Relacorilant With Pembrolizumab: A Phase 1b, Open-Label Study of a Selective Glucocorticoid Receptor Modulator Combined With a Checkpoint Inhibitor for Patients With Adrenocortical Carcinoma With Excess Glucocorticoid Production

Mouhammed Amir Habra, MD[1]; Gary D. Hammer, MD, PhD[2]; Electron Kebebew, MD[3]; Francis P. Worden, MD[4]; Nitya Raj, MD[5]; Julie E. Hallanger-Johnson, MD[6]; Andreas Grauer, MD[7]; Andreas Moraitis, MD[7]

[1]MD Anderson Cancer Center, Houston, TX, USA; [2]University of Michigan, Ann Arbor, MI, USA; [3]Stanford University, Palo Alto, CA, USA; [4]University of Michigan Rogel Cancer Center, Ann Arbor, MI, USA; [5]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [6]Moffitt Cancer Center, Tampa, FL, USA; [7]Corcept Therapeutics, Menlo Park, CA, USA

INTRODUCTION

- Adrenocortical carcinoma (ACC) is a rare and aggressive disease often associated with a poor prognosis, with overall 5-year survival rates between 16%-47% (5%-10% in metastatic disease)[1,2]
 - The recommended treatment regimen for advanced ACC – mitotane monotherapy or in combination with etoposide, doxorubicin, plus cisplatin – is associated with an objective response rate (ORR) less than 25% and a progression-free survival (PFS) duration of 5 months[3]
 - Thus, there is a need for improved therapeutic options
- Patients with ACC often present with excess secretion of hormones, with about 50%-80% of these patients exhibiting glucocorticoid (GC) excess[1,4]
 - The presence of GC excess is correlated with decreased survival in patients with ACC, potentially due to the immunosuppressive effects of GC[5]
 - GC excess (also referred to as hypercortisolism or Cushing syndrome) poses additional clinical challenges related to other comorbidities, such as hypertension, hyperglycemia/diabetes mellitus, muscle weakness, and hypercoagulability[6]
- Pembrolizumab is an immune checkpoint inhibitor approved to treat multiple advanced malignancies[7]
 - Specifically, pembrolizumab is a humanized monoclonal antibody that binds to the programmed cell death protein 1 (PD-1) receptor, an important receptor in regulating the immune response to tumor cells[7]
 - Blockade of PD-1 induces an anti-tumor immune response and results in tumor growth suppression[8]
- Pembrolizumab has shown only modest efficacy in ACC,[9] and evidence suggests that GCs may blunt the clinical response to checkpoint inhibitors[9,10]
- Previous clinical studies have demonstrated that antagonism of the GC receptor has the potential to increase immune-related transcripts, thus potentially promoting tumor immune response in patients with ACC and GC excess[11,12]

RELACORILANT

- Relacorilant (CORT125134, Corcept Therapeutics, **Figure 1**) is a highly selective glucocorticoid receptor modulator (SGRM) in clinical development for the treatment of patients with endogenous hypercortisolism (Cushing syndrome) and, in combination with chemotherapy, for the treatment of various solid tumors
- Relacorilant reverses the immunosuppresive effects of corticosteroids and interferes with the effects of endogenous cortisol[11]
- It is hypothesized that GC receptor antagonism by relacorilant will enhance sensitivity to treatment with pembrolizumab and may promote an anti-tumor immune response in patients with ACC, specifically when presenting with GC excess

Figure 1. Relacorilant Structure

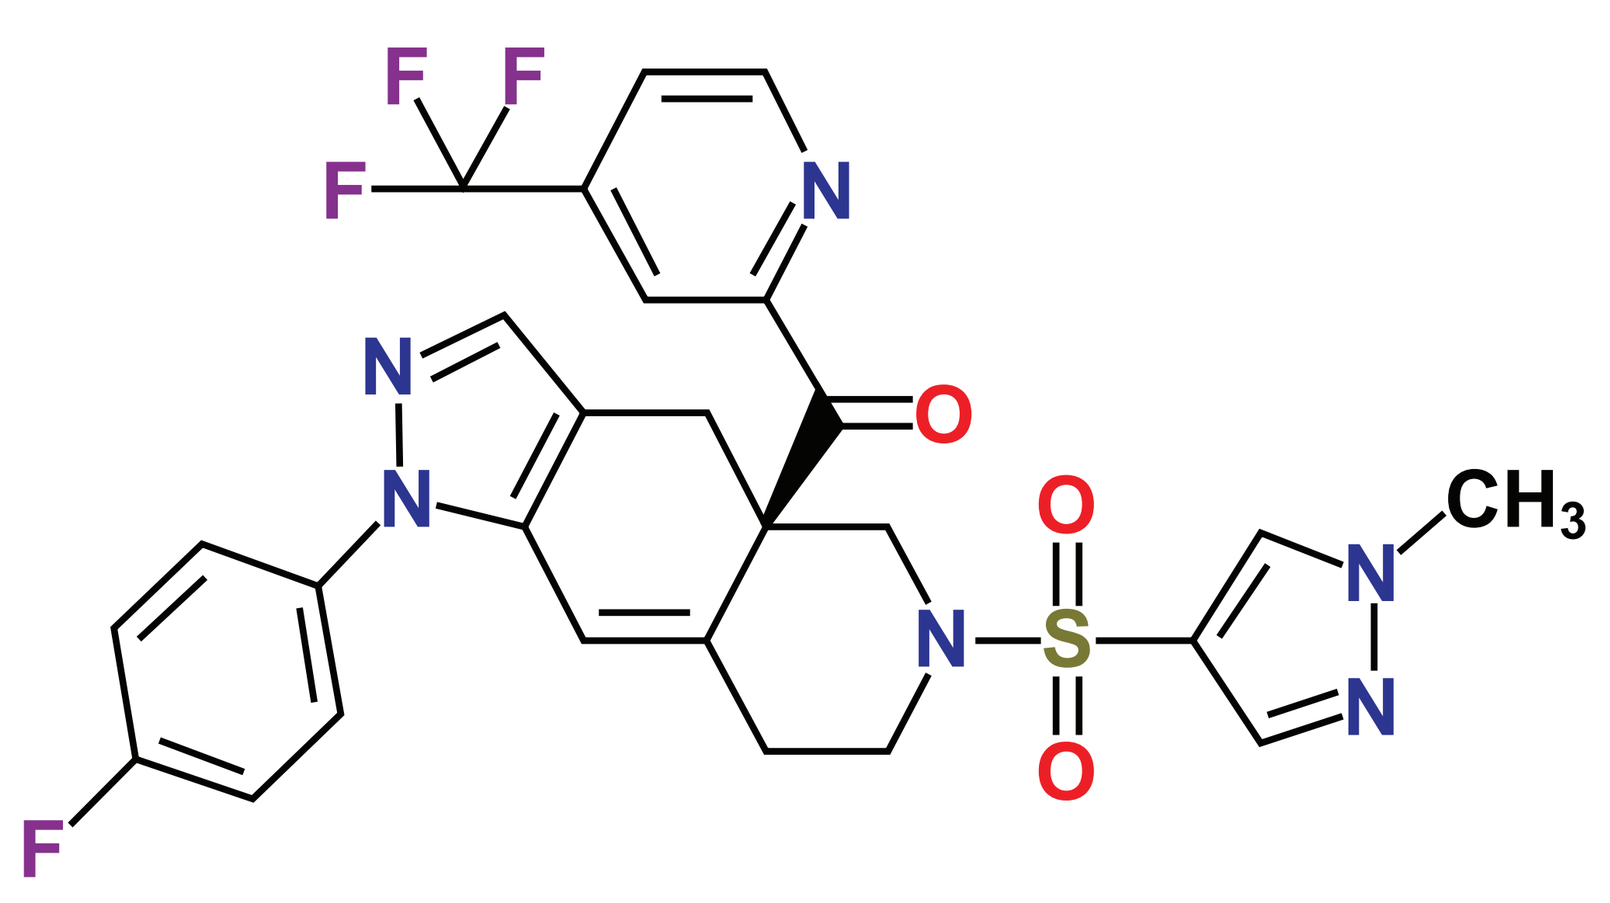
F
F
F
N
N
N
O
O
S
O
N
N
N
CH3
F

STUDY DESIGN AND METHODS

- This phase 1b, open-label, single-arm study (NCT04373265) is being conducted at multiple US study centers and is designed to evaluate the safety, efficacy, pharmacodynamics, and pharmacokinetics of relacorilant in combination with pembrolizumab
- The study includes a screening period, a lead-in period, and a single-arm combined treatment period, followed by a long-term follow-up period (**Figure 2**)

Figure 2. Study Design

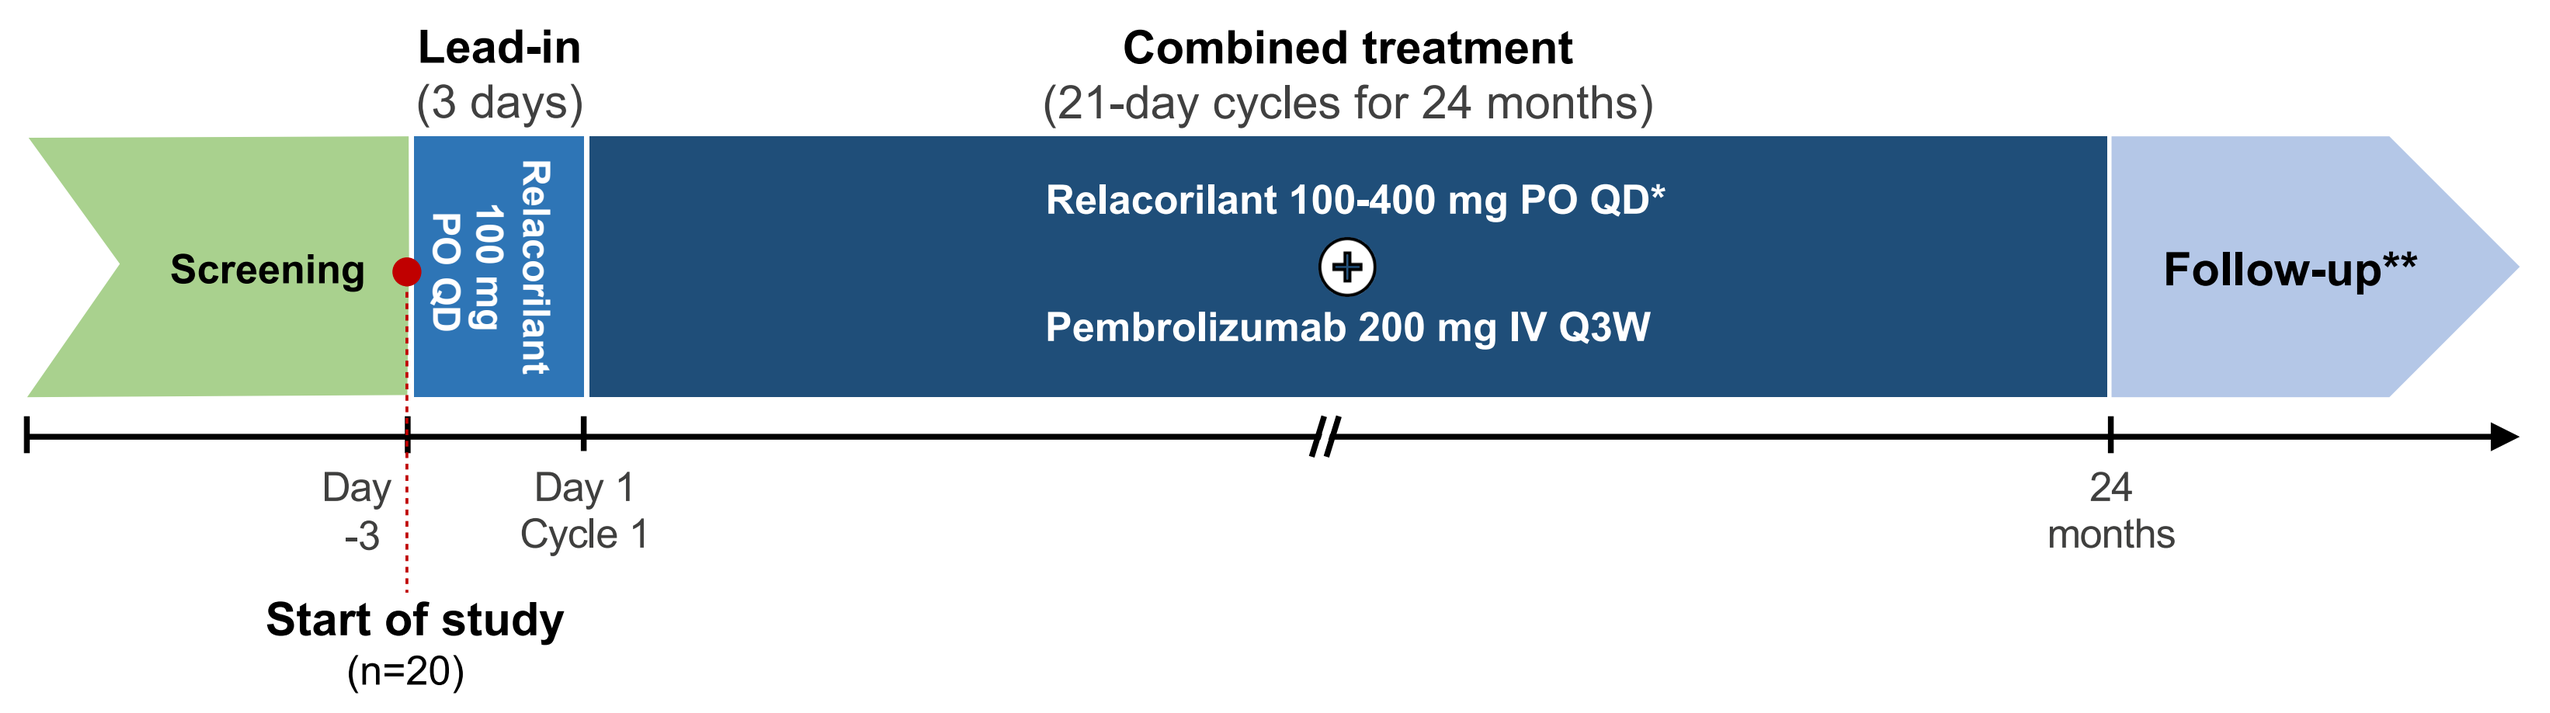
Lead-in
(3 days)
Combined treatment
(21-day cycles for 24 months)
Screening
Relacorilant 100 mg PO QD
Relacorilant 100-400 mg PO QD*
Pembrolizumab 200 mg IV Q3W
Follow-up**
Day -3
Day 1 Cycle 1
24 months
Start of study
(n=20)

*Relacorilant dose escalation in 100-mg increments, up to a maximum of 400 mg QD.
**Patients who continue to benefit from treatment may remain on treatment and will continue to be monitored. All patients will be followed for survival.
Q3W, once every 3 weeks; QD, once daily; IV, intravenous; PO, by mouth.

TREATMENT SCHEDULE

- **Screening period:** CT or MRI scans for baseline tumor assessment are performed prior to the first dose of relacorilant (Day -3)
- **Lead-in period:** Relacorilant 100 mg orally once daily, starting 3 days before the first pembrolizumab infusion
- **Combined treatment period:**
 - Pembrolizumab infusion administered on Day 1 of each 21-day cycle in combination with relacorilant once daily for 24 months or until confirmed disease progression
 - Relacorilant dose escalation in 100-mg increments up to 400 mg (≥3 weeks between each upward titration) will occur at each sequential cycle
- **Follow-up period:** Safety assessments will occur for 30 days, then every 9 weeks
- **Survival follow-up period:** Every 3 months after confirmed disease progression

STUDY OBJECTIVES

- The primary objective of the study is to determine the safety and efficacy of the recommended regimen of relacorilant in combination with pembrolizumab in patients with advanced ACC and GC excess
- Secondary objectives include: determination of the non-progression rate (NPR), PFS, overall survival (OS), duration of response (DOR), and an assessment of the effects of relacorilant and pembrolizumab on the clinical manifestations of GC excess

STUDY PARTICIPANTS

- Adults ≥18 years old with advanced ACC associated with GC excess are eligible to be enrolled

Table 1. Key Eligibility Criteria

INCLUSION CRITERIA

- Histologically or cytologically confirmed ACC (advanced unresectable and/or metastatic)
- Measurable disease per RECIST v1.1 and tumor accessible for biopsy
- Patients who have received mitotane must have a mitotane level <4 at screening
- ECOG performance status ≤2
- Adequate organ and bone marrow function
- Glucocorticoid excess must be documented by 2 of the following criteria:
 - 2 UFC >ULN
 - 2 LNSC >ULN
 - Cortisol >1.8 μg/dL post 1-mg DST

 Or
 - ACTH <10 pg/mL + cortisol >1.8 μg/dL post 1-mg DST

EXCLUSION CRITERIA

- Major surgery within 4 weeks of enrollment
- Untreated parenchymal brain metastases, uncontrolled CNS metastases, or any other concurrent cancer or history of another invasive malignancy within the last 3 years that has a likelihood of recurrence of >30% within the next 5 years
- Concomitant medication that is a strong CYP3A inducer or substrate with a narrow therapeutic window
- Prior treatment with either an anti-PD-1, anti-PD-L1, anti-PD-L2, anti-CD137, or CTLA-4 antibody therapy
- Treatment with any of the following therapies[a]
 - Monoclonal antibodies or anticancer vaccines within 60 days
 - Systemic corticosteroids within 28 days
 - Investigational products, systemic anticancer, or radiation therapies within 21 days
 - Mifepristone or GR antagonists within 21 days
 - Adrenostatic medications within 7-14 days

[a]These treatments are not allowed during the study.
ACC, adrenocortical carcinoma; ACTH, adrenocorticotropic hormone; CNS, central nervous system; CTLA, cytotoxic T-lymphocyte-associated protein; CYP, cytochrome P450; DST, dexamethasone suppression test; ECOG, Eastern clinical oncology group; GR, glucocorticoid receptor; LNSC, late-night salivary cortisol; UFC, urinary free cortisol; ULN, upper limit of normal; RECIST, response evaluation criteria in solid tumors.

STUDY ENDPOINTS

- Primary endpoints
 - Efficacy: ORR, defined as complete or partial remission (per RECIST v1.1) in patients who had measurable disease at baseline
 - Safety: Rate of dose-limiting toxicities (DLTs) during the first 5 cycles of treatment
- Secondary endpoints
 - Efficacy:
 - Best overall response per RECIST v1.1
 - NPR at 27 weeks, PFS, OS, and DOR per RECIST v1.1
 - Symptom improvement of GC excess
 - Safety as assessed through evaluation of treatment-emergent adverse events

SUMMARY

- A large proportion of patients with ACC present with GC excess and their prognosis is poor
- Controlling GC excess may be a key factor in improving patients' overall survival
- This is the first clinical study to evaluate whether GR antagonism may promote anti-tumor immune response and will help inform the potential benefit of relacorilant in combination with pembrolizumab in patients with ACC and GC excess
- The efficacy endpoints selected for this clinical study are relevant to ACC with GC excess and include ORR and clinical measures to assess improvement in the manifestations of GC excess

STUDY STATUS

- Multiple sites within the US are currently enrolling patients
- The anticipated sample size is N=20
- This study is registered at ClinicalTrials.gov (NCT04373265)
- Contact: Corcept Therapeutics, 650-327-3270, clinicalstudies@corcept.com

REFERENCES

1. Puglisi S, et al. *Endocrinol Metab Clin North Am.* 2018;47(2):395-407.
2. Stigliano A, et al. *J Endocrinol Invest.* 2015;39(1):103-21.
3. Fassnacht M, et al. *Eur J Endocrinol.* 2018;179(4):G1-G46.
4. Else T, et al. *Endocr Rev.* 2014;35(2):282-326.
5. Abiven G, et al. *J Clin Endocrinol Metab.* 91(7):2650-2655.
6. Pivonello R, et al. *Lancet Diabetes Endocrinol.* 2016;4(7):611-29.
7. KEYTRUDA® (pembrolizumab) injection, for intravenous use [prescribing information]. Whitehouse Station, NJ: Merck & Co. November 2020.
8. Alsaab HO, et al. *Front Pharmacol.* 2017;23(8):561.
9. Habra MA, et al. *J Immunother Cancer.* 2019;7(1):253.
10. Arbour KC, et al. *J Clin Oncol.* 2018;36, 2872-2883.
11. Hunt H, et al. *Clin Pharmacol Drug Dev.* 2018;7(4):408-421.
12. Gandhi S, et al. *Int J Mol Sci.* 2020;21(13):4635.

ACKNOWLEDGMENTS

This study is supported by Corcept Therapeutics. Funding for editorial, design, and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC, Princeton, NJ. The authors developed and revised the poster and provided approval of the final version.

DISCLOSURES

MAH: Consultant/advisor: Corcept, HRA Pharma; Research support: Corcept, Exelixis.
GDH: Other (provision of study materials): Corcept.
EK: Research support: Corcept.
FPW: Consultant/advisor: Merck; Research support: Corcept.
NR: Research support: Corcept, Novartis, Xencor.
JEHJ: Research support: Corcept.
AG: Employee: Corcept; Stock ownership: Corcept.
AM: Employee: Corcept.

Presented at the Endocrine Society Annual Meeting (ENDO 2021), March 20-23, 2021