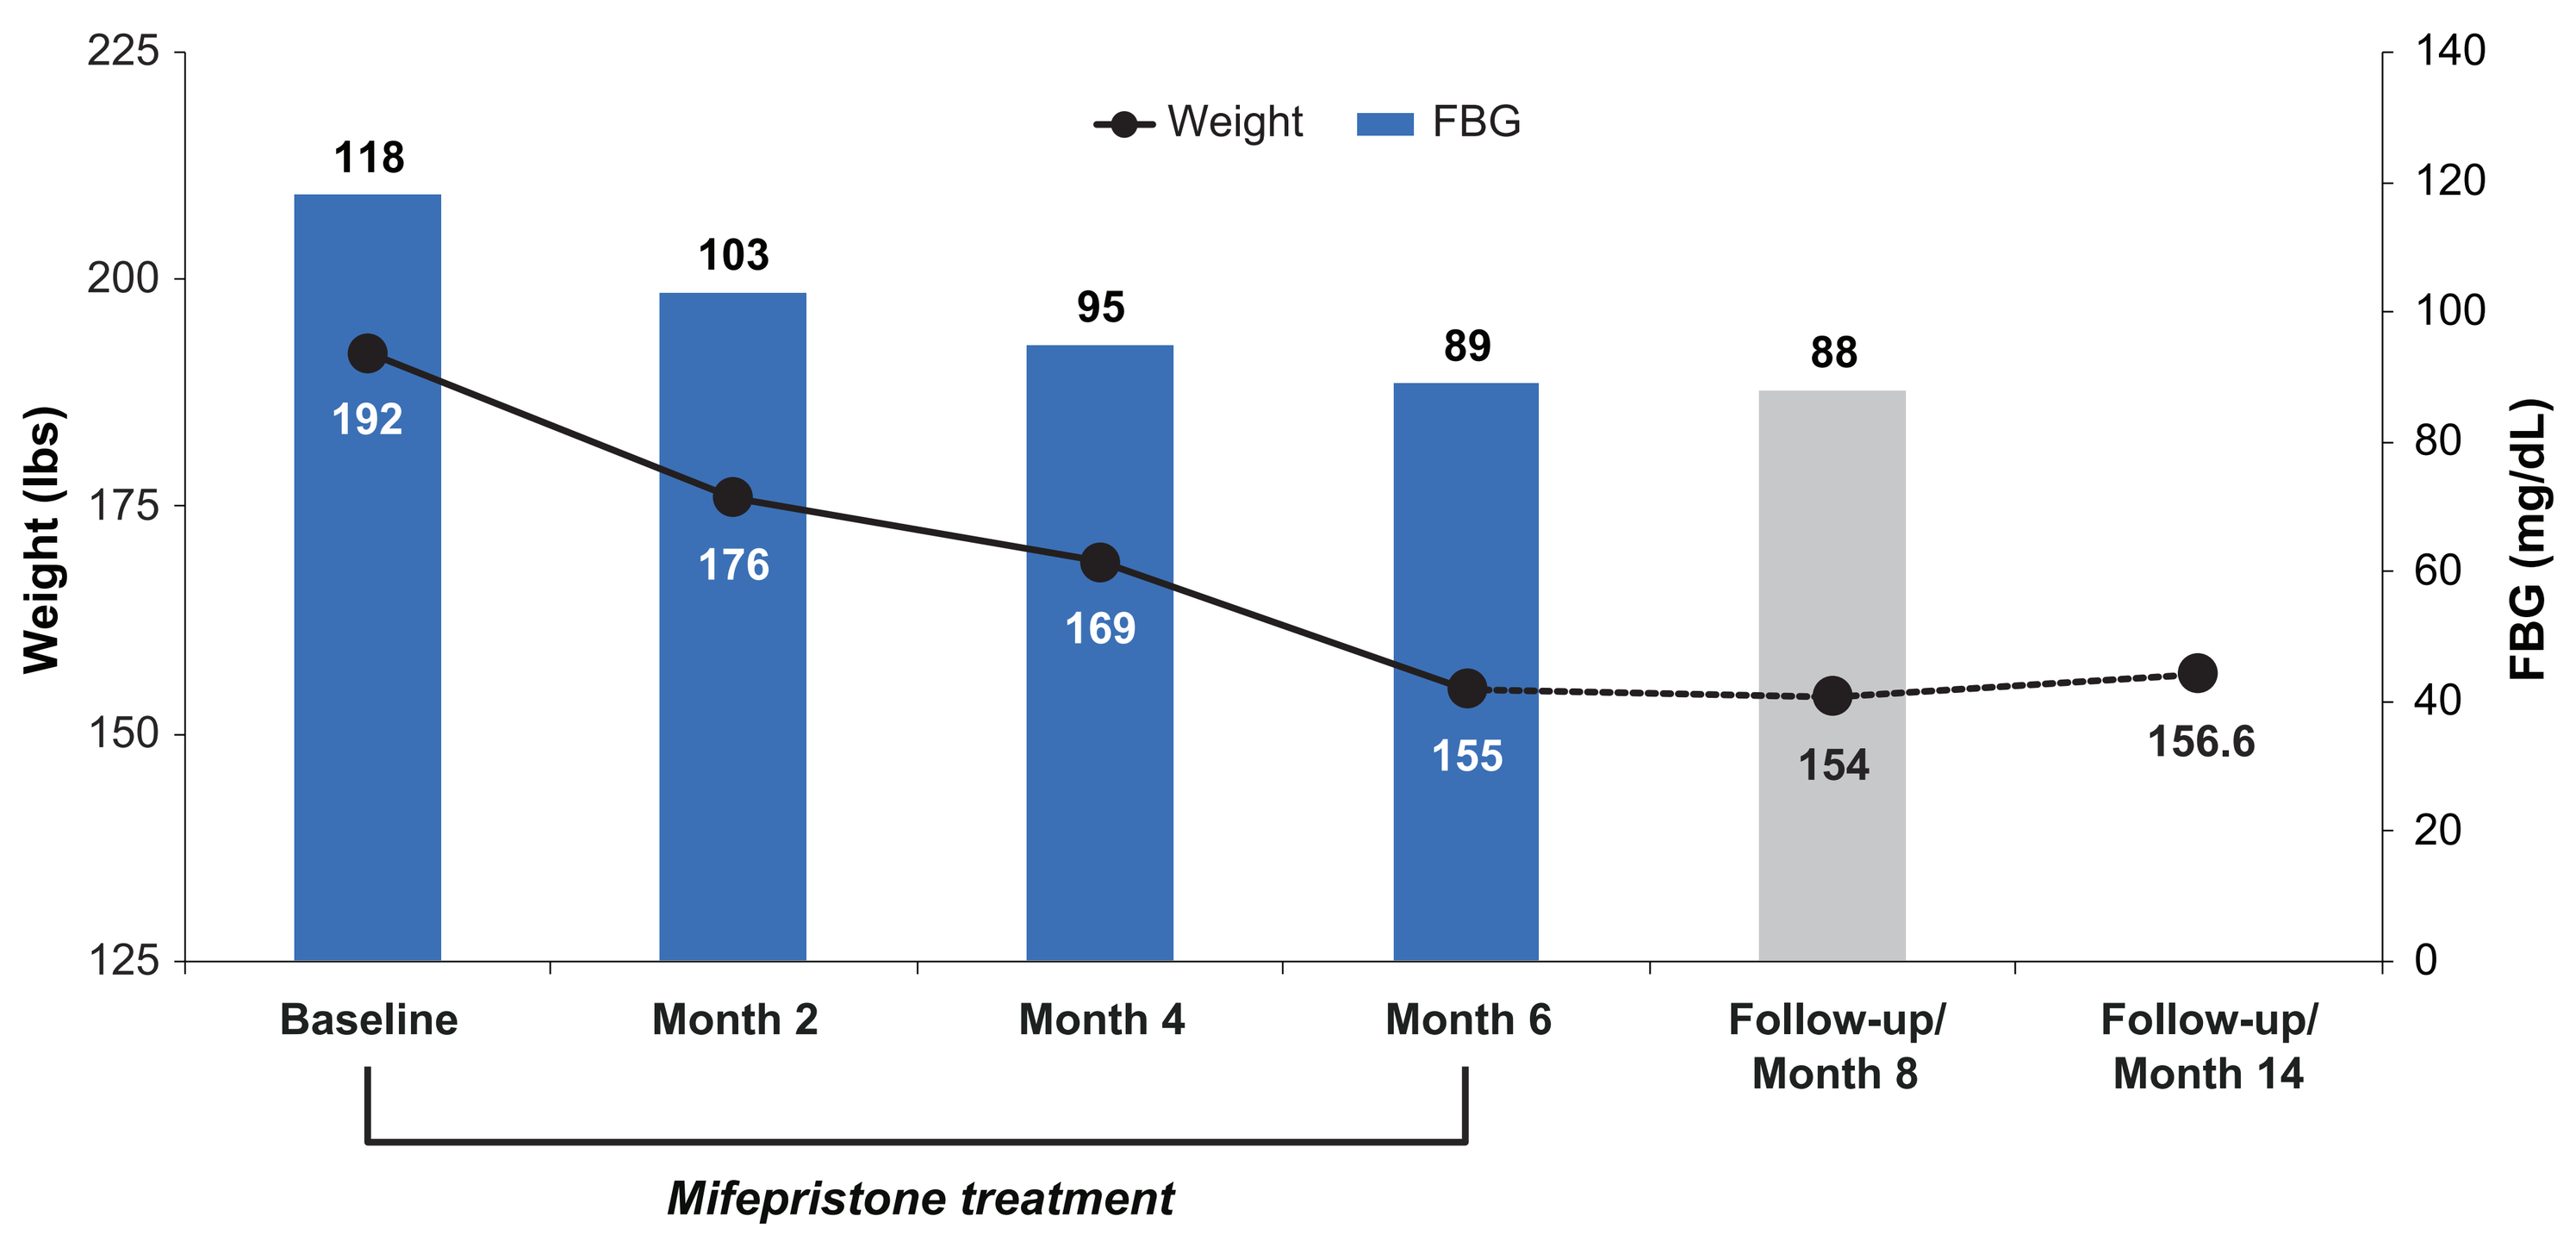
Weight
FBG
Weight (lbs)
FBG (mg/dL)
225
200
175
150
125
140
120
100
80
60
40
20
0
118
103
95
89
88
192
176
169
155
154
156.6
Baseline
Month 2
Month 4
Month 6
Follow-up/ Month 8
Follow-up/ Month 14
Mifepristone treatment

FBG, fasting blood glucose.

- Within 4 months, K+ levels decreased, BP increased, and edema developed, indicating that mifepristone may have increased cortisol levels and mineralocorticoid receptor activation (**Figure 3**)

Figure 3. Serum K+ (A) and Blood Pressure (B) During the Observation Period

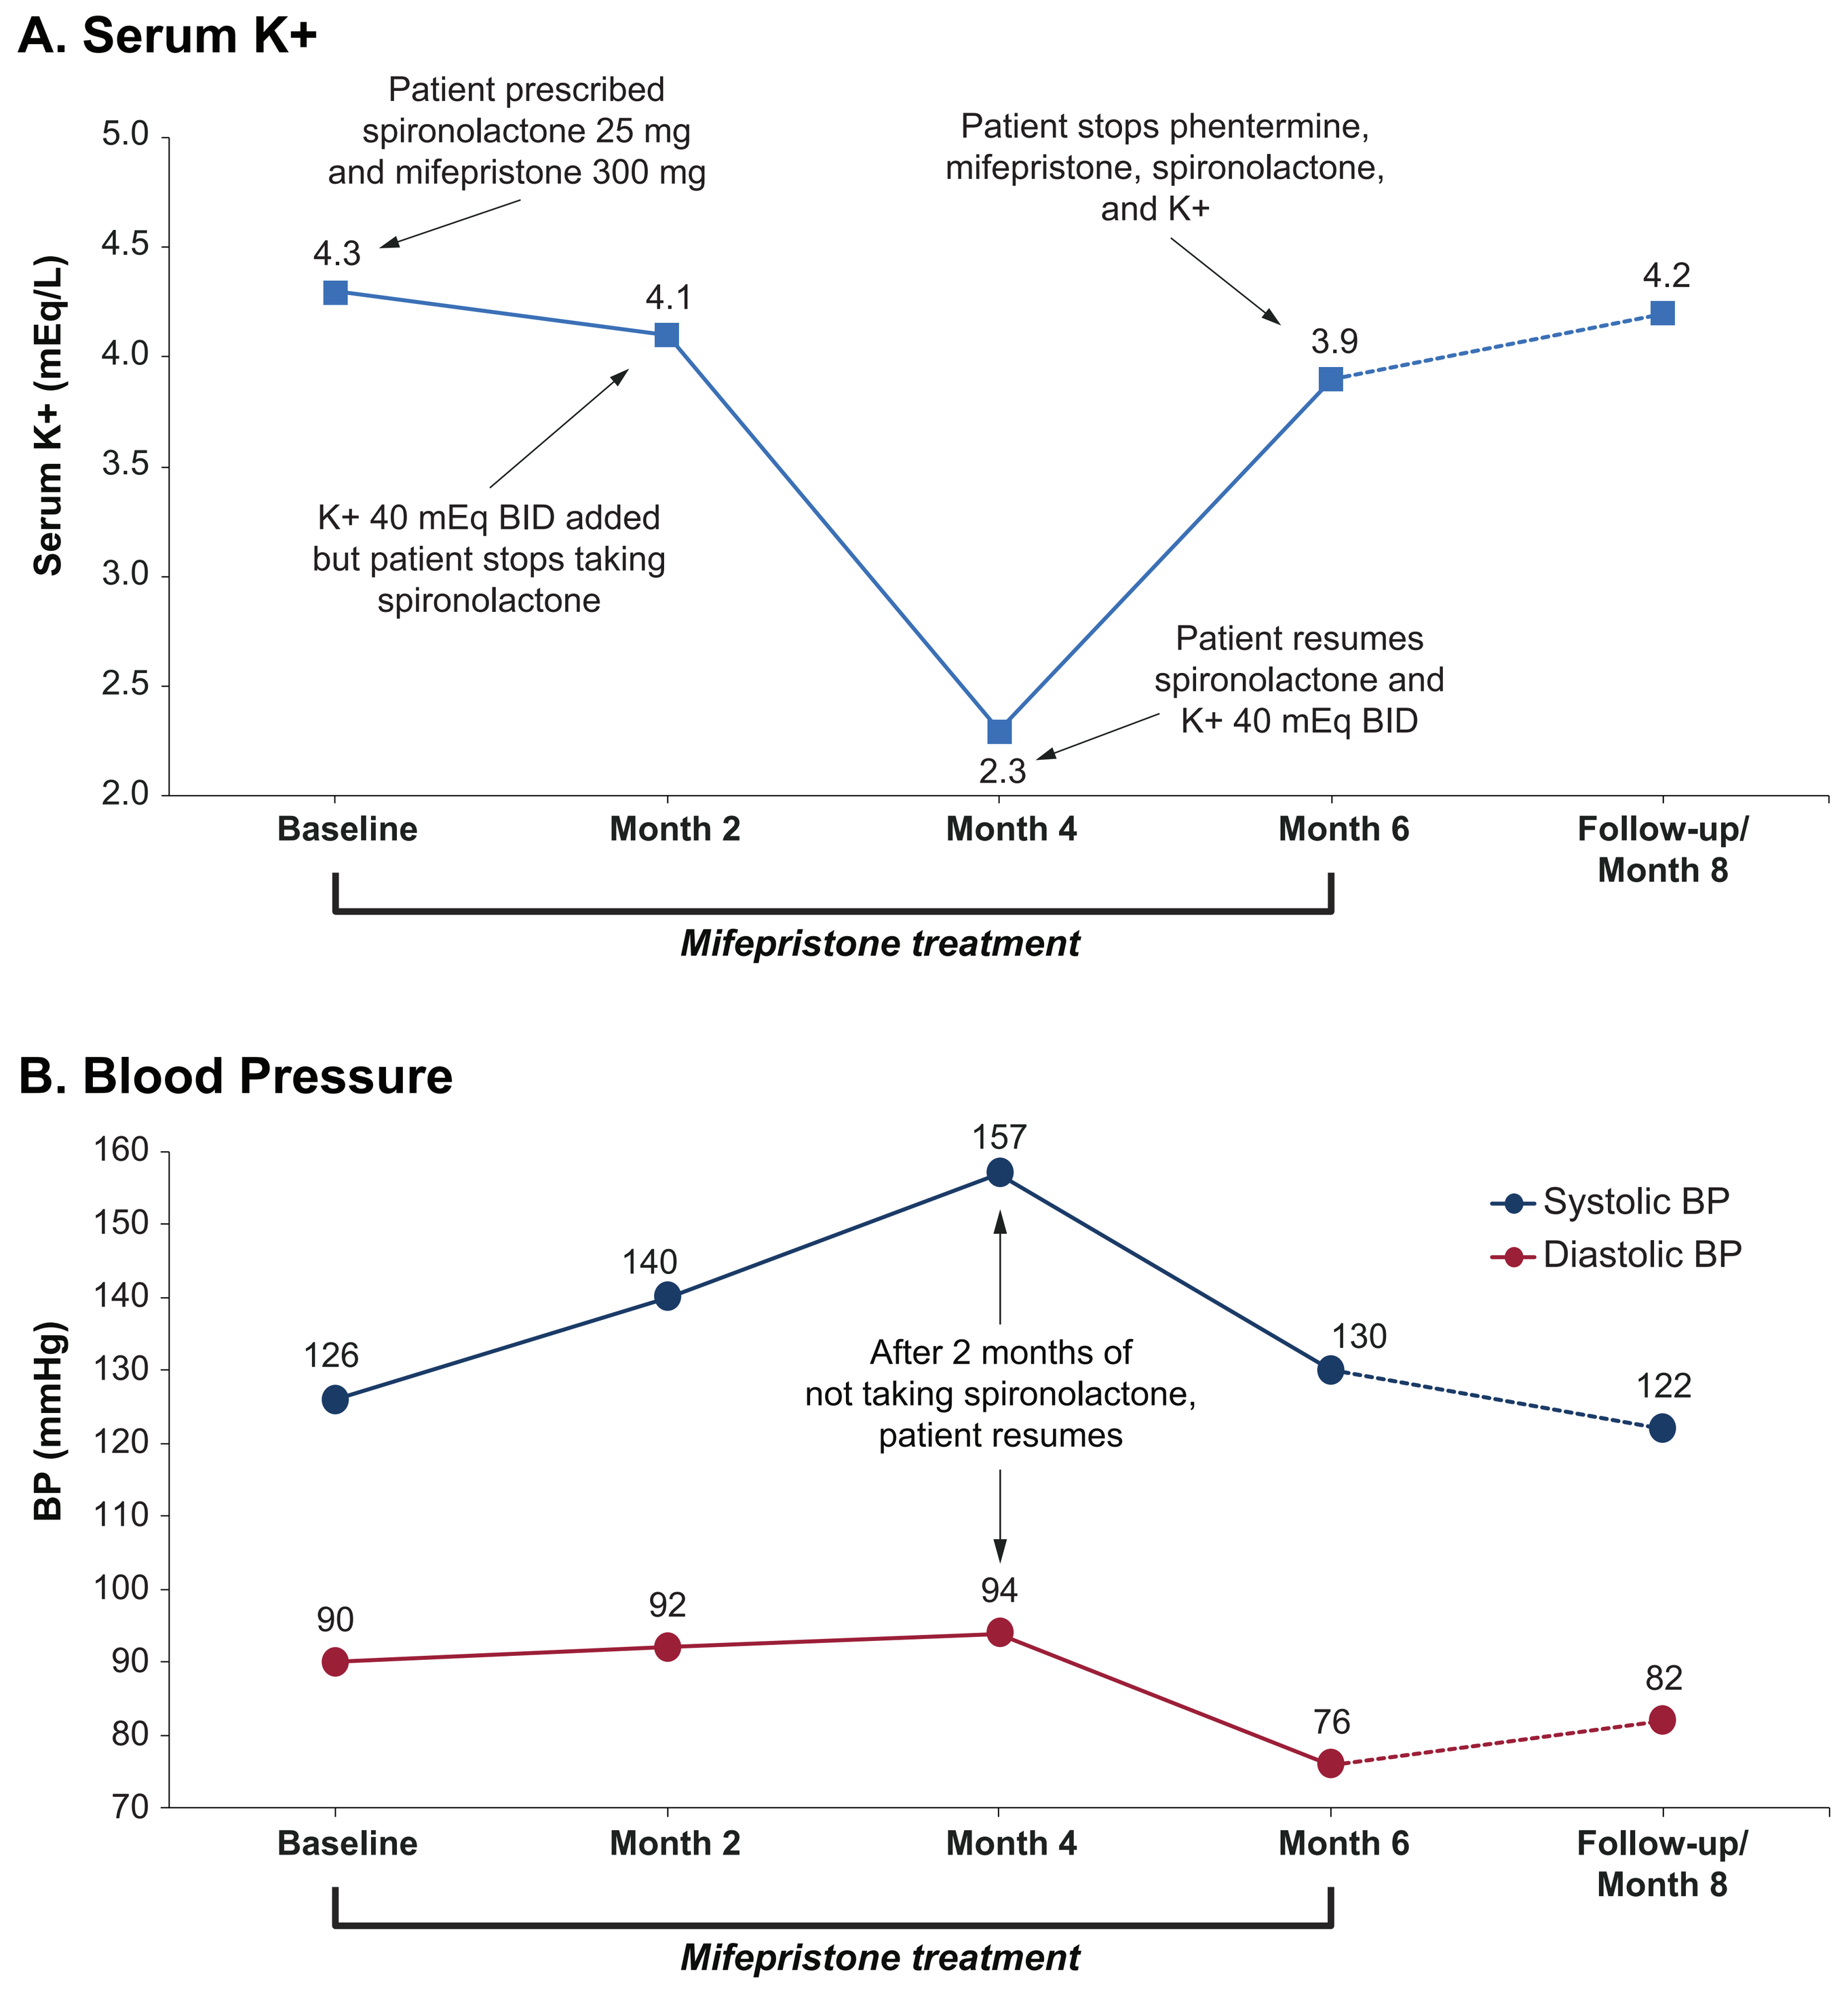
A. Serum K+
Serum K+ (mEq/L)
Patient prescribed spironolactone 25 mg and mifepristone 300 mg
Patient stops phentermine, mifepristone, spironolactone, and K+
K+ 40 mEq BID added but patient stops taking spironolactone
Patient resumes spironolactone and K+ 40 mEq BID
4.3
4.1
2.3
3.9
4.2
Baseline
Month 2
Month 4
Month 6
Follow-up/ Month 8
Mifepristone treatment
B. Blood Pressure
BP (mmHg)
Systolic BP
Diastolic BP
126
140
157
130
122
90
92
94
76
82
After 2 months of not taking spironolactone, patient resumes
Baseline
Month 2
Month 4
Month 6
Follow-up/ Month 8
Mifepristone treatment

BID, twice daily; BP, blood pressure.

- When the mineralocorticoid receptor antagonist spironolactone was added, both K+ and BP improved significantly, supporting the presence of excess cortisol levels
- At Month 6, her migraines, edema, and fatigue resolved; she discontinued mifepristone, spironolactone, and K+ supplementation; and her Synthroid and Cytomel doses were reduced
- At Month 8, thyroid function and FBG were normal, weight and sleep improvements continued, and regular heavy menses resumed
- By Month 8, she had lost 38 lbs from baseline (a 19.8% reduction) and her FBG decreased by 30 mg/dL (a 25% reduction)
- By Month 14, her weight continues to be maintained (156.6 lbs) and her blood pressure assessments are consistently between 120-130/70-80 mmHg
- Images of the patient before and after treatment with mifepristone are shown in **Figure 4**

Figure 4. The Patient Before and After Treatment With Mifepristone

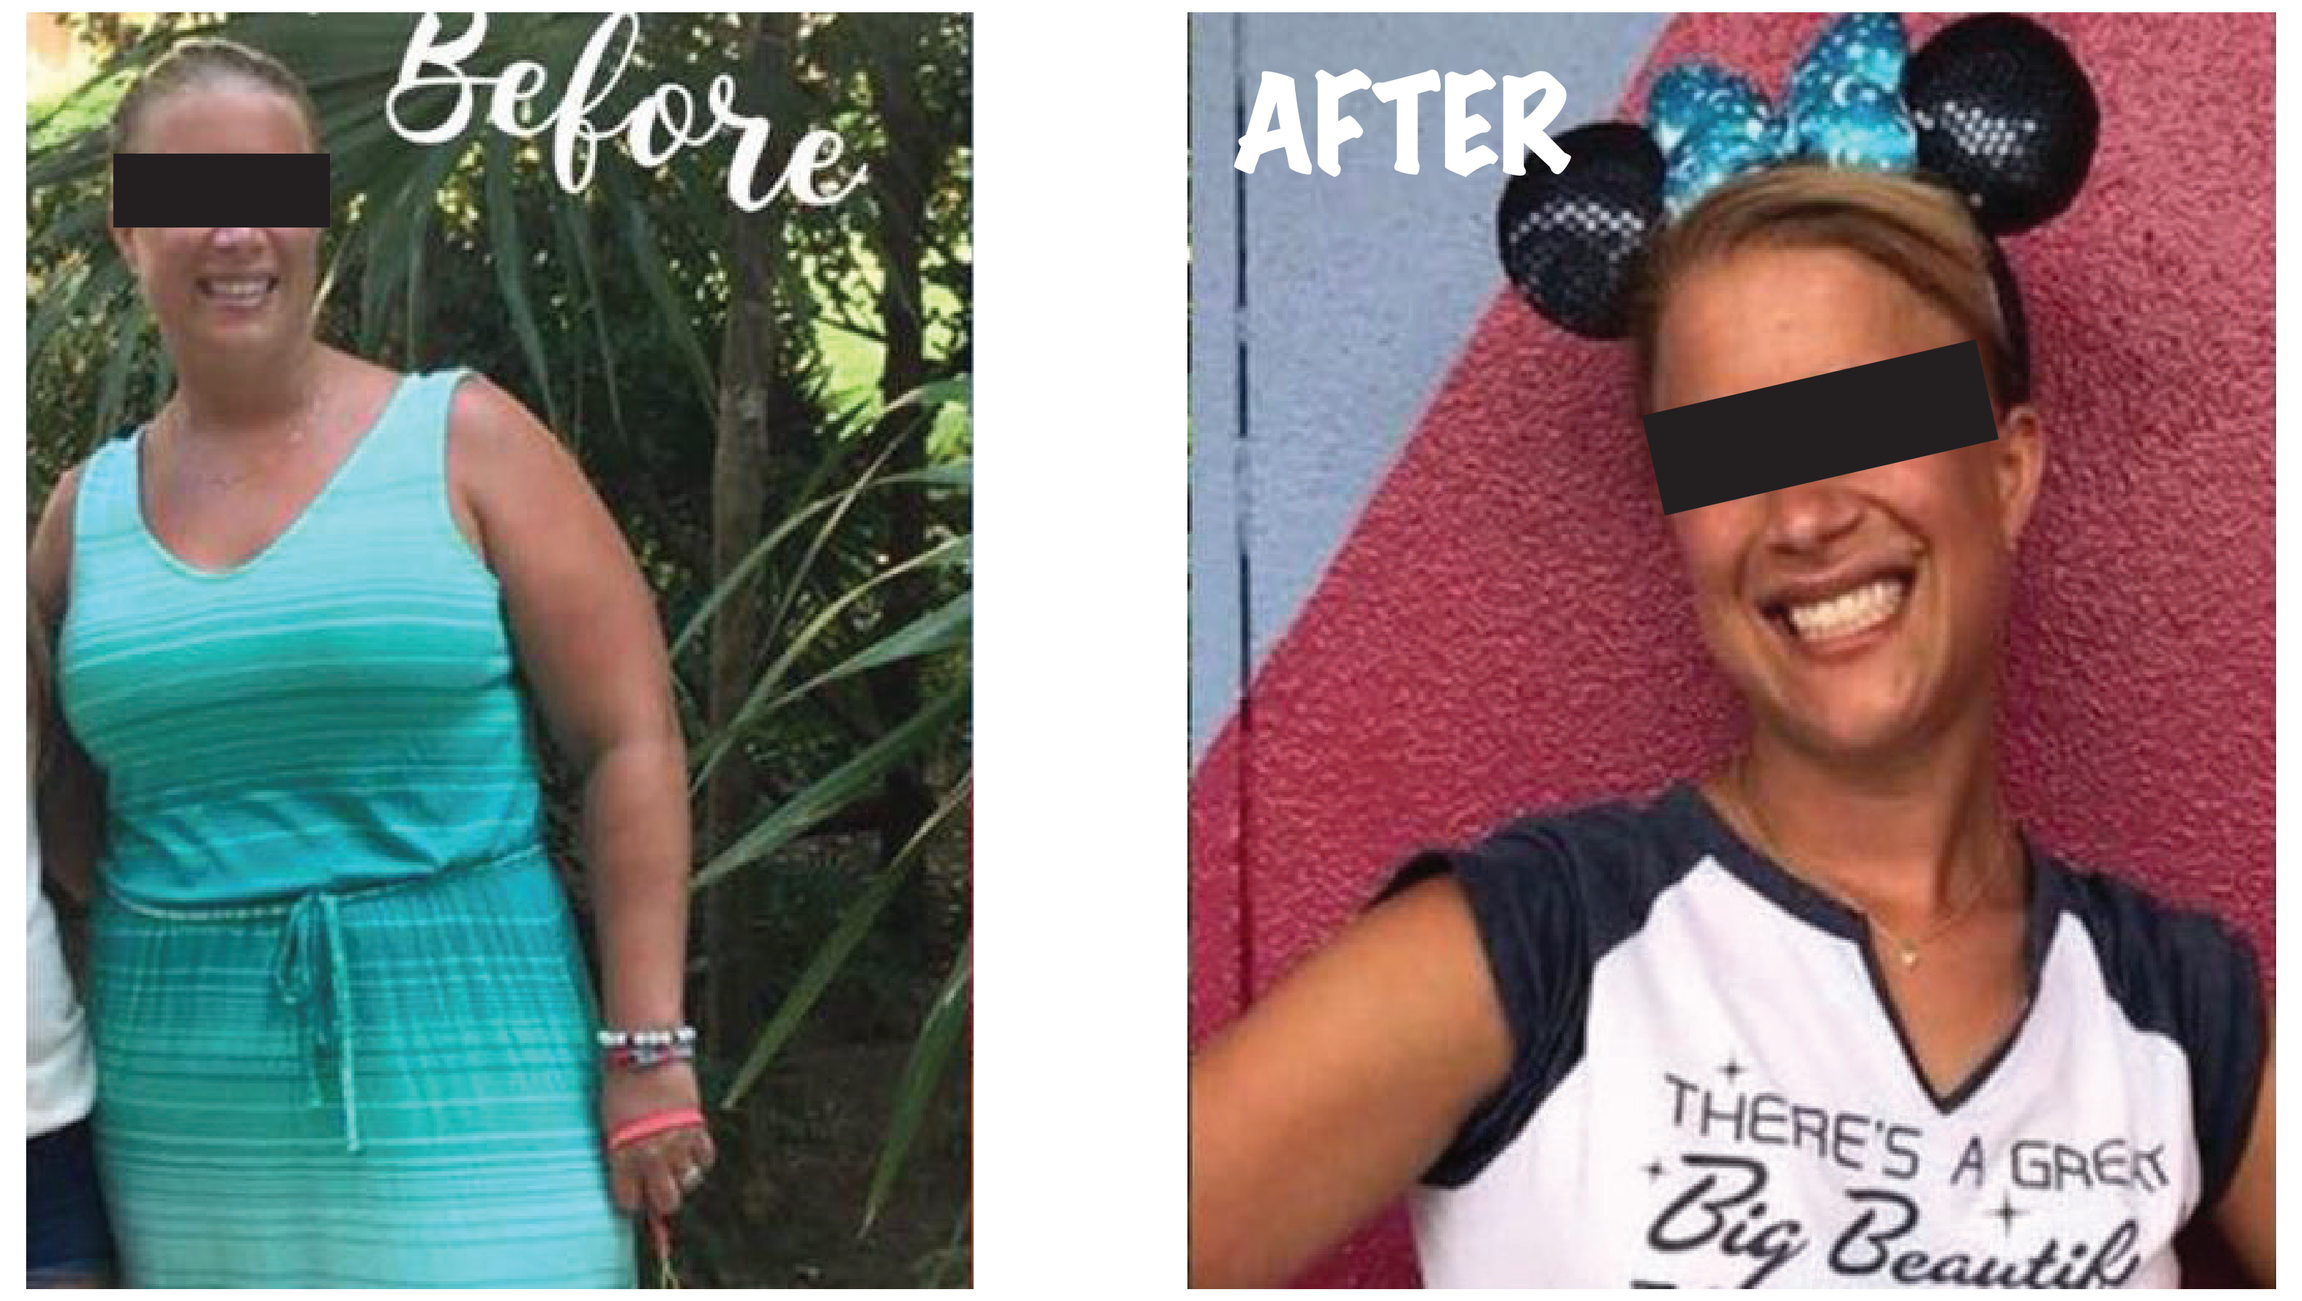
Before
AFTER

CONCLUSIONS

- This report details a patient with hypercortisolism who was treated with mifepristone for 6 months
- Hypercortisolism was suspected because of worsening clinical parameters (despite lifestyle and therapeutic interventions) and biochemical testing; however, the patient declined to undergo further diagnostic imaging work-up
- A trial of mifepristone was initiated because of its intracellular mechanism of action and the uncertainty of disease etiology
- The clinical and metabolic improvements noted while on therapy support the diagnosis of hypercortisolism
- We hypothesize that these findings, which are consistent with those observed in patients with more overt hypercortisolism,[6] may be indicative of a "reset" of the HPA axis
- Unexpectedly, the patient's improvements have been maintained up to 6 months after treatment discontinuation
- We postulate that the patient's history of stress and obesity might have contributed to autonomous non-neoplastic hypercortisolemia[7]
- These results suggest that a therapeutic trial of mifepristone can be useful in diagnosing and treating patients with suspected hypercortisolism when surgery is not warranted

REFERENCES

1. Nieman LK. *Endocrinol Metab (Seoul)*. 2018;33(2):139-46.
2. Dekkers OM, et al. *J Clin Endocrinol Metab*. 2013;98(6): 2277-84.
3. Valassi E, et al. *Eur J Endocrinol*. 2011;165(3):383-92.
4. Clayton RN, et al. *J Clin Endocrinol Metab*. 2011;96(3):632-42.
5. Reimondo G, et al. *Minerva Endocrinol*. 2019;44(1):33-42.
6. Fleseriu M, et al. *J Clin Endocrinol Metab*. 2012;97(6):2039-49.
7. Joseph JJ, Golden SH. *Ann N Y Acad Sci*. 2017;1391(1):20-34.

ACKNOWLEDGMENTS

Funding for editorial, design, and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster content and provided approval of the final version.

DISCLOSURES

RA: Consultant and speaker, Corcept Therapeutics
NP: Employee, Corcept Therapeutics.
AK: Nothing to disclose.

Presented at the Endocrine Society 2019 Annual Meeting, March 23-26, 2019, New Orleans, LA