MON-450

Predicting and Managing Hypertension and Hypokalemia in Cushing's Syndrome During Mifepristone Therapy

Richard J. Auchus,[1,2,*] Michael J. McPhaul,[2,*] James W. Findling,[3,*] Coleman Gross[4,*]

[1]Division of Metabolism, Diabetes, & Endocrinology, University of Michigan, Ann Arbor, MI; [2]Division of Endocrinology & Metabolism, University of Texas Southwestern Medical Center, Dallas, TX; [3]Endocrinology Center and Clinics, Medical College of Wisconsin, Milwaukee, WI; [4]Corcept Therapeutics, Menlo Park, CA.

Potential conflicts of interest exist, see author disclosures.

Abstract

Introduction:
Hypertension and hypokalemia are frequent manifestations of Cushing's syndrome (CS), because cortisol activates both the glucocorticoid receptor (GR) and the mineralocorticoid receptor (MR). We analyzed blood pressure (BP) and potassium data from the SEISMIC trial of 50 patients with CS who were treated with the GR antagonist mifepristone (300-1200 mg/d) to determine prevalence, predictive factors, and approaches to management.

Methods:
At baseline, 5 and 18 patients, respectively, required spironolactone or potassium supplements to maintain normokalemia, as specified by the protocol. During the trial, spironolactone was administered to 14 patients (25-400 mg/d), and 33 patients received potassium supplements (10-420 mEq/d) for hypokalemia.

Results:
One or more episodes of hypokalemia were noted in 20 participants, 16 (80%) of whom were normokalemic at final study visit. Those who required treatment for potassium wasting had higher urinary-free cortisol (UFC) values at baseline as well as at peak (Week 10) than those who were not treated:

Baseline:
Mean±standard deviation (SD) = 1371±3523 vs 317±228 nmol/24 h [497±1277 vs 115±83 μg/24 h]
Median [range, interquartile range (IQR)] = 289 [59-19739; 155-1017] vs 266 [85-1032; 229-324] nmol/24 h

Week 10:
Mean±SD = 3668±6094 vs 1103±1003 nmol/24 h [1330±2209 vs 400±364 μg/24 h]
Median [range, IQR] = 938 [41-23480; 286-4417] vs 749 [32-2630; 312-2131] nmol/24 h
(*P*=not significant (NS) for all)

Patients who required potassium treatment prior to starting mifepristone had higher UFC levels than those treated after starting mifepristone (mean±SD = 2194±4654 vs 383±387 nmol/24 h [795±1687 vs 139±140 μg/24 h], *P*=.03). Baseline UFC was >500 nmol/24 h (>180 μg/24 h) in 14 of 33 participants who required treatment for hypokalemia, but in only 2 out of 17 participants who did not.

Systolic BP (SBP) rose during the trial in 18 participants, 10 of whom had clinical evidence of mineralocorticoid excess. Overall, the mean change in SBP was -2.5 mm Hg during the trial (*P*=NS) in the 40 patients with hypertension, but changes in antihypertensive therapies among 19 subjects with diabetes confounded analysis of predictive factors. In repeated measures mixed models, UFC was positively correlated with SBP and negatively correlated with serum potassium levels (*P*=.02 for both).

Conclusion:
We conclude that hypertension and hypokalemia are common in CS patients requiring GR antagonist therapy with UFC >500 nmol/24 h (>180 μg/24 h). These patients require concomitant treatment with a MR antagonist and/or potassium supplements, which control BP and potassium in the majority of cases. Clinical findings of MR activation are less predictable in patients with UFC <500 nmol/24 h.

Introduction

Hypertension and hypokalemia are common in CS. Cortisol activates both GR and MR. Cortisol is converted to inactive cortisone by renal 11β-hydroxysteroid dehydrogenase type 2 (11β-HSD2) so that aldosterone is the main ligand for MR under normal physiological states. In CS, 11β-HSD2 cannot fully inactivate the excessive amounts of cortisol produced, resulting in MR activation, which can cause hypertension, hypokalemia, and edema. In the SEISMIC study, 24 weeks of treatment with mifepristone, a GR antagonist, improved the clinical and metabolic status in 50 CS patients.[1] In patients with Cushing's disease (CD), mifepristone causes increased cortisol production due to loss of glucocorticoid negative feedback, which further predisposes to hypokalemia, alkalosis, and edema due to a state of apparent mineralocorticoid excess. In SEISMIC, adverse events of decreased serum potassium (34%), edema (26%), and hypertension (24%) were common. We analyzed blood pressure and potassium data from SEISMIC to determine prevalence, predictive factors, and approaches to CS management.

Methods

- SEISMIC was a 24-week, open-label study of mifepristone in 50 patients with CS.
 - Main eligibility criteria:
 - Adrenocorticotropic hormone (ACTH)-dependent or ACTH-independent CS;
 - CS and type 2 diabetes mellitus/impaired glucose tolerance (C-DM), or;
 - CS and hypertension without diabetes mellitus/impaired glucose tolerance (C-HT).
 - SEISMIC subjects:
 - C-DM: N=29 (19 with coexisting hypertension).
 - ~ Changes in antihypertensive medications permitted.
 - C-HT: N=21.
 - ~ Changes in antihypertensive medications not permitted.
 - 43 Patients had CD.
 - 3 Patients had adrenal carcinoma.
 - 4 Patients had ectopic ACTH syndrome.
 - UFC was measured at baseline, Week 6, Week 10, Week 16, Week 24, and 6 weeks after discontinuation of mifepristone (6-week follow-up).
 - Assay: LC-MS/MS (normal range 5.5-117 nmol/24 h).
- Administration of spironolactone (MR antagonist) was allowed in SEISMIC to control hypokalemia that did not respond to potassium replacement alone.
- Descriptive statistics, least mean squares estimates, and Wilcoxon tests for unmatched and matched samples were used to compare group differences.
- Repeated measures mixed models analysis was used to examine predictive factors for changes in BP and low potassium.

Results

- 22 Patients experienced at least 1 potassium measurement below the lower limit of normal (3.5 mmol/L) **(Figure 1)**.
 - 20 Participants experienced at least 1 low potassium value during treatment with mifepristone.
 - 2 Participants had low potassium values prior to starting mifepristone, but not during mifepristone. Both patients required treatment for low potassium.
 - Low serum potassium values were episodic. The median nadir potassium value during the study was 3.1 mmol/L (range 2.1-3.4 mmol/L), and during mifepristone treatment the median nadir was 3.2 mmol/L (range 2.1-4.2 mmol/L).
- Prior to mifepristone, potassium supplementation was common (n=18), but MR antagonist therapy was uncommon (n=5). During mifepristone, the use of both potassium supplementation and MR antagonist therapy increased **(Table 1)**.
- Patients who required treatment for hypokalemia tended to have higher UFC levels at baseline and during mifepristone, but the differences were not statistically significant due to wide ranges **(Figure 2*)**.
- Patients who required potassium supplementation prior to mifepristone treatment had higher UFC levels than those who required potassium supplementation only while receiving mifepristone (*P*=.037) **(Table 2*, Figure 3*)**.
- Baseline UFC was >500 nmol/24 h in 42.4% of patients who required treatment for hypokalemia, but in only 11.7% who did not (*P*=.008) **(Table 3*)**.

*Note: To convert UFC to μg/24 h, divide by 2.759.

- SBP increased in 18 patients with an initial diagnosis of hypertension (n=40) and was associated with parameters of mineralocorticoid excess in 10 patients **(Table 4)**.
- At study end, there was no significant change from baseline in mean SBP in patients with a diagnosis of hypertension **(Table 5)**.
- The change in UFC during mifepristone treatment was positively correlated with the change in SBP and negatively correlated with the change in serum potassium **(Table 6)**. There was no significant interaction between UFC change from baseline, the need for potassium supplementation, or use of MR antagonist therapy.

Figure 1. Trend Lines of Serum Potassium in Individual Patients Who Experienced Hypokalemia During the Study

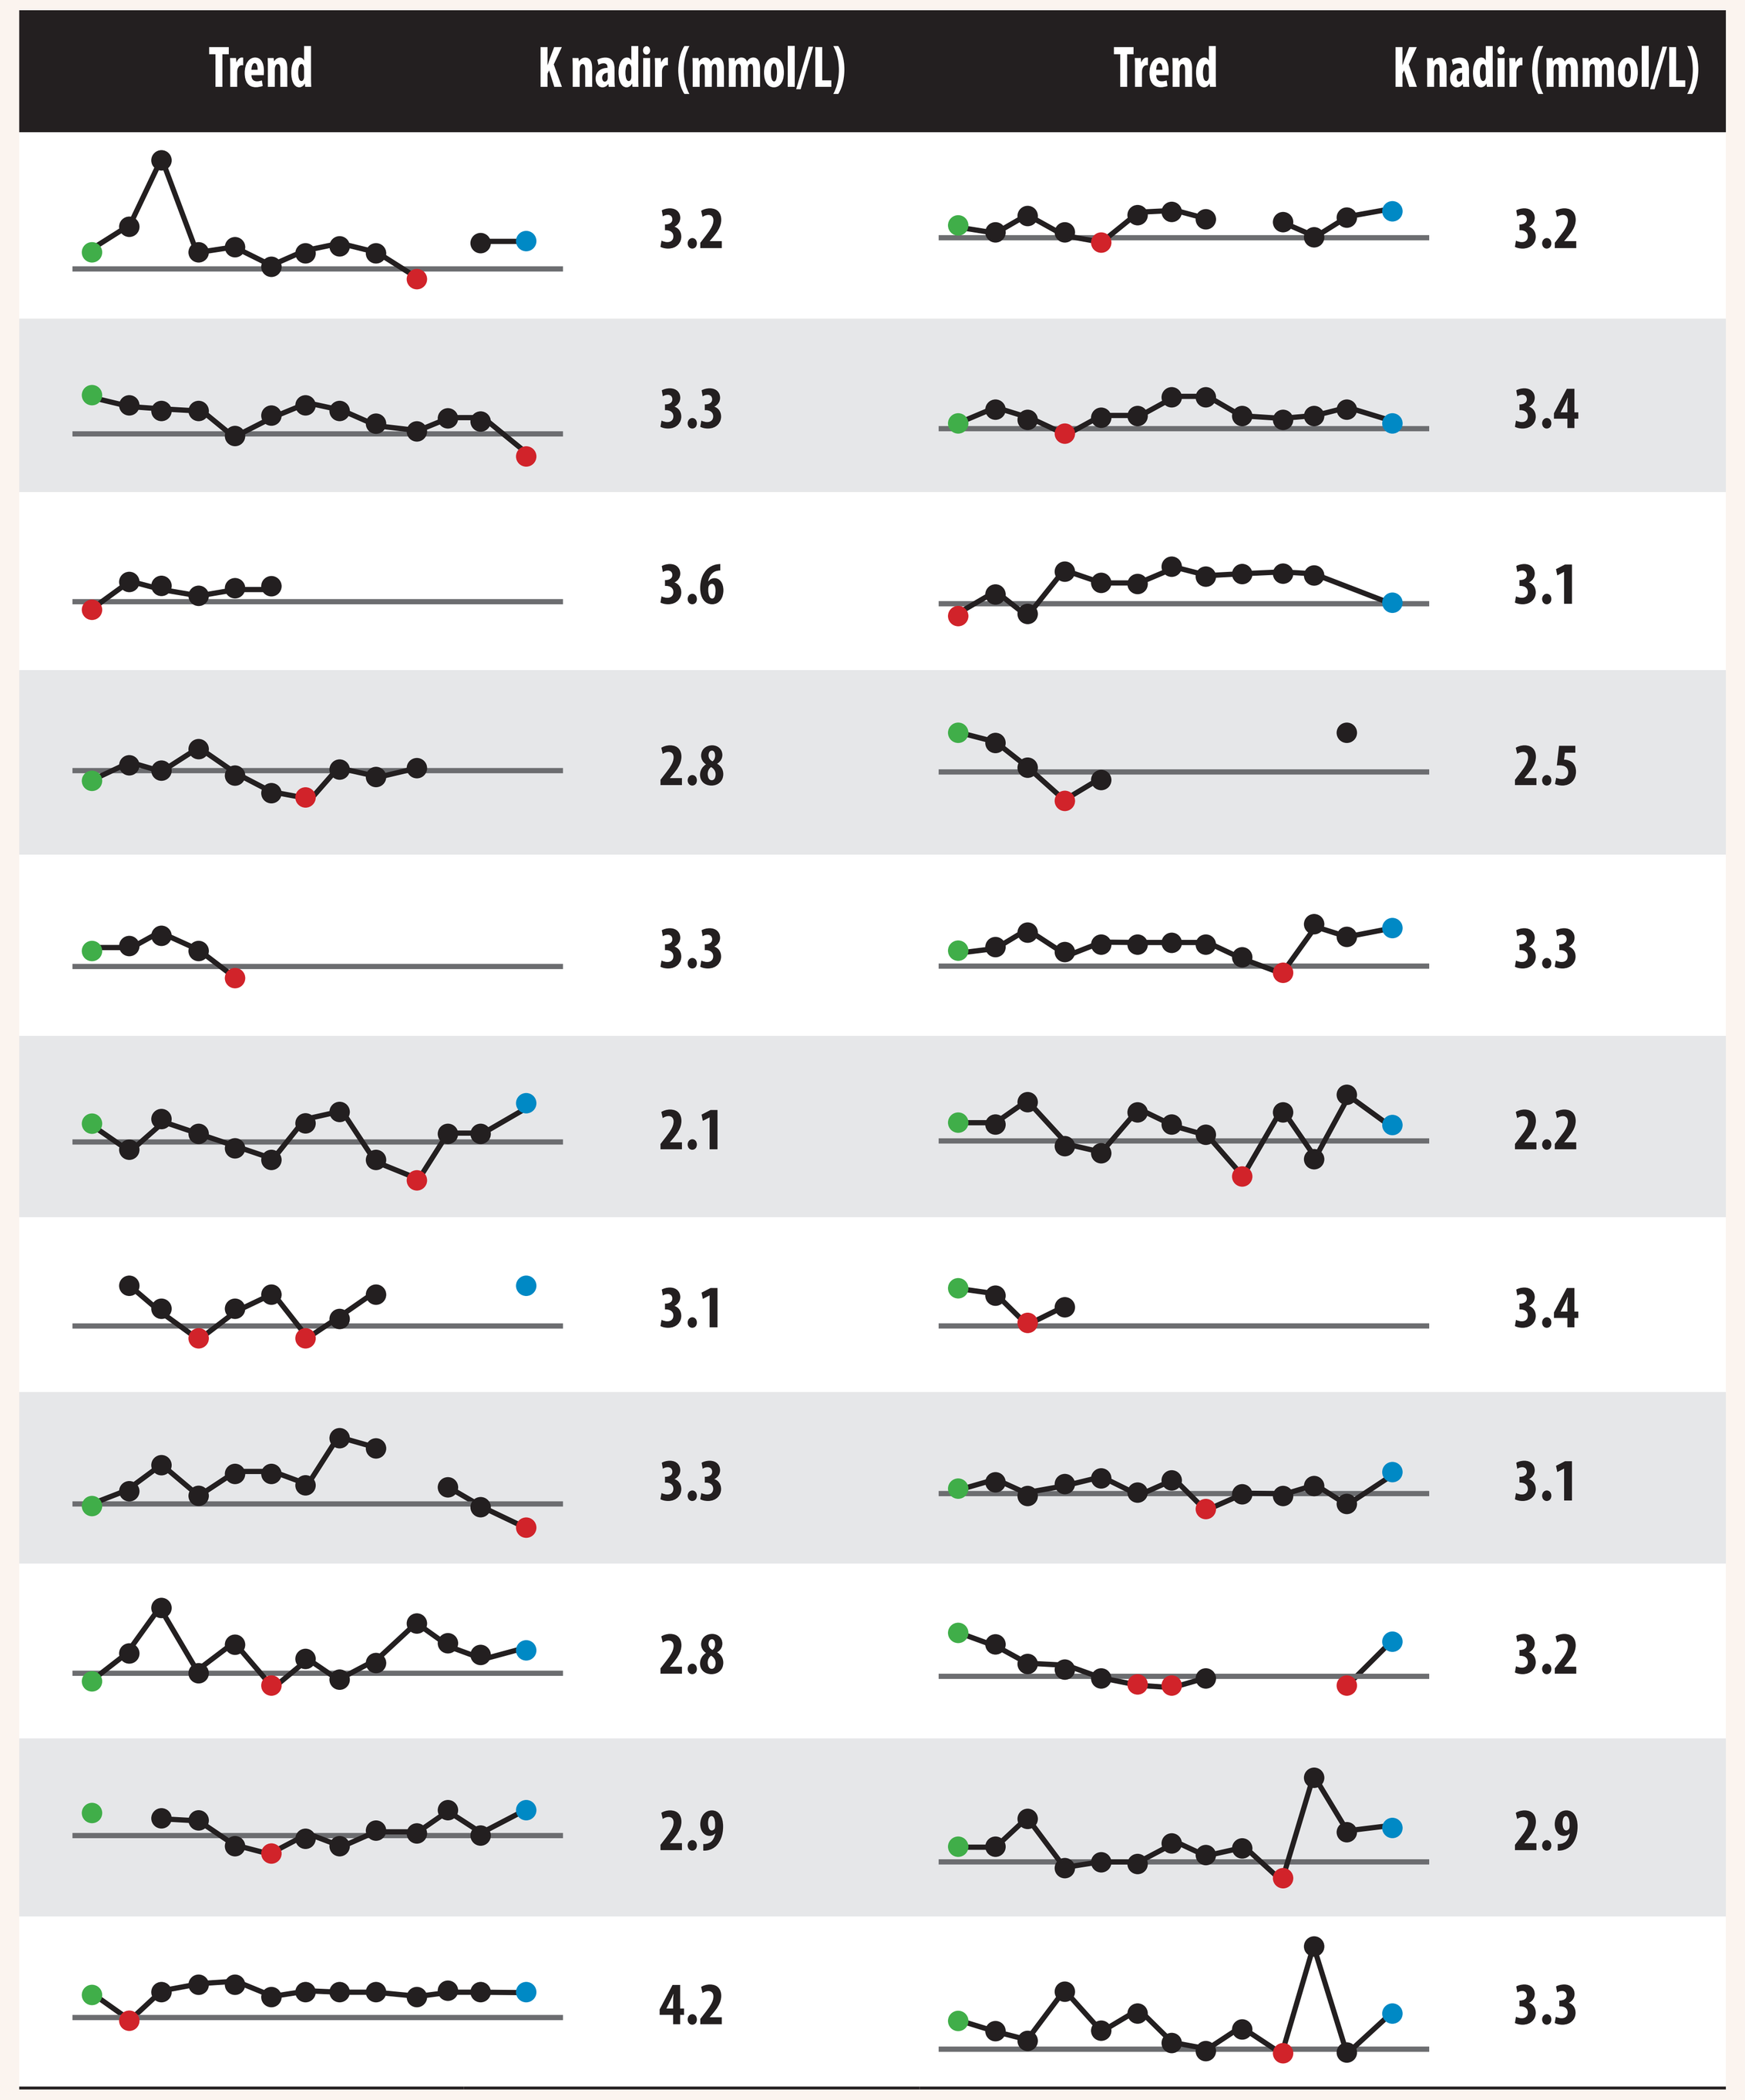

Trend
K nadir (mmol/L)
Trend
K nadir (mmol/L)
3.2
3.2
3.3
3.4
3.6
3.1
2.8
2.5
3.3
3.3
2.1
2.2
3.1
3.4
3.3
3.1
2.8
3.2
2.9
2.9
4.2
3.3

Potassium values plotted as function of time. Horizontal line indicates lower limit of normal potassium level (3.5 mmol/L). The nadir potassium level during mifepristone administration is shown. Each marker represents a potassium determination: ●=screening potassium; ●=during study (includes Day 1 pre-mifepristone); ●=off mifepristone (6-week follow-up); ●=lowest potassium measured. K=potassium.

Table 1. Potassium and Spironolactone Use

	Total Daily Dose of Potassium (mEq/d)		Total Daily Dose of Spironolactone (mg/d)	
	Prior to Mifepristone	During Mifepristone[a]	Prior to Mifepristone	During Mifepristone[a]
n	18	33	5[b]	14[c]
Mean (SD)	47 (49)	110 (110)	175 (166)	173 (118)
Median	20	80	100	175
Range	10–180	10–420	25–400	25–400
IQR	55	150	250	213

[a]Maximum dose during mifepristone; [b]2 participants with adrenal cancer, 2 with ectopic ACTH syndrome, 1 with CD; [c]1 participant received eplerenone (100 mg/d).

Figure 2. Urinary-Free Cortisol by Treatment With Potassium

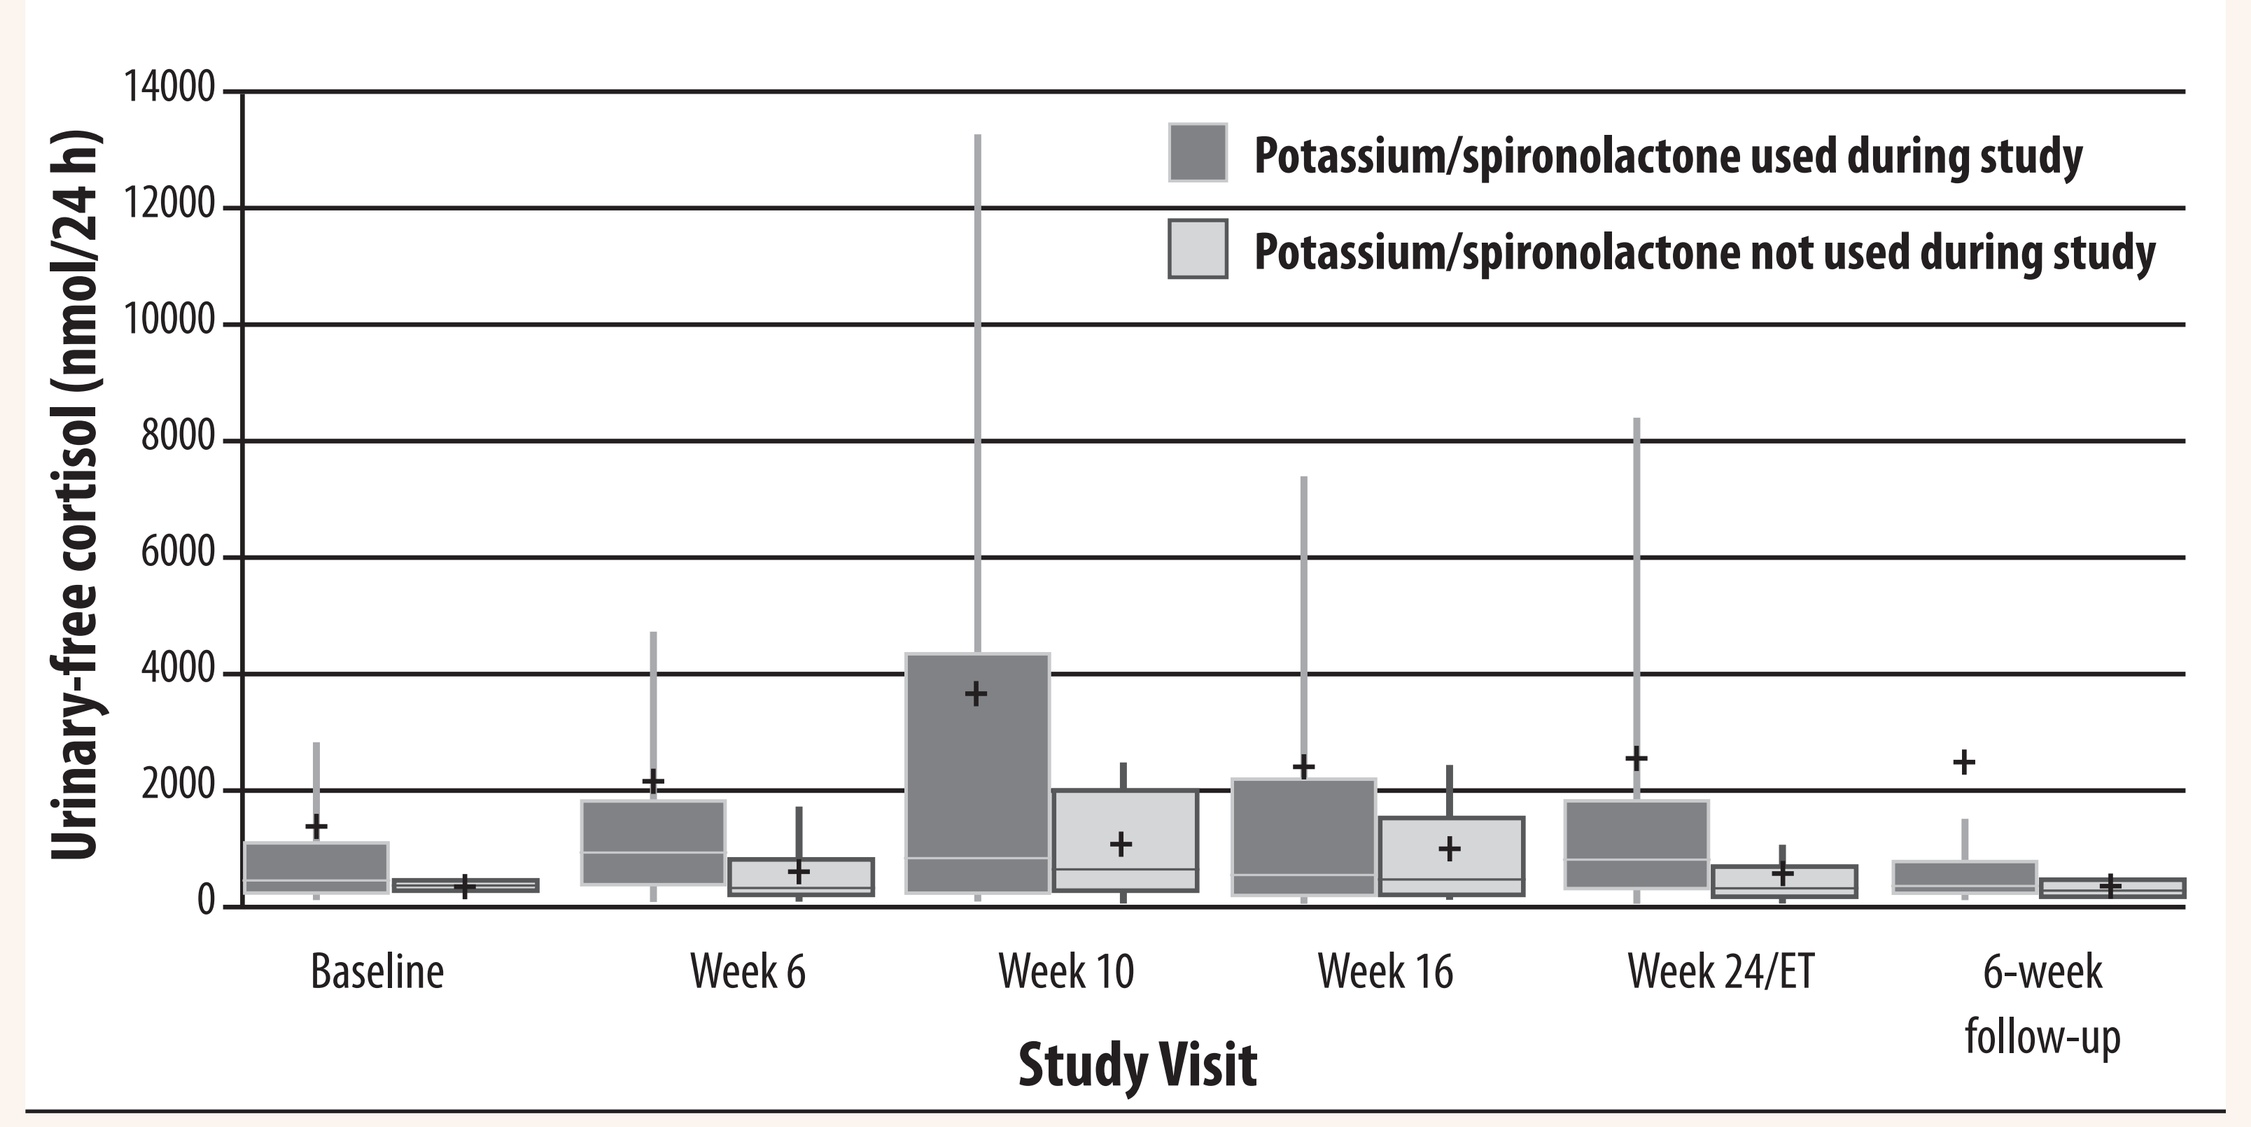

Urinary-free cortisol (nmol/24 h)
14000
12000
10000
8000
6000
4000
2000
0
Potassium/spironolactone used during study
Potassium/spironolactone not used during study
Baseline
Week 6
Week 10
Week 16
Week 24/ET
6-week follow-up
Study Visit

UFC values plotted by study visit. UFC values in patients who did (n=33) and who did not (n=17) receive treatment with potassium. All participants who were treated with spironolactone also received potassium (n=14). Crossline=median; box limits=interquartile range (25%-75%); whiskers=10% and 90%; + = mean. ET=early termination.

Table 2. Baseline Urinary-Free Cortisol[a] (nmol/24 h) by Treatment With Potassium

	Potassium Pretreatment[b] (n=18)	No Potassium Pretreatment[b] (n=15)
Mean (SD)	2194 (4654)	383 (387)
Median	587	203
Range	124–19739	59–1119
IQR	1561	396

[a]Last UFC measurement prior to mifepristone; [b]pretreatment refers to use of potassium prior to start of mifepristone that continued during study.

Figure 3. Baseline Urinary-Free Cortisol by Potassium Use Prior to Mifepristone Administration

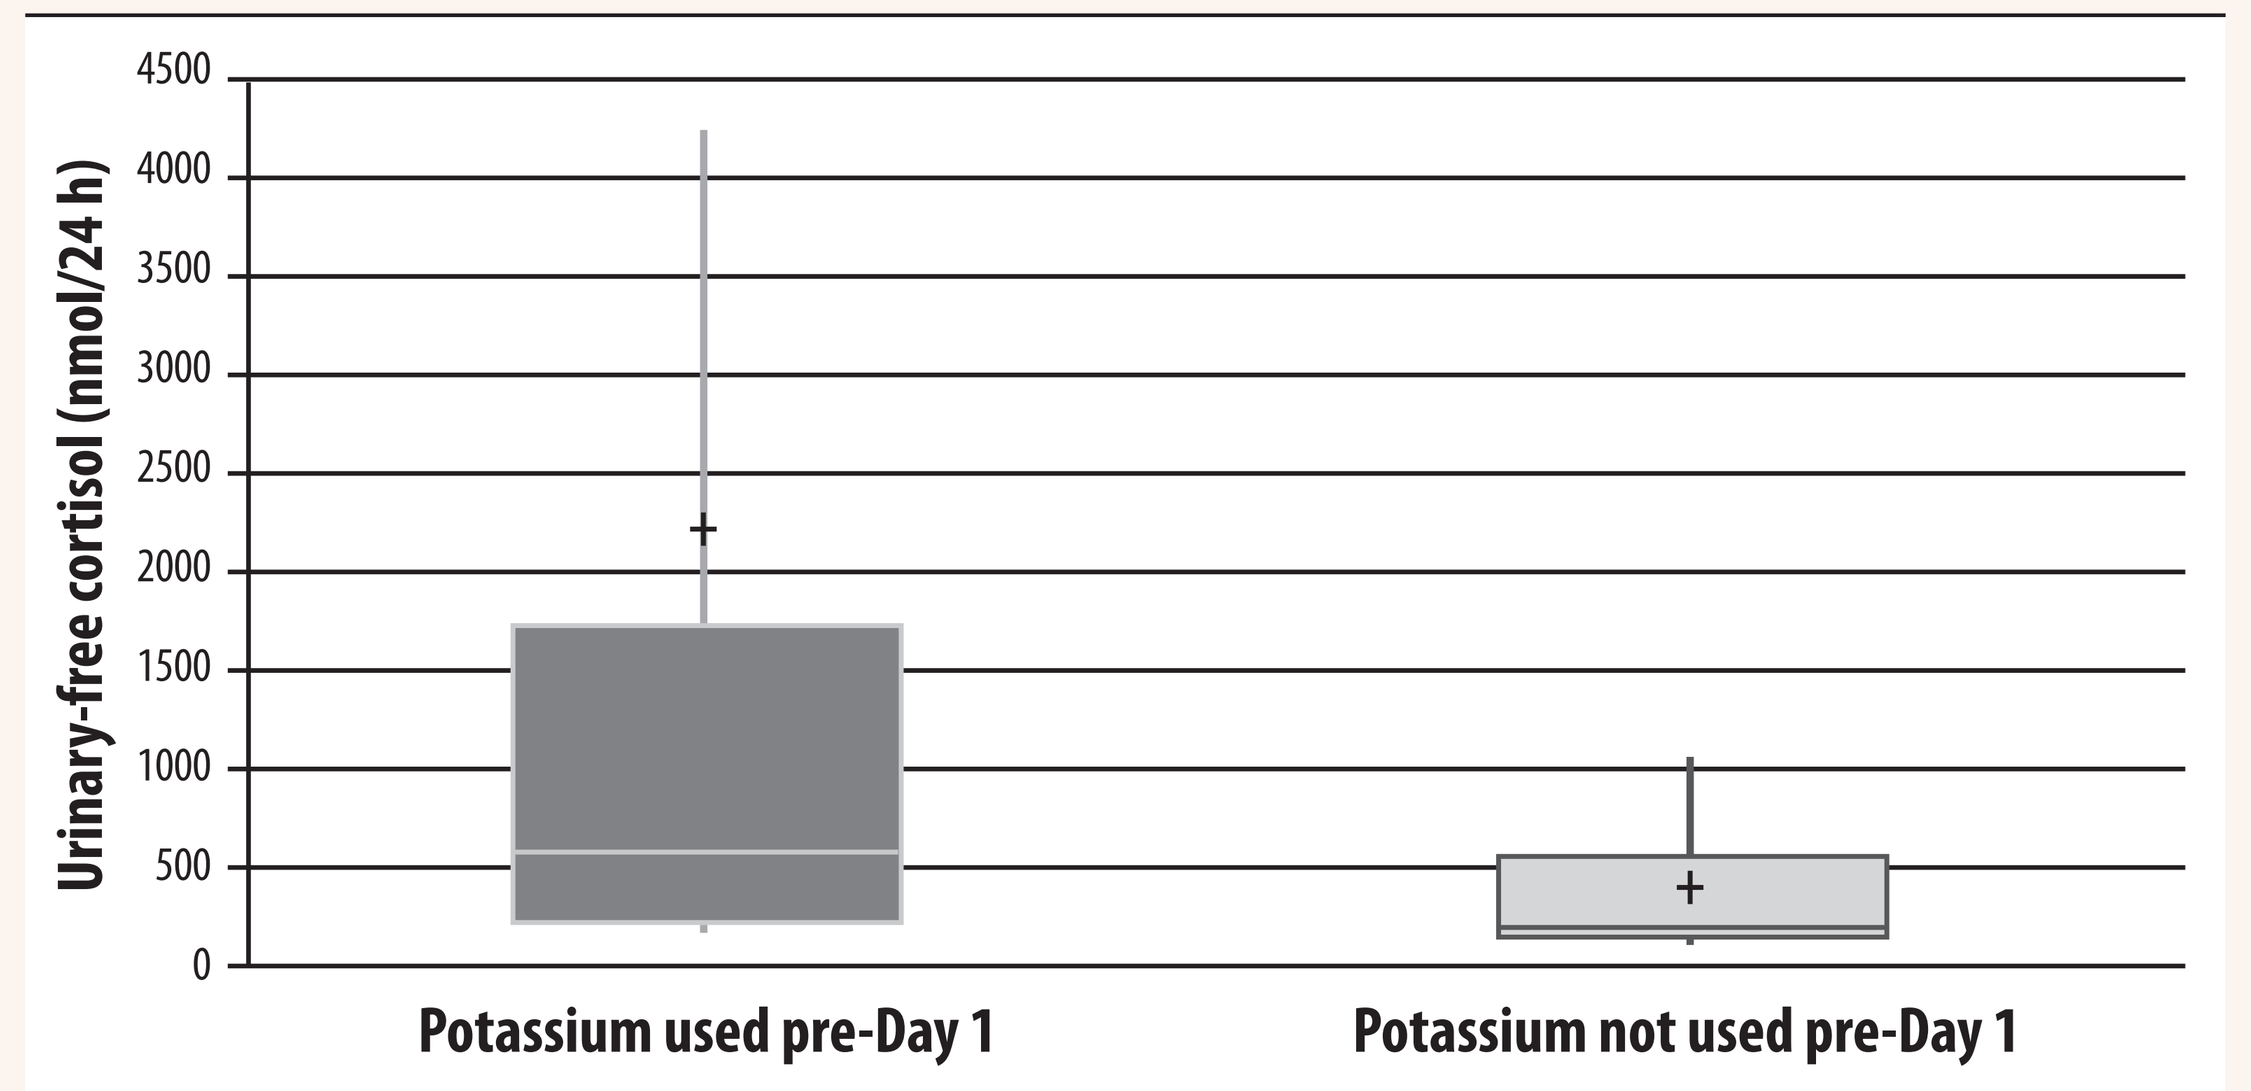

Urinary-free cortisol (nmol/24 h)
4500
4000
3500
3000
2500
2000
1500
1000
500
0
Potassium used pre-Day 1
Potassium not used pre-Day 1

UFC values in patients who did and who did not require potassium prior to mifepristone use. Crossline=median; box limits=interquartile range (25%-75%); whiskers=10% and 90%; + = mean.

Table 3. Baseline Urinary-Free Cortisol[a] (nmol/24 h) by Need for Potassium Treatment During Mifepristone Treatment

	Potassium Required (n=33)	Potassium Not Required (n=17)
Mean (SD)	1371 (4654)	317 (228)
Median	289	266
Range	59–19739	85–1032
IQR	862	94
n with UFC>250	19 (57.6%)	11 (64.7%)
n with UFC>500	14 (42.4%)	2 (11.7%)
n with UFC>750	10 (30.3%)	1 (5.9%)
n with UFC>1000	9 (27.3%)	1 (5.9%)

[a]Last UFC prior to mifepristone.

Table 4. Evidence of Mineralocorticoid Excess in Patients With Hypertension Taking Mifepristone[a]

Degree of Increase in SBP (mm Hg)	Patients With SBP Increase (n)	Patients With Evidence of Mineralocorticoid Excess[b] (n)	Patients Who Required Spironolactone (n)
Any increase	18	10	7
≥5	15	8	6
≥10	10	4	4
≥20	4	3	1

[a]Analysis limited to those with a diagnosis of hypertension at baseline (n=40); [b]defined by at least 2 of the following: hypokalemia, alkalosis, edema, worsening hypertension.

Table 5. SBP (mm Hg) in Patients With a Diagnosis of Hypertension (N=40)

	Mean (SD)	Median	Range (min, max)
Baseline	133.4 (20.5)	134	99, 181
Week 24/ET	131.0 (17.7)	129.5	94, 170
Change from baseline	-2.5 (19.1)	-2.5	-52, 39

Table 6. Regression Analyses: Potassium and SBP vs Change in Urinary-Free Cortisol (nmol/24 h)[a] (N=46)

	Slope (SE) [10^{-5}]	95% CI [10^{-5}]	*P* Value
Change in serum potassium (mmol/L)	-2.7 (1.12)	-5.0, 0.48	.0175
Change in SBP (mm Hg)	90.4 (39.26)	12.6, 168.3	.0232

Note: use of potassium or spironolactone not correlated to UFC. [a]Repeated measures mixed models. *P* value tests if slope estimate differs from 0. CI=confidence interval; SE=standard error.

Summary

- Hypokalemia, edema, and hypertension are common in CS, reflecting an apparent mineralocorticoid excess state, and can be exacerbated with mifepristone treatment, particularly in CD.
- UFC is higher in patients who require treatment for hypokalemia and are receiving mifepristone compared to those who do not require treatment.
- Hypokalemia with mifepristone therapy in CS is more common in patients with UFC values >500 nmol/24 h (>180 μg/24 h); BP may increase during mifepristone treatment.

Conclusions

- Hypertension and hypokalemia are common in patients with CS who require GR antagonist therapy with mifepristone and have high UFC levels (>500 nmol/24 h [>180 μg/24 h]) .
- Use of potassium supplements is often required to control hypokalemia, and in combination with MR antagonist medication, maintains normal serum potassium levels in the majority of patients treated for CS with mifepristone.

Reference
1. Fleseriu. *J Clin Endocrinol Metab*. 2012; 97(6):2039-2049.

Author Disclosures
Dr. Auchus is an investigator and consultant for and receives research funding support from Corcept Therapeutics and Novartis. Dr. McPhaul is an investigator for and receives research funding support from Corcept Therapeutics and Novartis. Dr. Findling is an investigator for and receives research funding support from Corcept Therapeutics and Novartis, and acts as a consultant for Corcept Therapeutics and Novartis Nutrition. Dr. Gross is an employee of Corcept Therapeutics.